00345

PGT121 AND VESATOLIMOD IN CHRONICALLY TREATED SHIV-INFECTED RHESUS MONKEYS

Noe Mercado[1], Abishek Chandrashekar[1], Erica N. Borducchi[1], **Joseph P Nkolola**[1], Romas Geleziunas[2], Brian Carr[2], Nathan Thomsen[2], Dan H. Barouch[1,3]
[1]Beth Israel Deaconess Medical Center (BIDMC), Boston, MA, USA, [2]Gilead Sciences, Foster City, CA, USA, [3]The Ragon Institute of MGH, MIT and Harvard, Cambridge, MA, USA

BACKGROUND

The latent HIV-1 viral reservoir in infected CD4+ T cells plays a central role in the viral rebound observed in the majority of infected individuals who discontinue anti-retroviral therapy (ART) and thus represents one of the key challenges for an HIV-1 cure. We have previously reported that intravenous (I.V.) administration of an HIV-1 V3 glycan-dependent broadly neutralizing antibody (bNAb) PGT121 together with the orally (P.O.) administered TLR7 agonist vesatolimod (VES) delayed or prevented viral rebound in Simian Human Immunodeficiency Virus (SHIV)-infected rhesus monkeys following ART discontinuation in animals that initiated ART early during acute infection [*Nature* (2018) 563(7731):360-364]. More recently, GS-9721 an Fc effector enhanced analog of PGT121 designed to improve human antibody-dependent cellular cytotoxicity (ADCC) and antibody-dependent cellular phagocytosis (ADCP) has been developed. Using PGT121 or GS-9721, both in combination with VES, we sought to explore the potential cure strategy of innate immune activation with immune-based elimination of the viral reservoir in the more stringent and clinically relevant model of chronically infected animals that initiated ART during chronic infection with extended ART. Such animals represent a higher efficacy bar due to larger and more diverse viral reservoirs than acutely infected animals placed on early ART.

METHODS

24 outbred rhesus monkeys (*Macca mulatta*) were intra-rectally infected with pathogenic SHIV-SF162.P3. After 12 months of chronic infection, a daily pre-formulated subcutaneous administered ART regimen (TDF/FTC/DTG) (5.1/40/2.5 mg/Kg) was initiated. Following 30 months of this continuous daily suppressive ART, animals received 10 bi-weekly administrations of PGT121 and VES or GS-9721 and VES at the doses and routes specified in figure 1. Sham control animals received placebo (saline) administrations. The first administration of VES was started on the day of third antibody administration. At week 42 following initial antibody dosing (24 weeks after the final combined antibody/VES doses) ART was discontinued and viral rebound was monitored for 140 days.

