FRI-213

Elbasvir/Grazoprevir Plus Sofosbuvir ± Ribavirin in Treatment-Naive And Treatment-Experienced People With Hepatitis C Virus Genotype 3 Infection and Compensated Cirrhosis: SVR24 Results of the C-ISLE Study

Presented at The International Liver Congress™ 2017; April 19-23, 2017; Amsterdam, The Netherlands

Foster GR[1]; Agarwal K[2]; Cramp M[3]; Moreea S[4]; Barclay S[5]; Collier J[6]; Brown AS[7]; Ryder SD[8]; Ustianowski A[9]; Forton DM[10]; Fox R[11]; Gordon F[12]; Rosenberg WM[13]; Mutimer DJ[14]; Du J[15]; Gilbert CL[15]; Robertson MN[15]; Barr E[15]; Haber B[15]

[1]The Royal London Hospital, London, UK; [2]Institute of Liver Studies, Kings College Hospital, London, UK; [3]South West Liver Unit, Derriford Hospital and Peninsula School of Medicine and Dentistry, Plymouth, UK; [4]Bradford Teaching Hospitals Foundation Trust, Bradford, UK; [5]Glasgow Royal Campus, Glasgow, UK; [6]John Radcliffe Hospital, Oxford, UK; [7]Imperial College Healthcare, London, UK; [8]NIHR Biomedical Unit in Gastrointestinal and Liver Diseases at Nottingham University Hospitals NHS Trust and The University of Nottingham, Nottingham, UK; [9]North Manchester General Hospital, Manchester, UK; [10]St. Georges University of London, London, UK; [11]Gartnavel General Hospital, Glasgow, UK; [12]Hepatology Joint Clinical Research Unit, Bristol, UK; [13]University College London, London, UK; [14]QE Hospital, Birmingham, UK; [15]Merck & Co., Inc., Kenilworth, NJ, USA

Background

- Genotype (GT)3 is a common hepatitis C virus (HCV) genotype, accounting for 30% of infections globally[1]
 - GT3 infection is associated with high rates of hepatocellular carcinoma and rapid progression to cirrhosis
- Despite recent advances, HCV GT3 infection and cirrhosis are challenging to treat with all-oral regimens
 - Treatment-experienced people or those with baseline NS5A resistance-associated substitutions (RASs) are particularly challenging
- Elbasvir (EBR) is a once-daily NS5A inhibitor and grazoprevir (GZR) is a once-daily HCV NS3/4A protease inhibitor (**Figure 1**)
 - The combination of EBR and GZR is approved in Europe, the United States, Canada, and other countries worldwide[2]
 - Broad activity vs most HCV genotypes in vitro[3-5]
 - Efficacious in treatment-naive and -experienced people, cirrhotic and noncirrhotic people, HIV/HCV co-infected people, and those with chronic kidney disease[6-9]
- Sofosbuvir (SOF) is an NS5B inhibitor indicated for the treatment of chronic HCV infection as a component of a combination antiviral treatment regimen (**Figure 1**)[10]
- The C-SWIFT study evaluated the safety and efficacy of EBR/GZR + SOF for 4-12 weeks in treatment-naive participants with HCV GT1 or 3 infection[11]
 - High sustained virologic response (SVR) rates (>90%) in HCV GT3-infected, treatment-naive participants receiving EBR/GZR + SOF for 12 weeks
 - Similar efficacy in cirrhotic participants treated for 12 weeks and noncirrhotic participants treated for 8 or 12 weeks

