1401

Drug Interactions With Once-Daily B/F/TAF in Combination With Once-Weekly Rifapentine

Priyanka Arora, Sean E. Collins, Hal Martin, Xu Zhang, Lily Mak, John Ling, Polina German — Gilead Sciences, Inc., Foster City, CA

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Introduction

- Bictegravir (BIC), emtricitabine (FTC), and tenofovir (TFV) alafenamide (B/F/TAF) is a guidelines-recommended, first-line, single tablet once-daily (qd) treatment for people living with HIV (PLWH),[1-3] which has a high barrier to resistance
 - BIC has a high mean inhibitory quotient (IQ) of 16.1 following administration of B/F/TAF in HIV-infected participants[4]
- Among people with latent tuberculosis (TB) infection (LTBI), PLWH are ~20 times more likely to develop active TB compared with those without HIV, making it a leading cause of death among PLWH[5,6]
 - Standard of care includes cotreatment of LTBI and HIV concomitantly
 - Guidelines-recommended LTBI treatments include once-weekly (qwk) rifapentine (RPT) + isoniazid for 12 wk
- BIC is metabolized by uridine diphosphate glucuronosyltransferase 1A1 (UGT1A1) and cytochrome P450 (CYP) 3A; TFV alafenamide (TAF) is a substrate of transporters P-glycoprotein (P-gp), organic anion-transporting polypeptide (OATP)-1B1/1B3, and breast cancer resistance protein
 - Coadministration of drugs that are strong inducers of these pathways is expected to decrease plasma concentrations of BIC and/or TAF
 - Rifampin, a drug commonly used in the treatment of TB, is a potent pan-inducer of CYP3A4, UGT1A1, and P-gp, which is contraindicated with B/F/TAF due to significantly lower BIC plasma exposure
- RPT is a strong inducer of CYP3A, but with induction potency less than that of rifampin; the inductive effect of RPT on P-gp is currently unknown
- The effects of RPT on B/F/TAF pharmacokinetics (PK) and safety have not been explored, thus precluding coadministration of these agents

Objectives

- **Primary:** to evaluate the effect of RPT qwk on B/F/TAF PK
- **Secondary:** to evaluate the safety and tolerability of multiple-dose B/F/TAF administered with RPT qwk
- **Exploratory:** to describe the PK of TFV diphosphate (TFV-DP; TAF/TFV active metabolite) in peripheral blood mononuclear cells (PBMCs)

Methods

Study Design

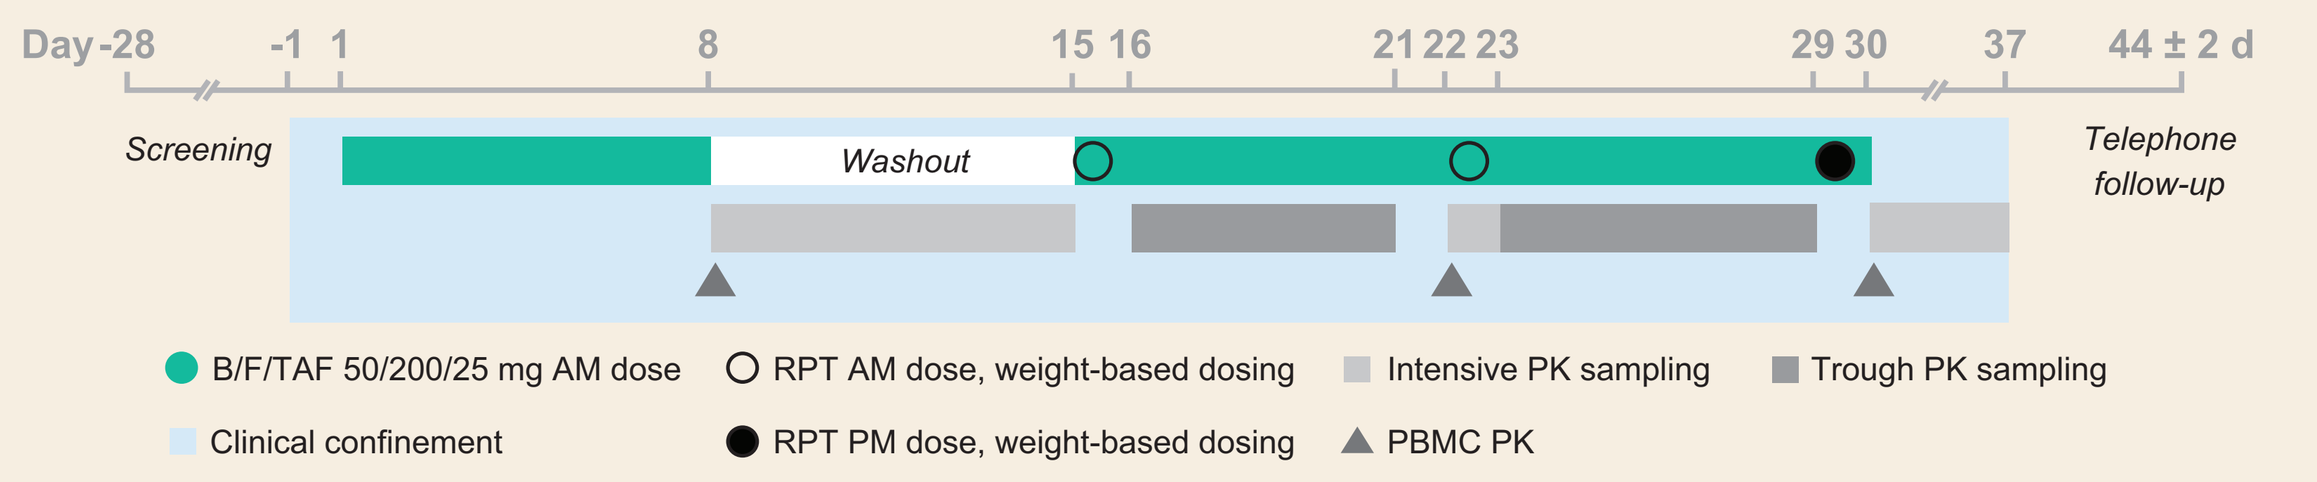
Day -28
-1 1
8
15 16
21 22 23
29 30
37
44 ± 2 d
Screening
Washout
Telephone follow-up
B/F/TAF 50/200/25 mg AM dose
RPT AM dose, weight-based dosing
Intensive PK sampling
Trough PK sampling
Clinical confinement
RPT PM dose, weight-based dosing
PBMC PK

- A Phase 1, open-label, 3-period fixed-sequence, multiple-dose, single-center study was conducted in 30 HIV-negative healthy volunteers
- An even distribution (1:1) of healthy men and nonpregnant, nonlactating women aged 18–45 y were enrolled
- Following completion of screening and admission assessments, all eligible participants began receiving B/F/TAF qd on Days 1–8 followed by a washout period from Days 9–14; participants then received B/F/TAF qd on Days 15–30 with RPT qwk co-dosed on Days 15 and 22, and 12 h before B/F/TAF on Day 29