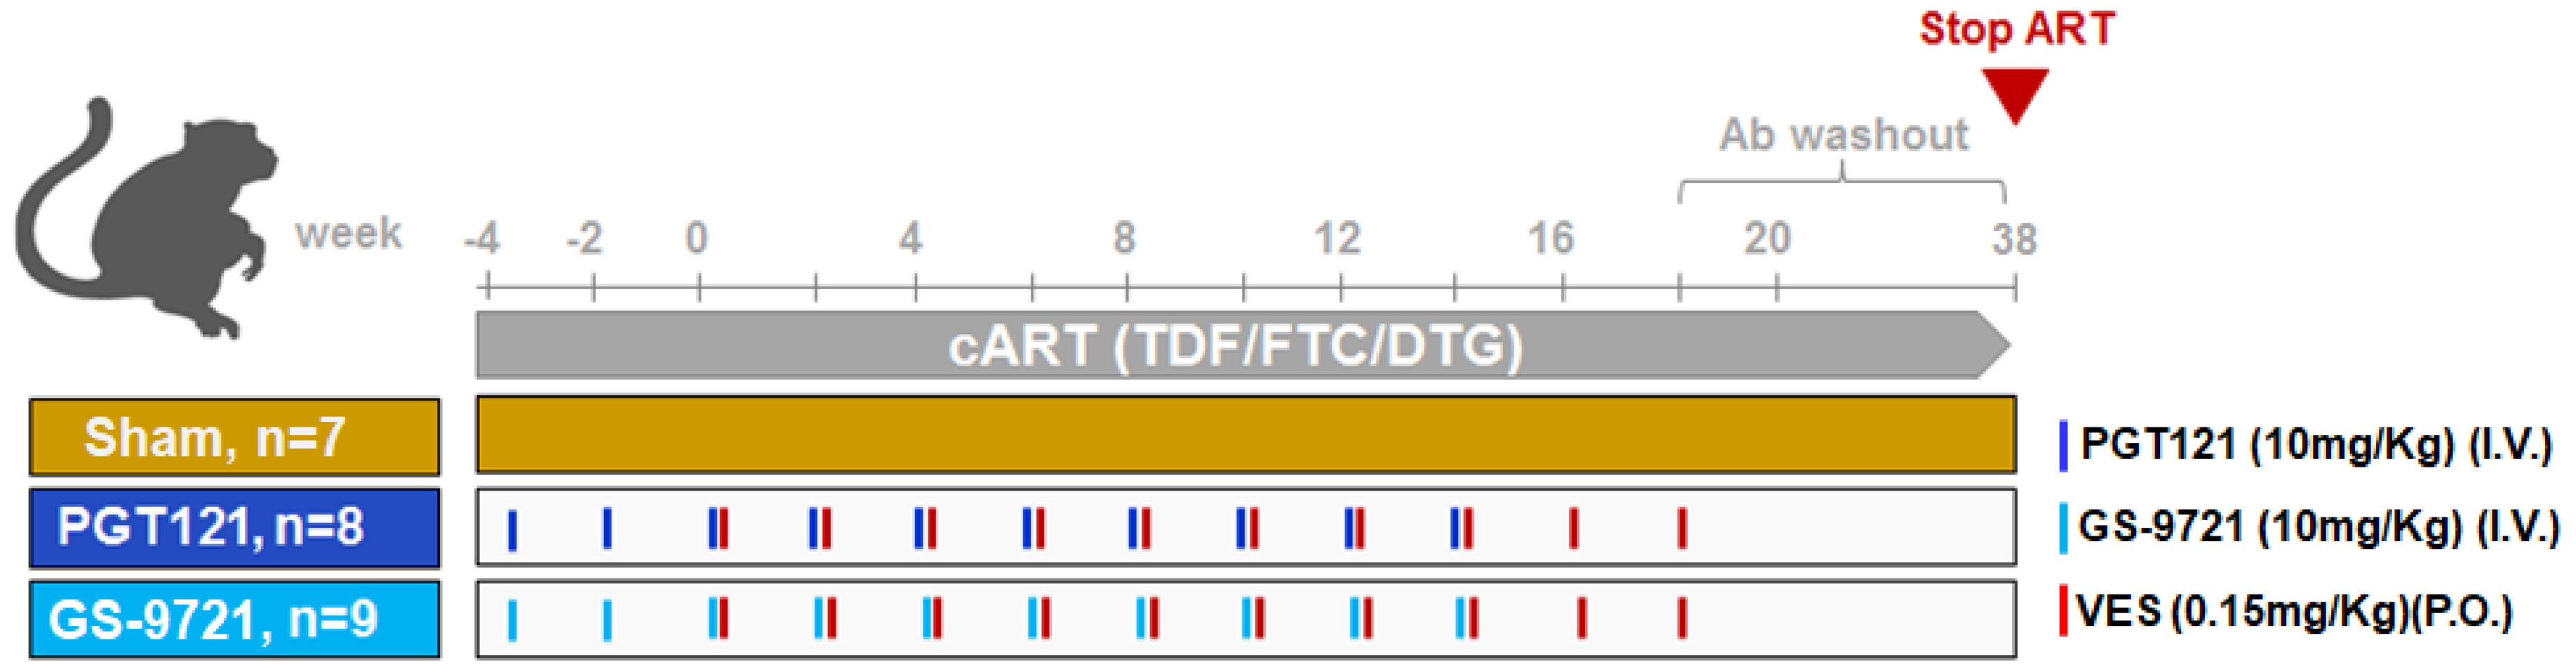
Stop ART
Ab washout
week
-4 -2 0 4 8 12 16 20 38
cART (TDF/FTC/DTG)
Sham, n=7
PGT121, n=8
GS-9721, n=9
PGT121 (10mg/Kg) (I.V.)
GS-9721 (10mg/Kg) (I.V.)
VES (0.15mg/Kg)(P.O.)

Figure 1. Study design

RESULTS

PGT121 and GS-9721 infusion resulted in 24 weeks of antibody levels followed by a decline to sub-therapeutic levels prior to ART discontinuation as shown in figure 2. Anti-Drug Antibodies (ADA) against PGT121 or GS-9721 were not observed during the period of antibody administrations. However, multiple animals administered GS-9721 had ADA which became detectable during the washout period (ADA data not shown).

