Poster #THU-246

EASL
The International Liver Congress
Amsterdam, The Netherlands

19-23 April 2017

Prevention of Liver-Related Complications With Elbasvir/Grazoprevir in Hepatitis C Infected Patients Who Are Receiving Opioid Agonist Therapy (OAT)

Shelby Corman[1]; Yiling Jiang[2]; Heather Loryn Platt[3]; Chizoba Nwankwo[3]

[1]Pharmerit International, Bethesda, MD, USA; [2]Merck, Sharp & Dohme Limited, Hoddesdon, Hertfordshire, UK; [3]Merck & Co., Inc., Kenilworth, NJ, USA

Background

Background

- The World Health Organization (WHO) estimates that 130-150 million people are infected with hepatitis C virus (HCV) worldwide,[1] with 1.1% of the world's population chronically infected[2]
- However, among people who inject drugs (PWID), the prevalence of HCV infection is estimated at 67%[3]
- WHO recommends that all adults and children with chronic HCV infection, including PWID, should be assessed for antiviral treatment[4]
- The C-EDGE CoSTAR trial compared immediate treatment with elbasvir/grazoprevir (EBR/GZR), a direct-acting antiviral, to delayed treatment (control group) in treatment-naïve patients with genotype (GT) 1, 4, or 6 chronic hepatitis C (CHC) who were receiving opioid agonist therapy (OAT)[5-6]

Objective

- The objective of this study was to model the long-term impact of EBR/GZR on the incidence of liver-related complications in patients receiving OAT by extending the results of C-EDGE CoSTAR over a 30-year time horizon

Methods

- A Markov model was constructed to evaluate the cost and effectiveness of EBR/GZR±ribavirin (RBV) over a 30-year time horizon
- The target population was patients infected with CHC GT1 or 4, stratified by presence of cirrhosis
- The model consists of 16 health states encompassing METAVIR fibrosis score (F0-F4), treatment success or failure, decompensated cirrhosis (DC), hepatocellular carcinoma (HCC), liver transplant, and liver-related death (**Figure 1**)