Study Methods

- Intensive plasma sampling occurred at the following time points for determination of BIC, FTC, TAF, and TFV (TAF major metabolite) PK:
 - Days 8 and 30 at predose, and 0.25, 0.5, 1, 1.5, 2, 3, 4, 6, 8, 12, 16, 24, 48, 72, 96, 120, 144, and 168 h postdose
 - Day 22 at predose, and 0.25, 0.5, 1, 1.5, 2, 3, 4, 6, 8, 12, 16, and 24 h postdose
- Plasma samples were collected on Days 16–21 and 23–29 between RPT doses at the following time points to examine the time course of drug interaction:
 - Predose, and 1, 1.5, and 2 h postdose
- PBMC sampling occurred on Days 8, 22, and 30 for determination of TFV-DP PK
- PK parameters were estimated by noncompartmental methods using Phoenix WinNonlin 8.2 (Certara, Princeton, NJ)
- Geometric least-squares mean (GLSM) ratios and corresponding 90% confidence intervals (CIs) for primary PK parameters (area under plasma concentration-time curve over dosing interval [AUC_{tau}], maximal concentration [C_{max}], and trough concentration [C_{tau}], as applicable) were used for statistical comparisons of exposures
 - Test: B/F/TAF qd coadministered with RPT qwk or administered 12 h after RPT
 - Reference: B/F/TAF qd alone
- Safety was evaluated throughout the study by assessment of adverse events (AEs) and laboratory monitoring

Results

Demographics and Baseline Characteristics

	N=30
Enrolled/completed, n/n	30/28
Mean age, y (SD)	36 (6.3)
Female at birth, n (%)	15 (50)
Race, n (%)	
American Indian or Alaska Native	1 (3)
Asian	1 (3)
Black or African-American	17 (57)
White	11 (37)
Hispanic or Latinx ethnicity, n (%)	6 (20)
Mean BMI, kg/m² (SD)	26.6 (2.5)
Mean $eGFR_{CG}$, mL/min (SD)	121 (19)

BMI, body mass index; $eGFR_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault formula; SD, standard deviation.

Safety

- Study treatments were safe and well tolerated
- Treatment-emergent AEs occurred in a larger proportion of participants receiving B/F/TAF qd + RPT qwk (48%) vs B/F/TAF qd alone (27%)
- Most AEs were mild (Grade 1)
 - One Grade 3 AE was observed: neck pain judged not related to study drugs
- There were no serious AEs
- There were no clinically significant or Grade ≥3 laboratory abnormalities

BIC PK Following B/F/TAF qd Alone vs Coadministered With or Administered 12 h After RPT qwk

BIC Plasma Concentrations

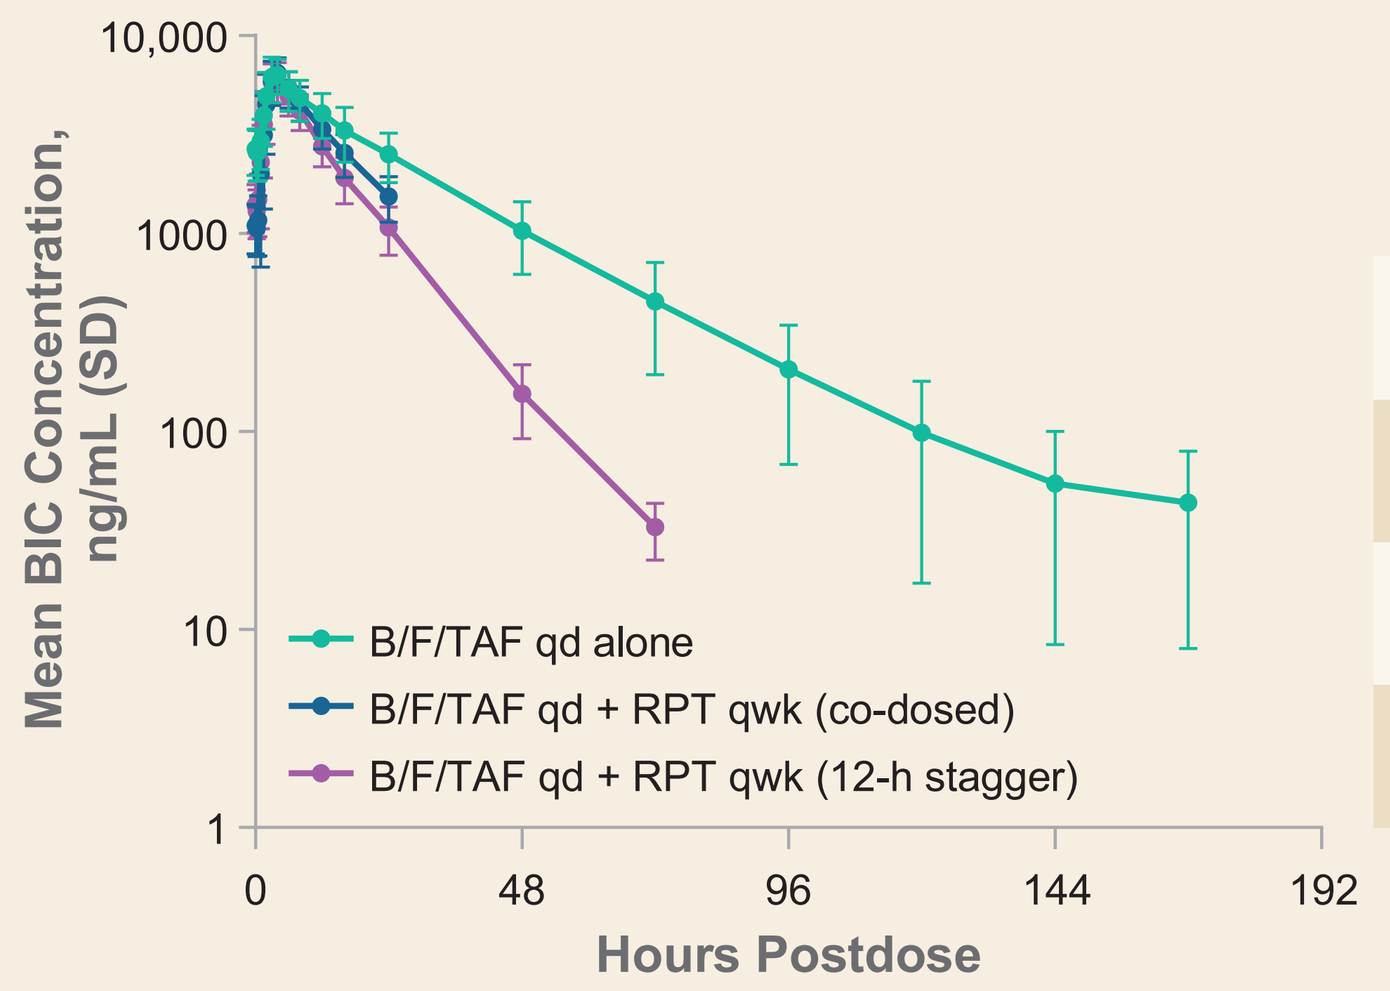
Mean BIC Concentration, ng/mL (SD)
10,000
1000
100
10
1
0
48
96
144
192
Hours Postdose
B/F/TAF qd alone
B/F/TAF qd + RPT qwk (co-dosed)
B/F/TAF qd + RPT qwk (12-h stagger)

