GILEAD
Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Viral and Biomarker Outcomes of an Engineered bNAb in ART-Suppressed Participants

1342

Yanhui Cai,[1] Lisa Selzer,[1] Peter Ruane,[2] Eric S. Daar,[3] Princy Kumar,[4] Laurie VanderVeen,[1] Liao Zhang,[1] Jeffrey J. Wallin,[1] Devi SenGupta,[1] Sean E. Collins,[1] Kimberly Workowski,[5] Joseph J. Eron,[6] Edwin DeJesus[7]

[1]Gilead Sciences, Inc., Foster City, CA; [2]Ruane Clinical Research Group Inc., Los Angeles, CA; [3]The Lundquist Institute, Torrance, CA; [4]MedStar Health, Washington, DC; [5]Emory University, Atlanta, GA; [6]The University of North Carolina at Chapel Hill; [7]Orlando Immunology Center, Orlando, FL

Introduction

- Broadly neutralizing antibodies (bNAbs) against HIV-1 may target and eliminate virally infected cells expressing viral envelope (Env) protein, reducing HIV-1 reservoir in people with HIV (PWH)
- Increased HIV-specific T-cell responses have been observed in PWH receiving bNAb therapy during antiretroviral therapy (ART) interruption[1] and simian-human immunodeficiency virus (SHIV)–infected macaques receiving bNAb during acute infection[2]; however, further investigations are needed in PWH on long-term ART receiving bNAb therapy[3]
- Elipovimab (EVM; GS-9722) is a bNAb targeting the V3 glycan motif of HIV gp120 engineered from PGT121 to enhance Fc-γ receptor binding[4]
- EVM in combination with the toll-like receptor-7 agonist vesatolimod has demonstrated efficacy via increasing post-treatment virologic control in SHIV-infected monkeys[5]
- EVM was well tolerated in healthy and HIV-infected participants, with a half-life of ~26 d when dosed every other week (QOW)[6]

Objectives

- To evaluate viral outcomes and HIV-specific T-cell responses in ART-suppressed PWH receiving EVM

Methods

GS-US-420-3902 Study Design

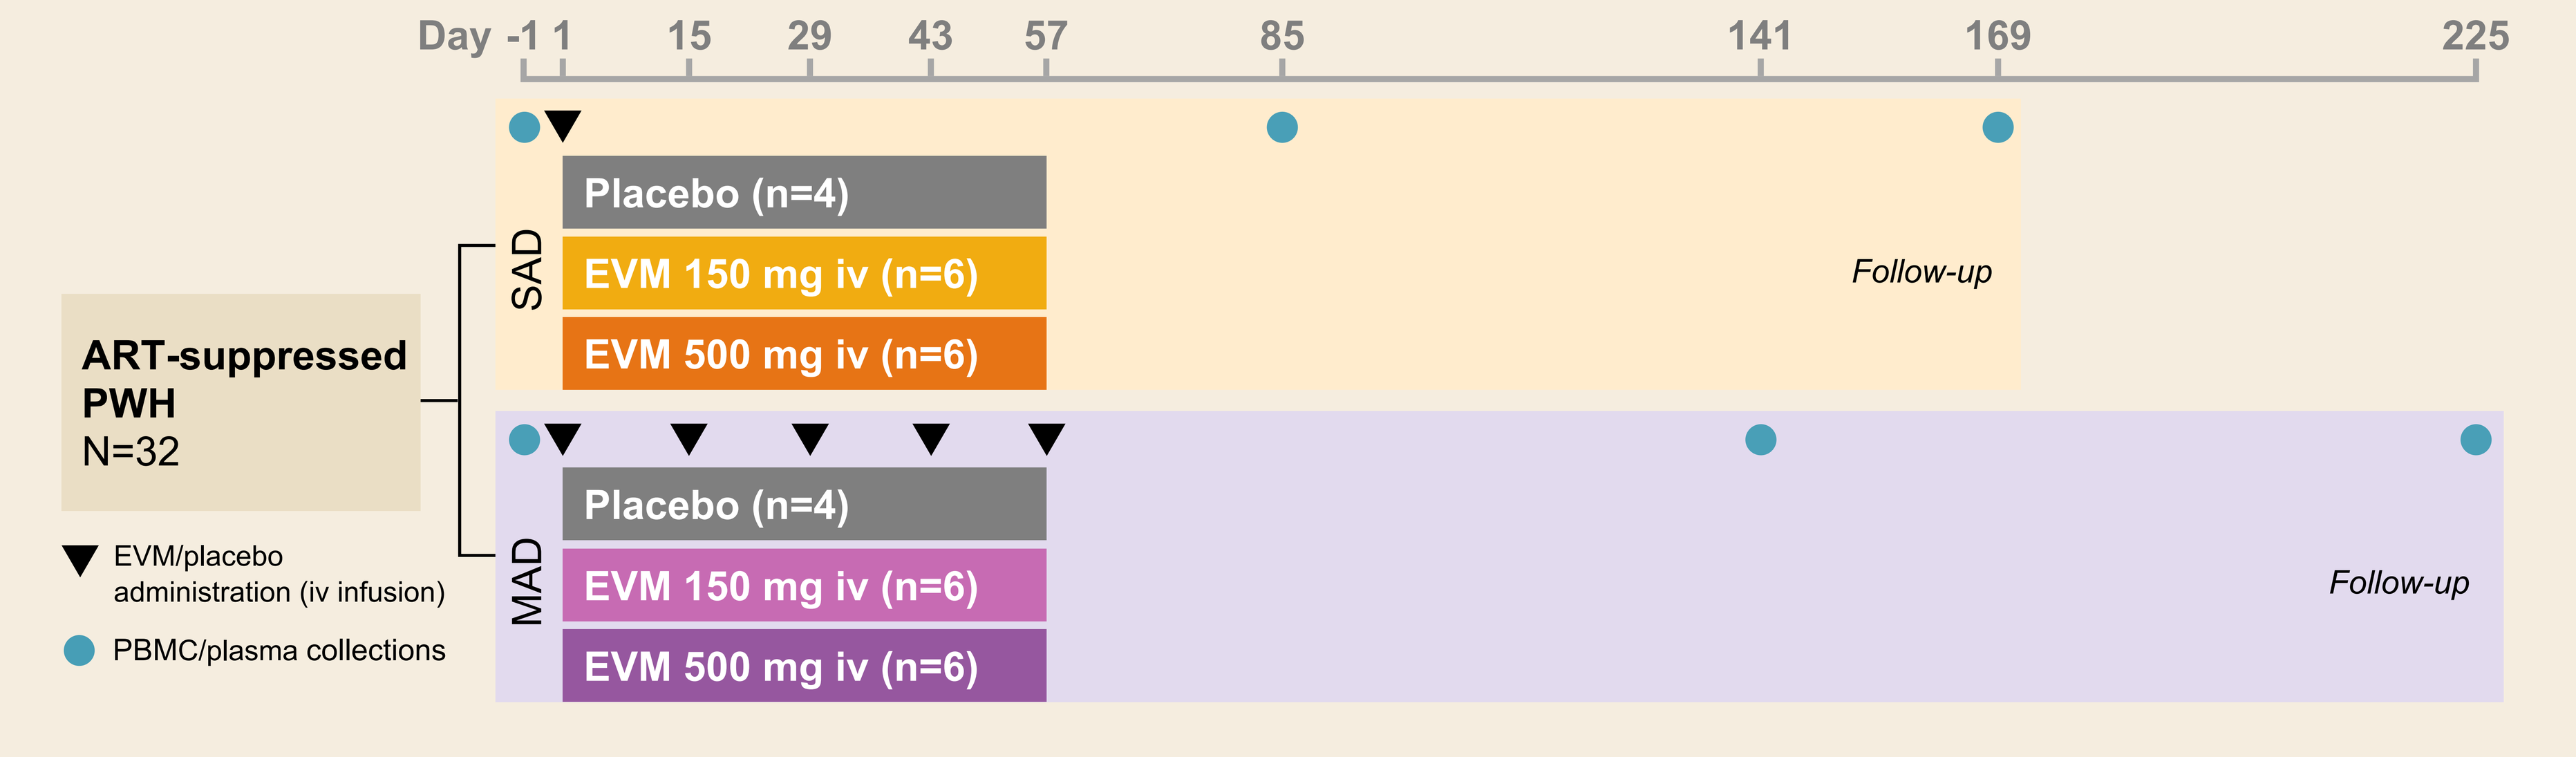
Day -1 1 15 29 43 57 85 141 169 225
ART-suppressed PWH
N=32
SAD
Placebo (n=4)
EVM 150 mg iv (n=6)
EVM 500 mg iv (n=6)
Follow-up
MAD
Placebo (n=4)
EVM 150 mg iv (n=6)
EVM 500 mg iv (n=6)
Follow-up
EVM/placebo administration (iv infusion)
PBMC/plasma collections

