1749

HIV-1 DNA Genotyping Is Often Variable in Repeat Testing From Single Blood Draws

Michelle L. D'Antoni, Kristen Andreatta, Rima Acosta, Hui Liu, Yongwu Shao, Kirsten L. White — Gilead Sciences, Inc., Foster City, CA

GILEAD
Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235
michelle.dantonibrogan@gilead.com

Introduction

- HIV-1 proviral DNA genotyping aims to detect archived drug resistance mutations (DRMs) in individuals with low or undetectable plasma HIV-1 RNA levels
- DHHS guidelines state that HIV-1 proviral DNA genotyping can be considered when conventional HIV-1 RNA drug resistance testing is unsuccessful or unavailable for patients initiating or failing therapy, and when optimizing antiretroviral (ARV) regimens in patients with viral suppression[1]
- DHHS guidelines also caution the interpretation of results as reports may miss some or all prior drug resistance[1]
 - In virologically suppressed individuals, concordance between previously detected DRMs and proviral genotyping by GenoSure Archive® (Monogram Biosciences, South San Francisco, CA) can range from 48% to 85%[2-6]
- Assays detecting archived mutations are insensitive due to sampling of small blood volumes and only assessing quasispecies that are circulating in the periphery, which also must be above the assay detection threshold

Dynamic Properties of the Latent Reservoir

Expansion and Contraction of HIV Clones and Detection by HIV-1 DNA Genotyping*

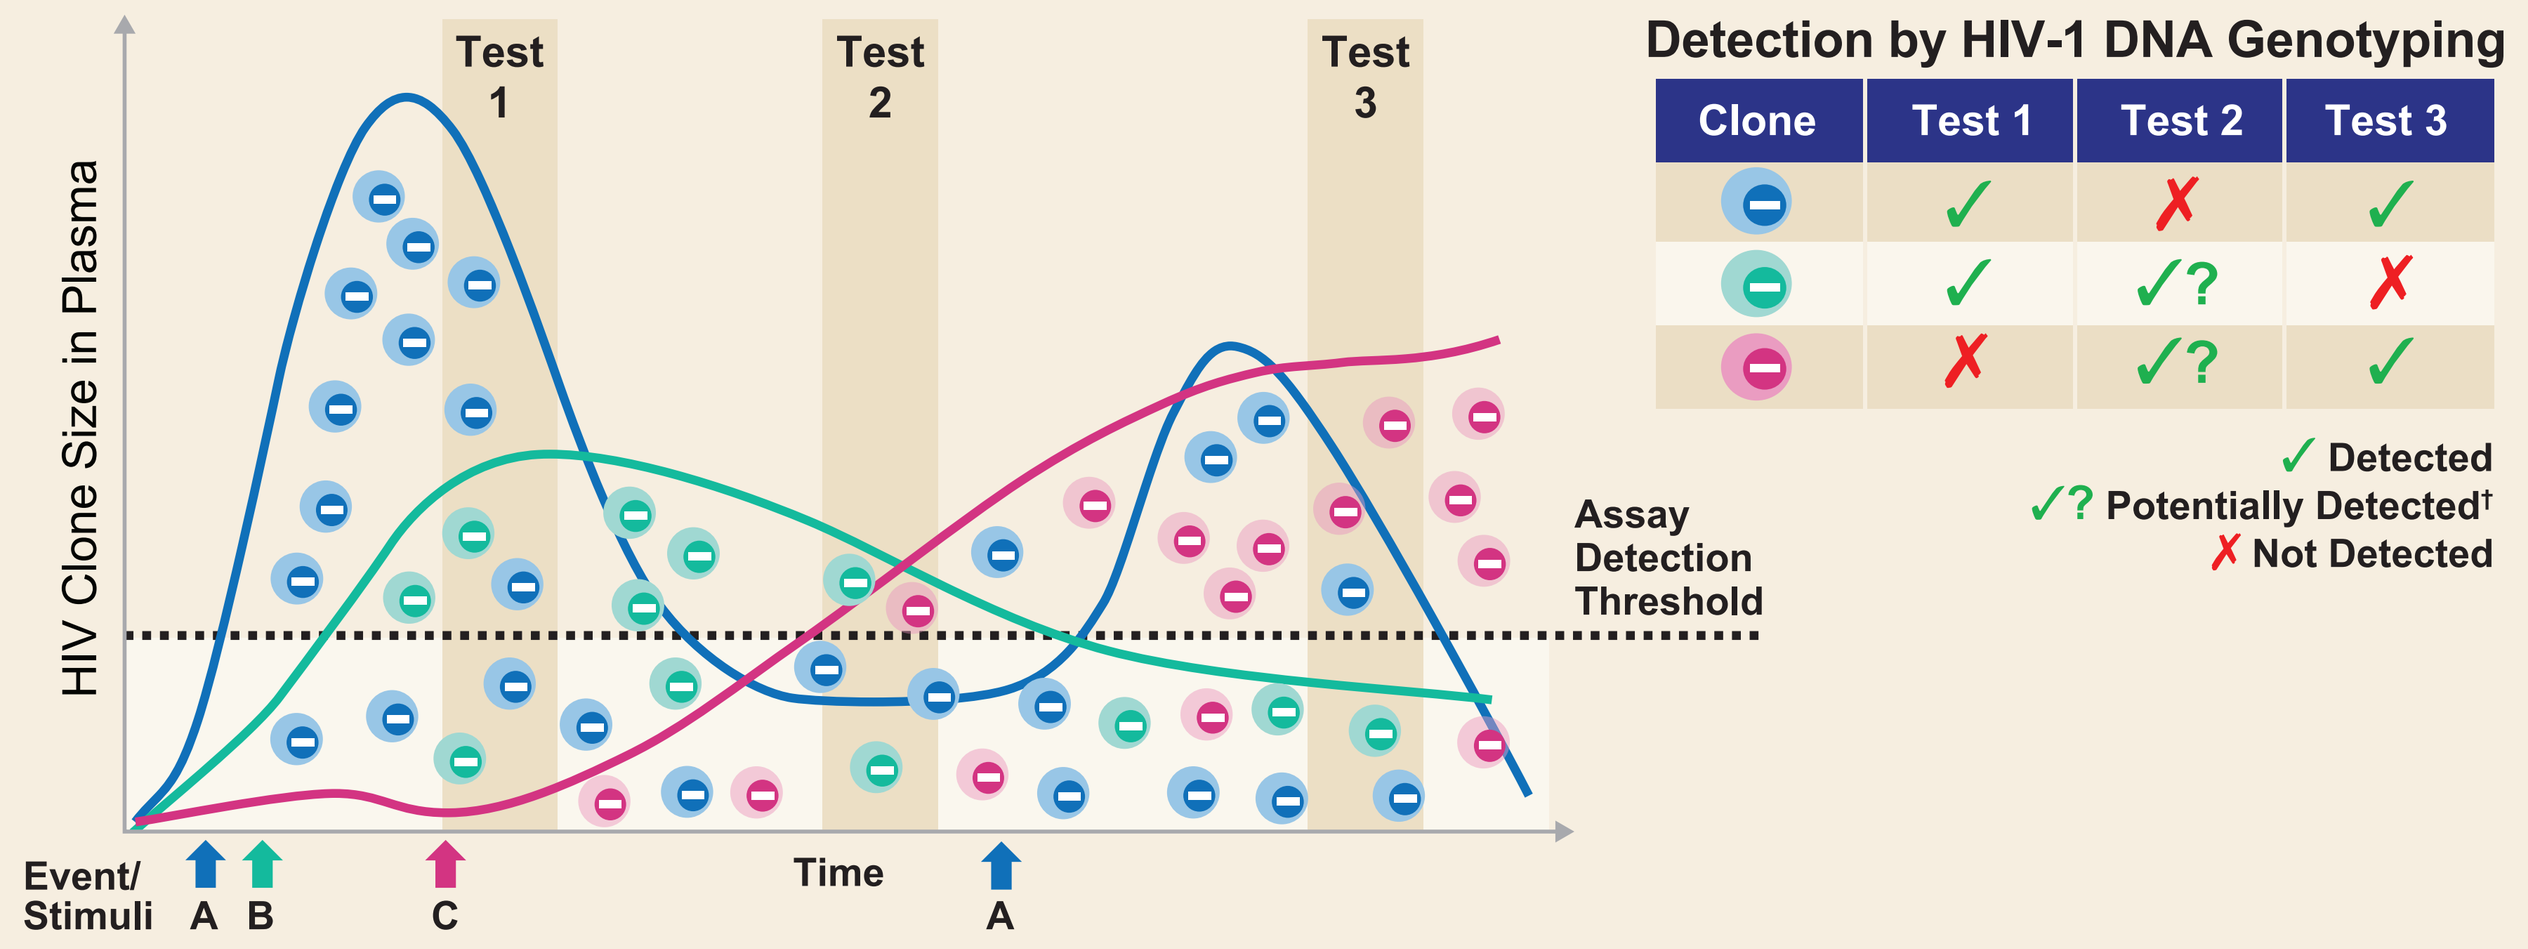
HIV Clone Size in Plasma
Test 1
Test 2
Test 3
Assay Detection Threshold
Event/ Stimuli
A B
C
Time
A
Detection by HIV-1 DNA Genotyping
Clone | Test 1 | Test 2 | Test 3
Blue: ✓ | ✗ | ✓
Green: ✓ | ✓? | ✗
Red: ✗ | ✓? | ✓
✓ Detected
✓? Potentially Detected†
✗ Not Detected

