1400

HIV Rebound in Controllers Is Associated With Specific Fecal Microbiome Profile

Yanhui Cai,[1] Steven G. Deeks,[2] Cynthia Brinson,[3] Moti Ramgopal,[4] Norman Jones,[2] Edwin DeJesus,[5] Anthony Mills,[6] Peter Shalit,[7] Brian Moldt,[1] Liao Zhang,[1] Elena Vendrame,[1] Diana M. Brainard,[1] Devi SenGupta,[1] Jeffrey Wallin,[1] Ondrej Podlaha[1]

[1]Gilead Sciences, Inc., Foster City, CA; [2]University of California San Francisco; [3]Central Texas Clinical Research, Austin, TX; [4]Midway Research Center, Fort Pierce, FL; [5]Orlando Immunology Center, Orlando, FL; [6]Men's Health Foundation, West Hollywood, CA; [7]Peter Shalit MD and Associates, Seattle, WA

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Introduction

- HIV infection negatively impacts gut immune homeostasis and frequently leads to dysbiosis, which can only be partially restored by antiretroviral therapy (ART) in people living with HIV (PLWH)
- Administration of a toll-like receptor-7 (TLR-7) agonist, in combination with a therapeutic vaccine, induced CD8 T-cell–mediated control of simian immunodeficiency virus in a nonhuman primate model[1]
- Vesatolimod (VES) is an oral, selective, TLR-7 agonist shown to be safe and well tolerated in PLWH
- In a Phase 1b, randomized, double-blind, placebo-controlled study of VES in HIV virologic controllers (VCs) on ART, VES was associated with:
 - A modest increase in time to viral rebound, as well as a decrease in viral set point and intact proviral DNA[2]
 - Induction of a dose-dependent interferon response and increased immune-cell activation[3]

Objectives

- To characterize the fecal microbiome in HIV VCs on ART and following VES treatment
- To investigate if there is any association between fecal microbiome and VES treatment outcomes

Methods

Study Design for Oral VES in HIV VCs on ART

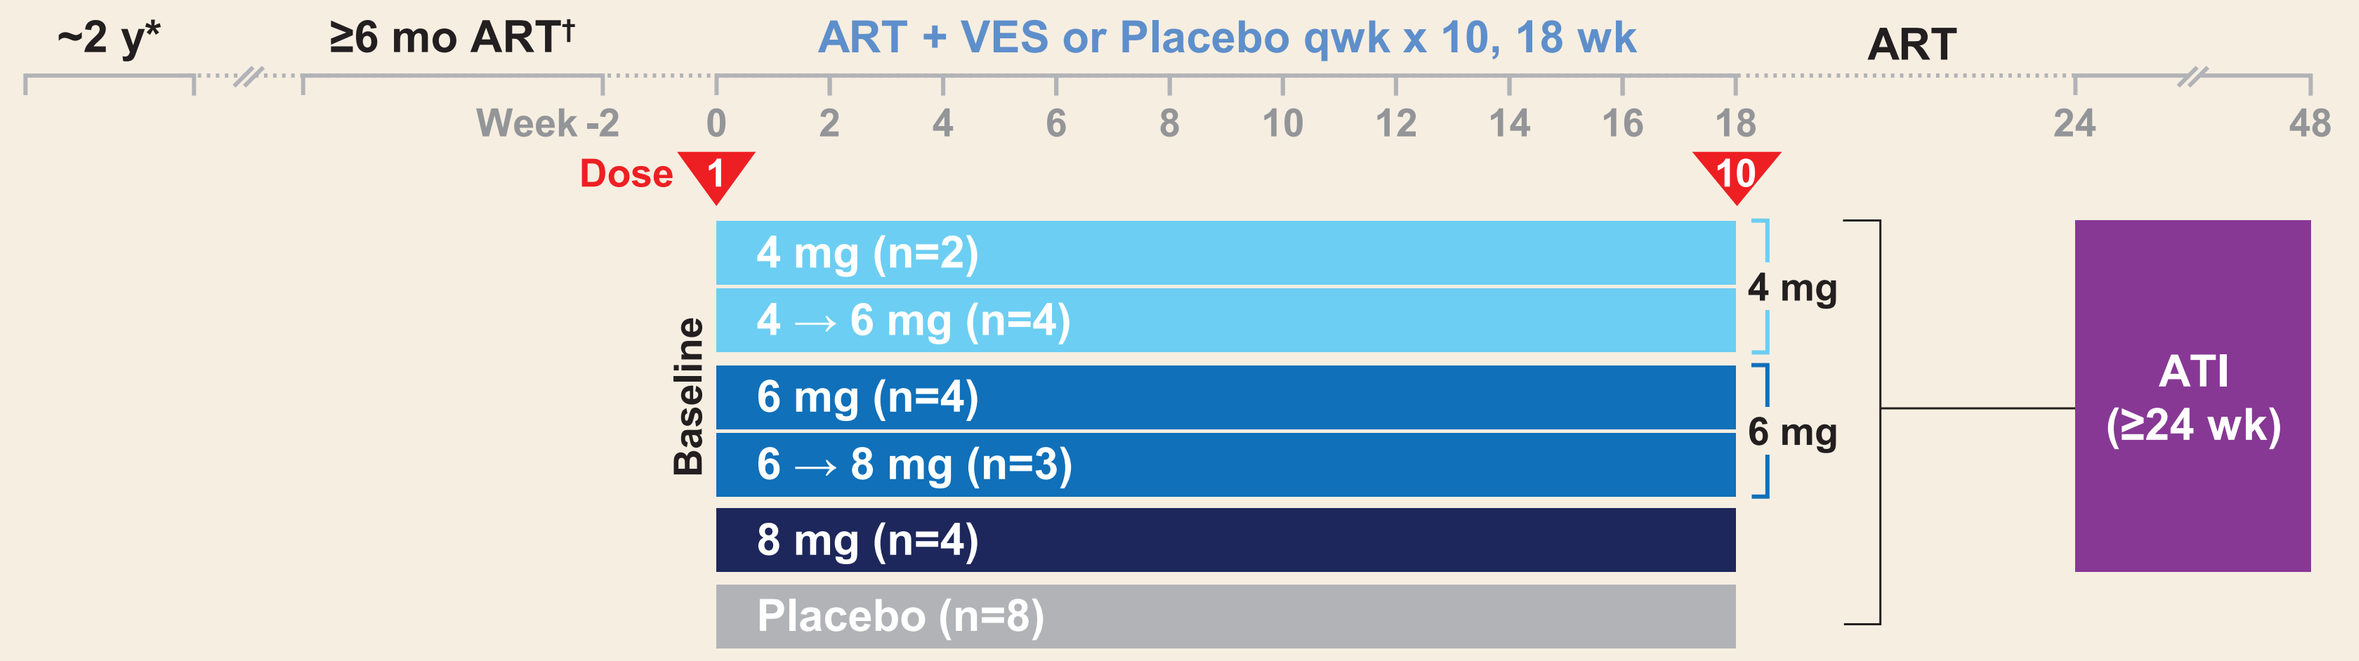
~2 y*
≥6 mo ART†
ART + VES or Placebo qwk x 10, 18 wk
ART
Week -2 0 2 4 6 8 10 12 14 16 18 24 48
Dose 1
10
Baseline
4 mg (n=2)
4 → 6 mg (n=4)
6 mg (n=4)
6 → 8 mg (n=3)
8 mg (n=4)
Placebo (n=8)
4 mg
6 mg
ATI
(≥24 wk)

*Set-point viral load (50–5000 copies/mL); †Viral load <50 copies/mL and CD4 >500 cells/μL. ATI, analytical treatment interruption.