Summary of BIC Plasma PK Parameter Estimates*

Mean PK Parameter (%CV)	B/F/TAF qd n=29	B/F/TAF qd + RPT qwk Co-Dosed n=29	B/F/TAF qd + RPT qwk 12-h Stagger n=28	%GLSM (90% CI) Co-Dosed vs Alone	%GLSM (90% CI) 12-h Stagger vs Alone
C_{max}, ng/mL	6870 (16.3)	6880 (16.9)	6590 (16.6)	100 (95.5, 105)	96.0 (91.8, 100)
AUC_{tau}, h·ng/mL	96100 (23.3)	81400 (17.9)	70800 (18.3)	85.5 (81.3, 89.8)	74.1 (70.2, 78.3)
C_{tau}, ng/mL	2510 (28.1)	1520 (26.6)	1080 (27.2)	60.4 (56.3, 64.7)†	42.5 (39.1, 46.2)†
Median $t_{1/2}$, h (Q1, Q3)	20.7 (18.5, 22.3)	10.3 (9.71, 11.2)	8.82 (8.07, 9.23)		

*Data shown to 3 significant digits; †Outside no-effect drug-drug interaction boundaries. CV, coefficient of variation; Q, quartile; $t_{1/2}$, half-life.

BIC Trough Concentrations Throughout Study Days Across Treatment Periods

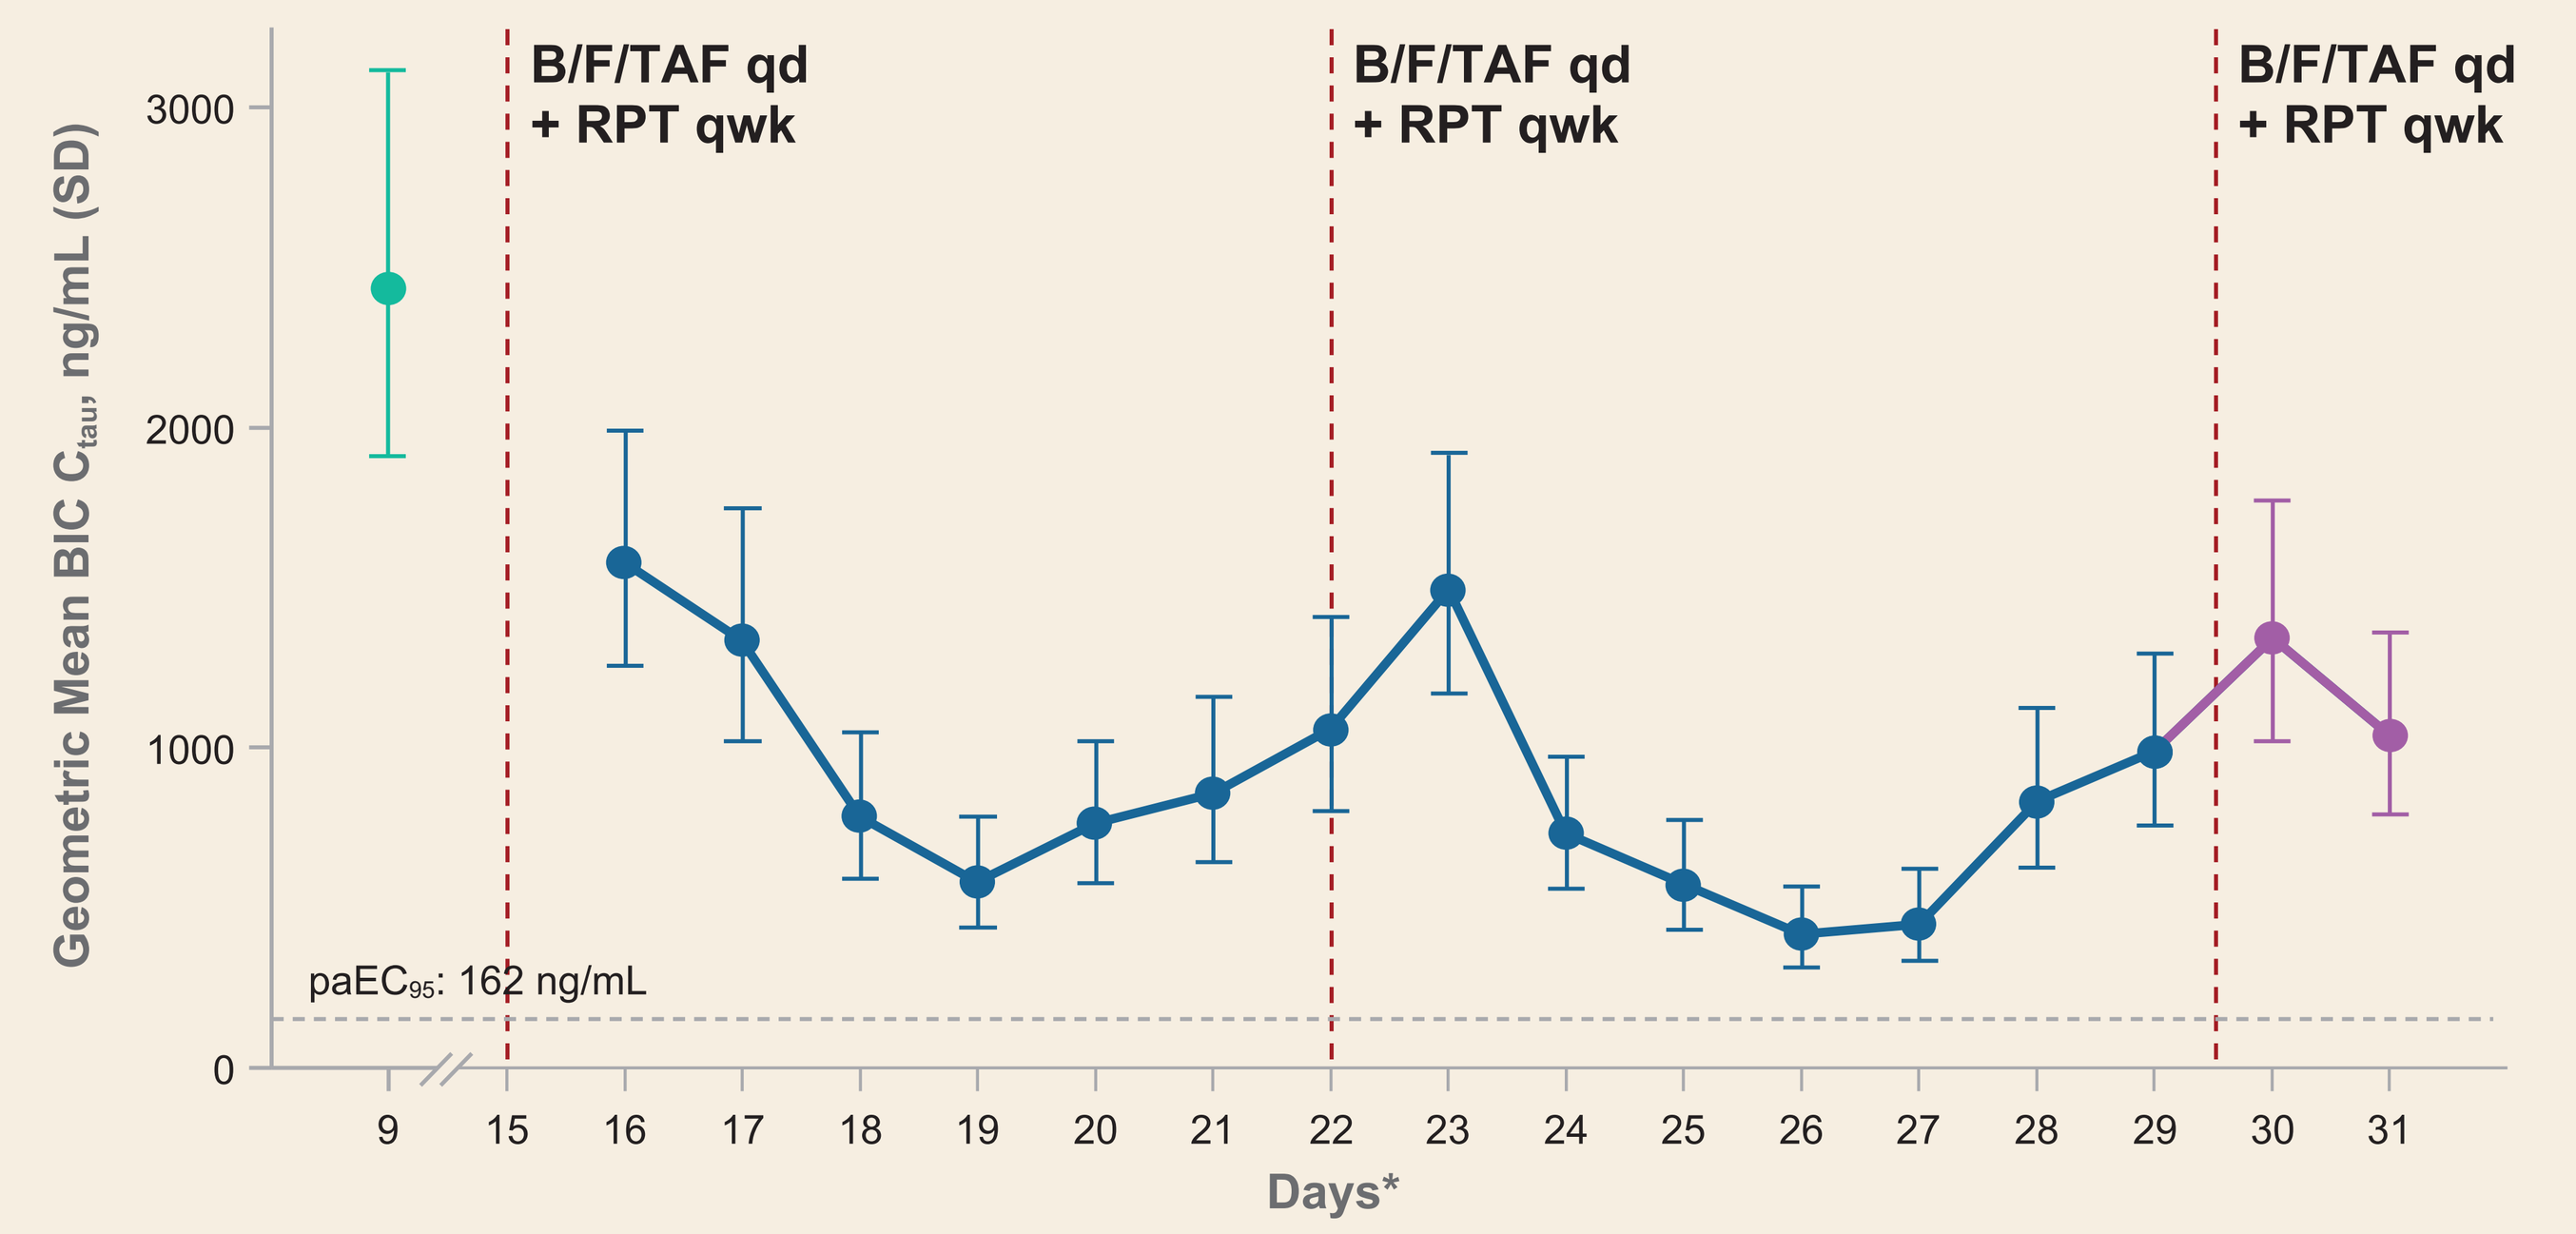
Geometric Mean BIC C_{tau}, ng/mL (SD)
3000
2000
1000
0
B/F/TAF qd + RPT qwk
B/F/TAF qd + RPT qwk
B/F/TAF qd + RPT qwk
paEC95: 162 ng/mL
9 15 16 17 18 19 20 21 22 23 24 25 26 27 28 29 30 31
Days*

*Relative to date of first study drug administration (Day 1); B/F/TAF qd was administered on Days 1–8 (reference); *red dashed lines* indicate RPT qwk administration with B/F/TAF qd on Days 15–30 (RPT was coadministered in AM with B/F/TAF on Days 15 and 22, and was administered in PM on Day 29, 12 h before AM administration of B/F/TAF on Day 30). $paEC_{95}$, protein-adjusted 95% effective concentration.

- BIC C_{tau} was reduced by as low as 83% by Day 4 post RPT dosing (nadir)
- BIC C_{tau} never recovered back to steady-state concentrations between RPT doses
- 12-h staggered (vs coadministration) of RPT qwk resulted in more pronounced decline in BIC C_{tau}

Distribution of BIC Trough Concentrations in HIV-Infected Participants in Phase 3 Studies Following Administration of B/F/TAF qd vs B/F/TAF qd + RPT qwk