Figure 1. EBR/GZR plus SOF

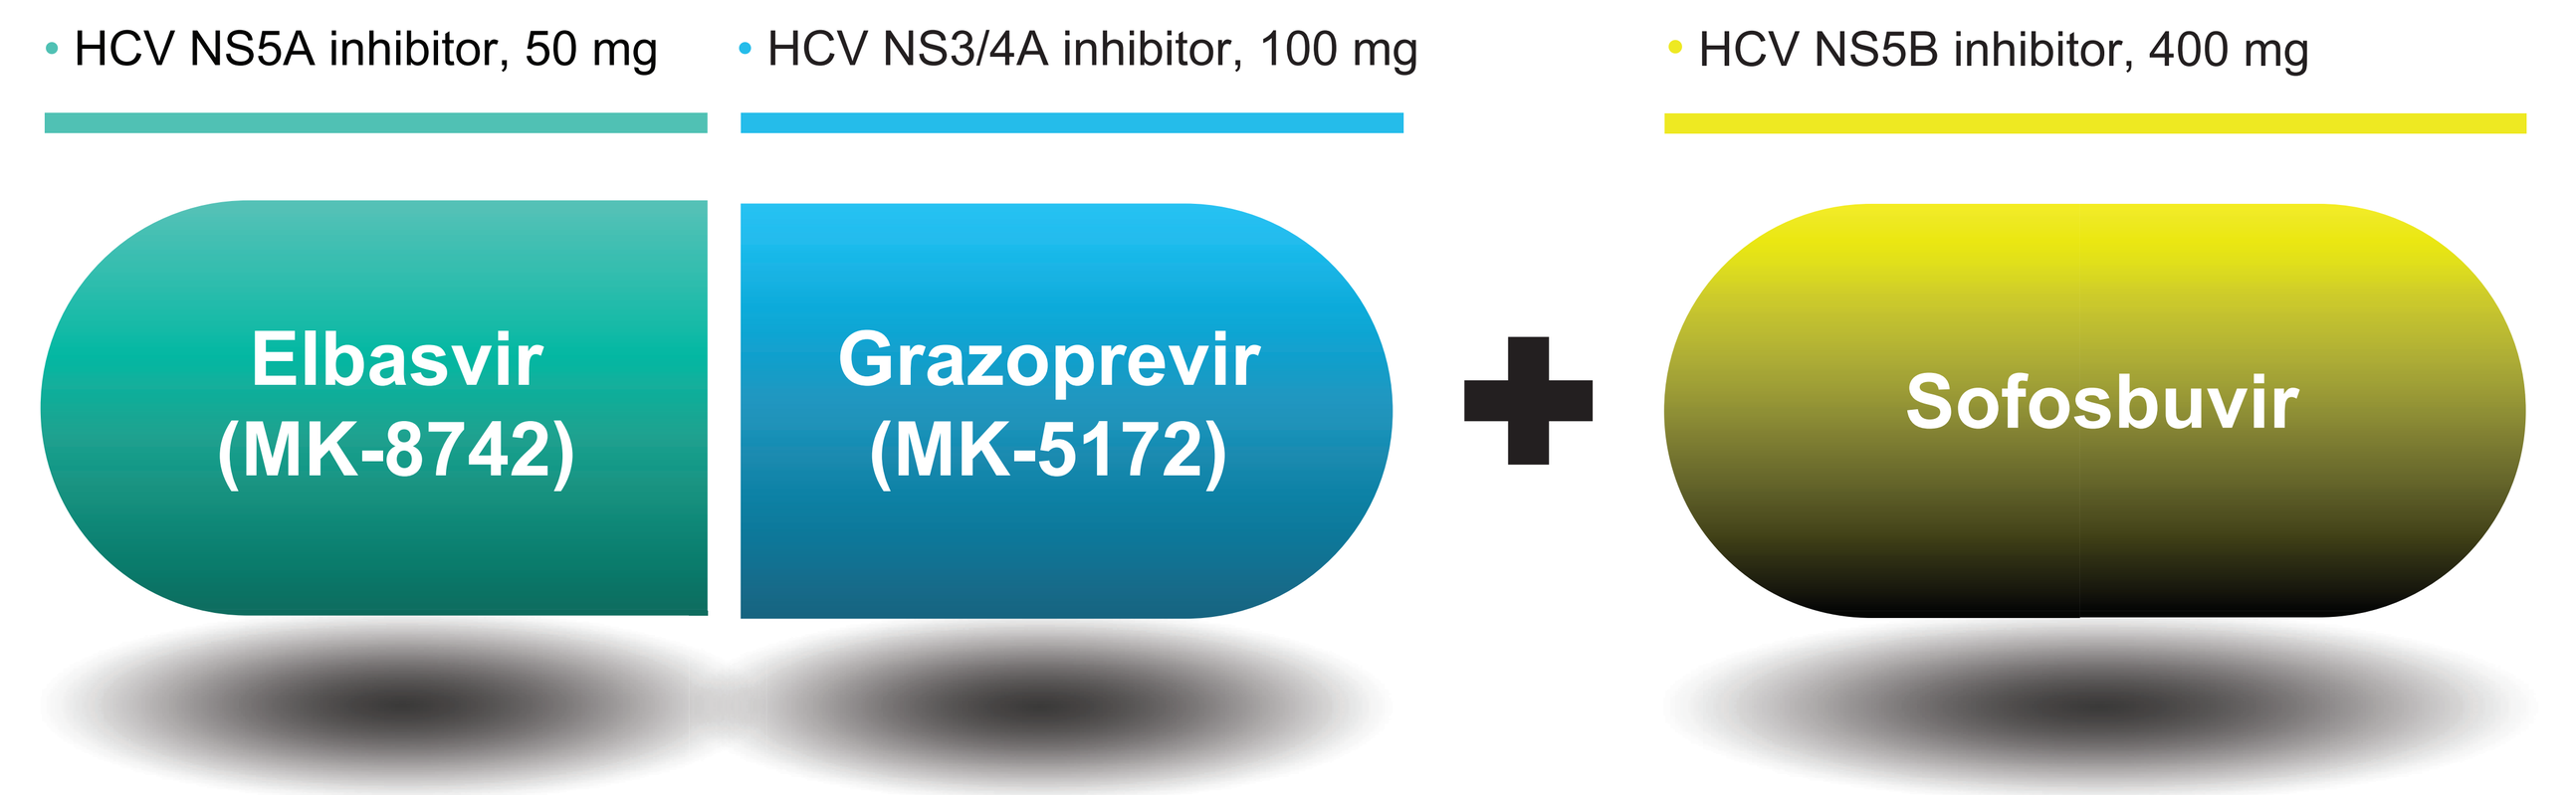

HCV NS5A inhibitor, 50 mg
HCV NS3/4A inhibitor, 100 mg
HCV NS5B inhibitor, 400 mg
Elbasvir (MK-8742)
Grazoprevir (MK-5172)
Sofosbuvir

Aim

- To evaluate the regimen of EBR/GZR + SOF ± ribavirin (RBV) in cirrhotic participants with HCV GT3 infection for durations ranging from 8-16 weeks

Participants and Methods

Study Design

- C-ISLE (NCT02601573; Protocol PN083-02) was a randomized, open-label, UK-based clinical trial in HCV GT3-infected participants with compensated cirrhosis (**Figure 2**)
- Adults with chronic HCV GT3 infection were included
 - Compensated liver cirrhosis defined by liver biopsy (METAVIR F4) or transient elastography (>12.5 kPa)
 - Treatment-naive, experienced to peginterferon/RBV, monoinfected or HIV co-infected
- All participants received EBR 50 mg/GZR 100 mg + SOF (400 mg as per prescribing information) ± RBV 800-1400 mg/day
 - Treatment-naive participants were treated for 8 or 12 weeks
 - Treatment-experienced participants were treated for 12 or 16 weeks
 - Randomization of treatment-experienced participants was stratified based on prior relapse vs nonrelapse (partial, null, interferon-intolerant)
 - Target enrollment was 25 participants per arm
 - The primary endpoint was SVR 12 weeks after completion of therapy (SVR12, HCV RNA <15 IU/mL [cobas® TaqMan® v2.0])
 - SVR 24 weeks after completion of therapy (SVR24) was assessed as a secondary endpoint
 - SVR12 data were presented at The Liver Meeting® 2016[12]; this presentation provides the final SVR24 data
- Efficacy analysis population
 - Full analysis set (FAS) included all participants who received ≥1 dose of study drug
 - Modified FAS (mFAS) excluded participants who discontinued for reasons unrelated to the treatment regimen, including discontinuation for non–drug-related AEs
- Resistance analysis was performed using next-generation sequencing: detection limit for variants of ≈1% or ≈15% prevalence
 - Resistance analysis population included all participants who had baseline sequencing available and a treatment outcome of either SVR12 or virologic failure
 - NS3 polymorphisms at positions 36, 54, 55, 56, 80, 107, 122, 132, 155, 156, 158, 168, 170, or 175
 - NS5A polymorphisms at positions 24, 28, 30, 31, 32, 38, 58, 62, 92, or 93
 - NS5B polymorphisms at positions 96, 142, 159, 282, 289, 316, 320, or 321