- 25 HIV VCs (pre-ART viral load 50–5000 copies/mL) on ART for ≥6 mo were enrolled
 - 17 participants received 10 biweekly VES doses and 8 received placebo prior to an ATI
- To avoid confounding factors due to gender discrepancy, fecal samples from male participants were assessed at baseline and 2 wk after the 10th dose for microbial abundance and diversity evaluation using metagenome sequencing method
 - Feces from an additional 14 male healthy volunteers (HVs) and 9 male ART-suppressed chronic HIV-infected participants (CHIs) were included to assess baseline differences due to HIV infection by Wilcoxon test
- Associations between immune biomarkers and bacterial abundance at the phylum level were measured by Spearman's rank correlation
- A univariate Cox proportional hazard regression model was used to explore the association between time to viral rebound and abundance of microbiota species
- Microbiome analysis plan:
 - Compare fecal microbiome α-diversity and abundance between PLWH and HVs
 - Characterize microbiome changes following VES treatment (baseline vs pre-ATI)
 - Investigate association between fecal microbiome abundance, immune biomarkers, and time to viral rebound after ATI

Results

Demographic Summary

	Baseline: CHIs: n=9	Baseline: VCs: n=6	Baseline: HVs: n=14	Baseline: p-Value*	Pre-ATI: VCs: n=13
Median age, y (IQR)	43 (31, 54)	36 (32, 46)	25 (23, 28)	0.001	44 (35, 52)
Race, n (%)				0.60	
Black or African-American	0	1 (17)	2 (14)		2 (15)
White	9 (100)	5 (83)	12 (86)		11 (85)
Median BMI, kg/m² (IQR)	24.7 (21.7, 26.7)	27.4 (25.1, 29.9)	26.0 (24.0, 31.0)	0.20	25.5 (24.4, 27.1)

*Kruskal-Wallis and Fisher's exact tests. BMI, body mass index; IQR, interquartile range.

Microbiome Abundance Between PLWH and HVs

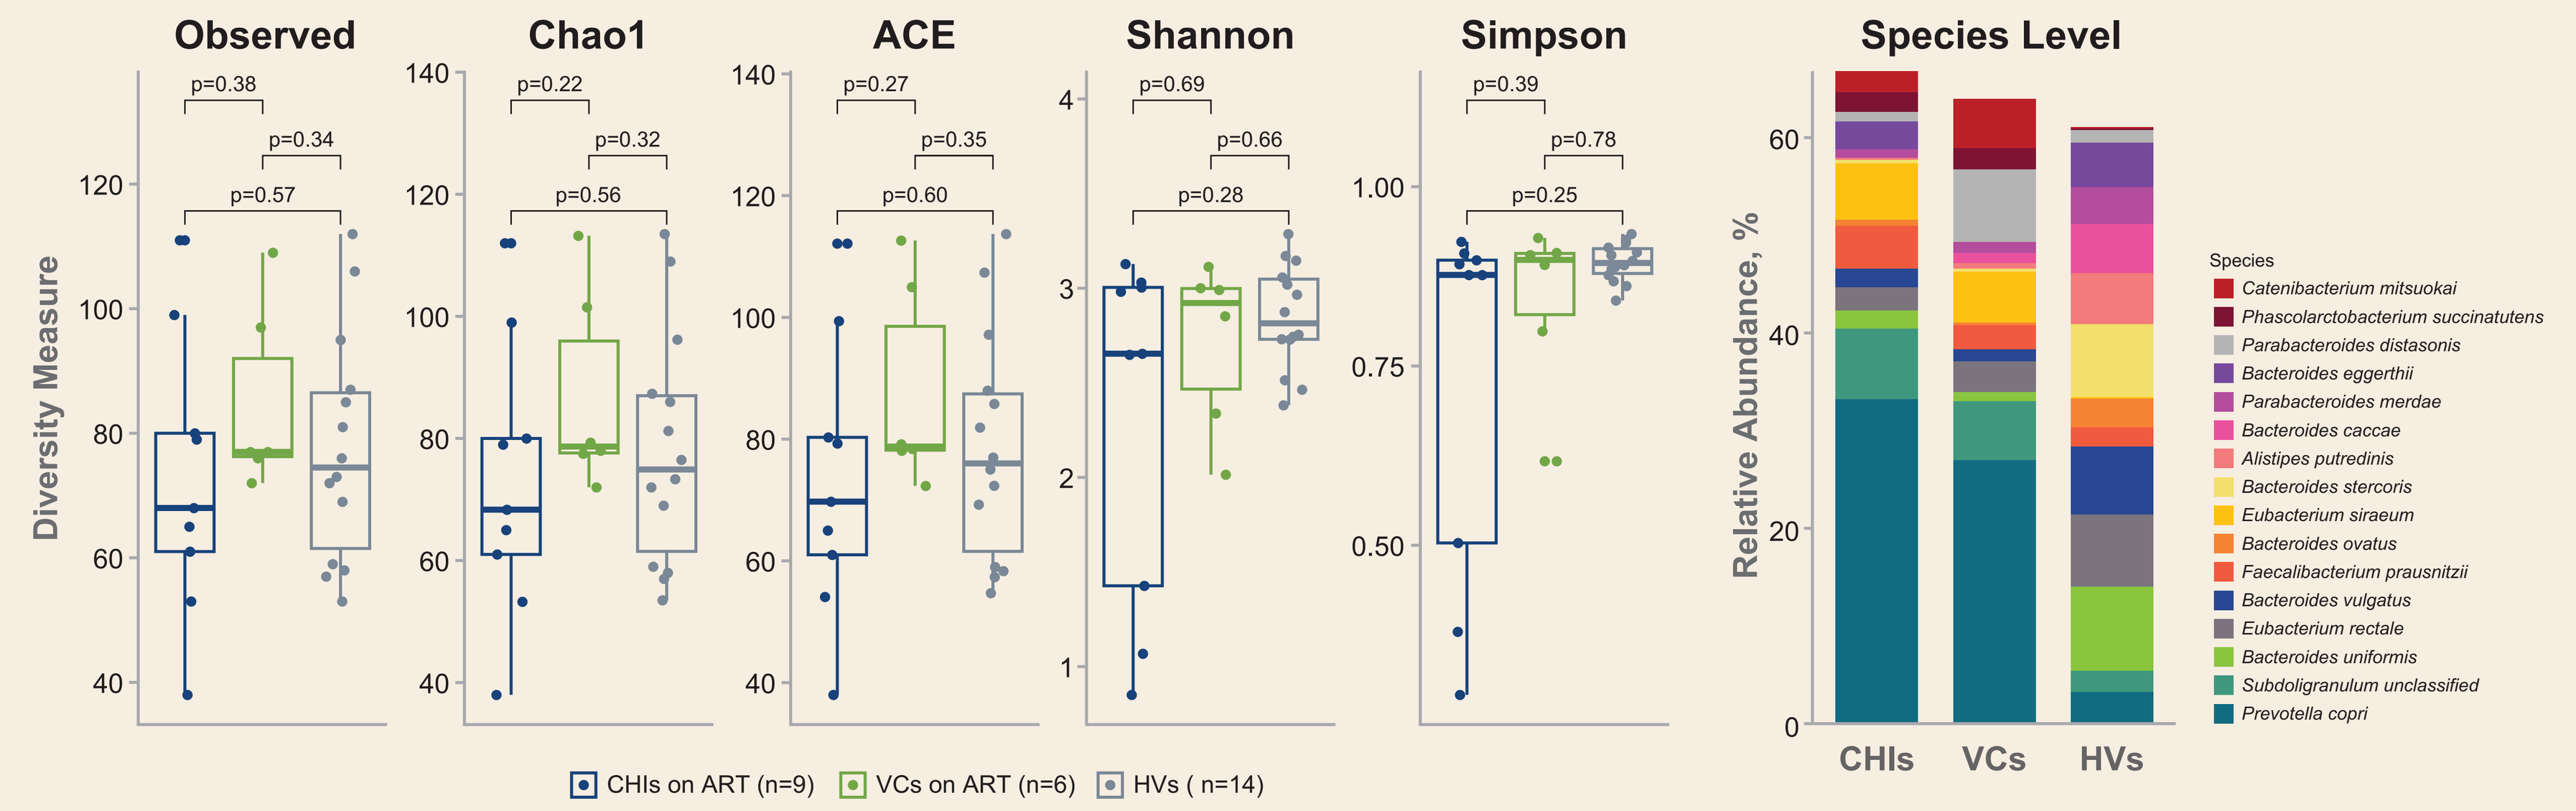
Observed
Chao1
ACE
Shannon
Simpson
Species Level
Diversity Measure
Relative Abundance, %
p=0.38
p=0.34
p=0.57
p=0.22
p=0.32
p=0.56
p=0.27
p=0.35
p=0.60
p=0.69
p=0.66
p=0.28
p=0.39
p=0.78
p=0.25
CHIs
VCs
HVs
Species
Catenibacterium mitsuokai
Phascolarctobacterium succinatutens
Parabacteroides distasonis
Bacteroides eggerthii
Parabacteroides merdae
Bacteroides caccae
Alistipes putredinis
Bacteroides stercoris
Eubacterium siraeum
Bacteroides ovatus
Faecalibacterium prausnitzii
Bacteroides vulgatus
Eubacterium rectale
Bacteroides uniformis
Subdoligranulum unclassified
Prevotella copri
CHIs on ART (n=9)
VCs on ART (n=6)
HVs (n=14)