Figure 1: State transition model for chronic HCV and liver disease model

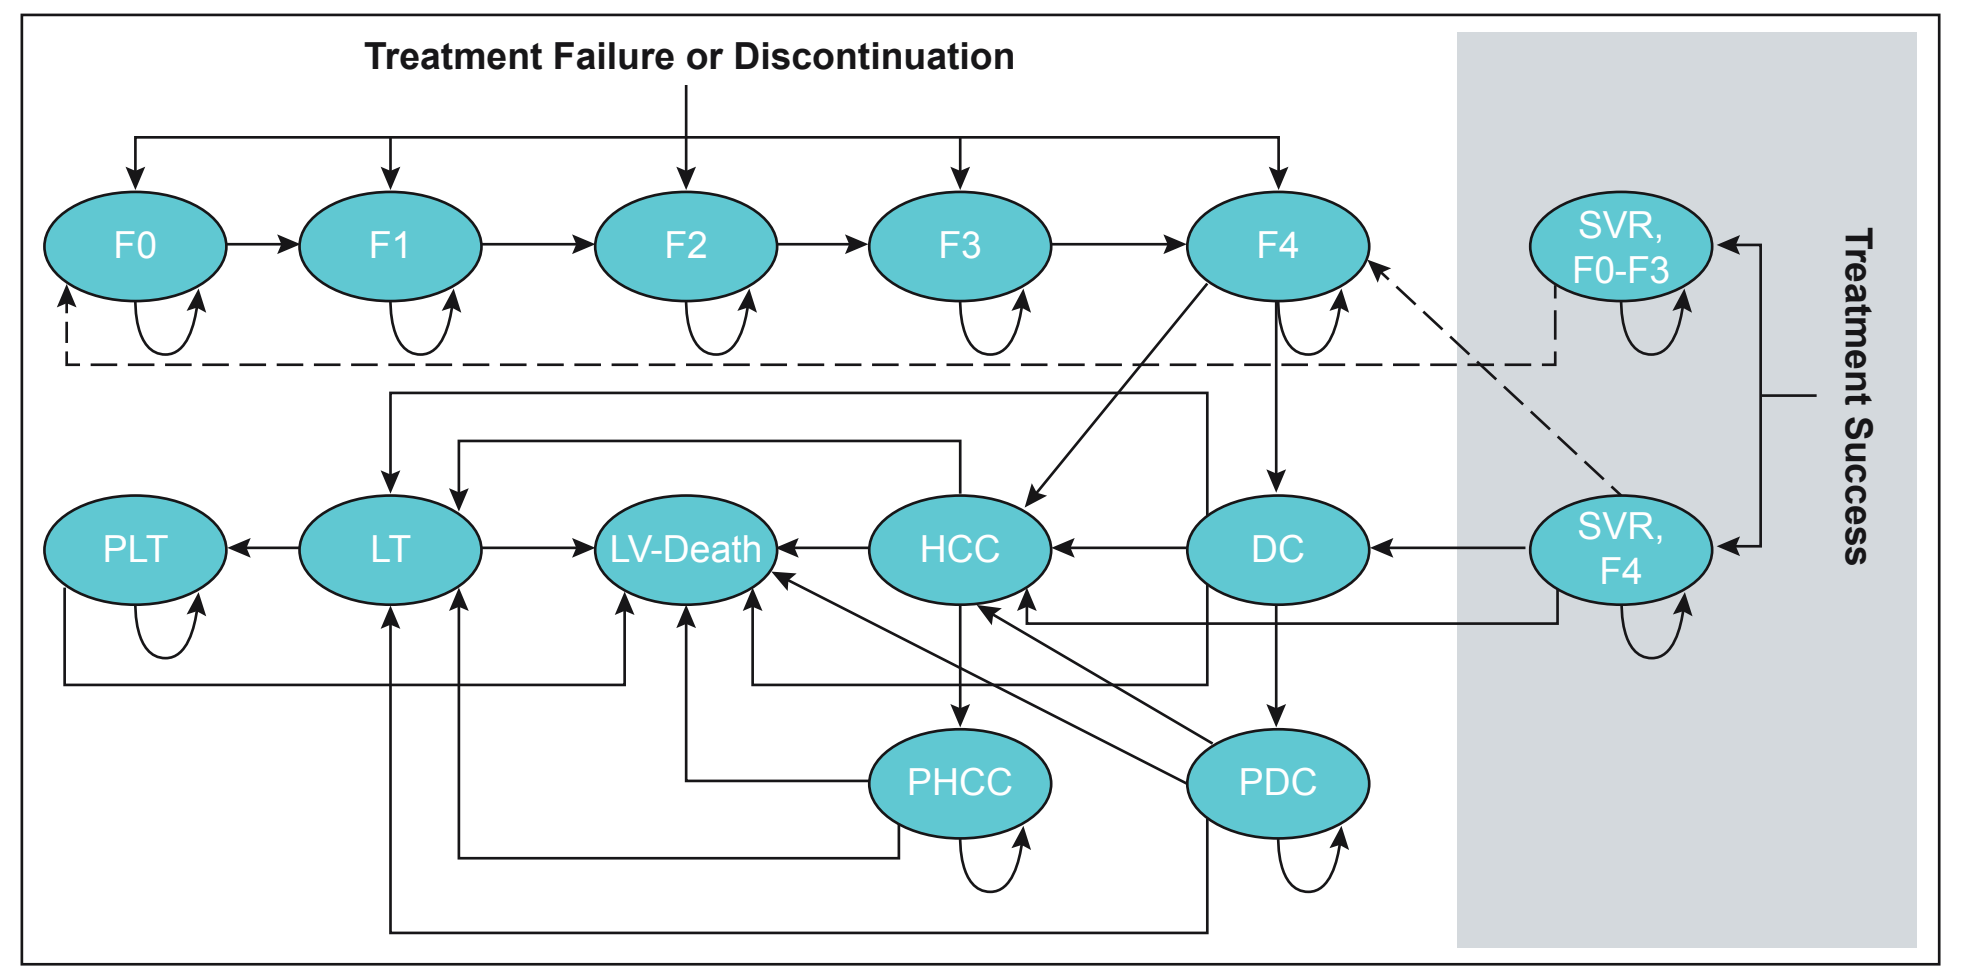
Treatment Failure or Discontinuation
F0
F1
F2
F3
F4
SVR, F0-F3
Treatment Success
PLT
LT
LV-Death
HCC
DC
SVR, F4
PHCC
PDC

Hepatic fibrosis stage was based on METAVIR fibrosis scoring system: F0 = no fibrosis; F1 = portal fibrosis without septa; F2 = portal fibrosis with few septa; F3 = portal fibrosis with numerous septa without cirrhosis; and F4 = compensated cirrhosis; DC = decompensated cirrhosis; HCC = hepatocellular carcinoma; PDC = one-year post decompensated cirrhosis; PHCC = one-year post hepatocellular carcinoma; SVR12 = sustained virologic response 12 weeks after cessation of treatment.