Figure 2. Study design

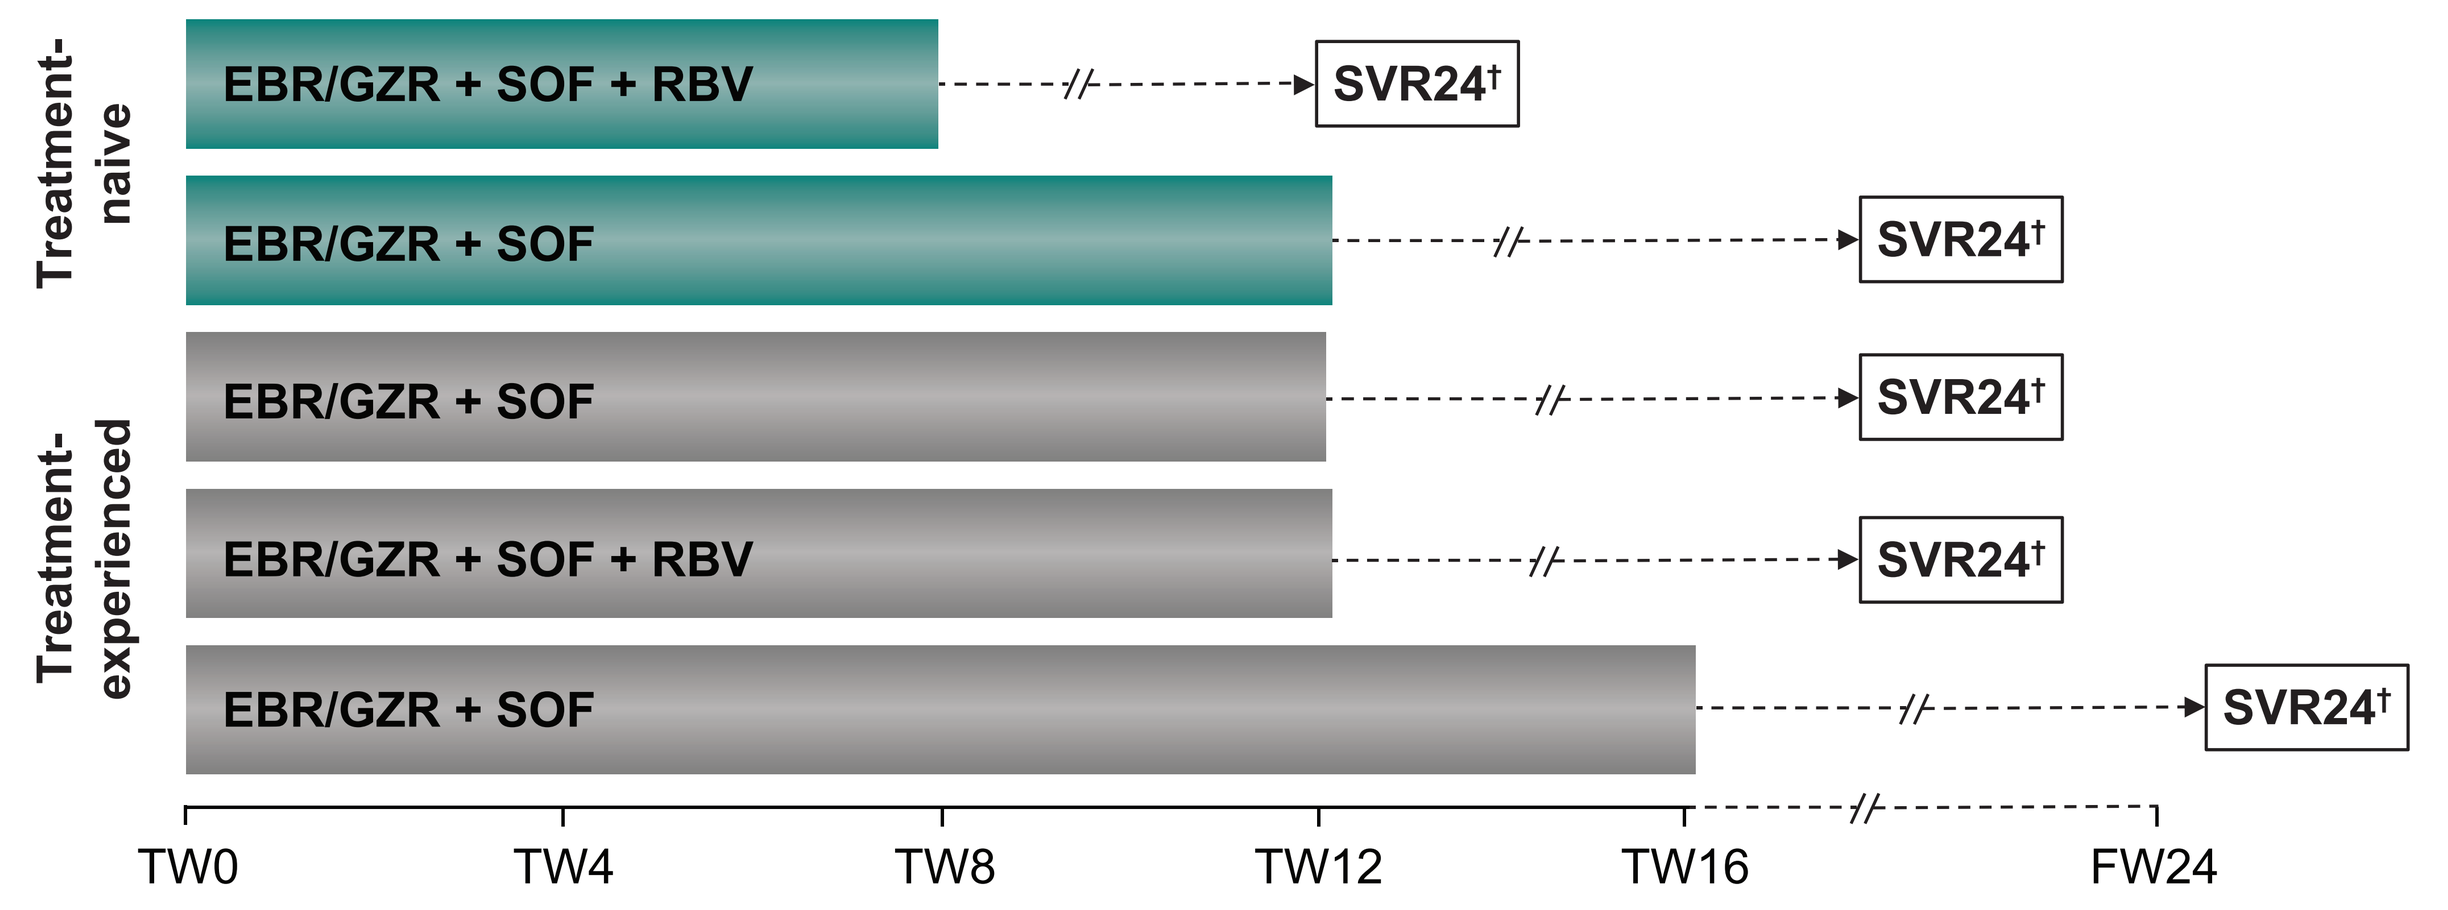

Treatment-naive
Treatment-experienced
EBR/GZR + SOF + RBV
EBR/GZR + SOF
EBR/GZR + SOF
EBR/GZR + SOF + RBV
EBR/GZR + SOF
SVR24†
TW0
TW4
TW8
TW12
TW16
FW24

FW, follow-up week; TW, treatment week.
†The primary endpoint was HCV RNA <15 IU/mL at FW12 (SVR12). SVR24 was assessed as a secondary endpoint.