ACE, abundance-based coverage estimator.

- No significant difference in fecal microbiome α-diversity was observed between PLWH and HVs
- VCs on ART tended to have greater microbiome diversity vs other CHIs on ART

HIV VCs on ART Presented Less Dysbiosis vs CHIs on ART*

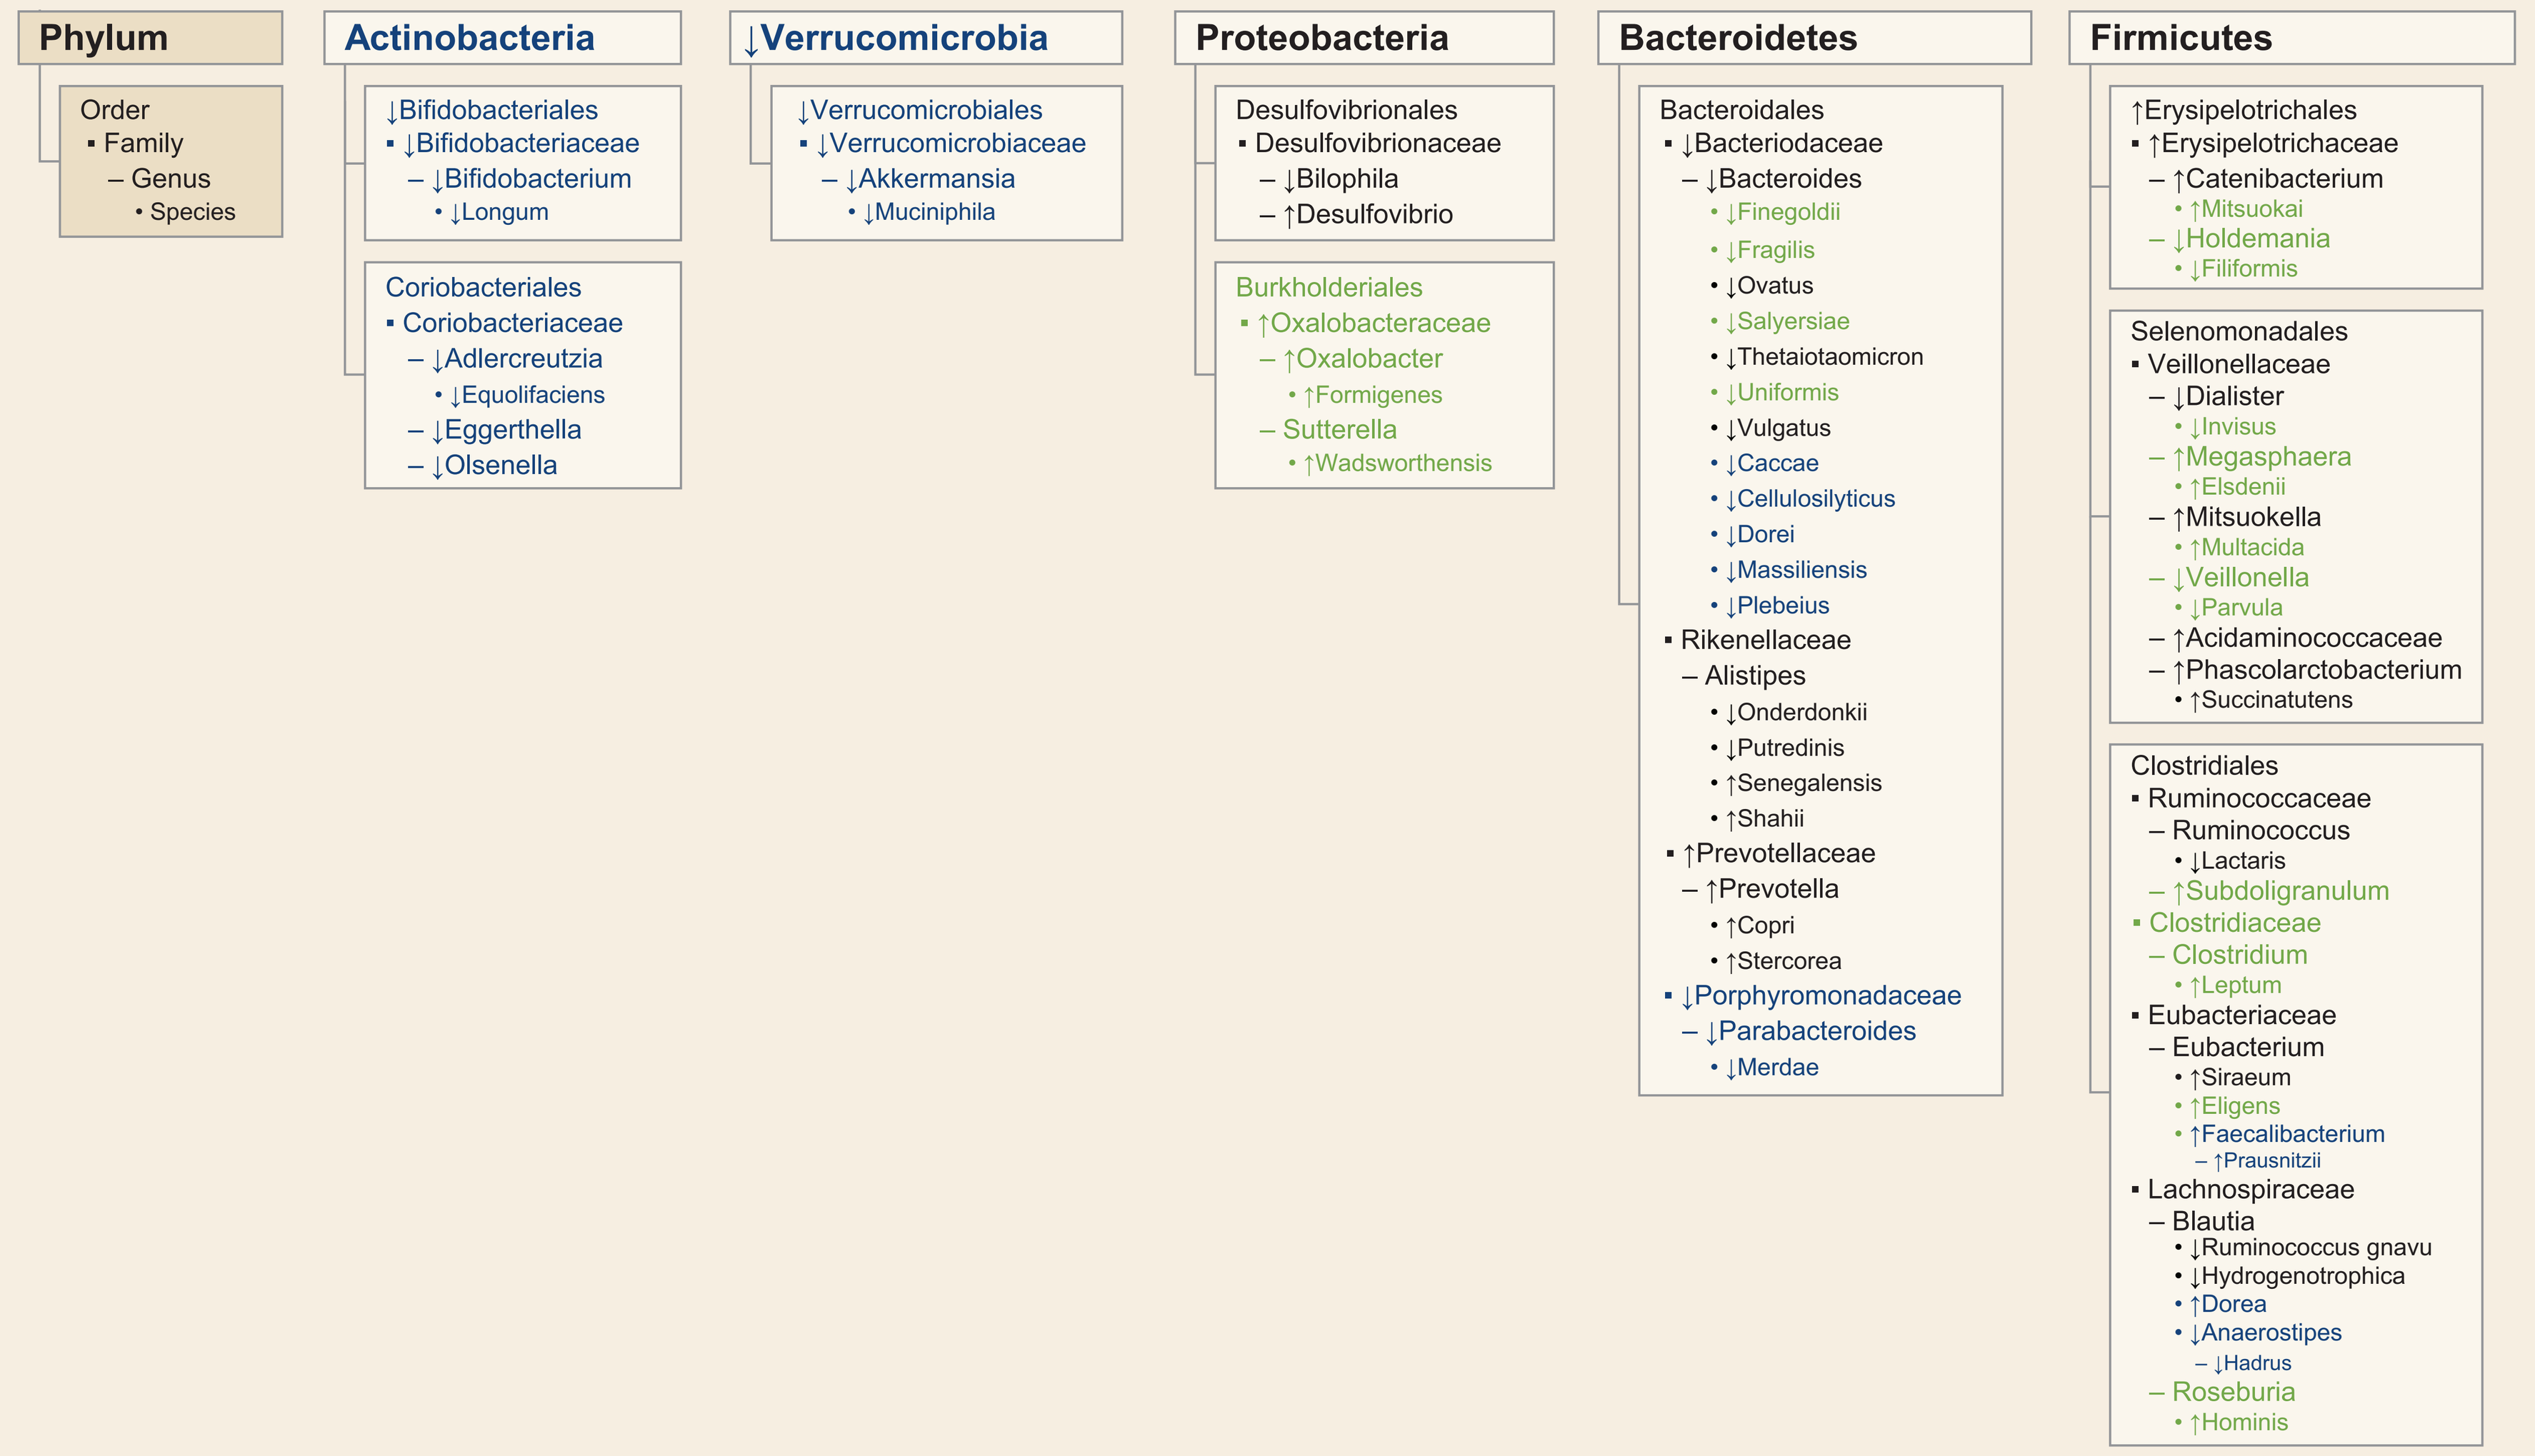
Phylum
Order
• Family
– Genus
• Species
Actinobacteria
↓Bifidobacteriales
• ↓Bifidobacteriaceae
– ↓Bifidobacterium
• ↓Longum
Coriobacteriales
• Coriobacteriaceae
– ↓Adlercreutzia
• ↓Equolifaciens
– ↓Eggerthella
– ↓Olsenella
↓Verrucomicrobia
↓Verrucomicrobiales
• ↓Verrucomicrobiaceae
– ↓Akkermansia
• ↓Muciniphila
Proteobacteria
Desulfovibrionales
• Desulfovibrionaceae
– ↓Bilophila
– ↑Desulfovibrio
Burkholderiales
• ↑Oxalobacteraceae
– ↑Oxalobacter
• ↑Formigenes
– Sutterella
• ↑Wadsworthensis
Bacteroidetes
Bacteroidales
• ↓Bacteriodaceae
– ↓Bacteroides
• ↓Finegoldii
• ↓Fragilis
• ↓Ovatus
• ↓Salyersiae
• ↓Thetaiotaomicron
• ↑Uniformis
• ↓Vulgatus
• ↓Caccae
• ↓Cellulosilyticus
• ↓Dorei
• ↓Massiliensis