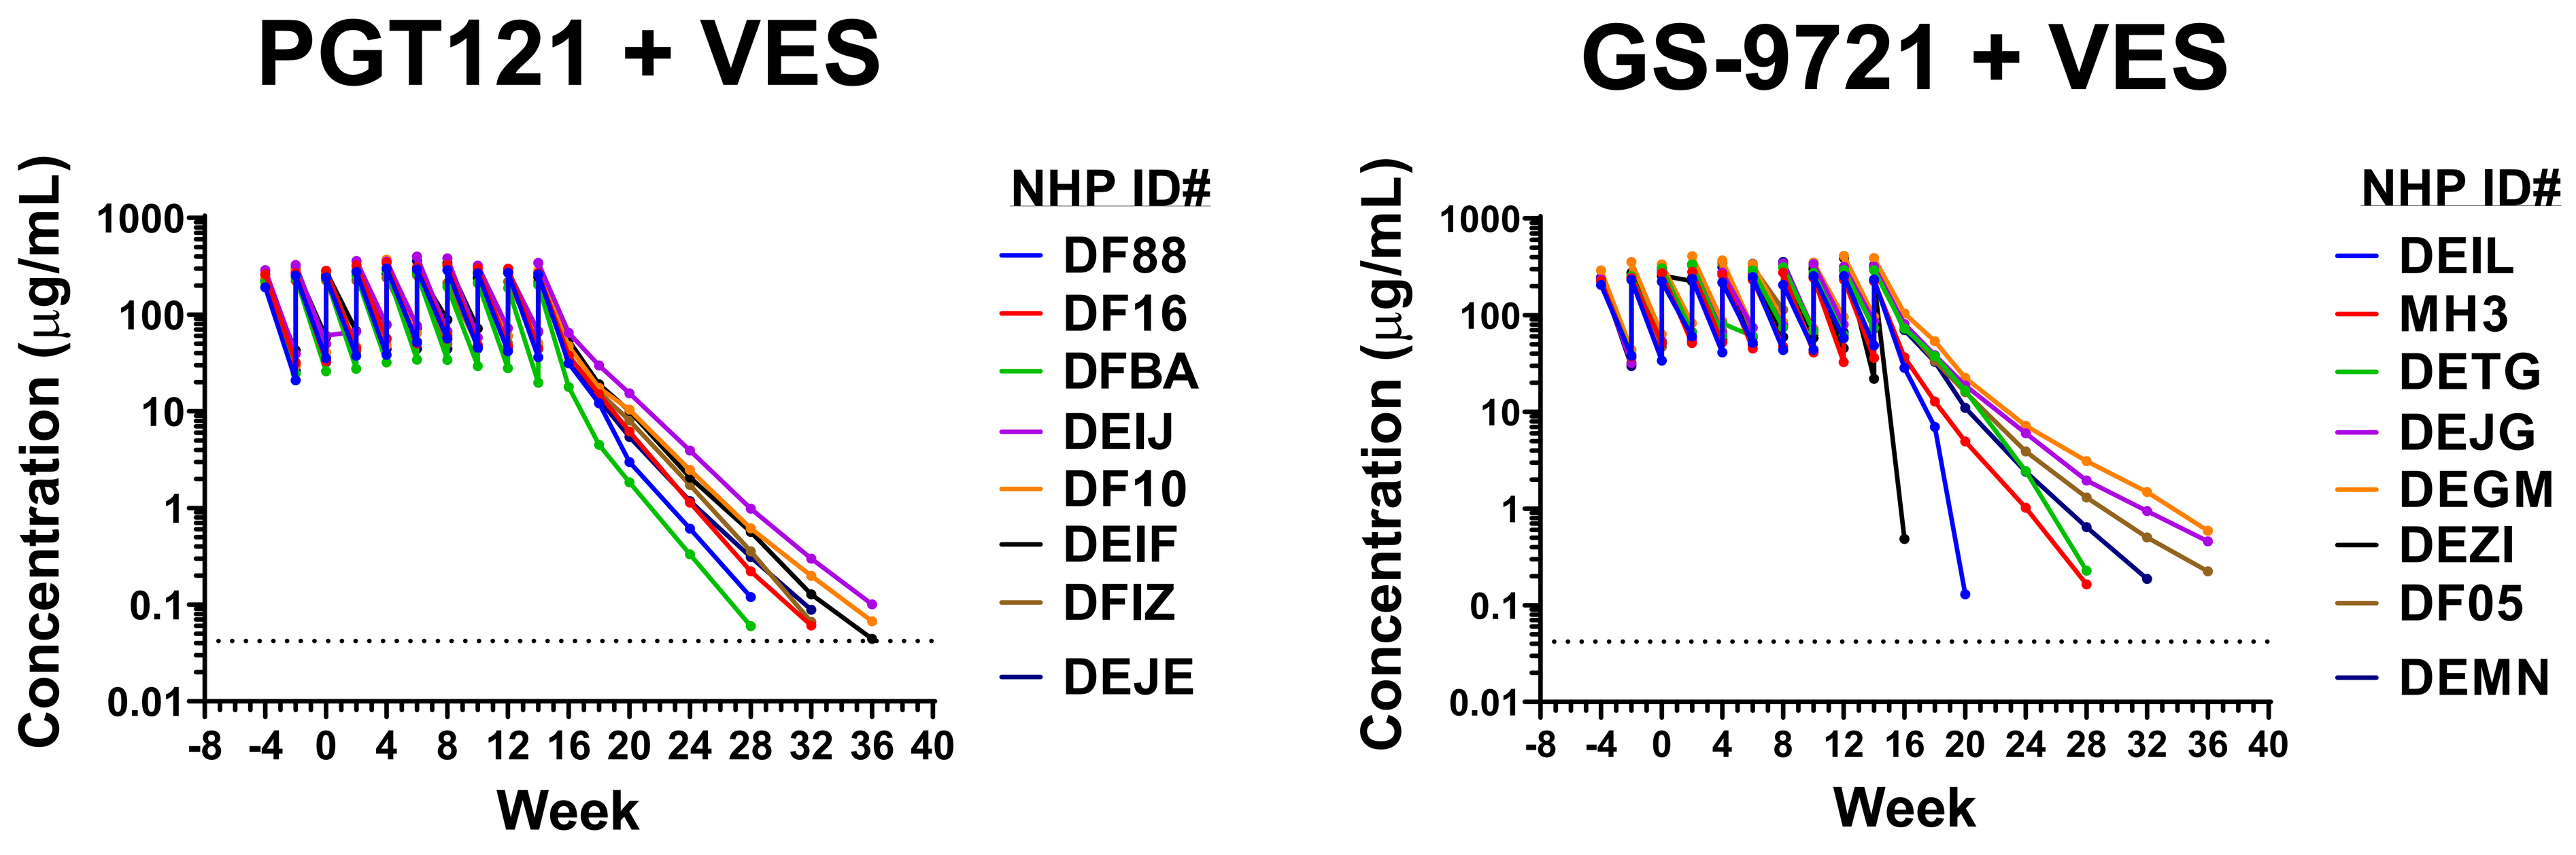
PGT121 + VES
GS-9721 + VES
Concentration (µg/mL)
1000 100 10 1 0.1 0.01
-8 -4 0 4 8 12 16 20 24 28 32 36 40
Week
NHP ID#
DF88
DF16
DFBA
DEIJ
DF10
DEIF
DFIZ
DEJE
NHP ID#
DEIL
MH3
DETG
DEJG
DEGM
DEZI
DF05
DEMN