Results

Demographics and Characteristics

- 100 participants with HCV GT3 infection and compensated cirrhosis were enrolled (**Table 1**)

Table 1. Participant demographics

	Cirrhotic GT3-infected participants (n = 100)
Male, n (%)	68 (68)
Race, n (%)	
Asian	29 (29)
White	69 (69)
Other	2 (2)
Age, years, mean (SD)	53.4 (8.7)
BMI ≥30 kg/m², n (%)	28 (28)
Cirrhosis diagnosis method	
Liver biopsy, n (%)	16 (16)
FibroScan®, n (%)	84 (84)
FibroScan® score, kPa, mean (SD)	25.4 (12.1)
Prior treatment history, n (%)	
Naive	47 (47)
PR-experienced	53 (53)
HCV RNA $\log_{10}$, IU/mL, mean (SD)	6.2 (0.7)
IL28B CC, n (%)	50 (50)
Albumin, g/dL, mean (SD)	3.6 (1.2)
ALT IU/L, median (range)	94 (21-389)
Total bilirubin, mg/dL, mean (SD)	0.7 (0.4)
Platelets × 10^3 cells/μL, median (range)	138 (46-396)
Platelet count <100 × 10^3 cells/μL, n (%)	24 (24)

ALT, alanine aminotransferase; BMI, body mass index; PR, peginterferon/ribavirin; SD, standard deviation.

Virologic Response

- In the FAS, SVR24 was achieved by 91% and 88% of treatment-naive participants treated for 8 and 12 weeks, respectively (**Figure 3**)
- SVR24 was achieved by 94%, 94% and 83% of treatment-experienced participants treated for 12 or 16 weeks
 - Two participants experienced virologic failure, both relapses in the 8-week treatment arm, with virologic recurrence prior to follow-up week (FW)12. There were no virologic failures after FW12
 - One participant discontinued treatment due to the drug-related adverse event (AE) of cellulitis
 - Seven participants were lost to follow-up: 2 prior to FW12 and 5 between FW12 and FW24