*Memory CD4+ T cells latently infected with various HIV clones (*blue, green and red*) are stimulated (event A, B, or C), leading to modulation of clone size (*blue, green, and red solid lines*); to be detected by HIV genotyping assays, clone size must be above assay detection threshold (*dotted black line*); not all clones are detected by tests (1–3 in table) depending on when whole blood is sampled; †Clones that are present at low frequencies may potentially be detected, but may also be missed. Figure adapted from Cohn et al.[7]

- The HIV reservoir is dynamic, with latently infected memory CD4+ T-cell populations expanding and contracting after exposure to various stimuli
 - Increases in cell number are driven by HIV integration site, homeostatic proliferation, and antigen recognition[7]
 - Detection of mutations is dependent on clone size
 - The archive is established early in acute HIV infection[8] and incorporates new variants during periods of viremia; infection is lifelong as HIV clones persist over time

Objective

- To characterize the variability of DNA genotyping by quantifying the reproducibility of mutation reporting from multiple assays from a single blood draw

Methods

Sample Population

- Baseline samples from 70 suppressed (plasma HIV-1 RNA <50 copies/mL for ≥3–6 mo prior to study entry) participants (67 with documented primary resistance and 3 without) from Studies 4449,[9] 4030,[10] and 4580[11] were analyzed
 - A subset of participants had a baseline and additional time-point sample (n=20)

Genotyping Analysis

- DNA genotyping of protease (PR), reverse transcriptase (RT), and integrase (IN) used GenoSure Archive from whole blood
 - Bioinformatic filters removed APOBEC-mediated, hypermutated, deep-sequence reads from GenoSure Archive results to prevent overreporting of E138K, M184I, and M230I in RT, and G163R in IN
 - 2–4 replicate GenoSure Archive assays from the same blood draw were evaluated

Substitutions Analyzed

- Primary PI-R, NRTI-R, NNRTI-R, and INSTI–R
- Other/non-R mutations were all nonprimary resistance mutations reported by the assay, which included secondary mutations and polymorphisms

Primary HIV-1 Drug-Resistance Substitutions*[12]

Primary PI-R	D30N, V32I, M46I/L, I47A/V, G48V, I50L/V, I54M/L, Q58E, T74P, L76V, V82A/F/L/S/T, N83D, I84V, N88S, L90M
Primary NRTI-R	K65R/E/N, T69 insertions, K70E, L74V/I, Y115F, Q151M, M184V/I, TAMs (M41L, D67N, K70R, L210W, T215F/Y, K219E/N/Q/R)
Primary NNRTI-R	L100I, K101E/P, K103N/S, V106A/M, V108I, E138A/G/K/Q/R, V179L, Y181C/I/V, Y188C/H/L,G190A/E/Q/S, H221Y, P225H, F227C, M230I/L
Primary INSTI-R	T66I/A/K, E92Q/G, T97A, F121Y, Y143R/H/C, S147G, Q148H/K/R, N155H/S, R263K

*Based on IAS-USA. INSTI, IN strand transfer inhibitor; NRTI, nucleos(t)ide RT inhibitor; NNRTI, non-NRTI; PI, PR inhibitor; R, resistance; TAMs, thymidine analogue mutations.

Reproducibility

- Reproducibility of individual substitutions were calculated as:

$$\text{Reproducibility (\%)} = \frac{\text{No. of times mutation was detected}}{\text{No. of assays run}}$$

Statistical Analysis

- Reproducibility of substitutions were reported as mean ± standard deviation
- Comparisons used Wilcoxon rank-sum tests
- Zero-truncated binomial model was used to estimate probability of mutation detection

Results

Clinical and Demographic Characteristics

	N=70
HIV-1 RNA <50 copies/mL, % (n)	96 (67)
HIV-1 RNA >50 copies/mL, n	3
Median CD4 count, cells/μL (IQR)	707 (473, 908)
HIV subtype B, % (n)	86 (60)
Male at birth, % (n)	79 (55)
Median age, y (range)	57 (19–77)

IQR, interquartile range.