Methods (continued)

Model Inputs

- Baseline patient characteristics, treatment regimens, and rates of sustained virologic response at 12 weeks (SVR12) and reinfection were obtained from C-EDGE CoSTAR (**Tables 1-2**)[5-6]
 – Results from the immediate and delayed treatment groups were pooled for use in the model
- Cost and utility inputs were obtained from published sources (**Table 3**)[7-8]
- Wholesale acquisition cost of $4,550 per week was used for EBR/GZR

Model Outputs

- The primary outcome was incremental cost-utility ratio (ICUR) for EBR/GZR vs no treatment
- Other outcomes included cumulative proportion of patients developing cirrhosis, DC, and HCC; receiving liver transplants; and dying of liver-related causes over the time horizon, and the number of these events prevented per 1000 patients treated with EBR/GZR

Table 1: Baseline characteristics, C-EDGE CoSTAR[5-6]

Characteristic	Proportion
Baseline fibrosis stage	
F0	0.244
F1	0.244
F2	0.244
F3	0.070
F4	0.199
Males	0.764
Age at baseline	
18-35	0.15
36-50	0.458
51-64	0.382
≥65	0.01

Table 2: Treatment and outcomes, C-EDGE CoSTAR[5-6]

Variable	Base Case (95% CI)
SVR12, base case (95% CI)	
GT1a	0.938 (0.898-0.966)
GT1b	0.932 (0.813-0.986)
GT4	0.944 (0.727-0.999)
Treatment discontinuation, base case (95% CI)	0.007 (0.001-0.024)
Reinfection, rate per 100 person-years	2.5 (0.8-5.9)

Table 3: Annual health state cost and utility inputs[7-8]

	Utility		Cost	
Input	Base Case	Range (±5%)	Base Case	95% CI
F0-F1	0.77	0.73-0.81	$793	$595-$991
F2	0.77	0.73-0.81	$803	$602-$1,004
F3	0.66	0.63-0.69	$1,630	$1,223-$2,038
F4	0.55	0.52-0.58	$1,901	$1,426-$2,376
DC	0.45	0.43-0.47	$21,122	$15,842-$26,403
HCC	0.45	0.43-0.47	$38,841	$29,131-$48,551
Post liver transplant, year 1	0.45	0.43-0.47	$112,217	$84,163-$140,271
Post liver transplant, subsequent years	0.67	0.64-0.70	$29,475	$22,106-$36,844
Post SVR, F0-F2	0.82	0.78-0.86	0	-
Post SVR, F3-F4	0.72	0.68-0.76	0	-
Annual discount rate	0.03	0-0.05	0.03	0-0.05

Results

- Over 30 years, the proportion of patients developing liver-related complications was substantially reduced in patients receiving EBR/GZR compared to no treatment (**Figures 2** and **3**)

Figure 2: Proportion of patients developing liver-related complications over 30-year time horizon