Figure 3. SVR24 in the FAS population

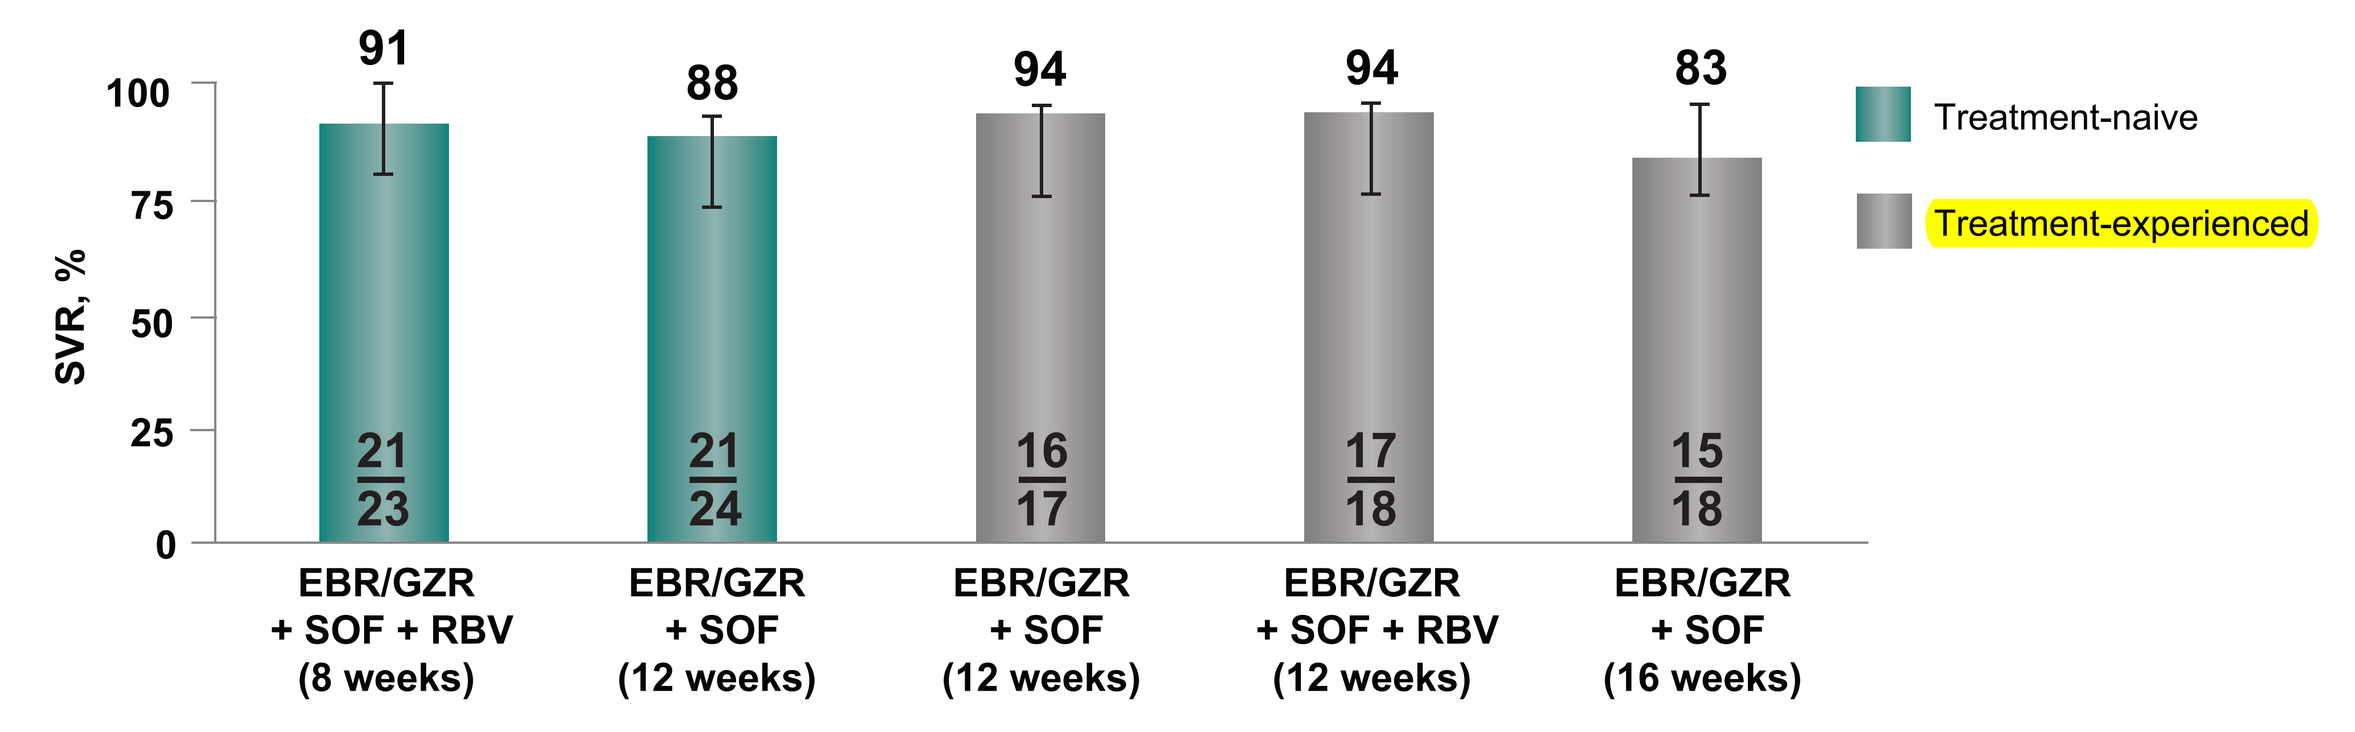

SVR, %
100
75
50
25
0
91
88
94
94
83
21/23
21/24
16/17
17/18
15/18
Treatment-naive
Treatment-experienced

	EBR/GZR + SOF + RBV (8 weeks)	EBR/GZR + SOF (12 weeks)	EBR/GZR + SOF (12 weeks)	EBR/GZR + SOF + RBV (12 weeks)	EBR/GZR + SOF (16 weeks)
Relapse					
EOT to FW12	2	0	0	0	0
FW12 to FW24	0	0	0	0	0
Nonvirologic failure†					
Prior to FW12	0	1	0	1	1‡
FW12 to FW24	0	2	1	0	2

EOT, end of treatment.
†All lost to follow-up or consent withdrawn, except 1 participant‡ who discontinued treatment due to a drug-related AE.

- In the mFAS, SVR24 was achieved by 100% of treatment-naive and -experienced participants receiving EBR/GZR + SOF for 12 weeks (**Figure 4**)