MAD, multiple ascending doses; PBMC, peripheral blood mononuclear cells; SAD, single ascending dose.

- Phase 1b randomized, double-blind, SAD/MAD study of EVM in 32 PWH chronically suppressed on ART (plasma viral load <50 copies/mL)
- Study endpoints
 - Primary objectives: safety and tolerability; pharmacokinetics
 - Secondary objectives: to determine if EVM induces antidrug antibodies and evaluate virologic effect of EVM
 - Exploratory objectives: to evaluate effect of EVM on HIV-1-specific responses and viral Env diversity
- Plasma and PBMCs were collected at baseline (BL) and multiple time points following treatment
- HIV-specific T-cell responses were evaluated with the interferon-γ enzyme-linked immunosorbent spot assay (ELISpot) using PBMCs stimulated with clade B HIV-1 consensus peptides (Env, Gag, Nef, and Pol; Cellular Technology Limited LLC, Shaker Heights, OH)
- Total HIV DNA and intact HIV proviral DNA in $CD4^+$ T cells were measured by multiplex digital droplet polymerase chain reaction (AccelevirDx, Baltimore, MD)[7]
- Baseline EVM sensitivity was defined using published HIV Env signatures with positive predictive values (PPVs) from 75% to 97%[8]
 - Proviral genotypes were determined using next-generation sequencing of the viral *Env* gene (Seq-IT GmbH & Co.KG, Kaiserslautern, Germany)
- Wilcoxon matched-pair rank test was used to compare the difference between 2 time points within the same group for biomarker and virologic data