Analysis Population

- For 90 blood draws from 70 participants, 257 genotype reports were analyzed
- All were suppressed except 3 participants with HIV-RNA 136, 105, and 107,000 copies/mL

Example of Reproducibility Reporting*

Type	Report 1	Report 2	Report 3
PR	T12T/P, I15V, K20K/T, D30D/N, M36M/I/T, I62I/V, L63P, I66I/L, A71A/T, V77V/I, N88N/D, L90L/M	T12T/P, I15V, K20K/T, D30D/N, M36M/I/L, L63P, A71A/T, N88N/D, L90L/M	I15V, K20K/T, D30D/N, M36M/I, L63P/S, A71A/T, V77V/I, N88N/D, L90L/M
RT	K20K/R, V35I, R83K, Q102K, K103K/N, D123D/E, I135I/T, C162S, Q174Q/R, I178I/L, M184M/V, P225P/H, K238R, V245V/E, A272P, R277K, T286A, V293I, G335D, R356K, M357K, K358R, A360A/V, K388K/T, K390R, K395K/R, A400A/T	K20K/R, V35I, R83K, Q102K, K103K/N, D123D/E, I135I/T, C162S, Q174Q/R, I178I/L, P225P/H, K238R, E248E/V, S251S/I, A272P, R277K, T286A, V293I, G335D, N348N/I, R356K, M357K, K358R, A360A/V, S379S/G, K390R, A400A/T	K20K/R, V35I, R83K, Q102K, K103K/N, M184M/V, T215T/N/S/Y, K238R, A272P, 277K, T286A, V293I, G335D, R356K, RM357K, K358R, A360A/V, T369T/A/I/V, K390R, A400A/T
IN	S17N, R20R/K, L28I, V72I, T112A, V113I, S119P, T124G, T125A, I141I/T, D207D/N, T218T/I, I220I/L, Q221Q/H, N222N/H, V234L, D256E	S17N, R20R/K, L28I, V72I, T112A, V113I, S119P, T124G, T125A, T206T/S, T218T/I, I220I/L, Q221Q/H, N222N/H, V234L, D256E, D288D/N	S17N, R20R/K, L28I, V72I, T112A, V113I, S119P, T124G, T125A, T206T/S, T218T/I, I220I/L, Q221Q/H, N222N/H, V234L, D256E, R269R/K

Type	Mutations: Report 1	Report 2	Report 3	Cumulative	No. of Times Mutation Was Detected	No. of Assays Run	% Reproducibility
Primary PI	D30N	D30N	D30N	D30N	3	3	100
Primary NRTI	M184V		M184V	M184V	2	3	67
			T215Y	T215Y	1	3	33
Primary NNRTI	K103N	K103N	K103N	K103N	3	3	100
Non-R INSTI	S119P	S119P	S119P	S119P	3	3	100

*Substitutions from summaries of GenoSure Archive reports were tabulated and reproducibility was calculated; examples of reproducibility for primary DRMs and non-R substitutions are shown: D30N in PR, K103N in RT, and S119 in IN were 100% reproducible, being detected in 3/3 reports; M184V in RT was 67% reproducible, being detected in 2/3 reports; and T215Y in RT was 33% reproducible, being detected in 1/3 reports.