• ↓Plebeius
• Rikenellaceae
– Alistipes
• ↓Onderdonkii
• ↓Putredinis
• ↑Senegalensis
• ↑Shahii
• ↑Prevotellaceae
– ↑Prevotella
• ↑Copri
• ↑Stercorea
• ↓Porphyromonadaceae
– ↓Parabacteroides
• ↓Merdae
Firmicutes
↑Erysipelotrichales
• ↑Erysipelotrichaceae
– ↑Catenibacterium
• ↑Mitsuokai
– ↓Holdemania
• ↓Filiformis
Selenomonadales
• Veillonellaceae
– ↓Dialister
• ↓Invisus
– ↑Megasphaera
• ↑Elsdenii
– ↑Mitsuokella
• ↑Multacida
– ↓Veillonella
• ↓Parvula
– ↑Acidaminococcaceae
– ↑Phascolarctobacterium
• ↑Succinatutens
Clostridiales
• Ruminococcaceae
– Ruminococcus
• ↓Lactaris
– ↑Subdoligranulum
• Clostridiaceae
– Clostridium
• ↑Leptum
• Eubacteriaceae
– Eubacterium
• ↑Siraeum
• ↑Eligens
• ↑Faecalibacterium
– ↑Prausnitzii
• Lachnospiraceae
– Blautia
• ↓Ruminococcus gnavu
• ↓Hydrogenotrophica
• ↑Dorea
• ↓Anaerostipes
– ↓Hadrus
– Roseburia
• ↑Hominis

**Blue:* statistical difference between CHIs and HVs; *green:* statistical difference between VCs and HVs; *black:* statistical difference between PLWH and HVs; Wilcoxon test.