Results

Prevalence of EVM Signatures at BL in ART-Suppressed Participants*

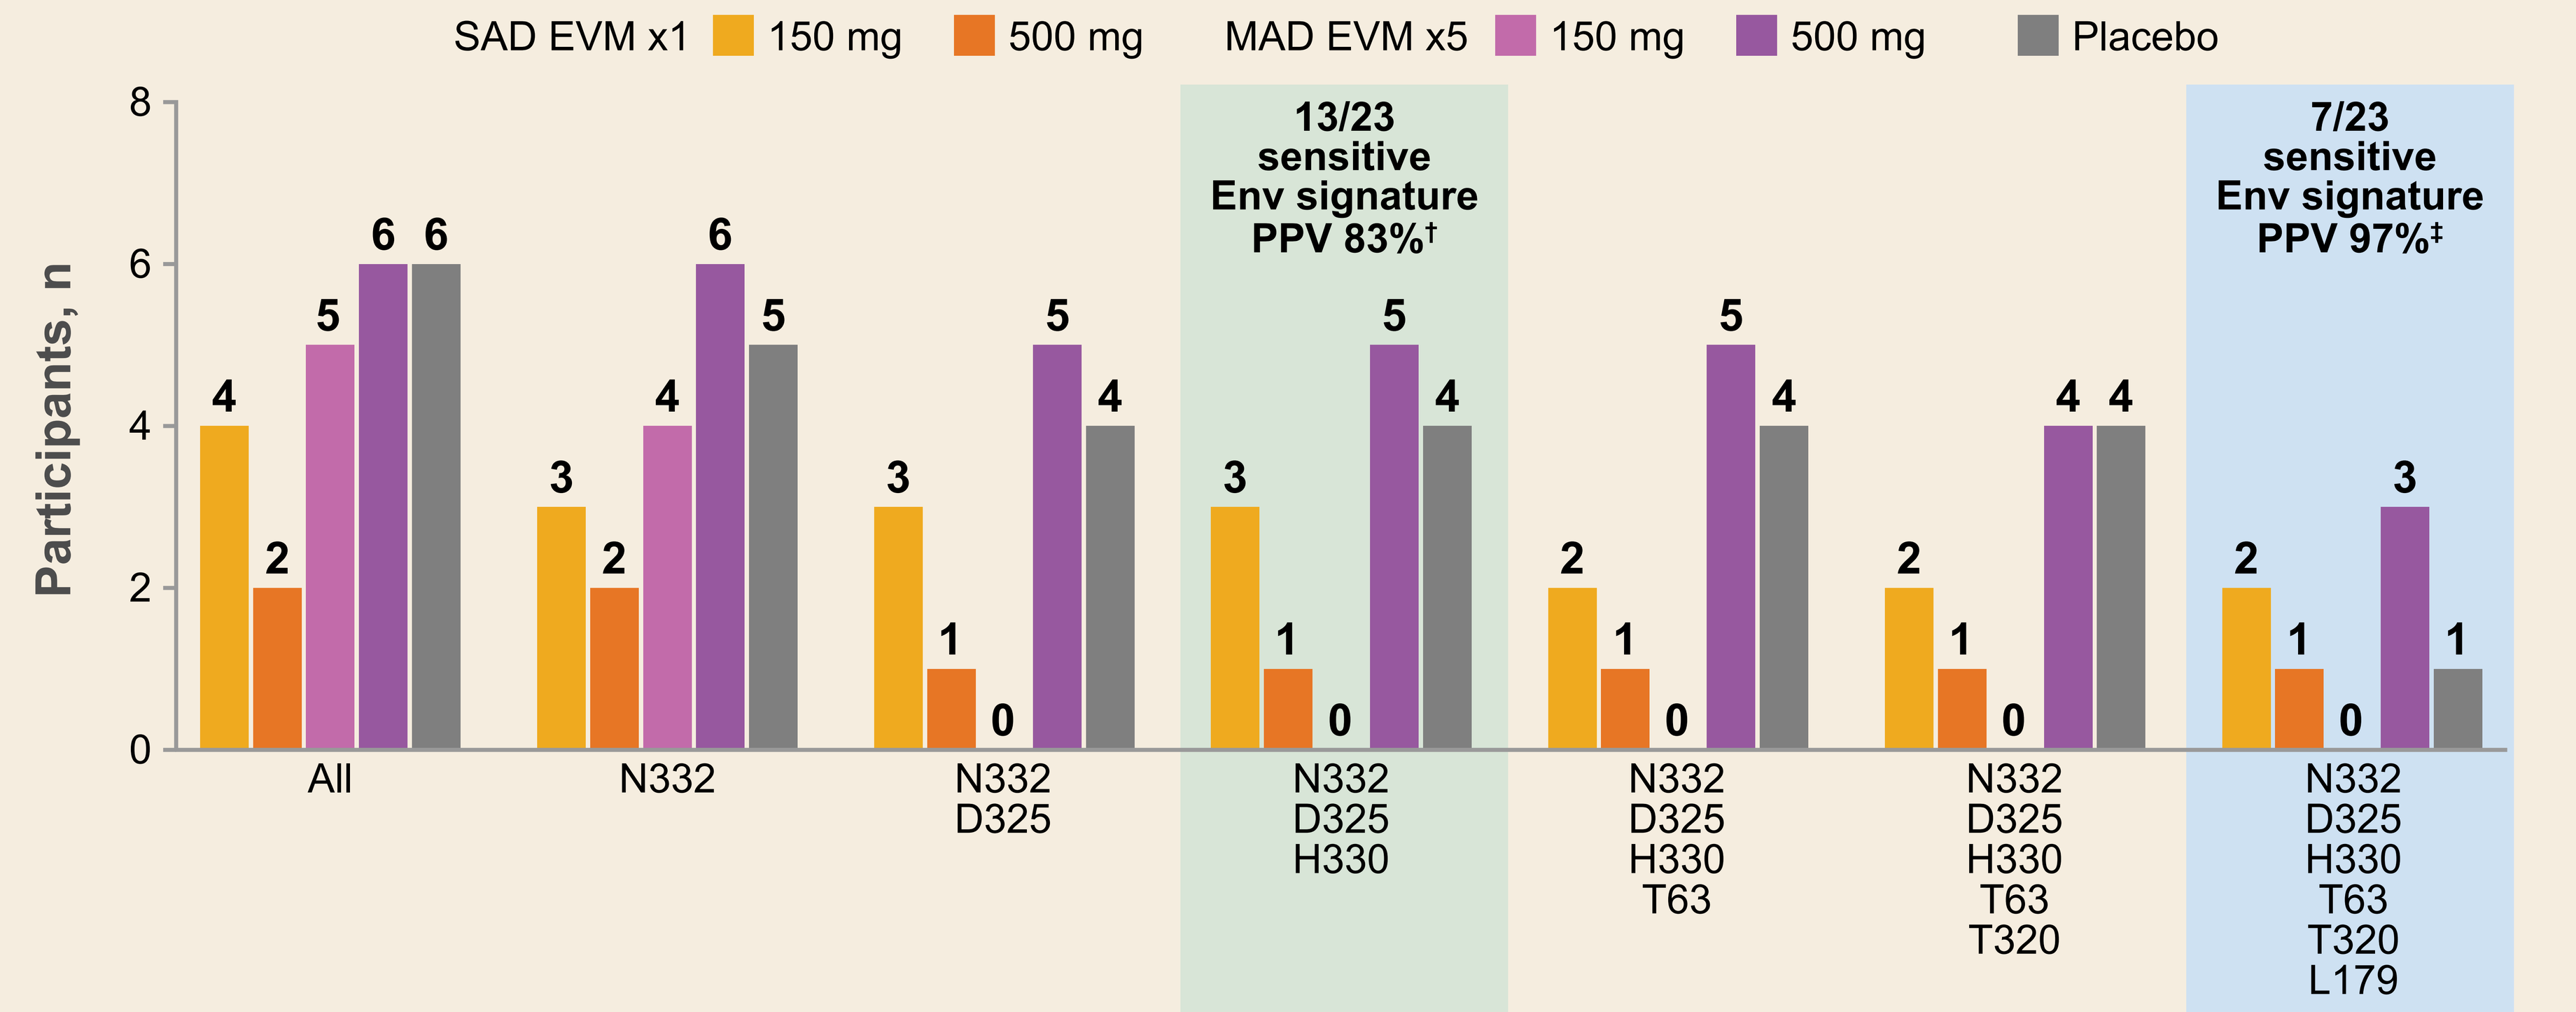
SAD EVM x1 150 mg 500 mg MAD EVM x5 150 mg 500 mg Placebo
Participants, n
13/23 sensitive Env signature PPV 83%†
7/23 sensitive Env signature PPV 97%‡
All
N332
N332 D325
N332 D325 H330
N332 D325 H330 T63
N332 D325 H330 T63 T320
N332 D325 H330 T63 T320 L179