Reproducibility of Substitutions in PR/RT/IN

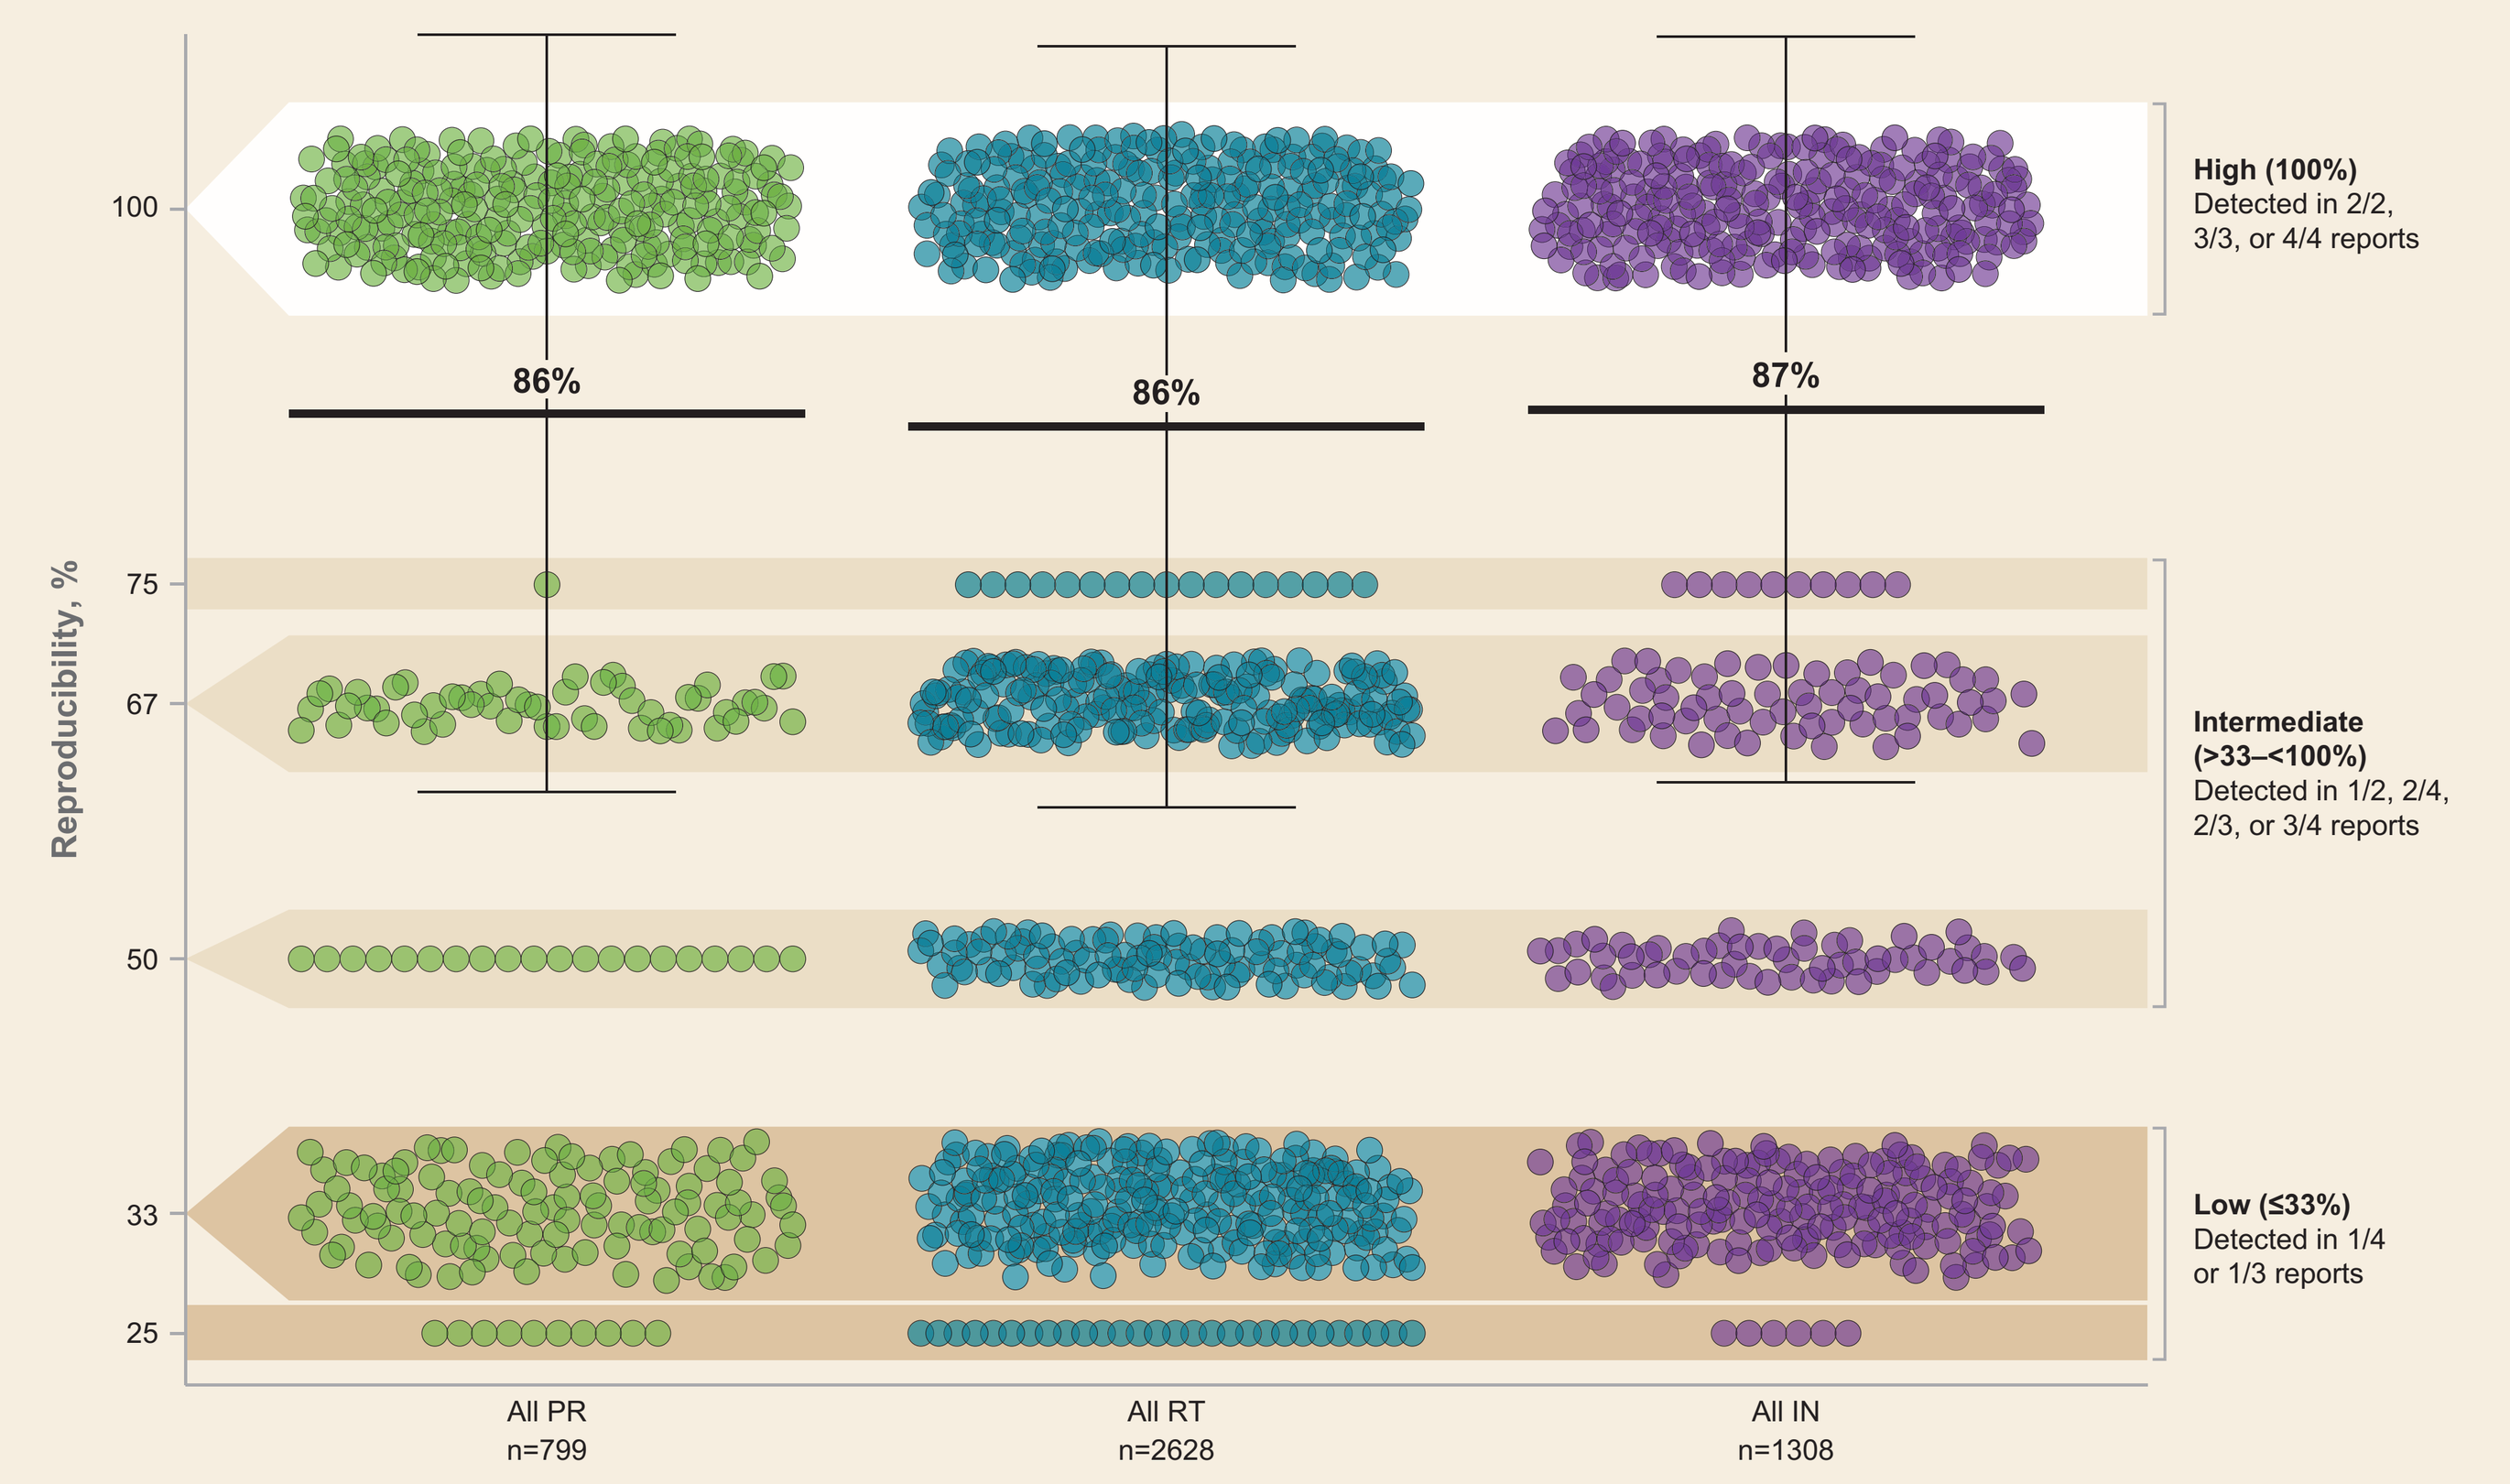
Reproducibility, %
100
75
67
50
33
25
86%
86%
87%
All PR n=799
All RT n=2628
All IN n=1308
High (100%) Detected in 2/2, 3/3, or 4/4 reports
Intermediate (>33–<100%) Detected in 1/2, 2/4, 2/3, or 3/4 reports
Low (≤33%) Detected in 1/4 or 1/3 reports

n, no. of observations of substitutions reported in PR, RT, and IN.