Figure 2. PGT121 and GS-9721 individual pharmacokinetics. (Horizontal dotted line depicts assay sensitivity cut-off of ~0.042µg/mL).

As illustrated in figure 3, VES was observed to induce activation of CD4+ T cells as evidenced by increased expression of CD69 one day following its first administration.

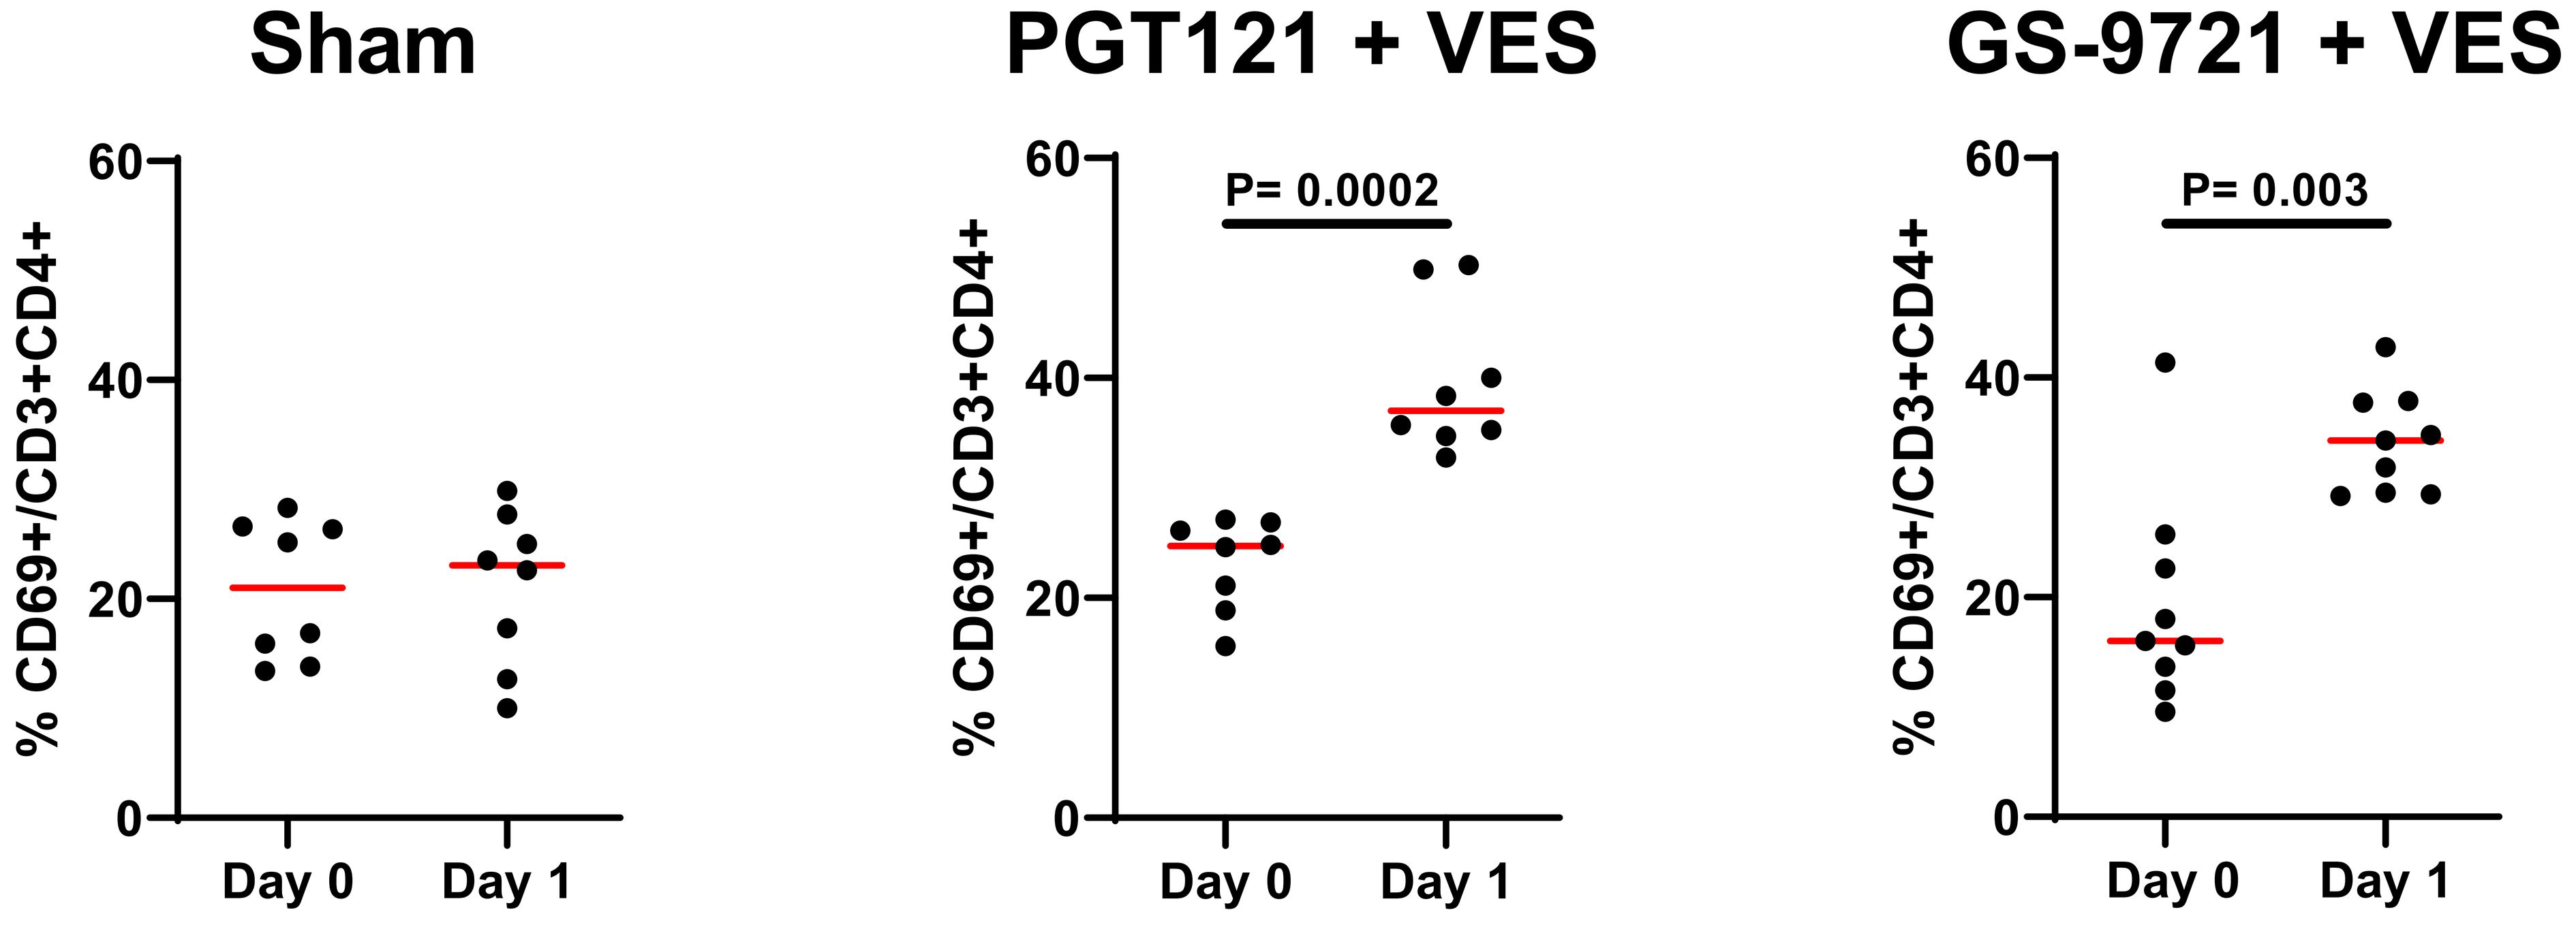
Sham
PGT121 + VES
GS-9721 + VES
% CD69+/CD3+CD4+
60 40 20 0
P= 0.0002
P= 0.003
Day 0
Day 1

Figure 3. CD4 cellular activation post-first VES administration (Red horizontal lines indicate median values. P-values reflect 2-sided Mann-Whitney tests).