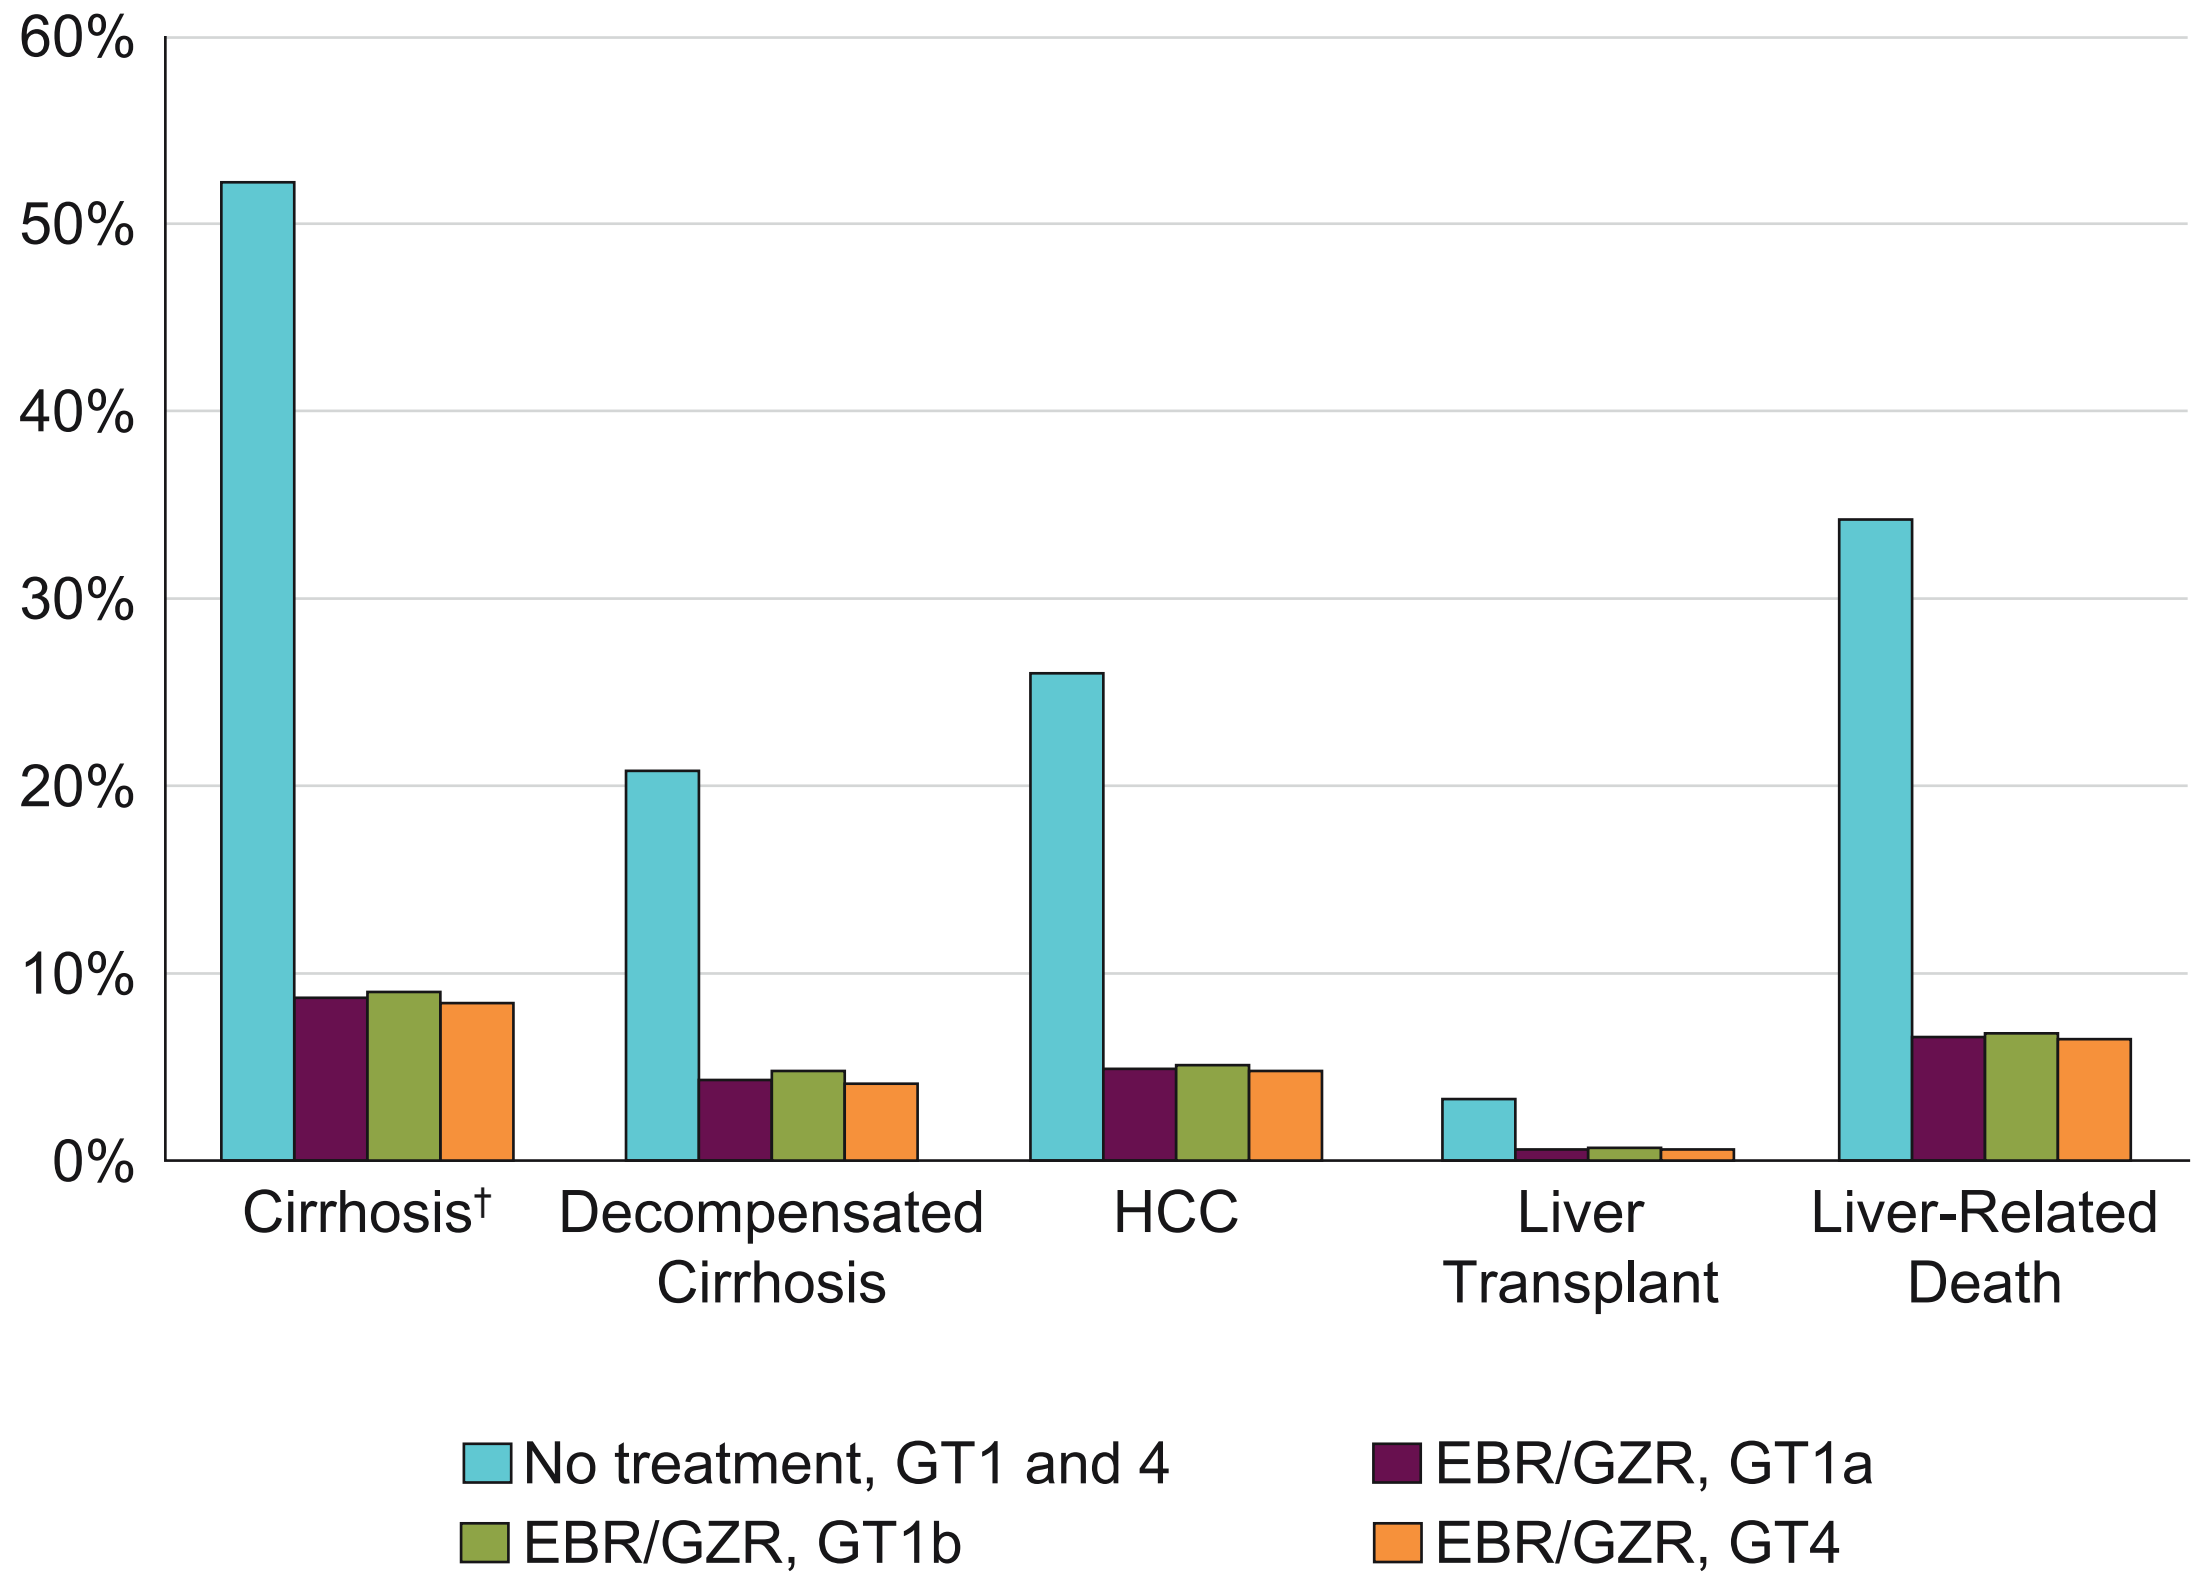
60%
50%
40%
30%
20%
10%
0%
Cirrhosis†
Decompensated Cirrhosis
HCC
Liver Transplant
Liver-Related Death
No treatment, GT1 and 4
EBR/GZR, GT1a
EBR/GZR, GT1b
EBR/GZR, GT4