- Reproducibility was similar for all amino-acid substitutions reported across the genes of PR (86 ± 25%), RT (86 ± 25%), and IN (87 ± 25%)
- In general, reproducibility was high but variable, with 14–15% of mutations being reported less than a 3rd or 4th of the time (low reproducibility)

Breakdown of Reproducibility of Substitutions in PR/RT/IN*

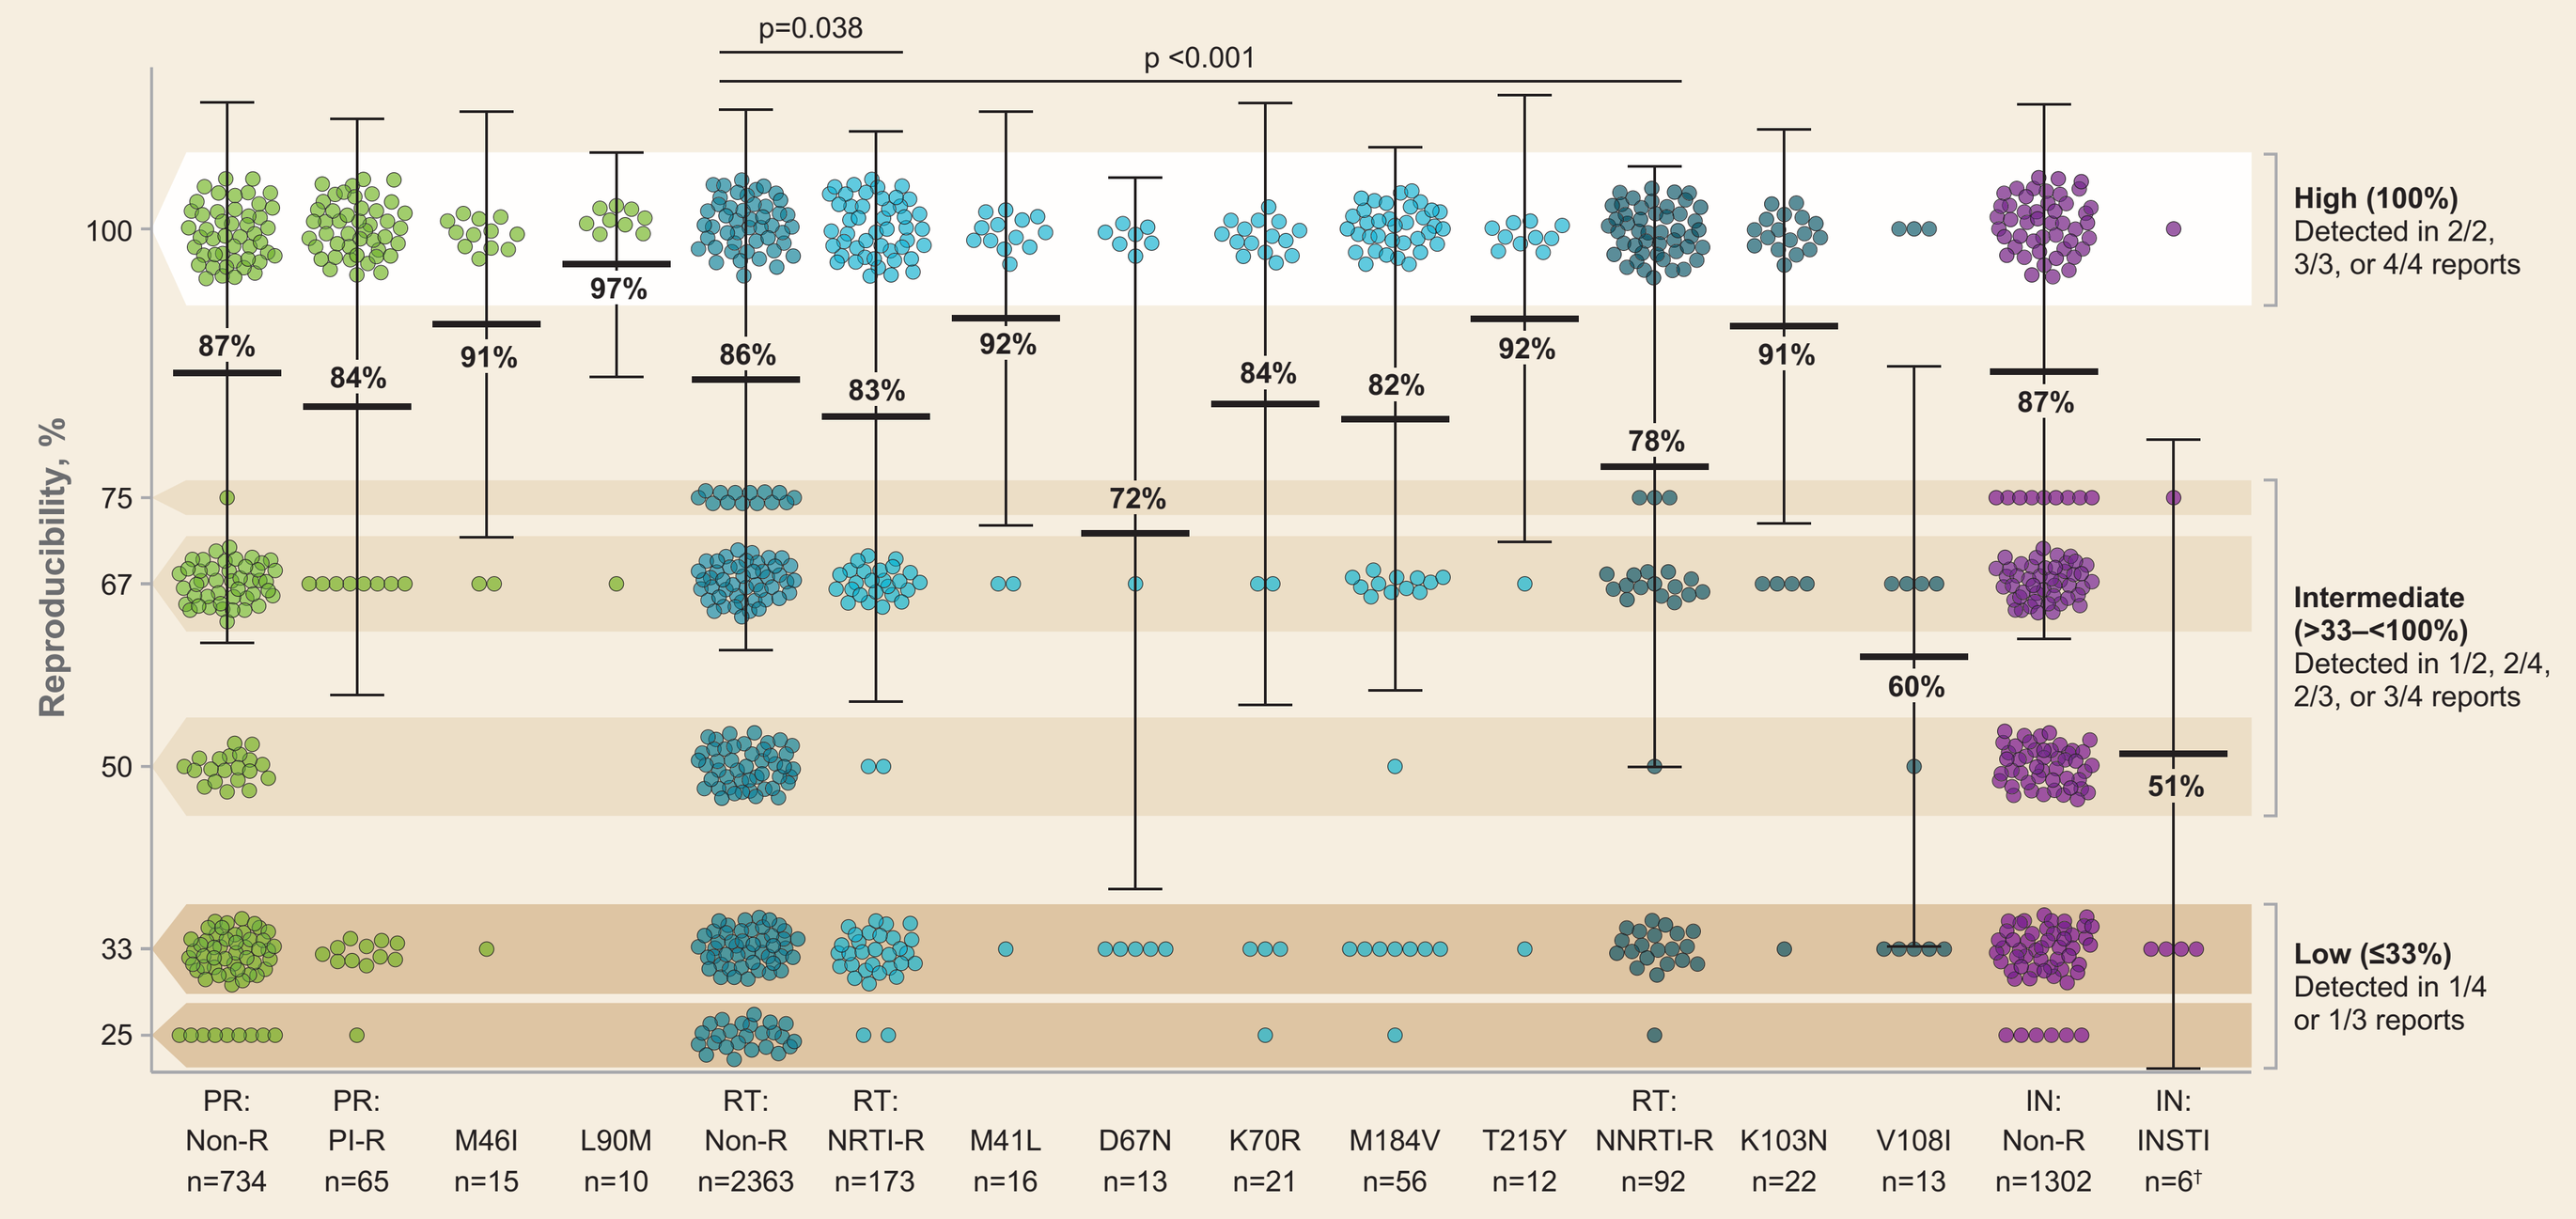
p=0.038
p <0.001
Reproducibility, %
100
75
67
50
33
25
87%
84%
91%
97%
86%
83%
92%
72%
84%
82%
92%
78%
91%
60%
87%
51%
PR: Non-R n=734
PR: PI-R n=65
M46I n=15
L90M n=10
RT: Non-R n=2363
RT: NRTI-R n=173
M41L n=16
D67N n=13
K70R n=21
M184V n=56
T215Y n=12
RT: NNRTI-R n=92
K103N n=22
V108I n=13
IN: Non-R n=1302
IN: INSTI n=6†
High (100%) Detected in 2/2, 3/3, or 4/4 reports
Intermediate (>33–<100%) Detected in 1/2, 2/4, 2/3, or 3/4 reports
Low (≤33%) Detected in 1/4 or 1/3 reports