Figure 4. SVR24 in the mFAS population

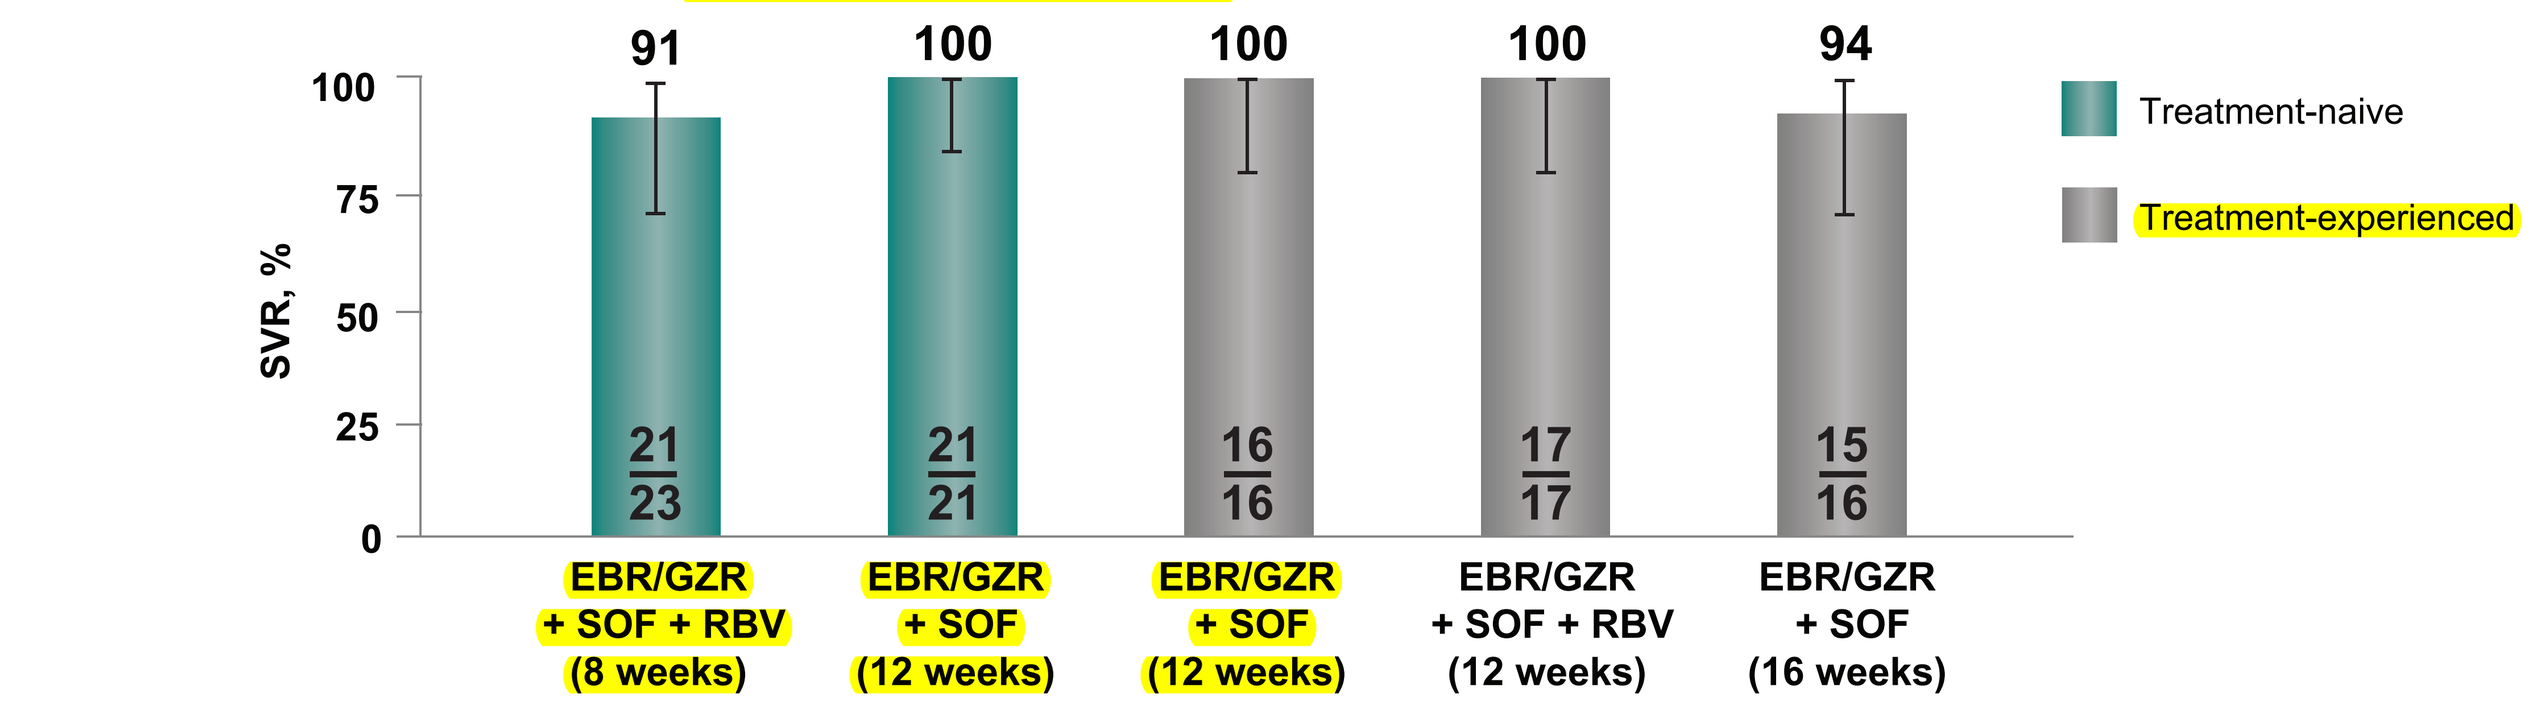

SVR, %
100
75
50
25
0
91
100
100
100
94
21/23
21/21
16/16
17/17
15/16
Treatment-naive
Treatment-experienced

	EBR/GZR + SOF + RBV (8 weeks)	EBR/GZR + SOF (12 weeks)	EBR/GZR + SOF (12 weeks)	EBR/GZR + SOF + RBV (12 weeks)	EBR/GZR + SOF (16 weeks)
Relapse					
EOT to FW12	2	0	0	0	0
FW12 to FW24	0	0	0	0	0
Discontinued due to AE	0	0	0	0	1
Excluded from mFAS analysis†	0	3	1	1	2

†All lost to follow-up or consent withdrawn.

- Two participants relapsed (**Table 2**)
 - At baseline, participant 1 had Y93H, P58S, and S62T present in 44%, 99% and 62% of the viral population, suggesting these 3 RASs were linked within the same virus

Table 2. Relapse participants

Participant	Treatment history	Treatment regimen	Time of relapse	NS5A RAVs Baseline	NS5A RAVs Failure
1	Naive	EBR/GZR + SOF + RBV (8 weeks)	FW4	Y93H, P58S, S62T	P58S, S62T
2	Naive	EBR/GZR + SOF + RBV (8 weeks)	FW8	WT	WT

Nonvirologic Failures

- Eight participants had nonvirologic failure (**Table 3**)

Table 3. Nonvirologic failures

Participant	Treatment history	Treatment regimen	Reason for discontinuation
Prior to FW12			
1	Naive	EBR/GZR + SOF (12 weeks)	LTFU after TW2
2	Experienced	EBR/GZR + SOF + RBV (12 weeks)	Withdrew consent after day 7
3	Experienced	EBR/GZR + SOF (16 weeks)	Discontinued after day 7 due to cellulitis
Between FW12 and FW24			
4	Naive	EBR/GZR + SOF (12 weeks)	LTFU/withdrew consent
5	Naive	EBR/GZR + SOF (12 weeks)	LTFU/withdrew consent
6	Experienced	EBR/GZR + SOF (12 weeks)	LTFU/withdrew consent
7	Experienced	EBR/GZR + SOF (16 weeks)	LTFU/withdrew consent
8	Experienced	EBR/GZR + SOF (16 weeks)	LTFU/withdrew consent

LTFU, lost to follow-up.

Resistance analyses

- The presence of baseline NS3 or NS5A RASs had no impact on SVR12 (**Figure 5**)