VES Restored Abundance of Some Microbiome Species to Level Close to HVs

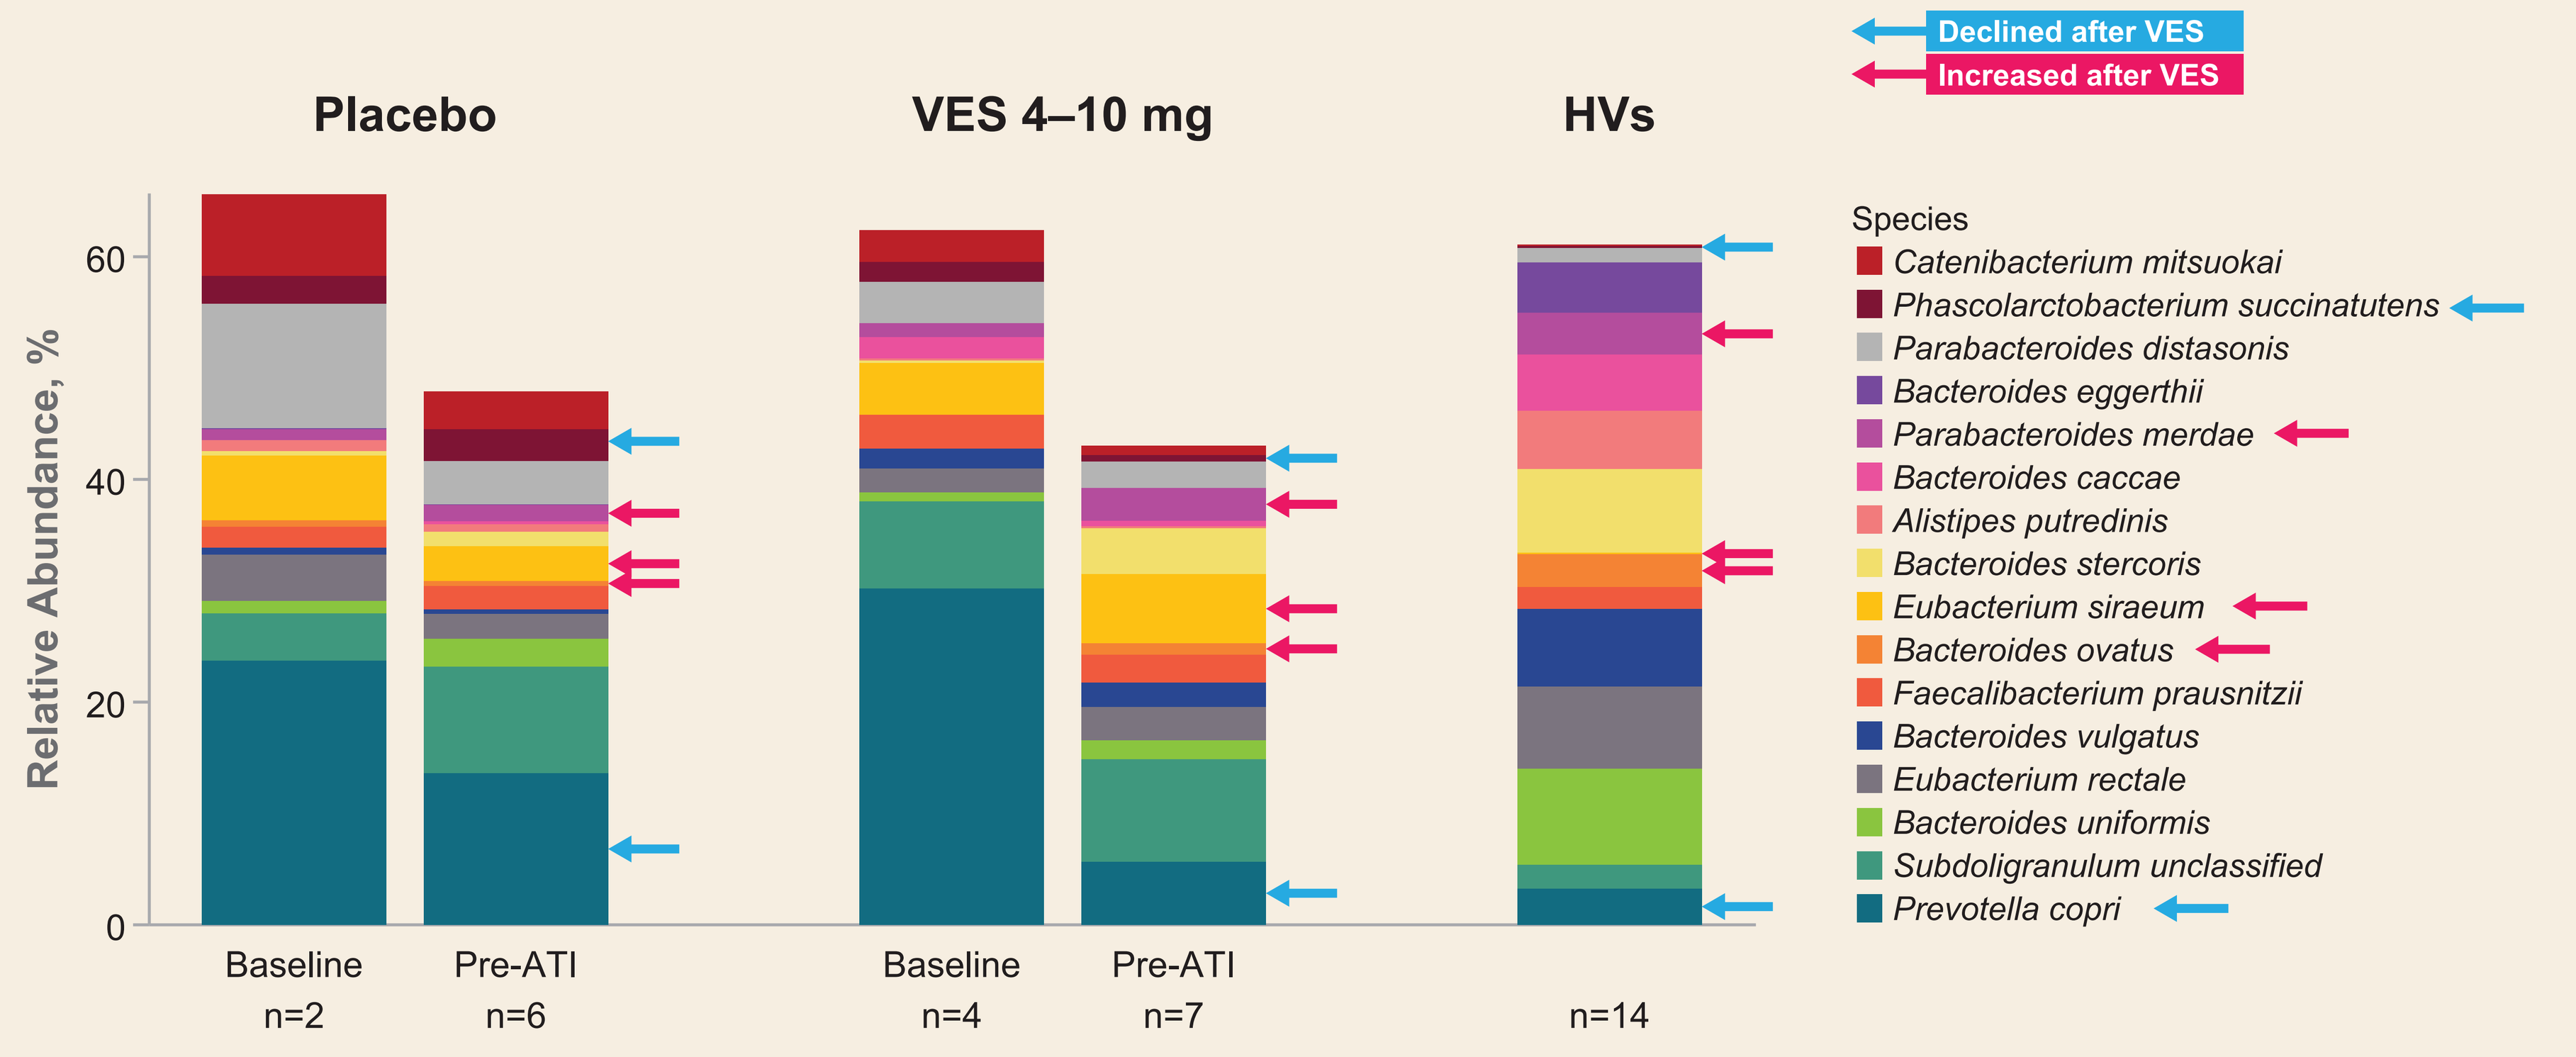
Declined after VES
Increased after VES
Placebo
VES 4–10 mg
HVs
Relative Abundance, %
Baseline
n=2
Pre-ATI
n=6
Baseline
n=4
Pre-ATI
n=7
n=14
Species
Catenibacterium mitsuokai
Phascolarctobacterium succinatutens
Parabacteroides distasonis
Bacteroides eggerthii
Parabacteroides merdae
Bacteroides caccae
Alistipes putredinis
Bacteroides stercoris
Eubacterium siraeum
Bacteroides ovatus
Faecalibacterium prausnitzii
Bacteroides vulgatus
Eubacterium rectale
Bacteroides uniformis
Subdoligranulum unclassified
Prevotella copri