As shown in figure 4A-C, following ART discontinuation 100% (7 of 7) of sham controls exhibited rapid viral rebound with a median rebound time of 21 [IQR 14-21] days. In contrast, only 50% (4 of 8) of PGT121 + VES treated animals and 66% (6 of 9) of GS-9721 + VES treated animals rebounded by day 140 after ART discontinuation (P=0.05, Fisher's exact test compared with sham controls) and showed a delay in the median rebound time of 28 [IQR 21-140+] days.

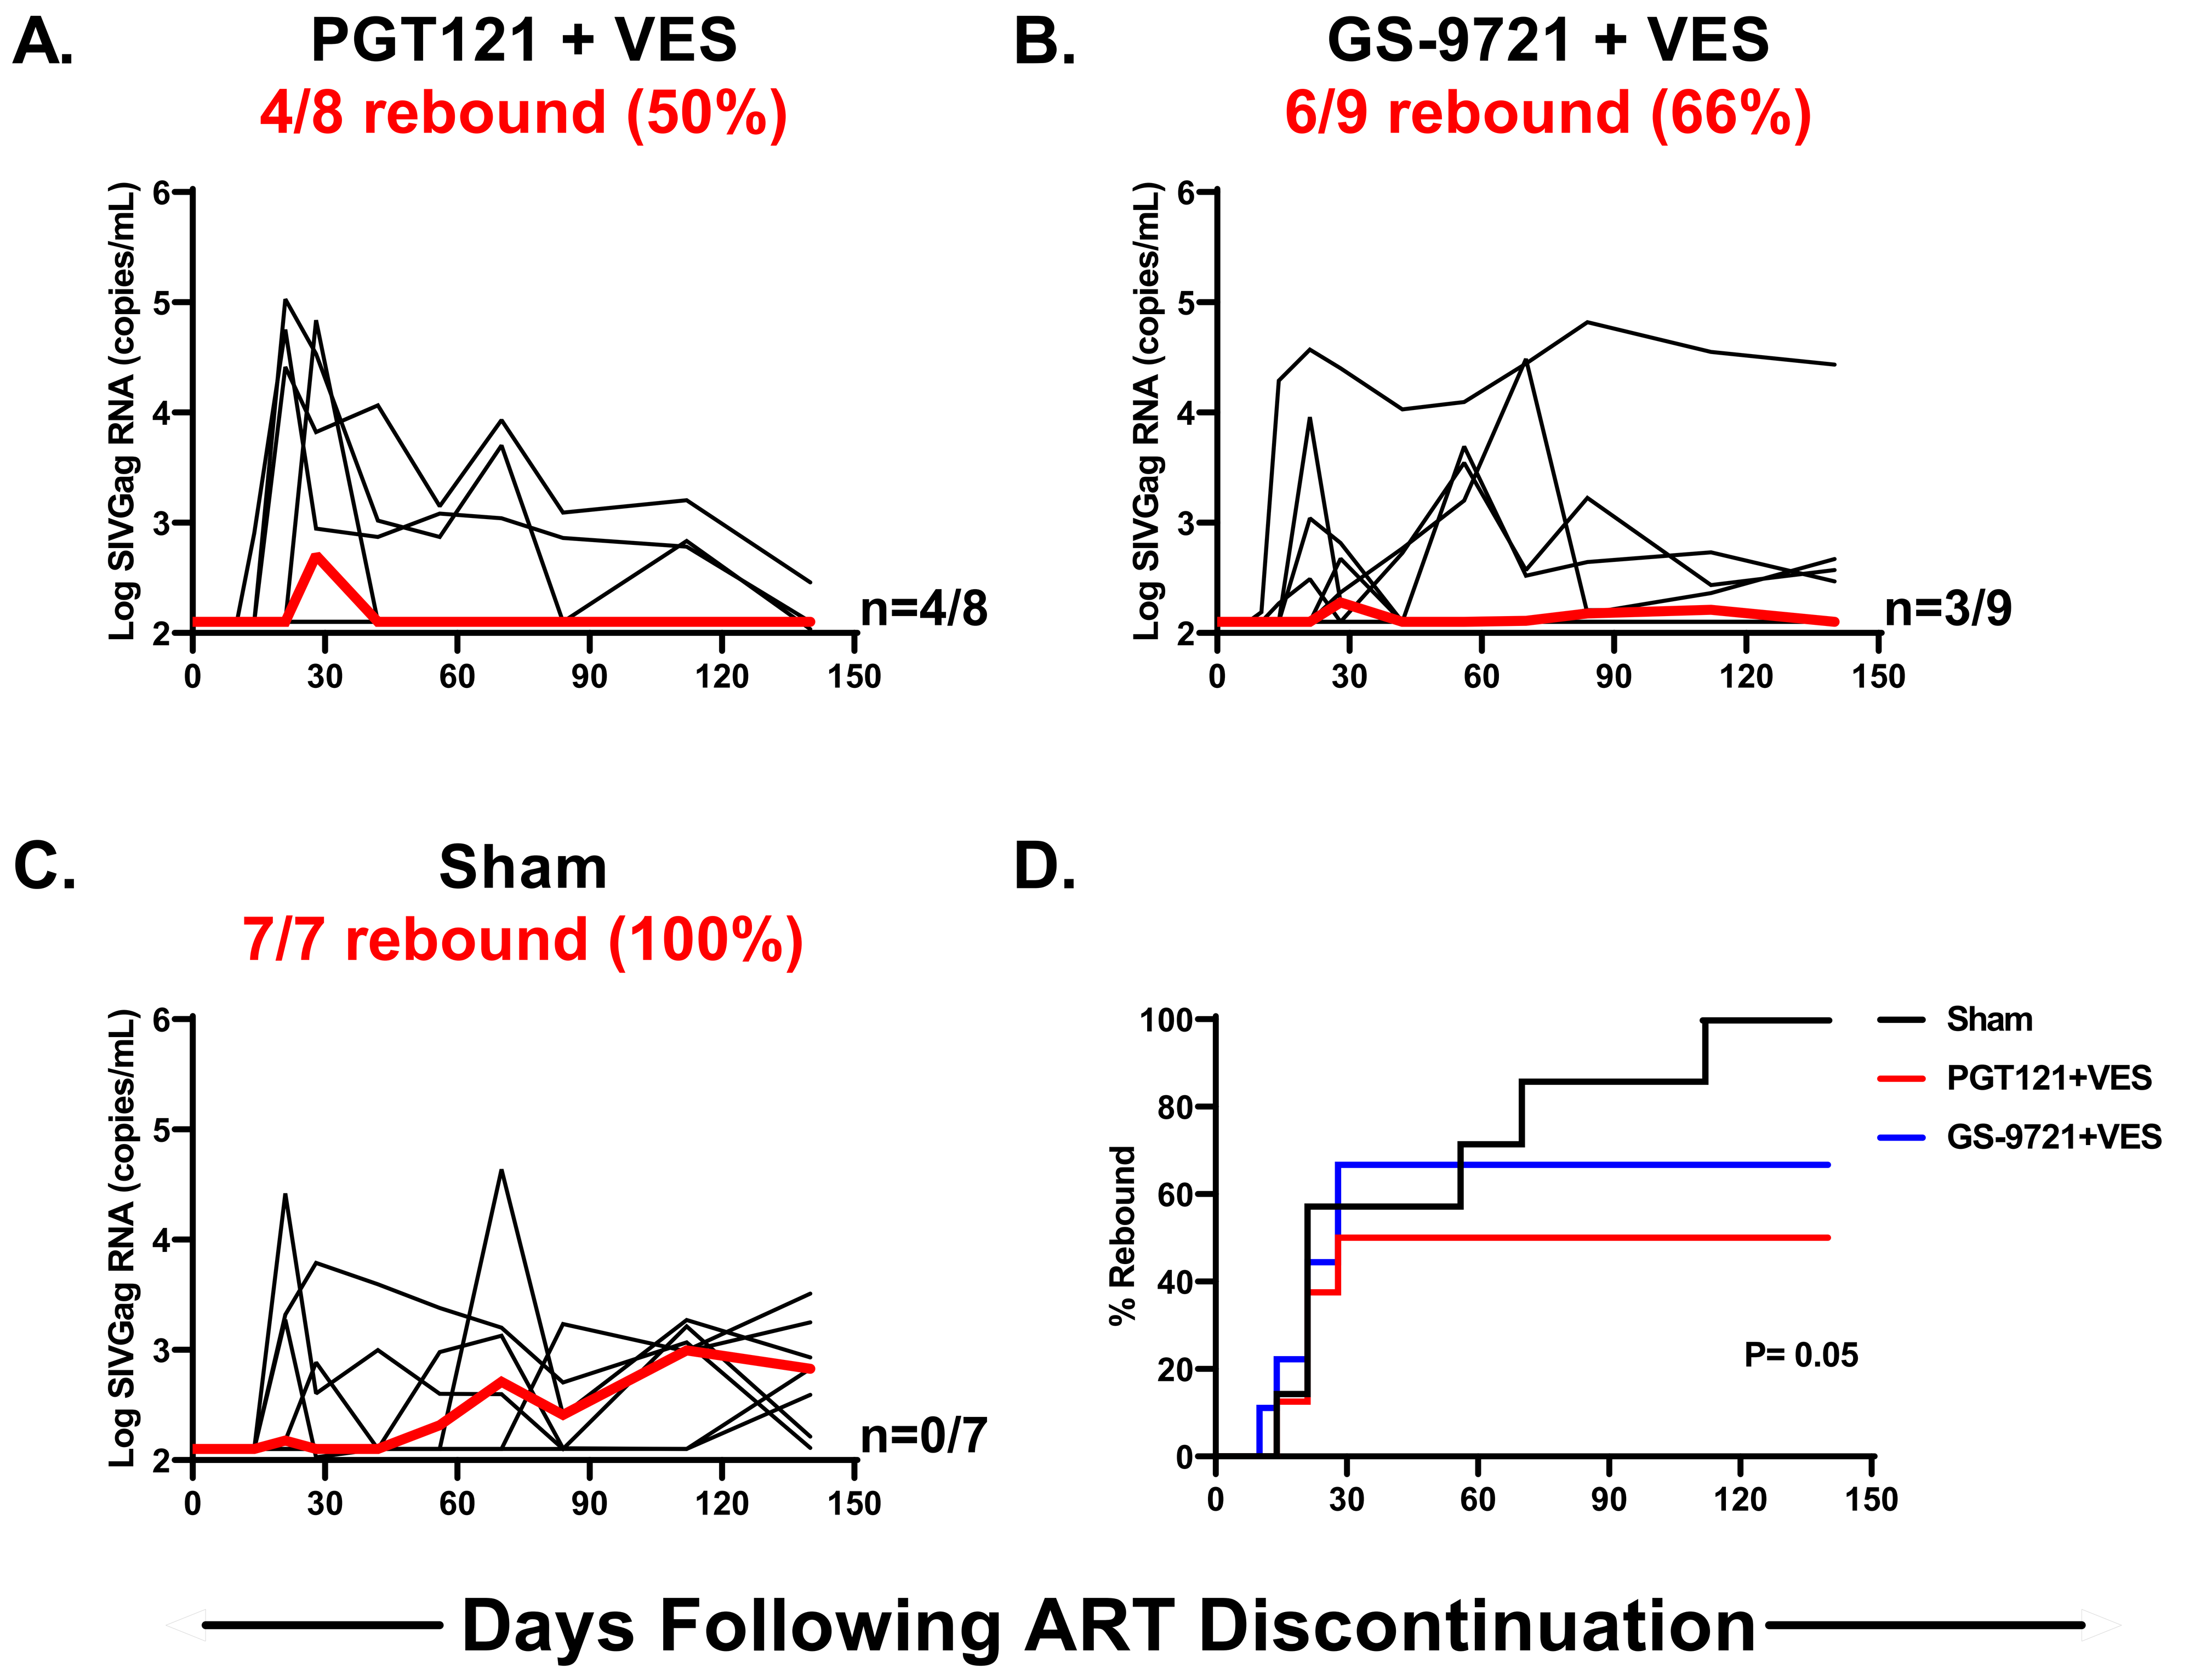
A.
PGT121 + VES
4/8 rebound (50%)
n=4/8
B.
GS-9721 + VES
6/9 rebound (66%)
n=3/9
C.
Sham
7/7 rebound (100%)
n=0/7