Figure 5. Prevalence and impact on SVR12 of NS3 and NS5A RASs

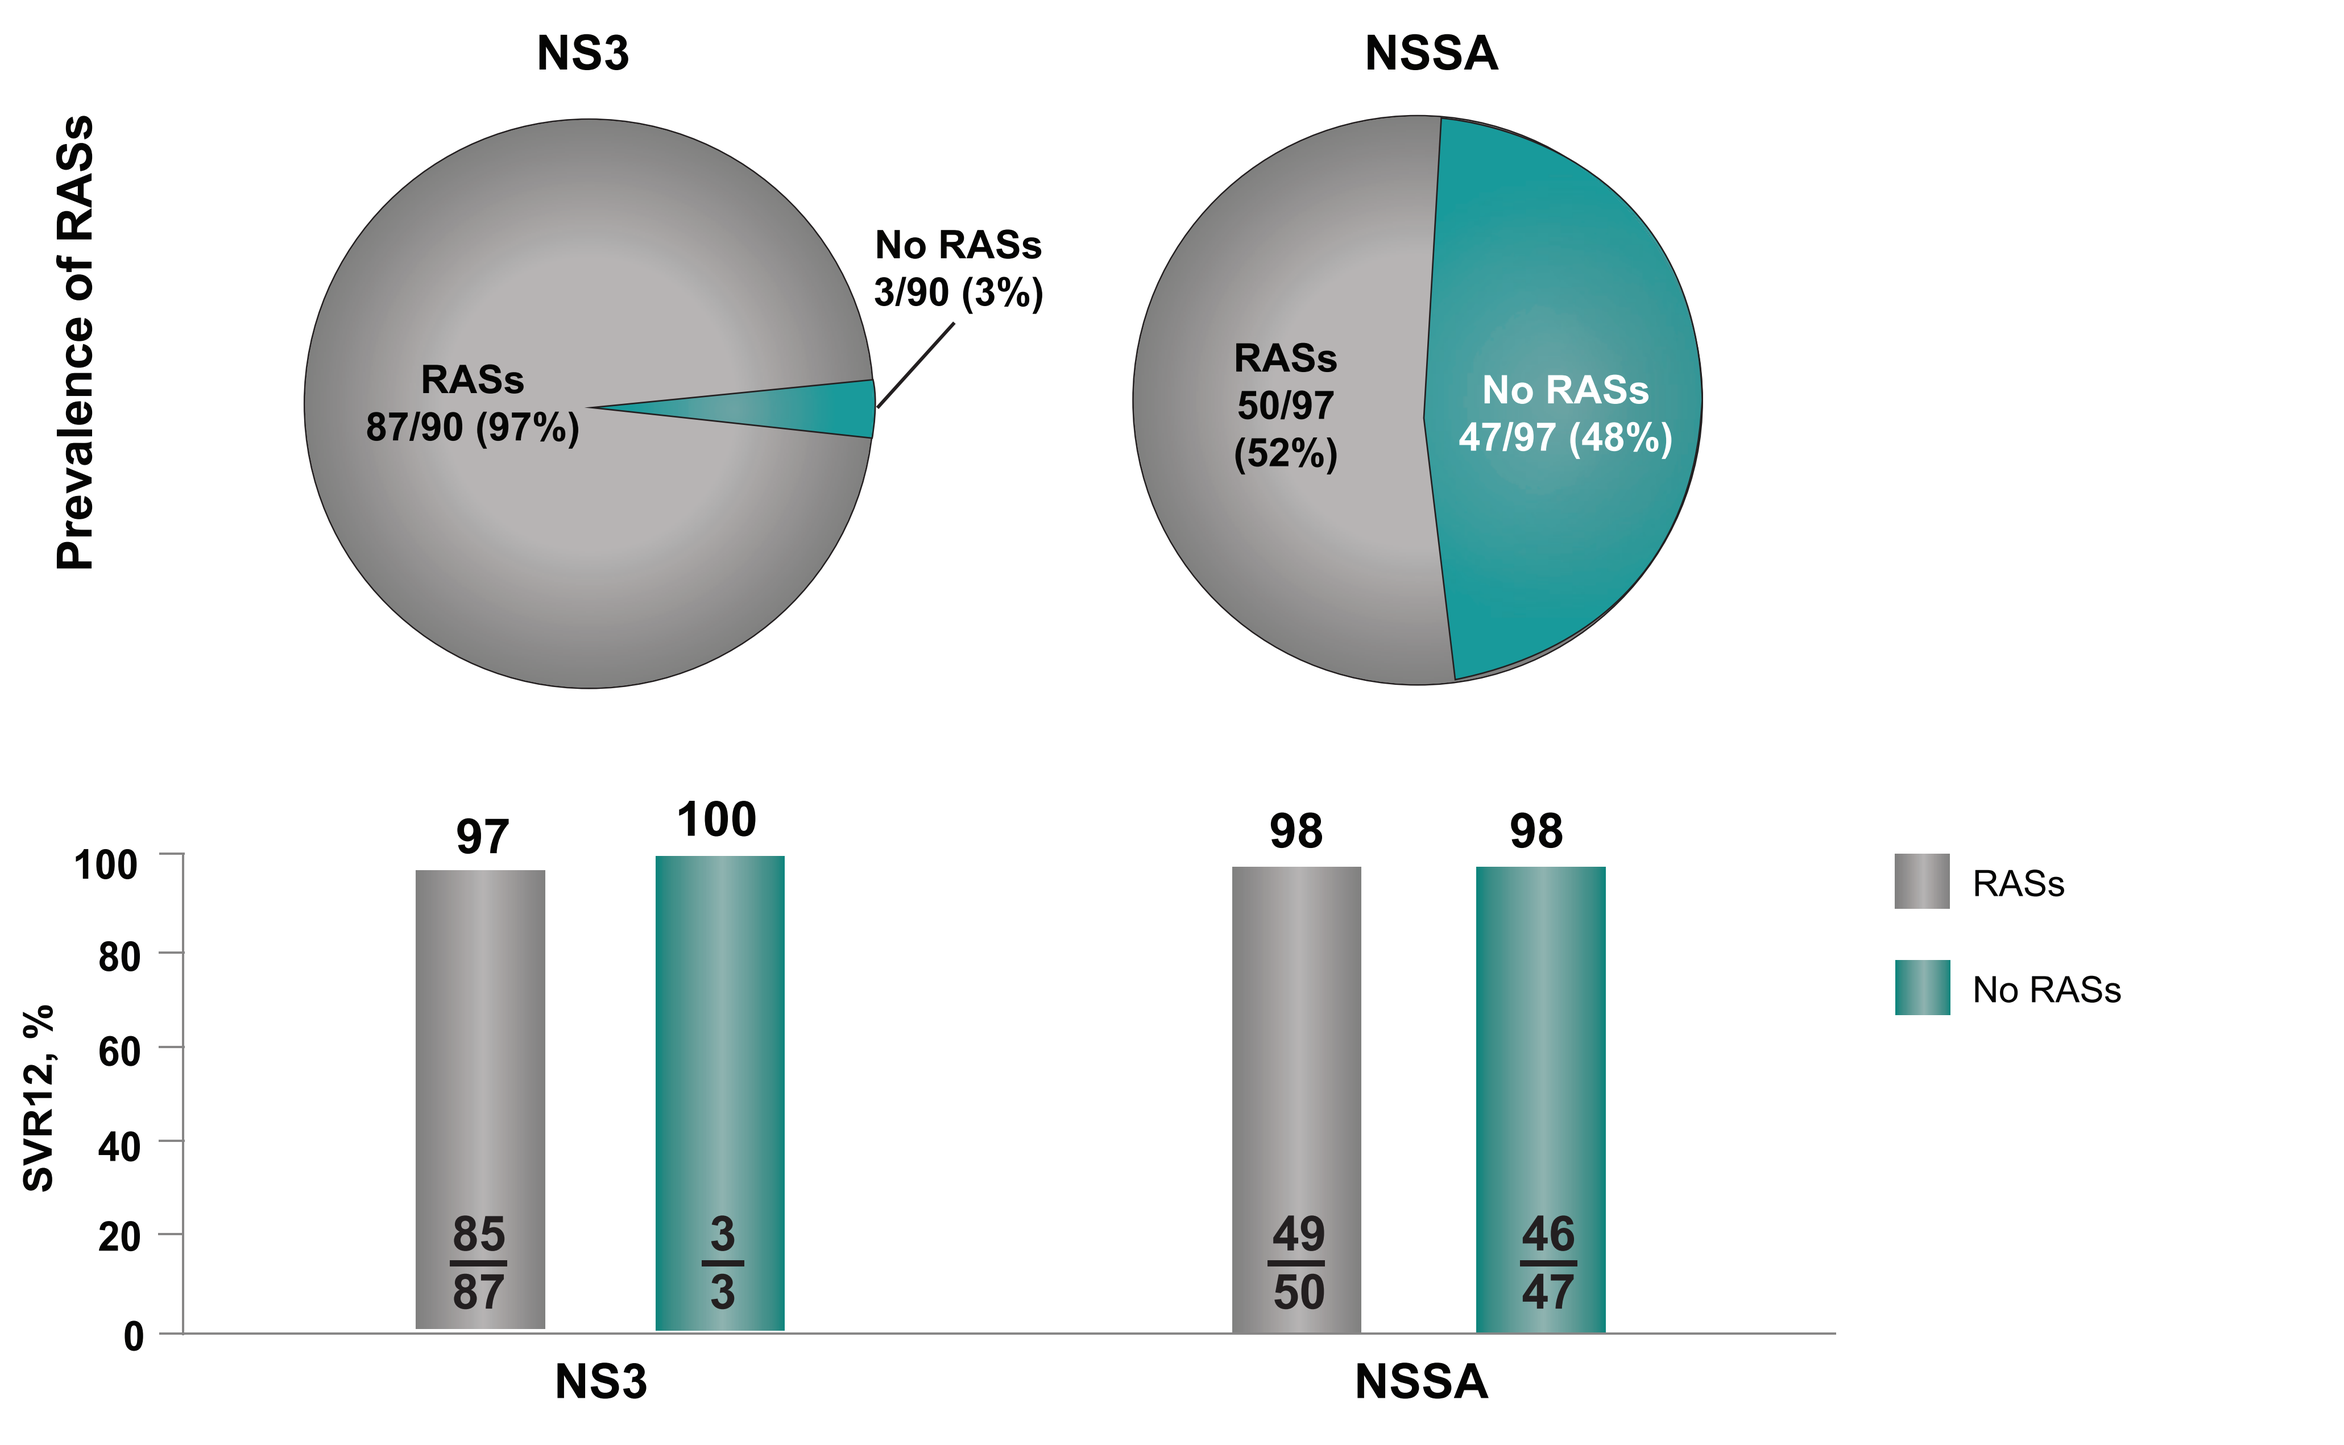

Prevalence of RASs
NS3
NSSA
RASs 87/90 (97%)
No RASs 3/90 (3%)
RASs 50/97 (52%)
No RASs 47/97 (48%)
SVR12, %
100
80
60
40
20
0
97
100
98
98
85/87
3/3
49/50
46/47
NS3
NSSA
RASs
No RASs

Next-generation sequencing: detection limit for variants of 15% prevalence.

- Based on next-generation sequencing with a detection limit of 1%, 9 participants had Y93 RASs present at baseline (**Table 4**)
 - Four participants had Y93 present in ≥11% of the circulating viral population

Table 4: Presence of Y93 RASs at baseline (next-generation sequencing 1%)

Participant	Treatment history	Treatment	Y93H in viral population (%)	Other NS5A RASs (% in total viral population)	Treatment outcome
1	TN	EBR/GZR + SOF + RBV for 8 weeks	1	A30V (2)	SVR12
2	TN	EBR/GZR + SOF + RBV for 8 weeks	44	P58S (99); S62A (3) S62T (62)	Relapse
3	TN	EBR/GZR + SOF for 12 weeks	87	A30L (7); A30S (5) A30V (2); S62T (90) S621T (62)	SVR12
4	TN	EBR/GZR + SOF for 12 weeks	11	A30M (14)	SVR12
5	TE	EBR/GZR + SOF for 12 weeks	2	S62T (99)	SVR12
6	TE	EBR/GZR + SOF + RBV for 12 weeks	1	A30V (1)	SVR12
7	TE	EBR/GZR + SOF + RBV for 12 weeks	28	A30F (17); A30V (75) S62T (11); S62L (7)	SVR12
8	TE	EBR/GZR + SOF + RBV for 12 weeks	2	P58T (1)	SVR12
9	TE	EBR/GZR + SOF for 16 weeks	7	P58H (1); P58T (1)	SVR12

TE, treatment-experienced; TN, treatment-naive.

Adverse events

- AEs are shown in **Table 5**

Table 5. Adverse events

	EBR/GZR + SOF + RBV 8 weeks (n = 23)	EBR/GZR + SOF 12 weeks (n = 41)	EBR/GZR + SOF + RBV 12 weeks (n = 18)	EBR/GZR + SOF 16 weeks (n = 18)
Drug-related AEs, n (%)				
Fatigue	3 (13.0)	9 (22.0)	9 (50.0)	5 (27.8)
Nausea	4 (17.4)	4 (9.8)	6 (33.3)	3 (16.7)
Skin rash/pruritus	2 (8.7)	3 (7.3)	8 (44.4)	2 (11.1)
Serious AE (SAE), n (%)†	0	1 (2.4)	3 (16.7)	1 (5.6)
Discontinued study medication due to AE, n (%)‡	0	0	0	1 (5.6)
Hemoglobin <10 g/dL, n (%)¶	0	1 (2.4)	2 (11.1)	0
Total bilirubin >5× baseline, n (%)	0	0	0	0