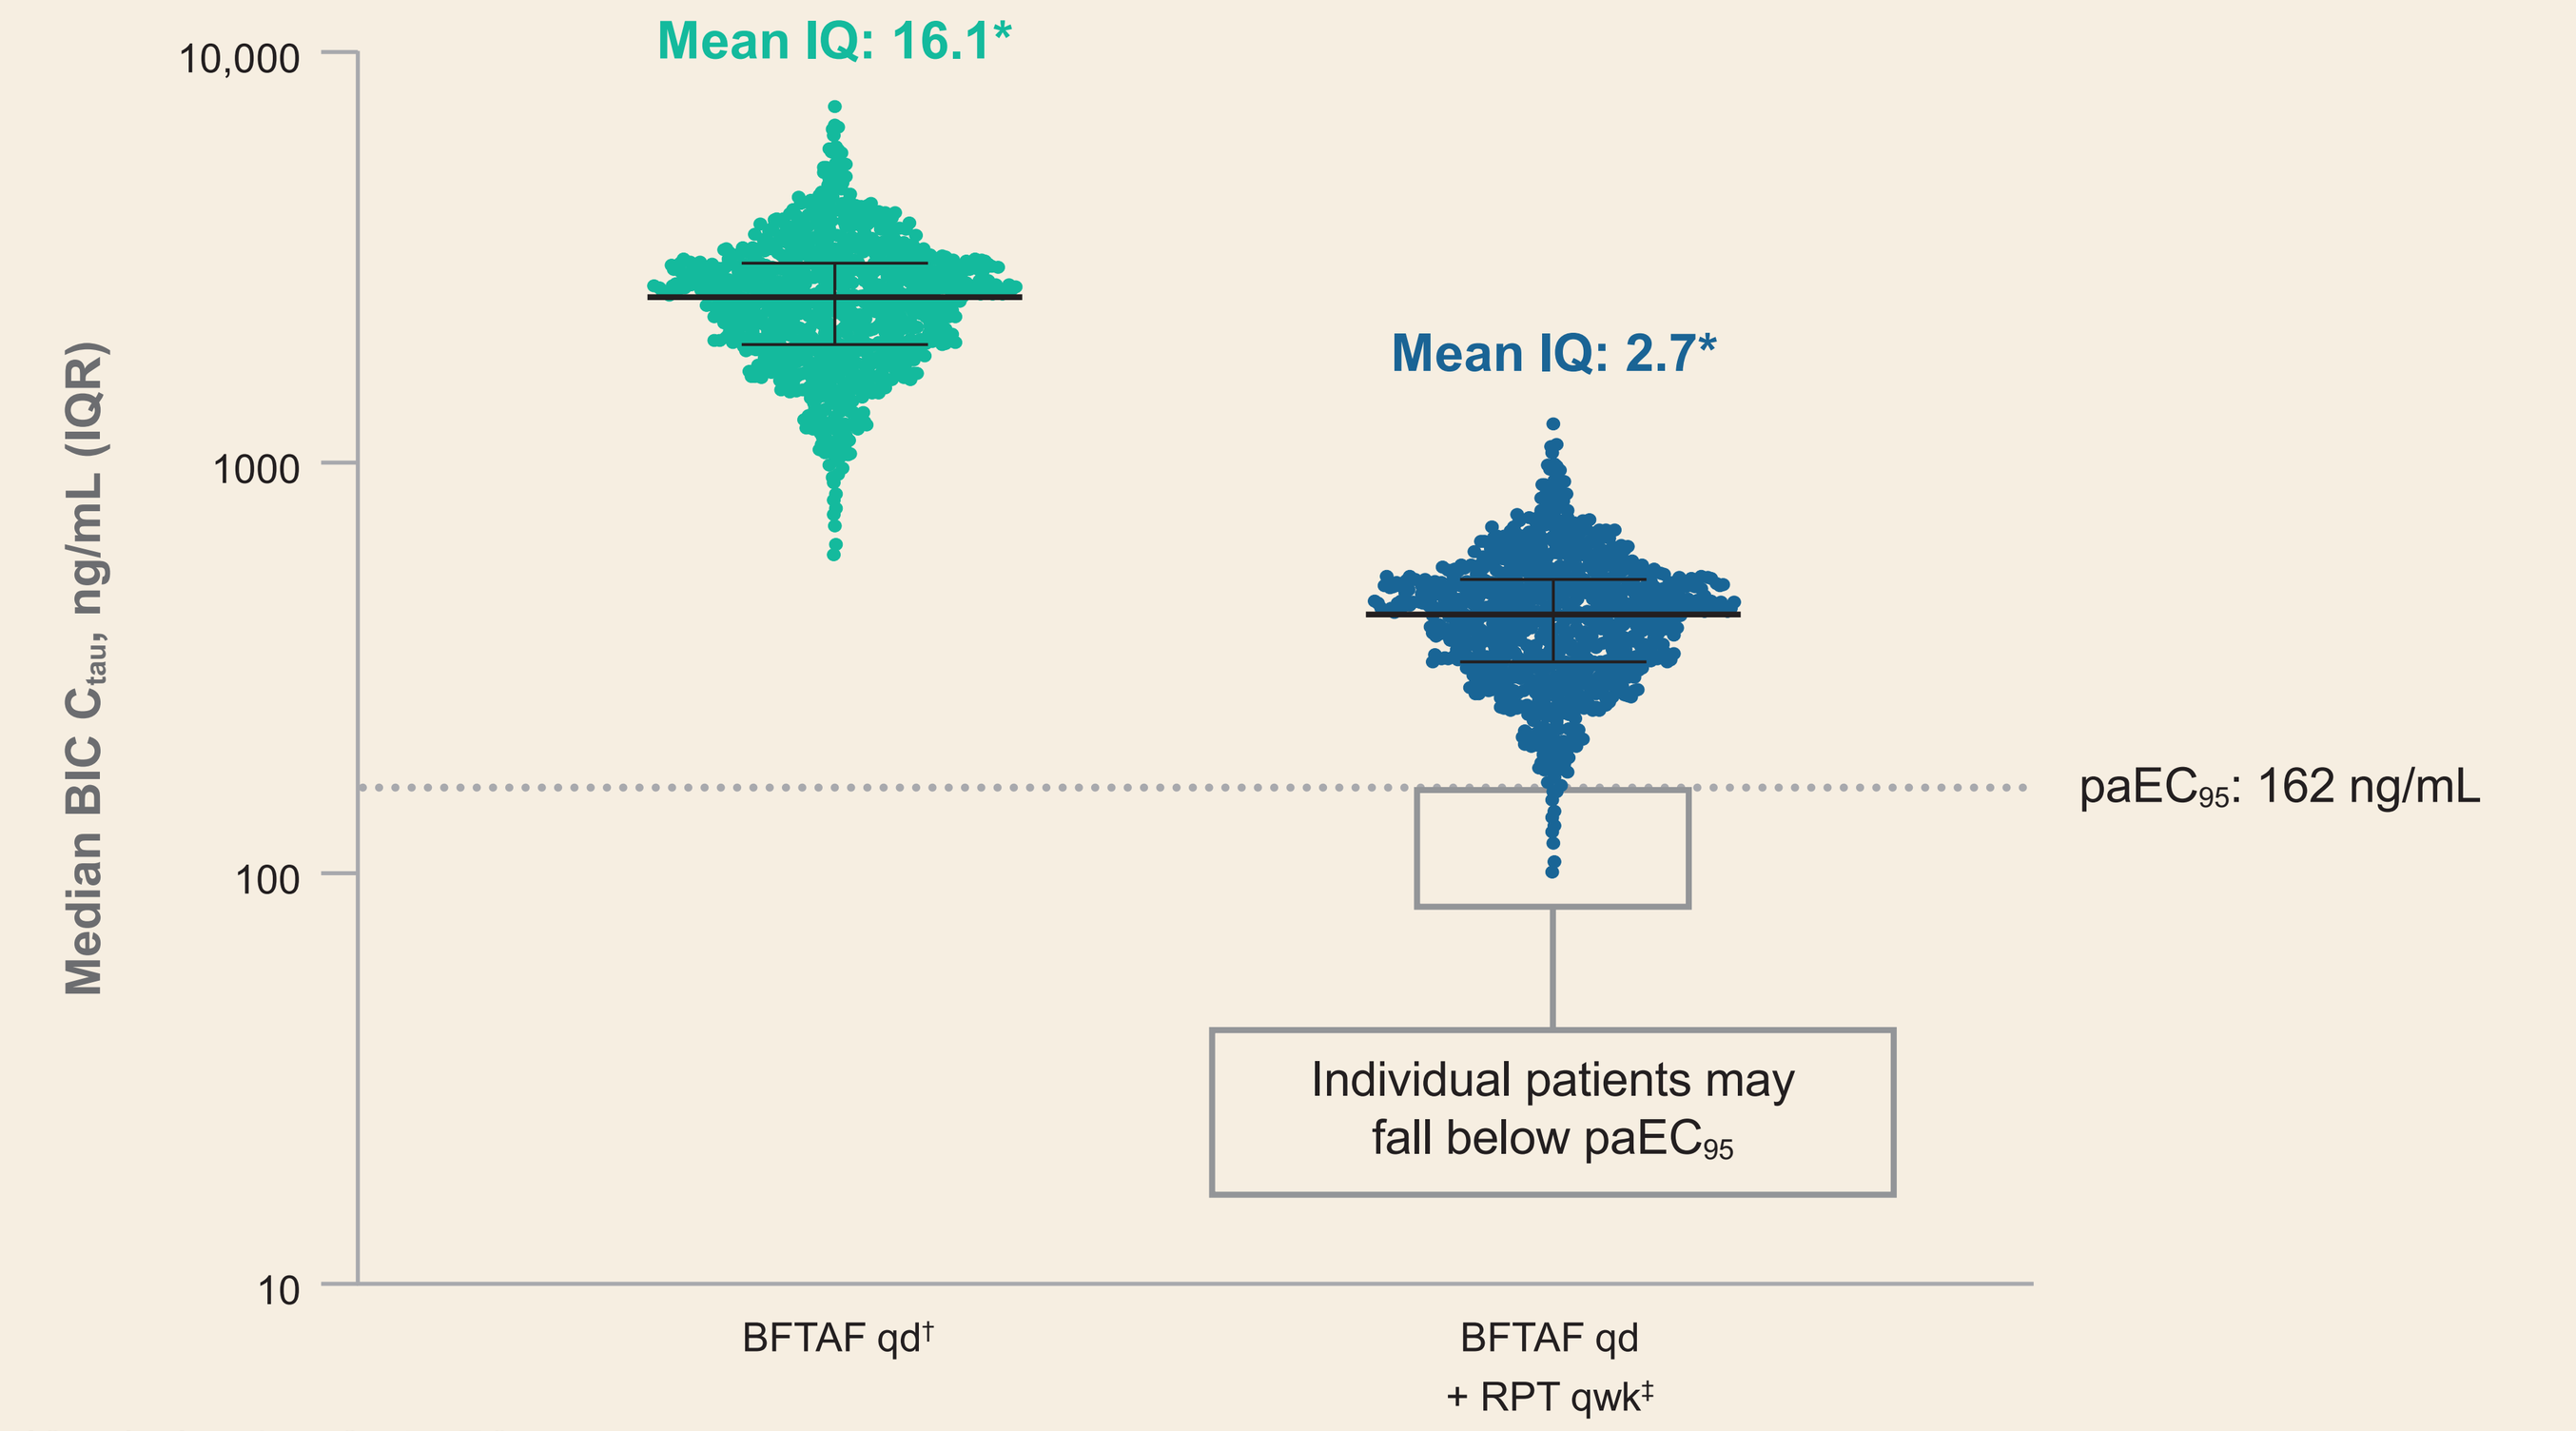
Median BIC C_{tau}, ng/mL (IQR)
10,000
1000
100
10
Mean IQ: 16.1*
Mean IQ: 2.7*
paEC95: 162 ng/mL
Individual patients may fall below paEC95
BFTAF qd†
BFTAF qd + RPT qwk‡