*BL HIV Env sequencing data were available for 25 participants; HXB2 numbering used for HIV Env amino-acid positions: N332, N332 glycan N-X-S/T; †Based on presence of N332 glycan, and D325 and H330 residues in HIV Env; ‡Based on presence of N332 glycan, and D325, H330, T63, T320, and L179 residues (N332glycan/D325/H330/T63/T320/L179) in HIV Env.

- Viral sensitivity to EVM for participants with available data was determined based on presence of HIV Env signatures

HIV-Specific T Cells Following Multiple High Doses of EVM in ART-Suppressed Participants*

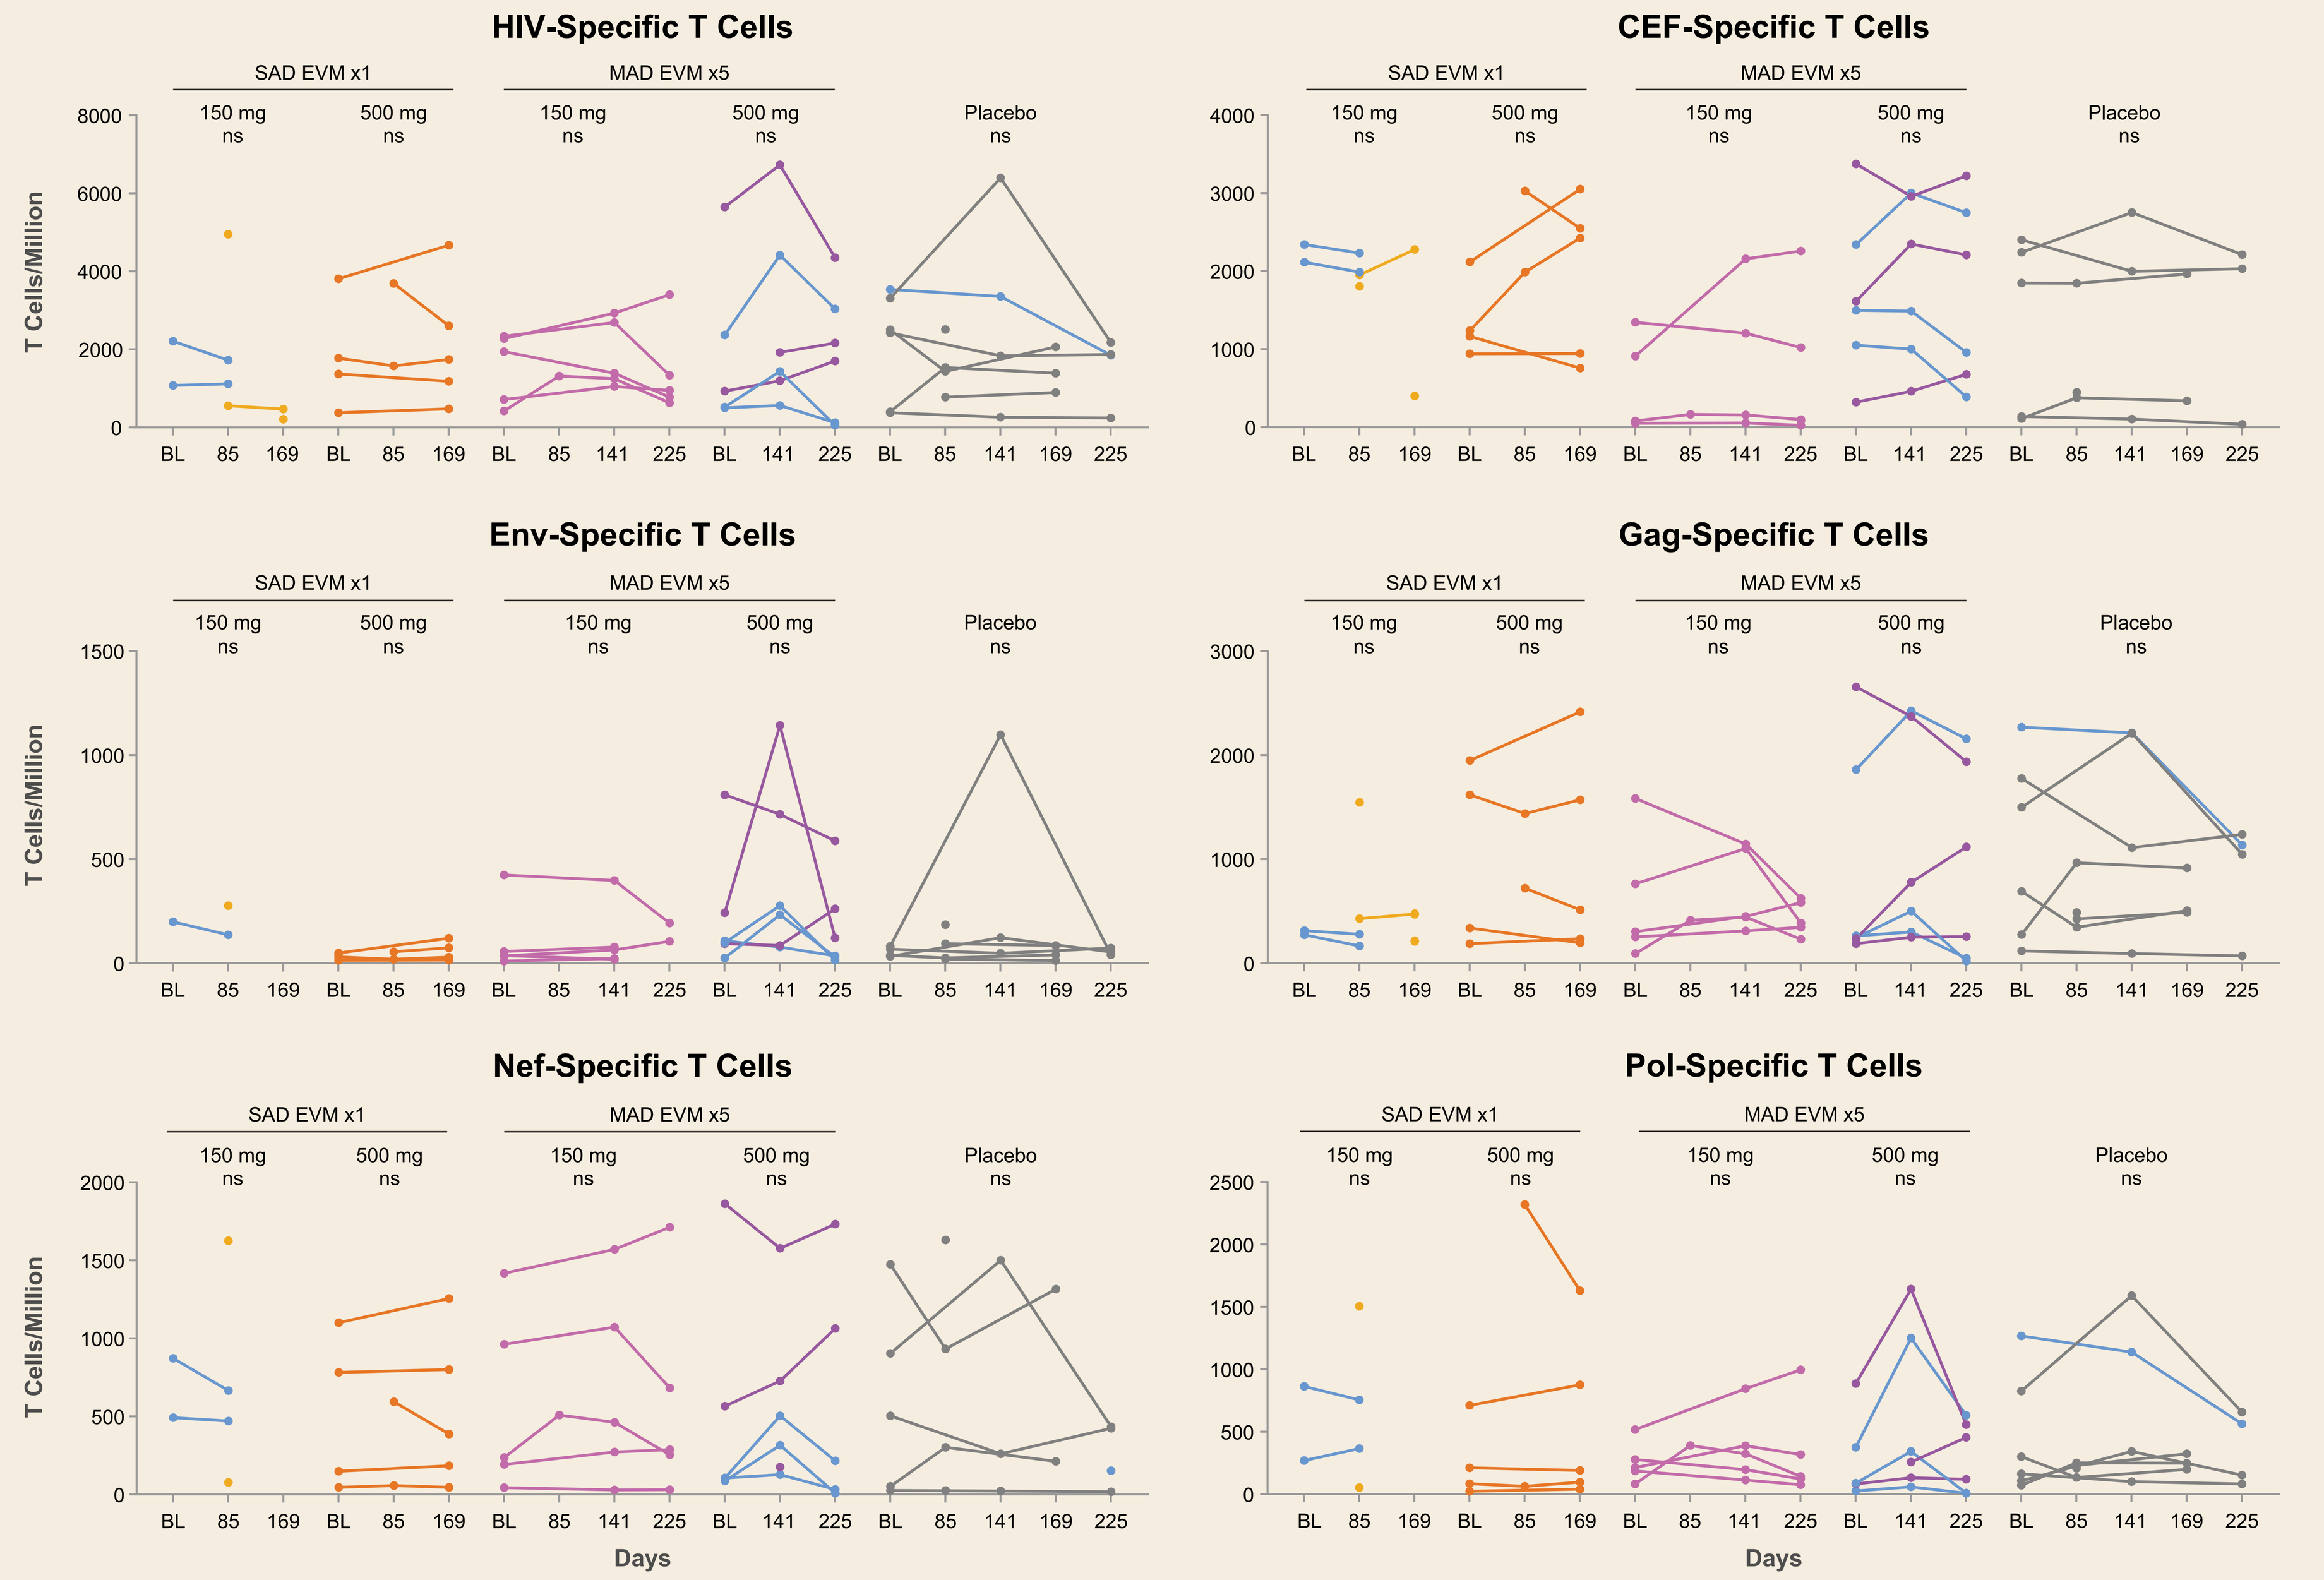
HIV-Specific T Cells
CEF-Specific T Cells
Env-Specific T Cells
Gag-Specific T Cells
Nef-Specific T Cells
Pol-Specific T Cells
SAD EVM x1
MAD EVM x5
150 mg ns
500 mg ns
Placebo ns
T Cells/Million
Days