ALT/AST >5× ULN, n (%)	0	0	0	0

AST, aspartate aminotransferase; ULN, upper limit of normal.
†SAEs of lung infection, creatinine increased, chest pain, opiate overdose, and cellulitis were reported.
‡One participant had a drug-related SAE of vomiting on day 4 and then subsequently discontinued treatment at day 7 due to drug-related cellulitis.
¶Lowest hemoglobin level was 8.9 g/dL.

Conclusions

- High efficacy of EBR/GZR was demonstrated in treatment-naive and -experienced cirrhotic HCV GT3-infected participants
 - In the mFAS analysis, SVR12 was 100% among participants receiving EBR/GZR + SOF for 12 weeks
 - There were no virologic failures among participants receiving EBR/GZR + SOF ± RBV for 12 or 16 weeks
- Treatment duration longer than 12 weeks is not needed
- High efficacy regardless of presence of baseline NS5A RASs or participant characteristics
- Generally safe and well tolerated

References

1. Messina JP et al. *Hepatology.* 2015;61:77-87.
2. Zepatier [package insert]. Whitehouse Station, NJ: Merck Sharp & Dohme Corp.; 2017.
3. Summa V et al. *Antimicrob Agents Chemother.* 2012;56:4161-4167.
4. Harper S et al. *ACS Med Chem Lett.* 2012;3:332-336.
5. Coburn CA et al. *ChemMedChem.* 2013;8:1930-1940.
6. Kwo P et al. *Gastroenterology.* 2017;152:164-175.e4.
7. Roth D et al. *Lancet.* 2015;386:1537-1545.
8. Rockstroh JK et al. *Lancet HIV.* 2015;2:e319-e327.
9. Zeuzem S et al. *Ann Intern Med.* 2015;163:1-13.
10. Sovaldi [package insert]. Foster City, CA: Gilead Sciences; 2017.
11. Lawitz E et al. *Hepatology.* 2017;65(2):439-450.
12. Foster GR et al. *Hepatology.* 2016;64(suppl 1):39A.

Acknowledgments

- We extend our gratitude to the participants, their families, investigators, and site personnel who participated in this study
- Medical writing assistance was provided by Tim Ibbotson, PhD, of ApotheCom (Yardley, PA, USA) and funded by Merck & Co., Inc., Kenilworth, NJ, USA.

Copyright © 2017 Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc. All rights reserved.