*IQ calculated as $C_{tau}/paEC_{95}$.
†Observed BIC C_{tau} in HIV-infected participants in Phase 3 registrational studies (N=1193) following administration of B/F/TAF qd.
‡Predicted BIC C_{tau} calculated after accounting for ~83% reduction in BIC C_{tau} at nadir following administration of B/F/TAF qd with RPT qwk.

TAF Pharmacokinetics

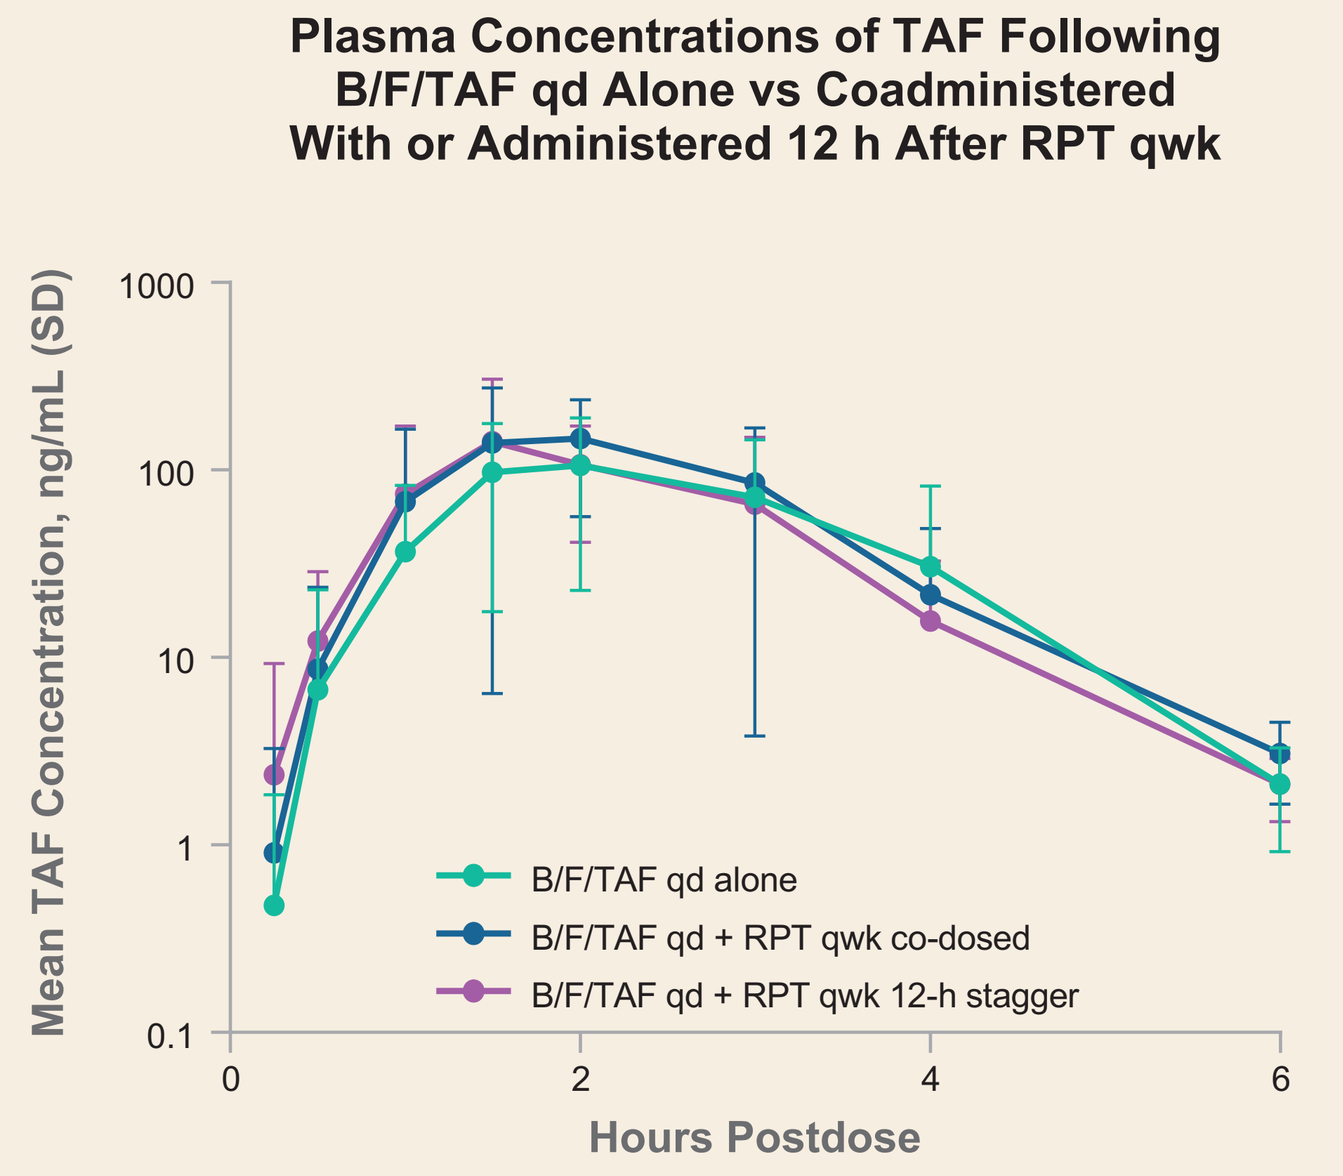
Plasma Concentrations of TAF Following B/F/TAF qd Alone vs Coadministered With or Administered 12 h After RPT qwk
Mean TAF Concentration, ng/mL (SD)
1000
100
10
1
0.1
0
2
4
6
Hours Postdose
B/F/TAF qd alone
B/F/TAF qd + RPT qwk co-dosed
B/F/TAF qd + RPT qwk 12-h stagger

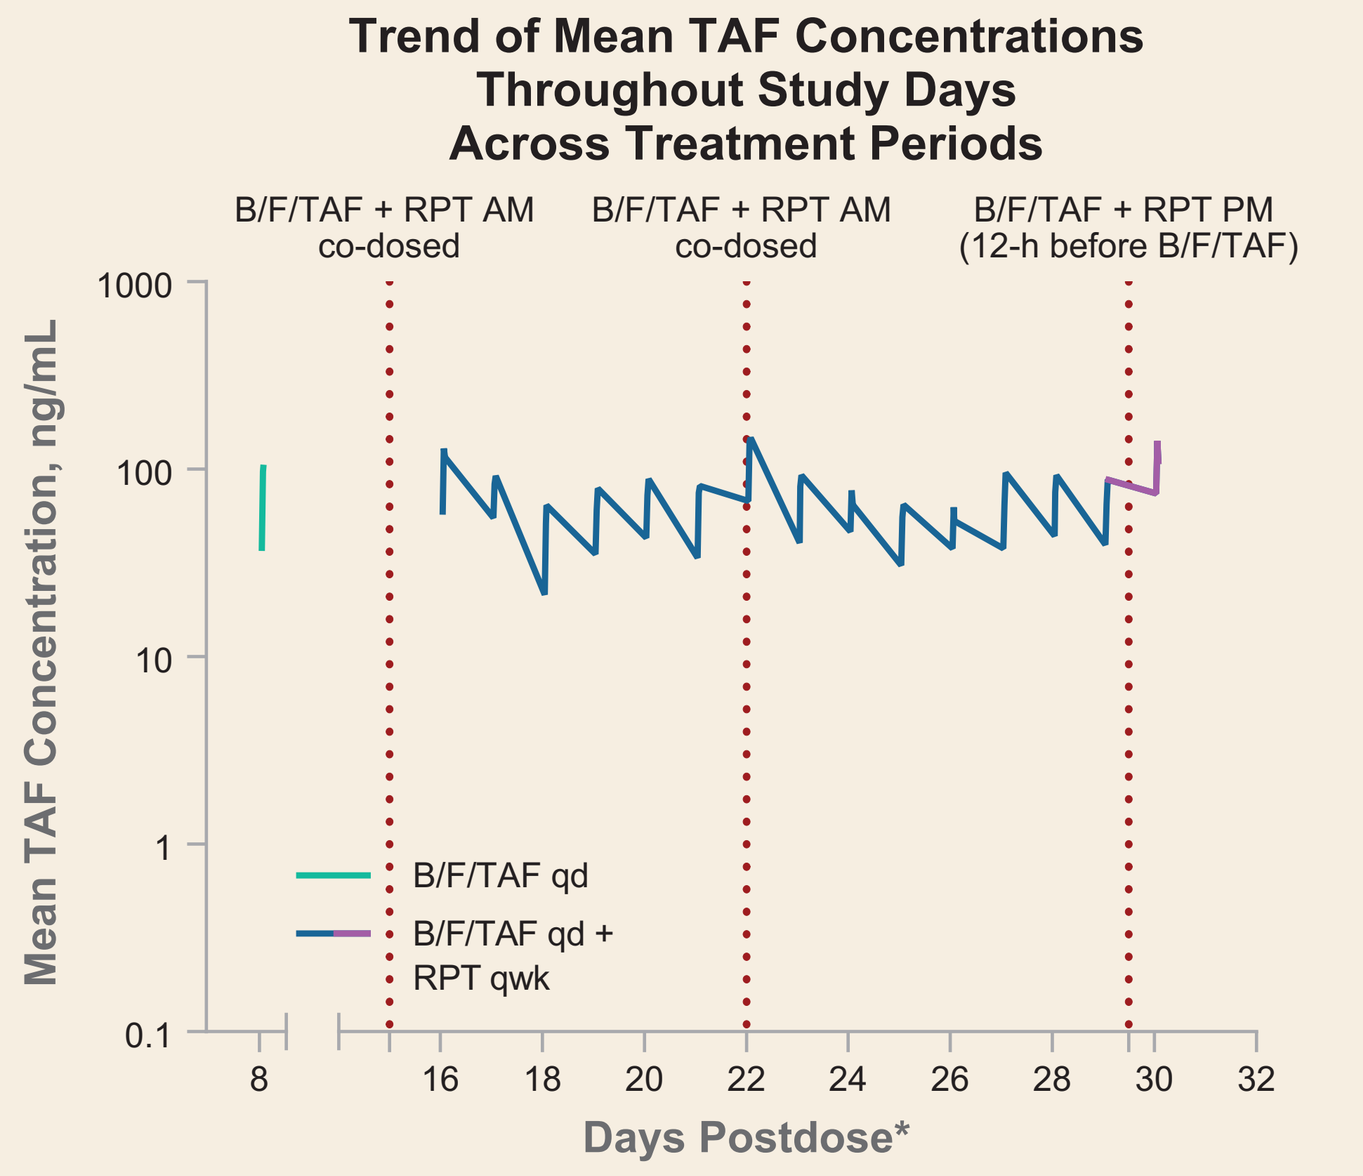
Trend of Mean TAF Concentrations Throughout Study Days Across Treatment Periods
B/F/TAF + RPT AM co-dosed
B/F/TAF + RPT AM co-dosed
B/F/TAF + RPT PM (12-h before B/F/TAF)
Mean TAF Concentration, ng/mL
1000
100
10
1
0.1
8 16 18 20 22 24 26 28 30 32
Days Postdose*
B/F/TAF qd
B/F/TAF qd + RPT qwk