*Participants sensitive to EVM signature N332glycan/D325/H330/T63/T320/L179[8] indicated in *blue*; HIV-specific T cells refer to sum of T cells responding to ex vivo stimulation with each clade B HIV-1 consensus peptide pool (Env, Gag, Nef, and Pol). CEF, cytomegalovirus, Epstein-Barr virus, and influenza virus; ns, not significant.

- Modest increases in HIV-specific T-cell responses were observed 12 wk (Day 141) after the last EVM dose in participants who received multiple doses of EVM 500 mg, which returned to BL after 24 wk (Day 225); no significant change in HIV-specific T cells was observed in the other 3 cohorts

Greater Increases in HIV-Specific T Cells Were Observed Following Multiple High Doses of EVM in bNAb-Sensitive Participants

ID	Sensitive to bNAb*	Fold Change of Antigen-Specific T Cells: Day 141 vs BL					
		HIV	CEF	Env	Gag	Nef	Pol
1	Yes	1.1	1.0	0.7	1.1	1.2	2.4
2	No	1.3	1.4	0.9	1.3	1.3	1.6
3	Yes	1.9	1.3	9.0	1.3	4.8	3.3
4	Yes	2.8	1.0	2.8	2.0	3.6	3.8
5†	No	N/A†	1.5	0.9	3.3	N/A†	N/A†
6	No	1.2	0.9	4.7	0.9	0.8	1.8
Median		1.3	1.2	1.9	1.3	1.3	2.4
IQR		1.2–2.4	1.0–1.4	0.9–5.8	1.1–2.3	1.0–4.2	1.7–3.6

*Viral sensitivity at BL based on presence of HIV Env signature N332glycan/D325/H330/T63/T320/L179[8]; †BL ELISpot data are not available (N/A) using criterion "cutoff 3-fold above background with 70% viability." IQR, interquartile range.