*For NRTI-, NNRTI-, and PI-R, only primary mutations with n >10 are graphed; †INSTI-R occurrence was too low for further analyses. n, no. of observations of substitutions reported in PR, RT, and IN.

- Both primary DRMs and non-R substitutions had overall high levels of reproducibility, but certain replicates of reports were variable
 - The NRTI DRM M184V had a reproducibility of 82 ± 25%, 14% of cases were detected in 1/4 or 1/3 reports, 23% in 1/2, 2/4, or 2/3 reports, and 63% in all reports
- Lower reproducibility may reflect more rare quasipecies
- Reproducibility was high and similar across PI, NRTI, and NNRTI drug classes
- Reproducibility of non-R RT substitutions was significantly higher than for primary NRTI and NNRTI DRMs
 - Polymorphisms can be more stable than primary mutations due to their minimal impact on viral fitness and potential role as immune escape variants

DRM Summary

- When aggregating all observations from repeat testing reports, a total of 15 PI, 18 NRTI, 19 NNRTI, and 3 INSTI individual DRMs were detected in 21, 43, 31, and 6 participants, respectively
 - M184V was detected at 1 time point for 20 participants and 2 time points each for 18 participants (n=56)

Reproducibility of Primary DRMs According to Historic Data

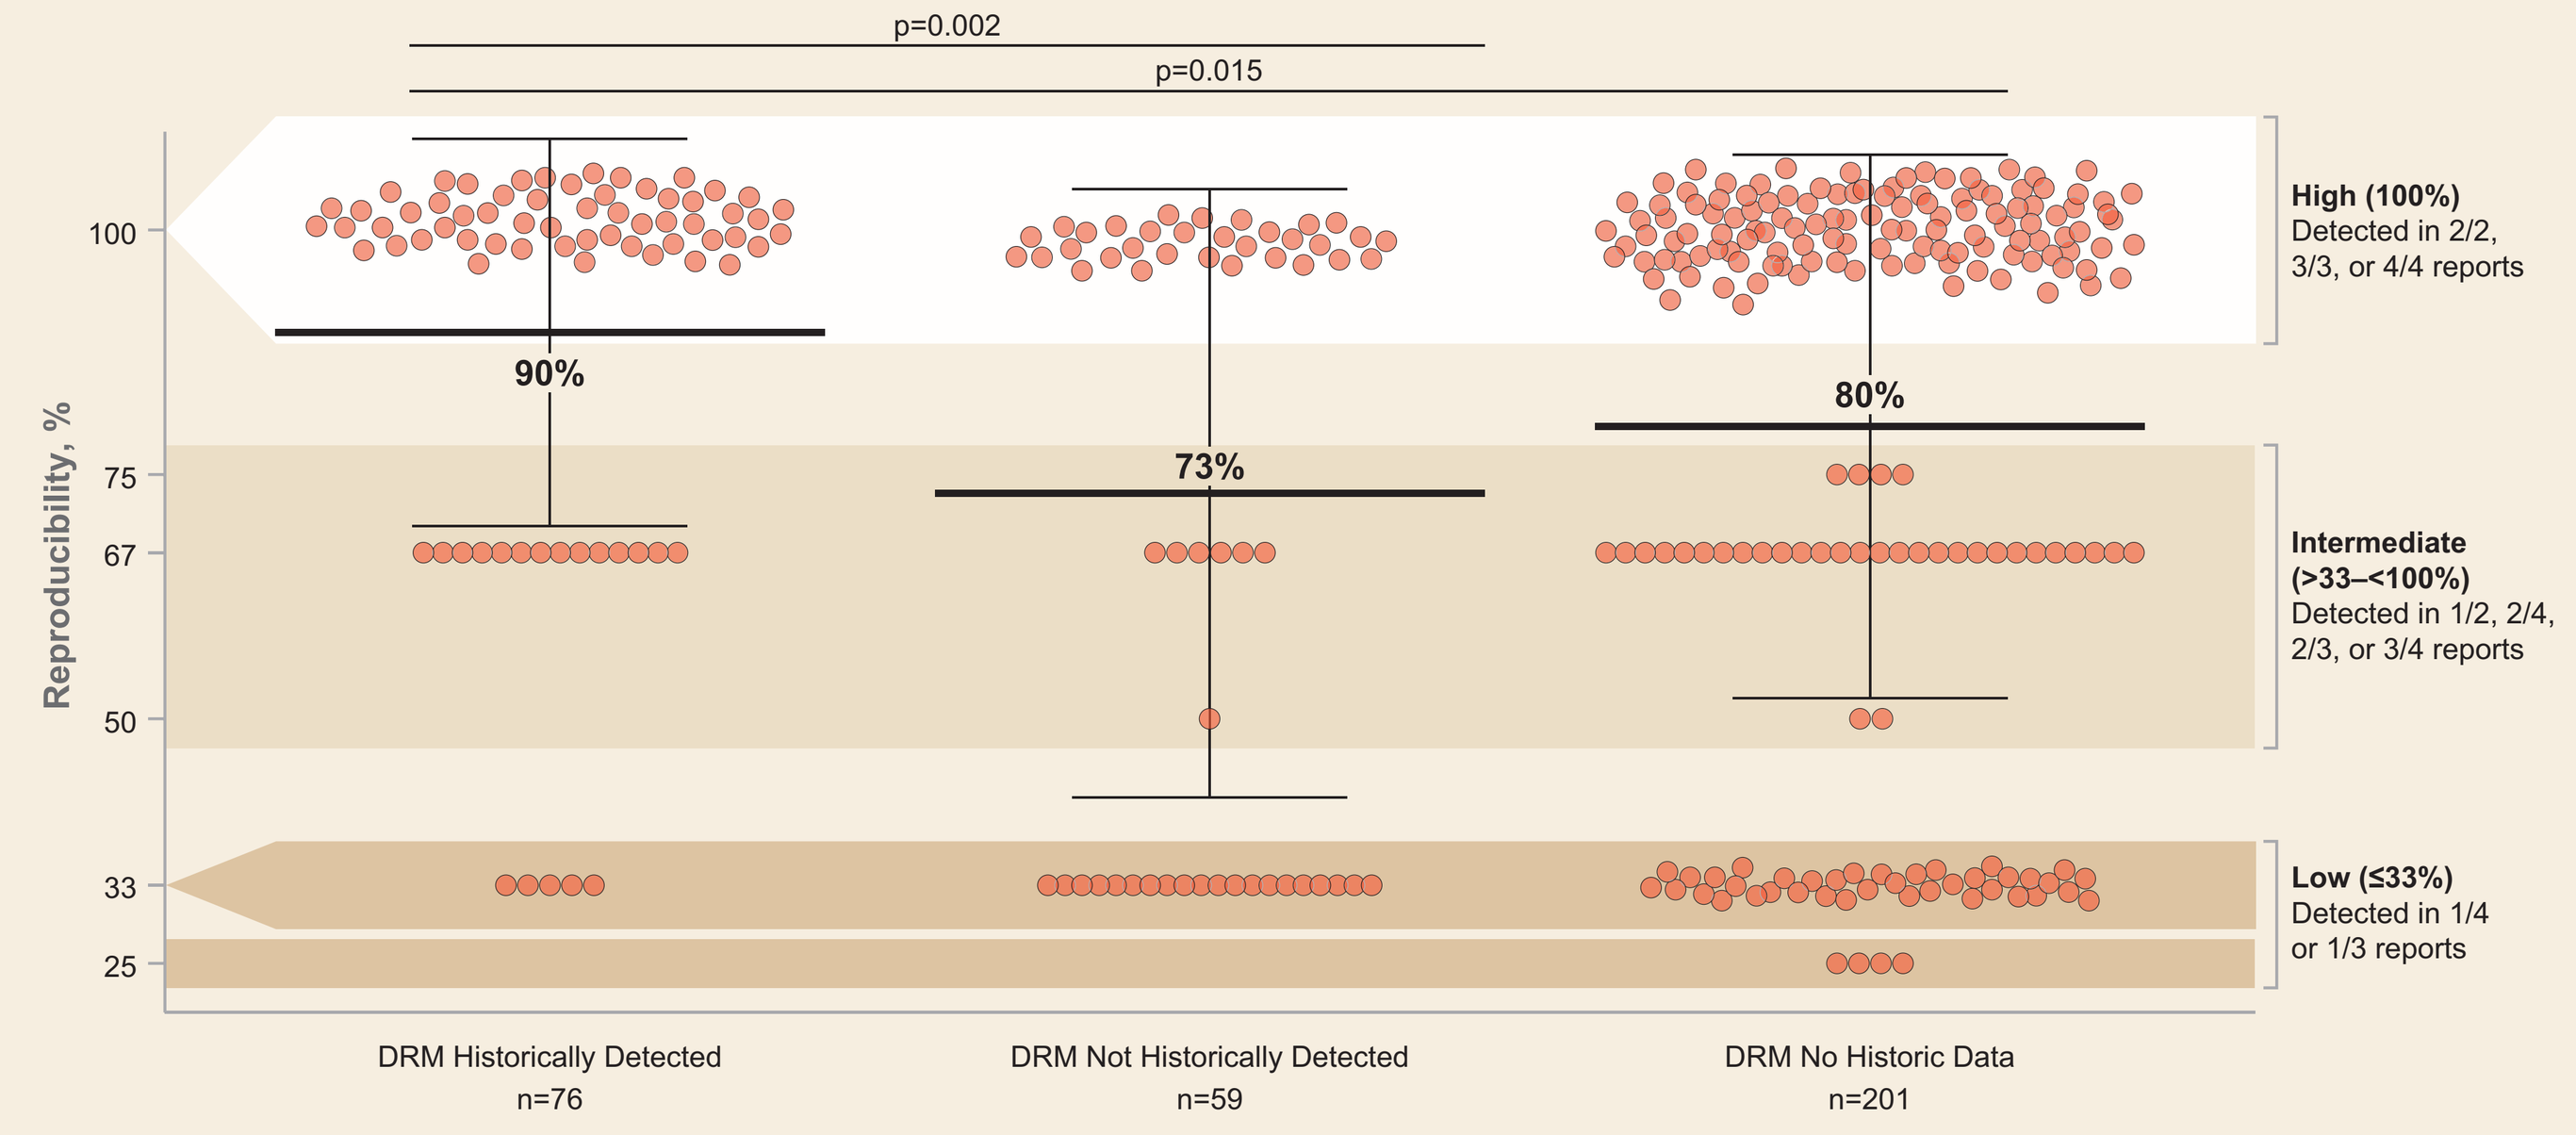
p=0.002
p=0.015
Reproducibility, %
100
75
67
50
33
25
90%
73%
80%
DRM Historically Detected n=76
DRM Not Historically Detected n=59
DRM No Historic Data n=201
High (100%) Detected in 2/2, 3/3, or 4/4 reports
Intermediate (>33–<100%) Detected in 1/2, 2/4, 2/3, or 3/4 reports
Low (≤33%) Detected in 1/4 or 1/3 reports