Proteobacteria Abundance Trended to Be Positively Correlated With ISG Expression Level and CD4 T-Cell Activation

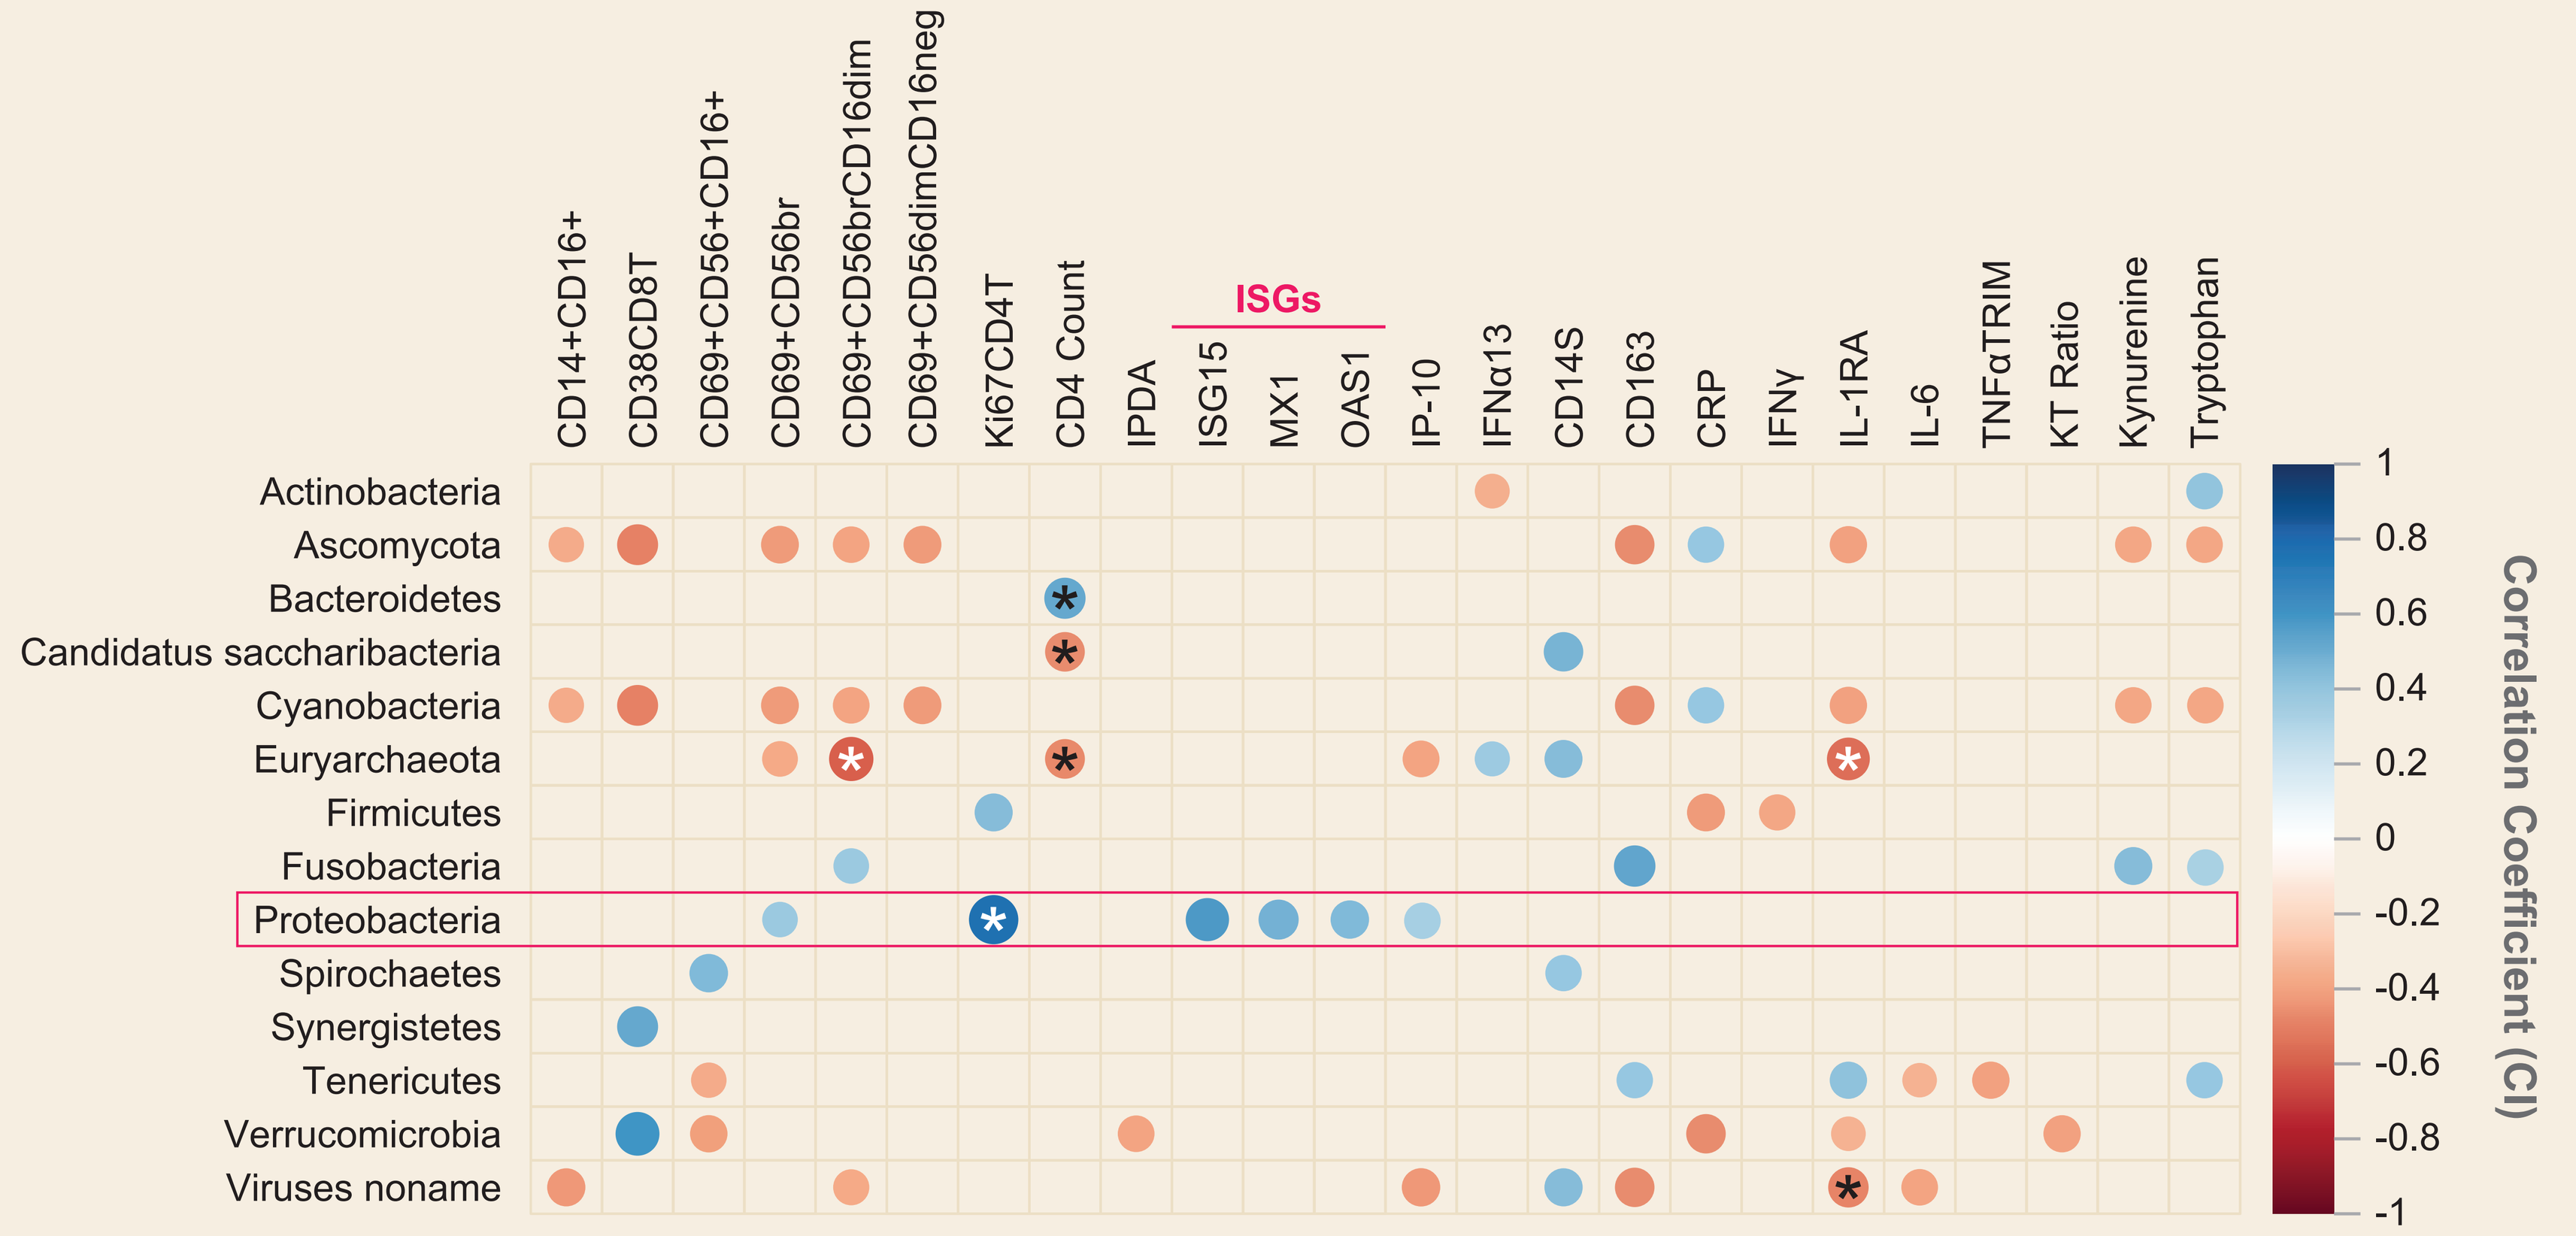
CD14+CD16+
CD38CD8T
CD69+CD56+CD16+
CD69+CD56br
CD69+CD56brCD16dim
CD69+CD56dimCD16neg
Ki67CD4T
CD4 Count
IPDA
ISGs
ISG15
MX1
OAS1
IP-10
IFNα13
CD14S
CD163
CRP
IFNγ
IL-1RA
IL-6
TNFαTRIM
KT Ratio
Kynurenine
Tryptophan
Actinobacteria
Ascomycota
Bacteroidetes
Candidatus saccharibacteria
Cyanobacteria
Euryarchaeota
Firmicutes
Fusobacteria
Proteobacteria
Spirochaetes
Synergistetes
Tenericutes
Verrucomicrobia
Viruses noname
Correlation Coefficient (CI)