No Effect on HIV Reservoir Was Observed Following EVM Treatment in ART-Suppressed Participants*

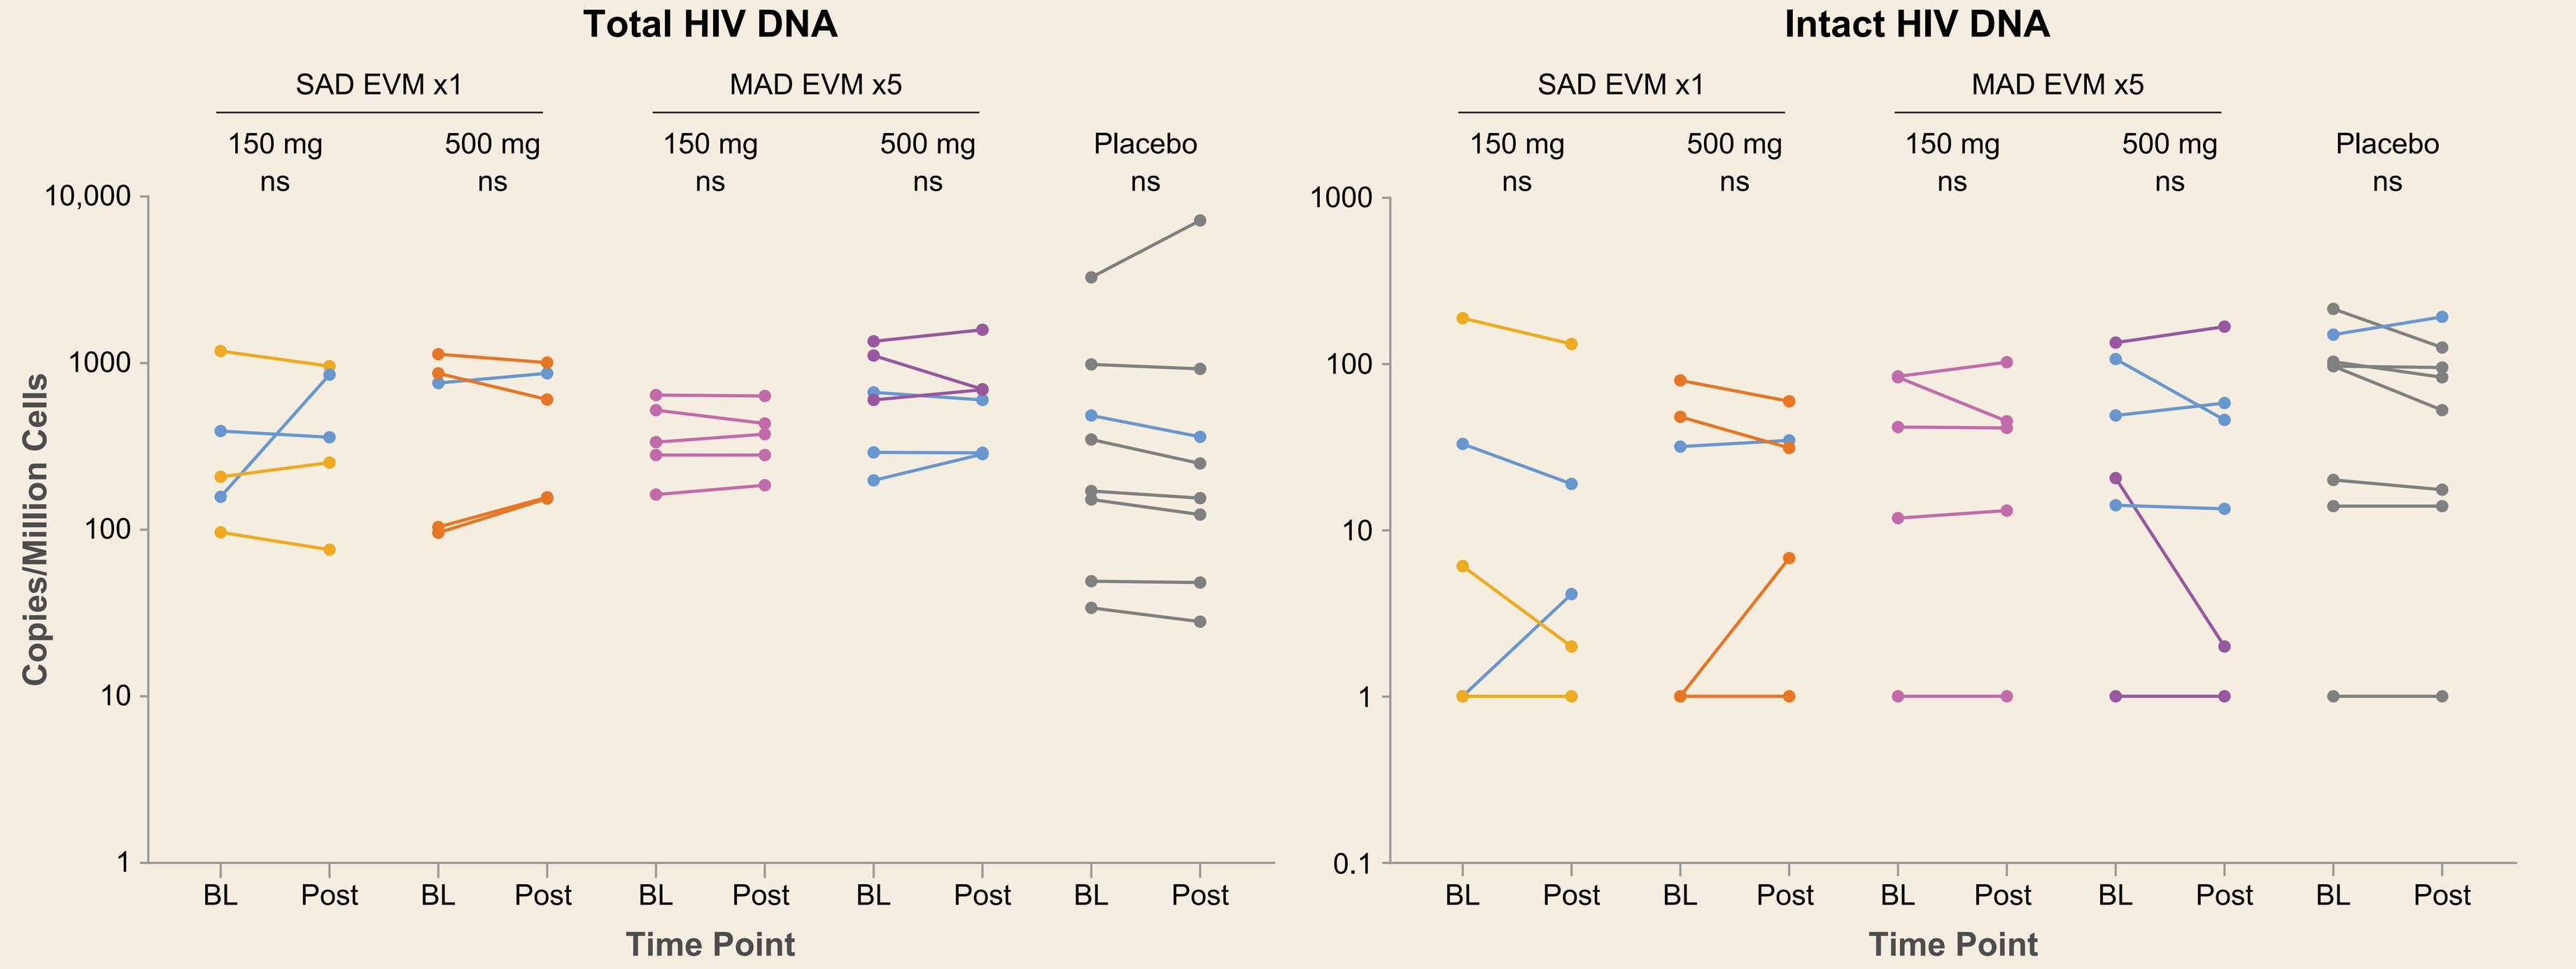
Total HIV DNA
Intact HIV DNA
SAD EVM x1
MAD EVM x5
150 mg ns
500 mg ns
Placebo ns
Copies/Million Cells
BL Post
Time Point