n, no. of observations of total DRMs reported in PR, RT, and IN.

- Reproducibility of DRMs (PI, NRTI, and NNRTI combined) was 10–17% higher when detected historically compared with being undocumented or not having historic data
- Of DRMs detected, 77% were previously undocumented or lacking historic data

Zero-Truncated Binomial Model for Mutation Detection

Primary DRM	Probability of Testing Positive	95% CI
NRTI	0.798	0.671, 0.884
NNRTI	0.763	0.593, 0.877
PI	0.800	0.637, 0.901
Historically detected	0.887	0.830, 0.927
Not historically detected	0.589	0.465, 0.702
No historic data	0.751	0.661, 0.824

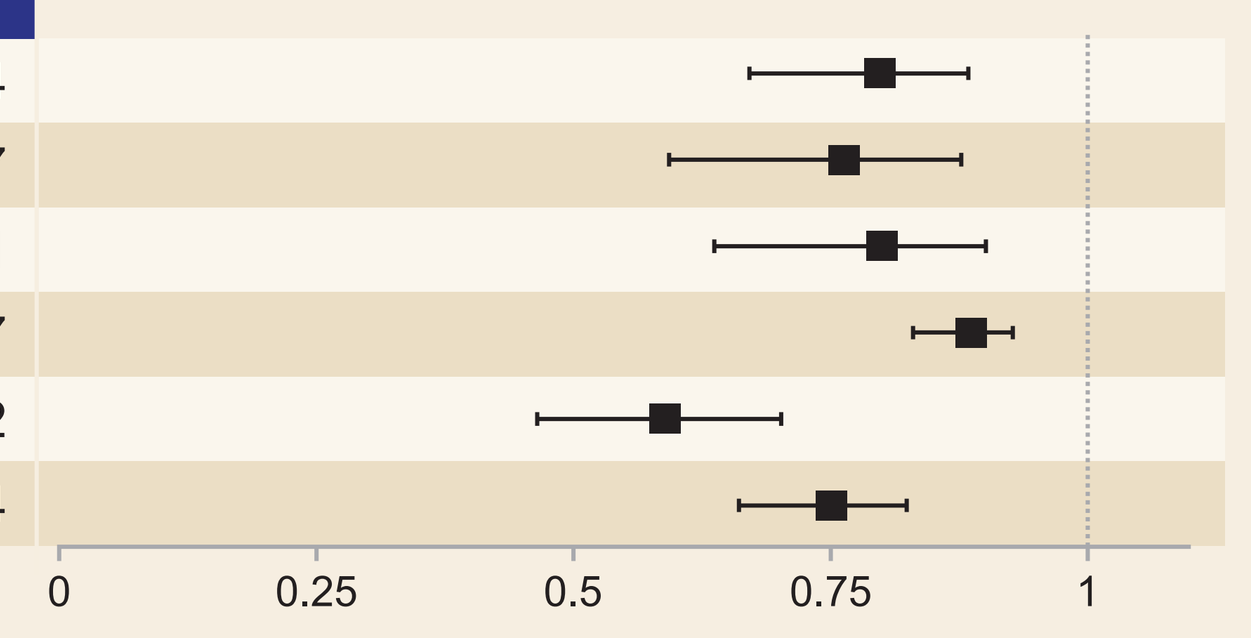
0
0.25
0.5
0.75
1

CI, confidence interval.

- By zero-truncated binomial modelling, if a participant had a PI, NRTI, or NNRTI DRM, the probability of it being reported by the proviral genotyping assay was 76–80%
 - The probability of a primary DRM being detected by the proviral genotyping assay increased to 89% if historically detected and decreased to 59% if not detected by a previous report
- All observations were similar when only baseline samples were analyzed (ie, 1 series of replicate reports/participant)

Conclusions

- The utility of HIV-1 proviral DNA genotyping for identification of archived DRMs is clinically relevant, especially in individuals with limited available clinical history; it can be beneficial in guiding regimen selection when mutations are present and to rule out certain ARVs, but can be insensitive as resistance substitutions are not always consistently detected
 - Proviral DNA genotypic susceptibility score can be a useful tool to predict virologic outcome in virologically suppressed people[13]
 - Primary DRM reproducibility of ~80% with standard deviations of ~25% indicates that detection of mutations is variable; low reproducibility still confirms the presence of the mutation
 - Reporting of a mutation as negative cannot confirm that the mutation is absent
 - A limitation of DNA genotyping is that current assays cannot determine whether DRM is on an intact, infectious HIV genome
- DNA genotyping may aid clinicians when switching ARV regimens, but these data reinforce the need to interpret tests with caution, as not all mutations may be reported; the individual's treatment history, including ARV therapy, virologic failures, and all past resistance test results, should be evaluated and considered

References: **1.** DHHS. Guidelines for the Use of Antiretroviral Agents in Adults and Adolescents With HIV; 12/18/19; **2.** Curanovic D, et al. IDWeek 2020, poster 691; **3.** Margot N, et al. J Antimicrob Chemother 2020;75:2249-52; **4.** Porter DP, et al. HIV Clin Trials 2016;17:29-37; **5.** Singh HK, et al. IDWeek 2016, abstr 1507; **6.** Toma J, et al. ICAAC 2015; **7.** Cohn LB, et al. Cell Host Microbe 2020;27:519-30; **8.** Coffin JM, et al. JCI Insight. 2019;4:e128432; **9.** Maggiolo F, et al. IAS 2019, abstr MOPEB238; **10.** Sax PE, et al. Clin Infect Dis 2020;ciaa988; **11.** Hagins DP, et al. IDWeek 2020, poster 1046; **12.** Wensing AM, et al. Top Antivir Med 2017;24:132-41; **13.** Armenia D, et al. J Clin Virol 2018;104:61-4.
Acknowledgments: We extend our thanks to the participants, their families, and all participating investigators and staff. This study was funded by Gilead Sciences, Inc.

Presented at *Virtual* CROI 2021, March 6–10, 2021

©2021 Gilead Sciences, Inc. All rights reserved.