*Statistically significant (p ≤0.05); data were obtained from male participants in HIV VC study, including both treatment arms and both time points (n=19); only confidence intervals (CIs) ≥0.2 are color coded. ISG, interferon-stimulated gene.

Fecal *Ruminococcus gnavus* Abundance at Pre-ATI Was Negatively Associated With Time to Viral Rebound in HIV VCs

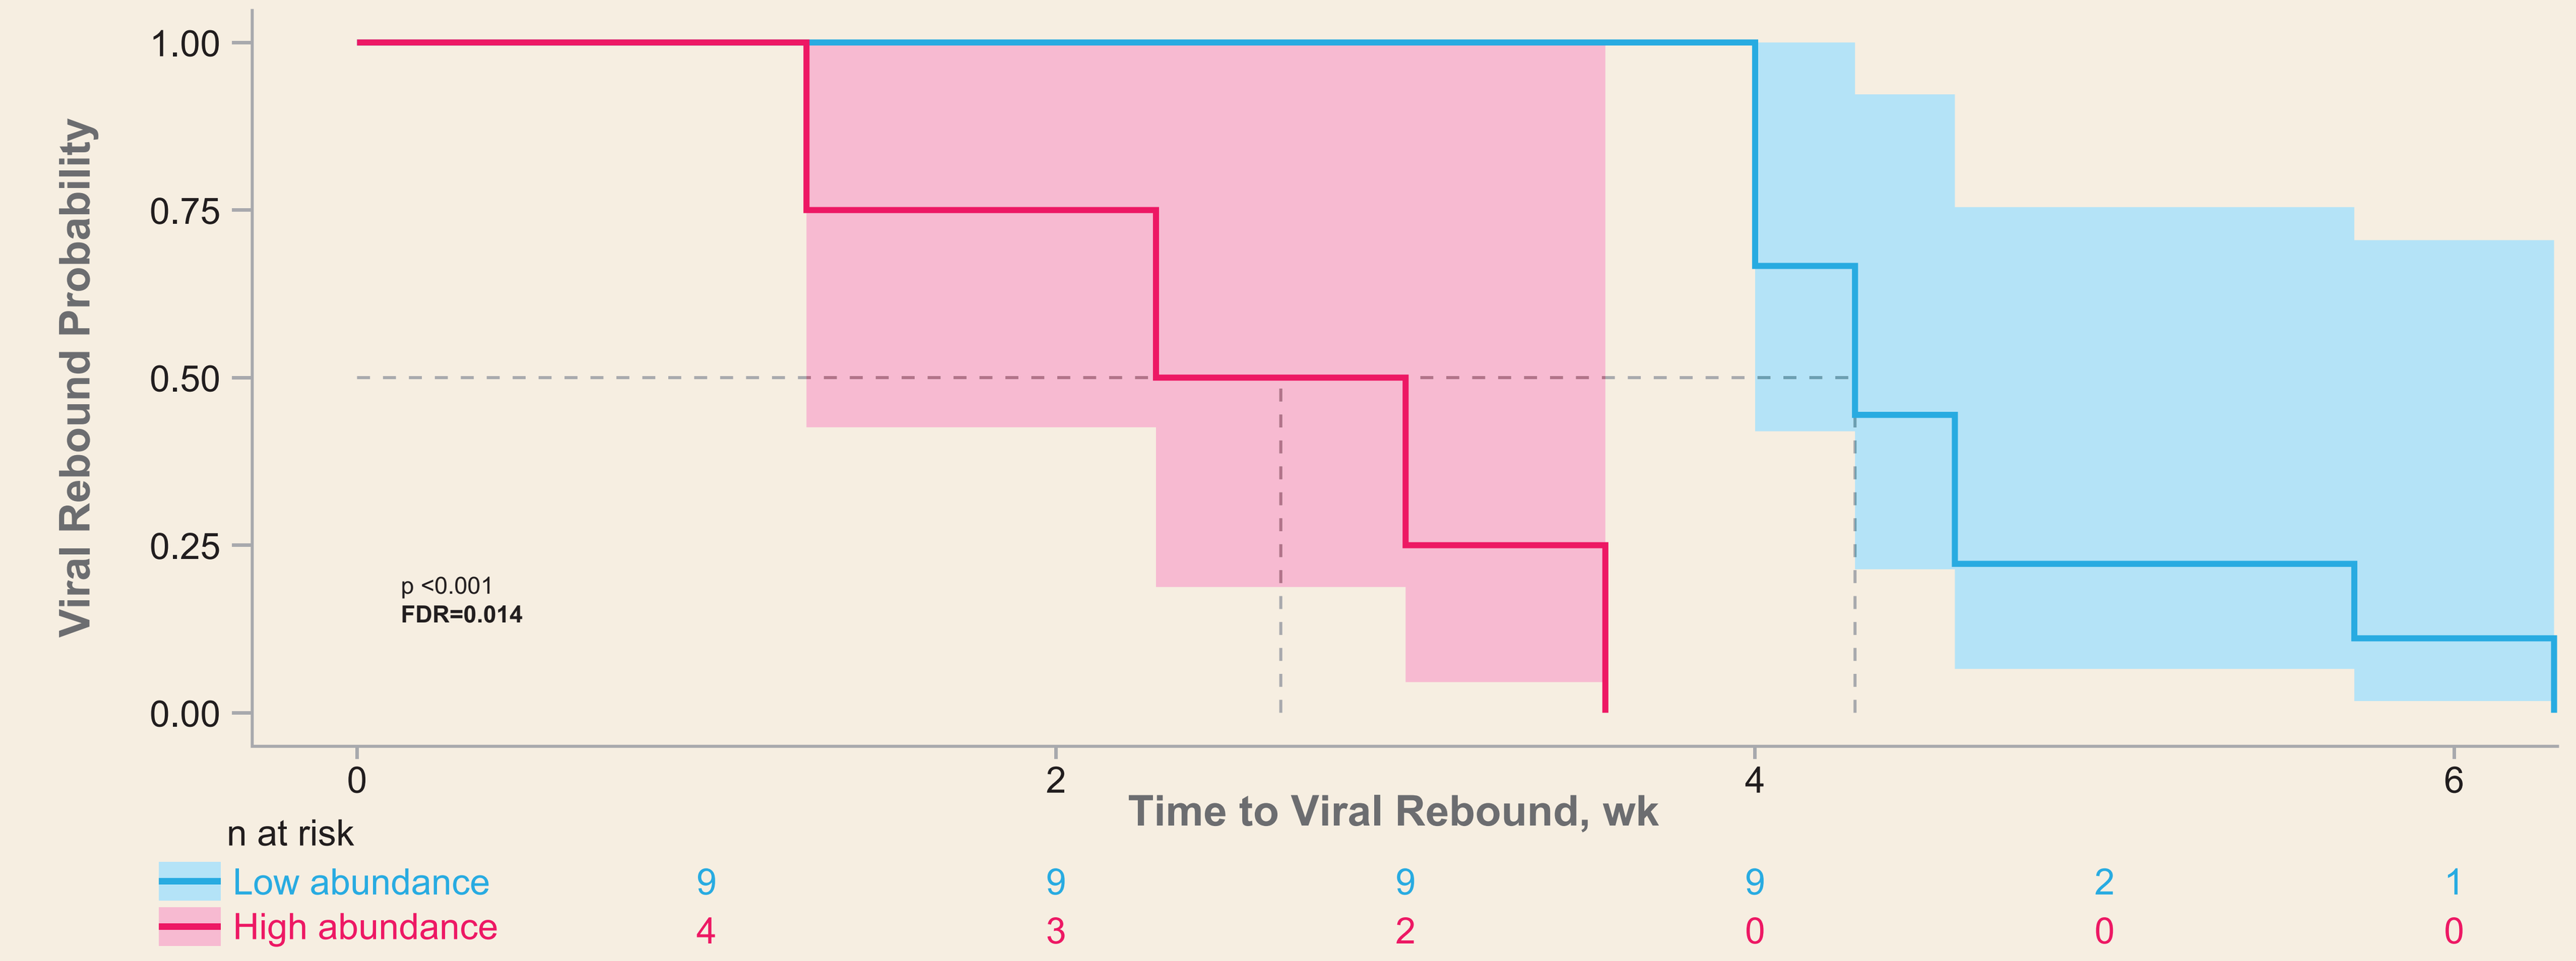
Viral Rebound Probability
p <0.001
FDR=0.014
Time to Viral Rebound, wk
n at risk
Low abundance 9 9 9 9 2 1
High abundance 4 3 2 0 0 0

FDR, false discovery rate.

- Higher abundance of *R. gnavus* at pre-ATI was associated with shorter time to rebound
- Low and high abundance for given taxa was determined by medium abundance
- These data need to be independently validated given small sample size

Conclusions

- VES restored the abundance of some microbiome species, such as *Prevotella copri*, to a level that was close to that of HVs
- Fecal proteobacteria abundance potentially reflects systemic immune activation and increased antiviral responses in VES-treated HIV VCs
- Enrichment of *R. gnavus* at pre-ATI was negatively associated with time to HIV rebound, suggesting a negative association between *R. gnavus* and HIV reservoir and viral persistence (*R. gnavus* probably led to increased oxidative stress and inflammation)
- Future studies are needed to understand the possible mechanisms driving gut dysbiosis, and investigate the abundance of some microbiome species in predicting antiviral responses and viral reservoir in HIV cure studies, as well as expanding these studies to include women living with HIV

References: 1. Borducchi E, et al. Nature 2016;540:284-7; 2. SenGupta D, et al. CROI 2020, abstr 3982; 3. Wallin J, et al. AIDS 2020, abstr OAB0205.
Acknowledgments: We extend our thanks to the participants, their partners, and families. Special thanks to the study teams: **Principal Investigator:** Steven Deeks; **Clinical Investigators:** Moti Ramgopal, Cynthia Brinson, Edwin DeJesus, Anthony Mills, Peter Shalit; **Biomarker/Microbiome Analysis:** *UCSF Core Immunology Lab:* Norman Jones, Valerie Girling; *Accelevir Diagnostics:* Gregory Laird; *Gilead:* Ondrej Podlaha, Jeff Wallin, Liao Zhang, Peter Shweh, Brian Moldt, Romas Geleziunas, Elena Vendrame, Diana Brainard, Devi SenGupta. This study was funded by Gilead Sciences, Inc.

Presented at *Virtual* CROI 2021, March 6–10, 2021

©2021 Gilead Sciences, Inc. All rights reserved.