*Postdose: Day 85 or 169 (SAD cohorts), or 225 (MAD cohorts); BL and postdose comparisons by Wilcoxon matched-pair signed rank test at 2-sided 0.05 significance level; participants sensitive to EVM signature N332glycan/D325/H330/T63/T320/L179[8] indicated in *blue*.

- No significant changes were observed in levels of total and intact HIV DNA after administration of EVM or placebo

Conclusions

- EVM may augment HIV-specific T-cell responses in PWH harboring bNAb-sensitive viruses
 - 3/6 participants in Cohort 4 (MAD) receiving higher doses of EVM were identified as EVM sensitive by genotyping, including 2 with the highest fold increases in HIV-specific T cells
- EVM did not have a measurable effect on levels of total and intact HIV DNA in ART-suppressed PWH
- Whether bNAbs can facilitate clearance of the replication-competent latent HIV reservoir remains an area of interest, and a combination of bNAbs may increase the breadth of coverage of diverse viruses

References: **1.** Niessl J, et al. Nat Med 2020;26:222-7; **2.** Nishimura Y, et al. Nature 2017;543:559-63; **3.** Stephenson KE, et al. Nat Med 2021;27:1718-24; **4.** Thomsen ND, et al. CROI 2019, poster 3852; **5.** Borducchi EN, et al. Nature 2018;263:360-4; **6.** Ruane P, et al. CROI 2020, abstr 39; **7.** Bruner KM, et al. Nature 2019;566:120-5; **8.** Moldt B, et al. J Acquir Immune Defic Syndr. 2021;88:61-9.

Acknowledgments: We extend our thanks to the participants, their partners, and families. Special thanks to the study teams and contract research organizations (Cellular Technology Limited LLC, AccelevirDx, and Seq-IT GmbH & Co.KG). This study was funded by Gilead Sciences, Inc. Editorial and production assistance were provided by BioScience Communications, New York, NY, funded by Gilead.

Presented at *Virtual* CROI 2022, February 12–16, 2022

© 2022 Gilead Sciences, Inc. All rights reserved.