*Relative to date of first study drug administration (Day 1); B/F/TAF qd was administered Days 1–8 (reference); RPT qwk was administered with B/F/TAF qd Days 15–30.

- Coadministration of RPT qwk had no clinically significant effect on TAF PK
- A minor increase in TAF plasma concentrations was observed following coadministration or 12-h staggered administration of B/F/TAF qd with RPT qwk compared with B/F/TAF qd alone; given that RPT has been shown to inhibit OATP1B1-mediated uptake in vitro,[7] this transient increase in TAF exposure may be attributed to inhibition of OATP1B1 activity
- TAF concentrations gradually decreased across study days between RPT doses, with the nadir occurring ~3–4 d post RPT dosing; TAF levels recovered by the ~7th d prior to the next RPT qwk dose, suggesting a weak time-dependent P-gp induction potential of RPT
- Data show weak effect of RPT on TAF PK, possibly due to mixed inhibition and induction of OATP1B1 and P-gp, respectively

Summary of FTC, TAF, TFV, and TFV-DP Parameter Estimates Across Study Treatments

Mean PK Parameter (%CV)	B/F/TAF qd n=29	B/F/TAF qd + RPT qwk Co-Dosed n=29	B/F/TAF qd + RPT qwk 12-h Stagger n=28	%GLSM (90% CI) Co-Dosed vs Alone	%GLSM (90% CI) 12-h Stagger vs Alone
Plasma FTC					
C_{max}, ng/mL	1860 (19.6)	1960 (18.0)	1940 (20.0)	106 (99.1, 113)	105 (97.5, 112)
AUC_{tau}, h·ng/mL	9690 (14.6)	11,000 (14.6)	11,300 (13.6)	113 (110, 117)	117 (114, 120)
C_{tau}, ng/mL	72.5 (21.5)	66.0 (31.8)	86.5 (22.4)	88.9 (82.6, 95.5)	119 (115, 124)
Plasma TAF					
C_{max}, ng/mL	174 (42.2)	231 (44.6)	211 (71.4)	131 (113, 152)	109 (89.4, 133)
AUC_{tau}, h·ng/mL	255 (39.2)	324 (49.2)	288 (45.1)	112 (93.9, 134)	98.4 (91.8, 105)
Plasma TFV					
C_{max}, ng/mL	14.9 (18.7)	13.3 (19.2)	12.1 (18.1)	89.3 (84.9, 93.9)	81.5 (77.6, 85.6)
AUC_{tau}, h·ng/mL	224 (16.6)	195 (16.6)	197 (17.3)	87.2 (84.6, 89.8)	88.0 (85.0, 91.0)
C_{tau}, ng/mL	7.58 (19.4)	6.60 (15.8)	6.84 (17.3)	87.4 (84.1, 90.9)	90.5 (86.7, 94.5)
PBMC TFV-DP					
C_{max}, fmol/10^6 cells	1070 (46.5)	959 (35.8)	874 (46.8)		
AUC_{tau}, h·fmol/10^6 cells	20,000 (43.2)	19,900 (35.0)	15,700 (34.8)		
C_{tau}, fmol/10^6 cells	735 (46.2)	782 (33.0)	600 (37.0)		

Data shown to 3 significant digits.

Conclusions

- All study treatments were safe and well tolerated
- BIC C_{tau} was ~35–83% lower following administration of B/F/TAF qd + RPT qwk, indicating a significant impact of RPT on BIC PK due to potent induction of CYP3A4
- After accounting for ~83% reduction in BIC C_{tau} at the nadir and variability observed in the Phase 3 population, trough levels are predicted to fall below $paEC_{95}$ (IQ of 1) in some patients if daily B/F/TAF is administered with weekly RPT
- No clinically significant changes in the PK of FTC, TAF, TFV, or TFV-DP were observed with coadministration of weekly RPT
- From a mechanistic perspective, this study suggests that RPT may be a weak OATP inhibitor and P-gp inducer, in addition to being a strong CYP3A inducer
- Based on the substantial reduction in BIC C_{tau}, use of single tablet regimen B/F/TAF with weekly RPT is not recommended

References: **1.** AIDSinfo. Guidelines for the Use of Antiretroviral Agents in Adults and Adolescents Living with HIV; 12/19; **2.** EACS Guidelines Version 10.0 November 2019; **3.** Saag MS, et al. JAMA 2018;320:379-96; **4.** Biktarvy [package insert]. Foster City, CA: Gilead Sciences, Inc.; 2/21; **5.** Centers for Disease Control and Prevention. Fact Sheets; 11/12/20; **6.** World Health Organization. Tuberculosis; 2021; **7.** Parvez MM, et al. Antimicrob Agents Chemother 2016;60:3096-105.

Acknowledgments: We extend our thanks to the participants, their families, and all participating study investigators and staff. This study was funded by Gilead Sciences, Inc.

Presented at *Virtual* CROI 2021, March 6–10, 2021

©2021 Gilead Sciences, Inc. All rights reserved.