Log SIVGag RNA (copies/mL)
D.
% Rebound
Sham
PGT121+VES
GS-9721+VES
P= 0.05
0 30 60 90 120 150
Days Following ART Discontinuation

Figure 4 (A-C) Viral loads among groups post-ART discontinuation (red lines depict median levels; n = undetectable animals) **(D)** Kaplan-Meier curves comparing time to viral rebound among groups.

CONCLUSIONS

Administration of PGT121 or GS-9721 (Fc effector enhanced PGT121) HIV-1 bNAbs with the TLR-7 agonist vesatolimod prevented viral rebound in 41% (7 out of 17) of animals following ART discontinuation in SHIV-infected rhesus monkeys that initiated ART after 1 year of chronic infection and that were virologically suppressed with ART for 2.5 years. These data suggest efficacy of bNAbs with TLR7 stimulation in targeting the viral reservoir in chronic SHIV infection in rhesus monkeys. Acknowledging several caveats of the study that include some ADA formation and rhesus versus human FcRs, additional investigation will likely be required to ascertain cure versus control in animals that did not rebound. Furthermore, the lack of difference between PGT121 and GS-9721, suggesting effector enhancement may not be necessary, may require further exploration.

ACKNOWLEDGEMENTS: Bioqual Inc., Gilead Sciences Inc., BIDMC