†Among those noncirrhotic at baseline.

Figure 3: Cases of liver complications prevented per 1000 GT1 and 4 patients treat with EBR/GZR vs no treatment over 30-year horizon

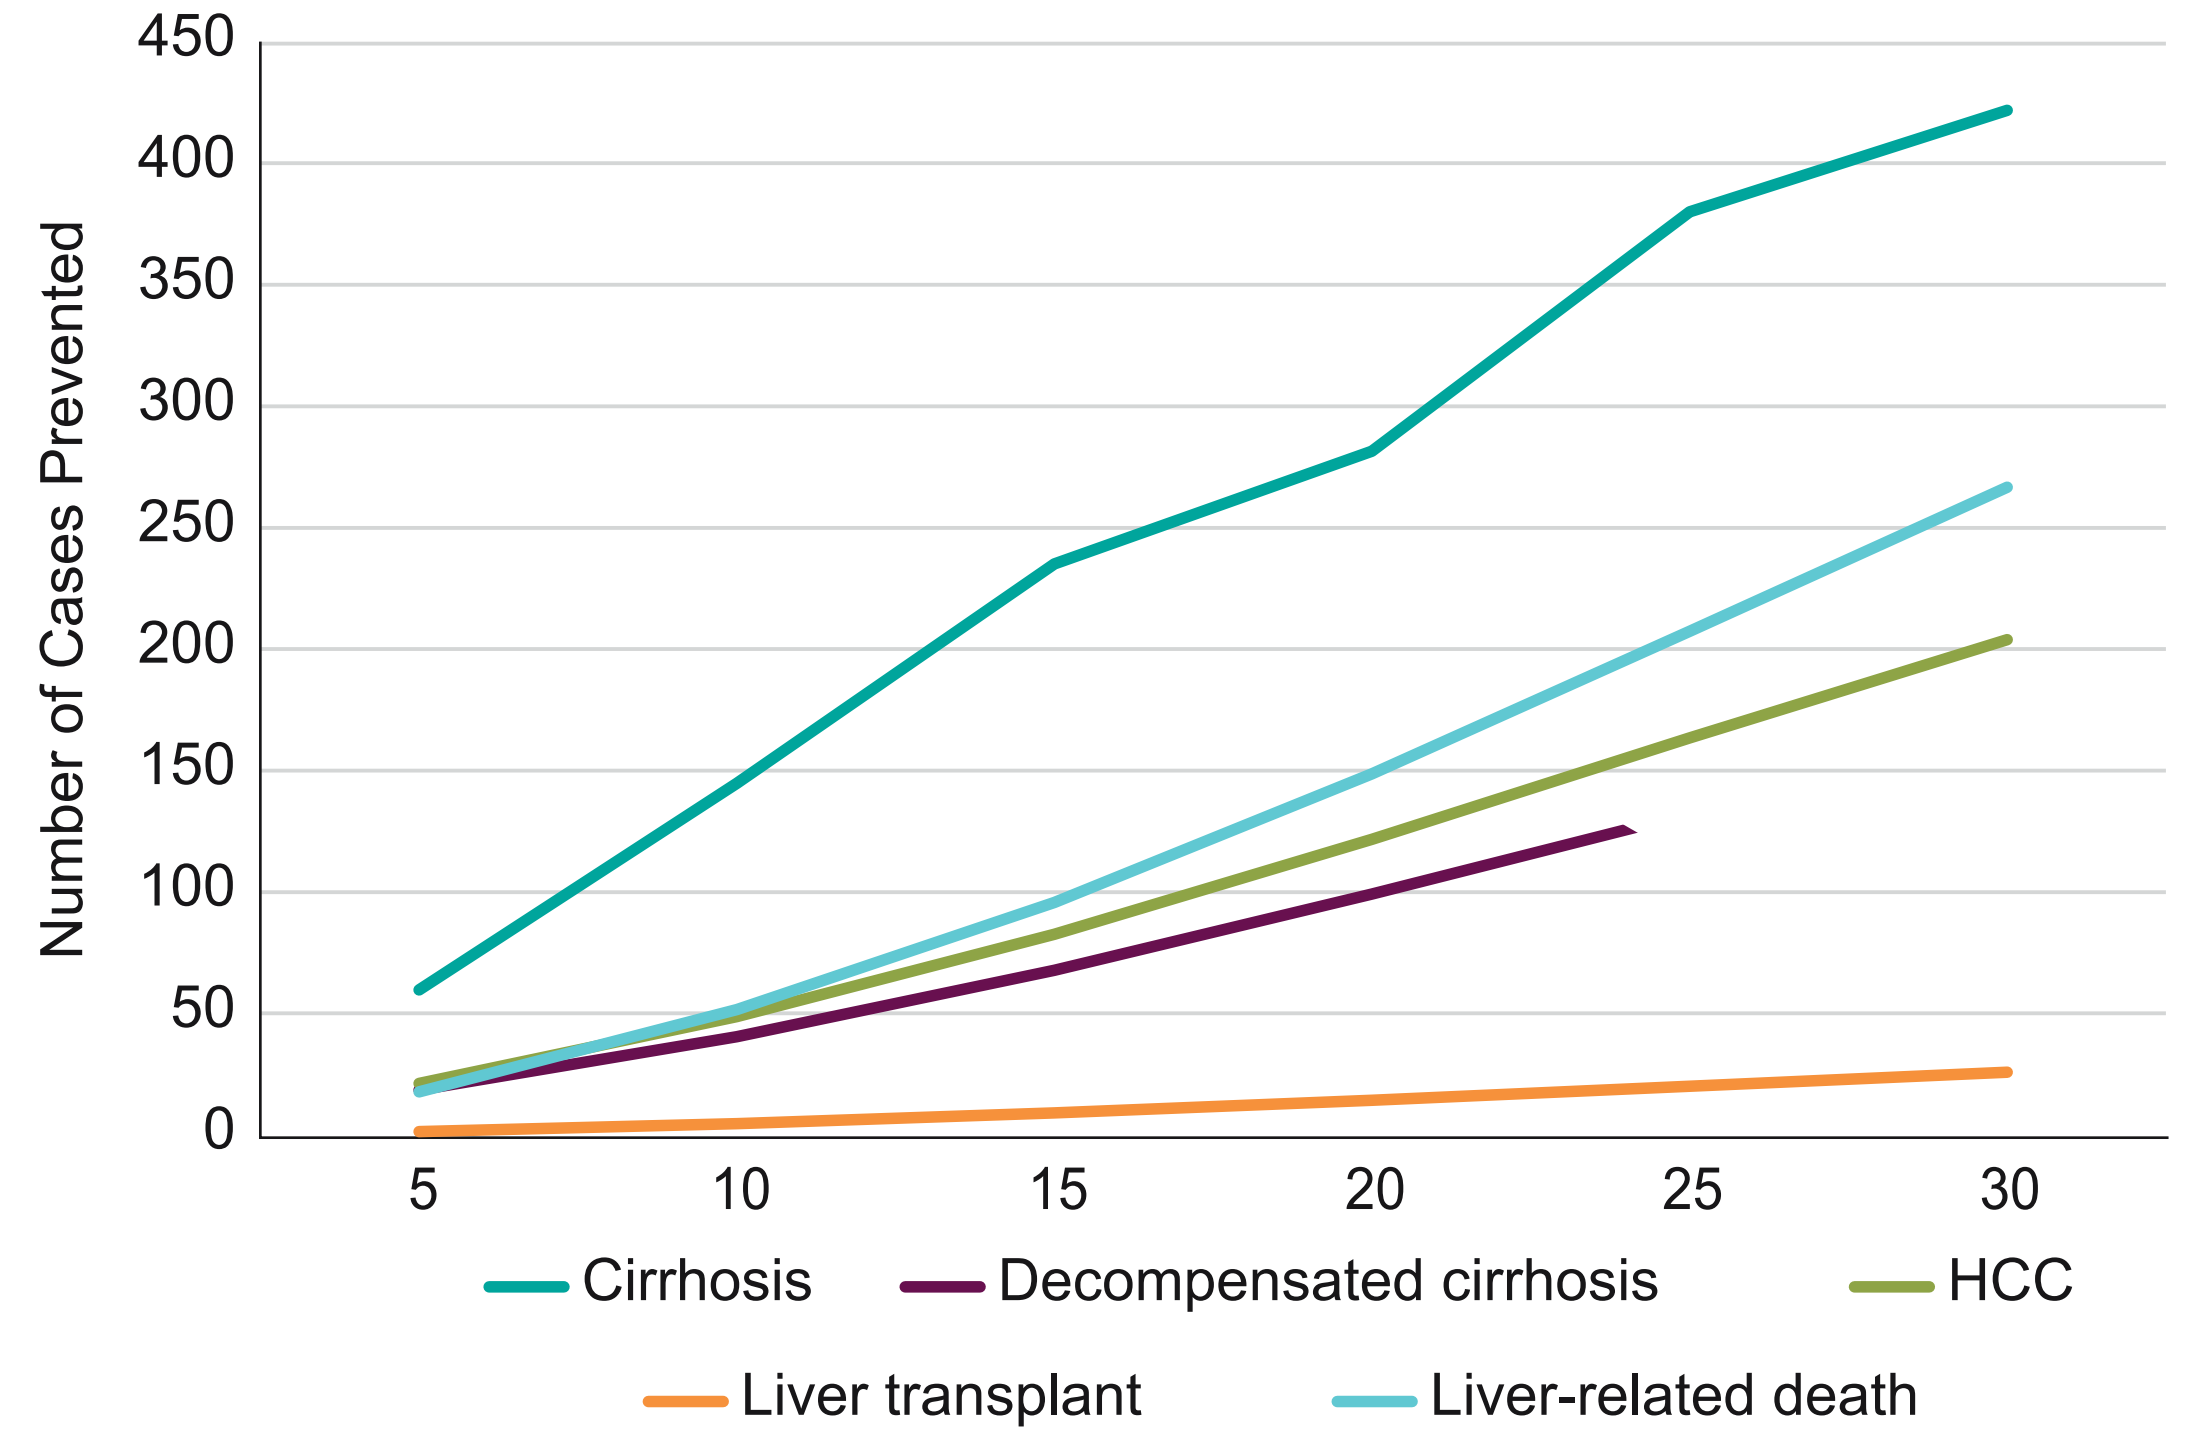
Number of Cases Prevented
450
400
350
300
250
200
150
100
50
0
5
10
15
20
25
30
Cirrhosis
Decompensated cirrhosis
HCC
Liver transplant
Liver-related death

- Of all subgroups, cirrhotic patients had the highest cumulative disease incidence (**Figure 3**) and therefore accrue the highest cumulative disease cost (**Figure 4**)

Figure 4: 30-year disease costs, overall and by subgroup GT1 and 4 patients treated with EBR/GZR vs no treatment

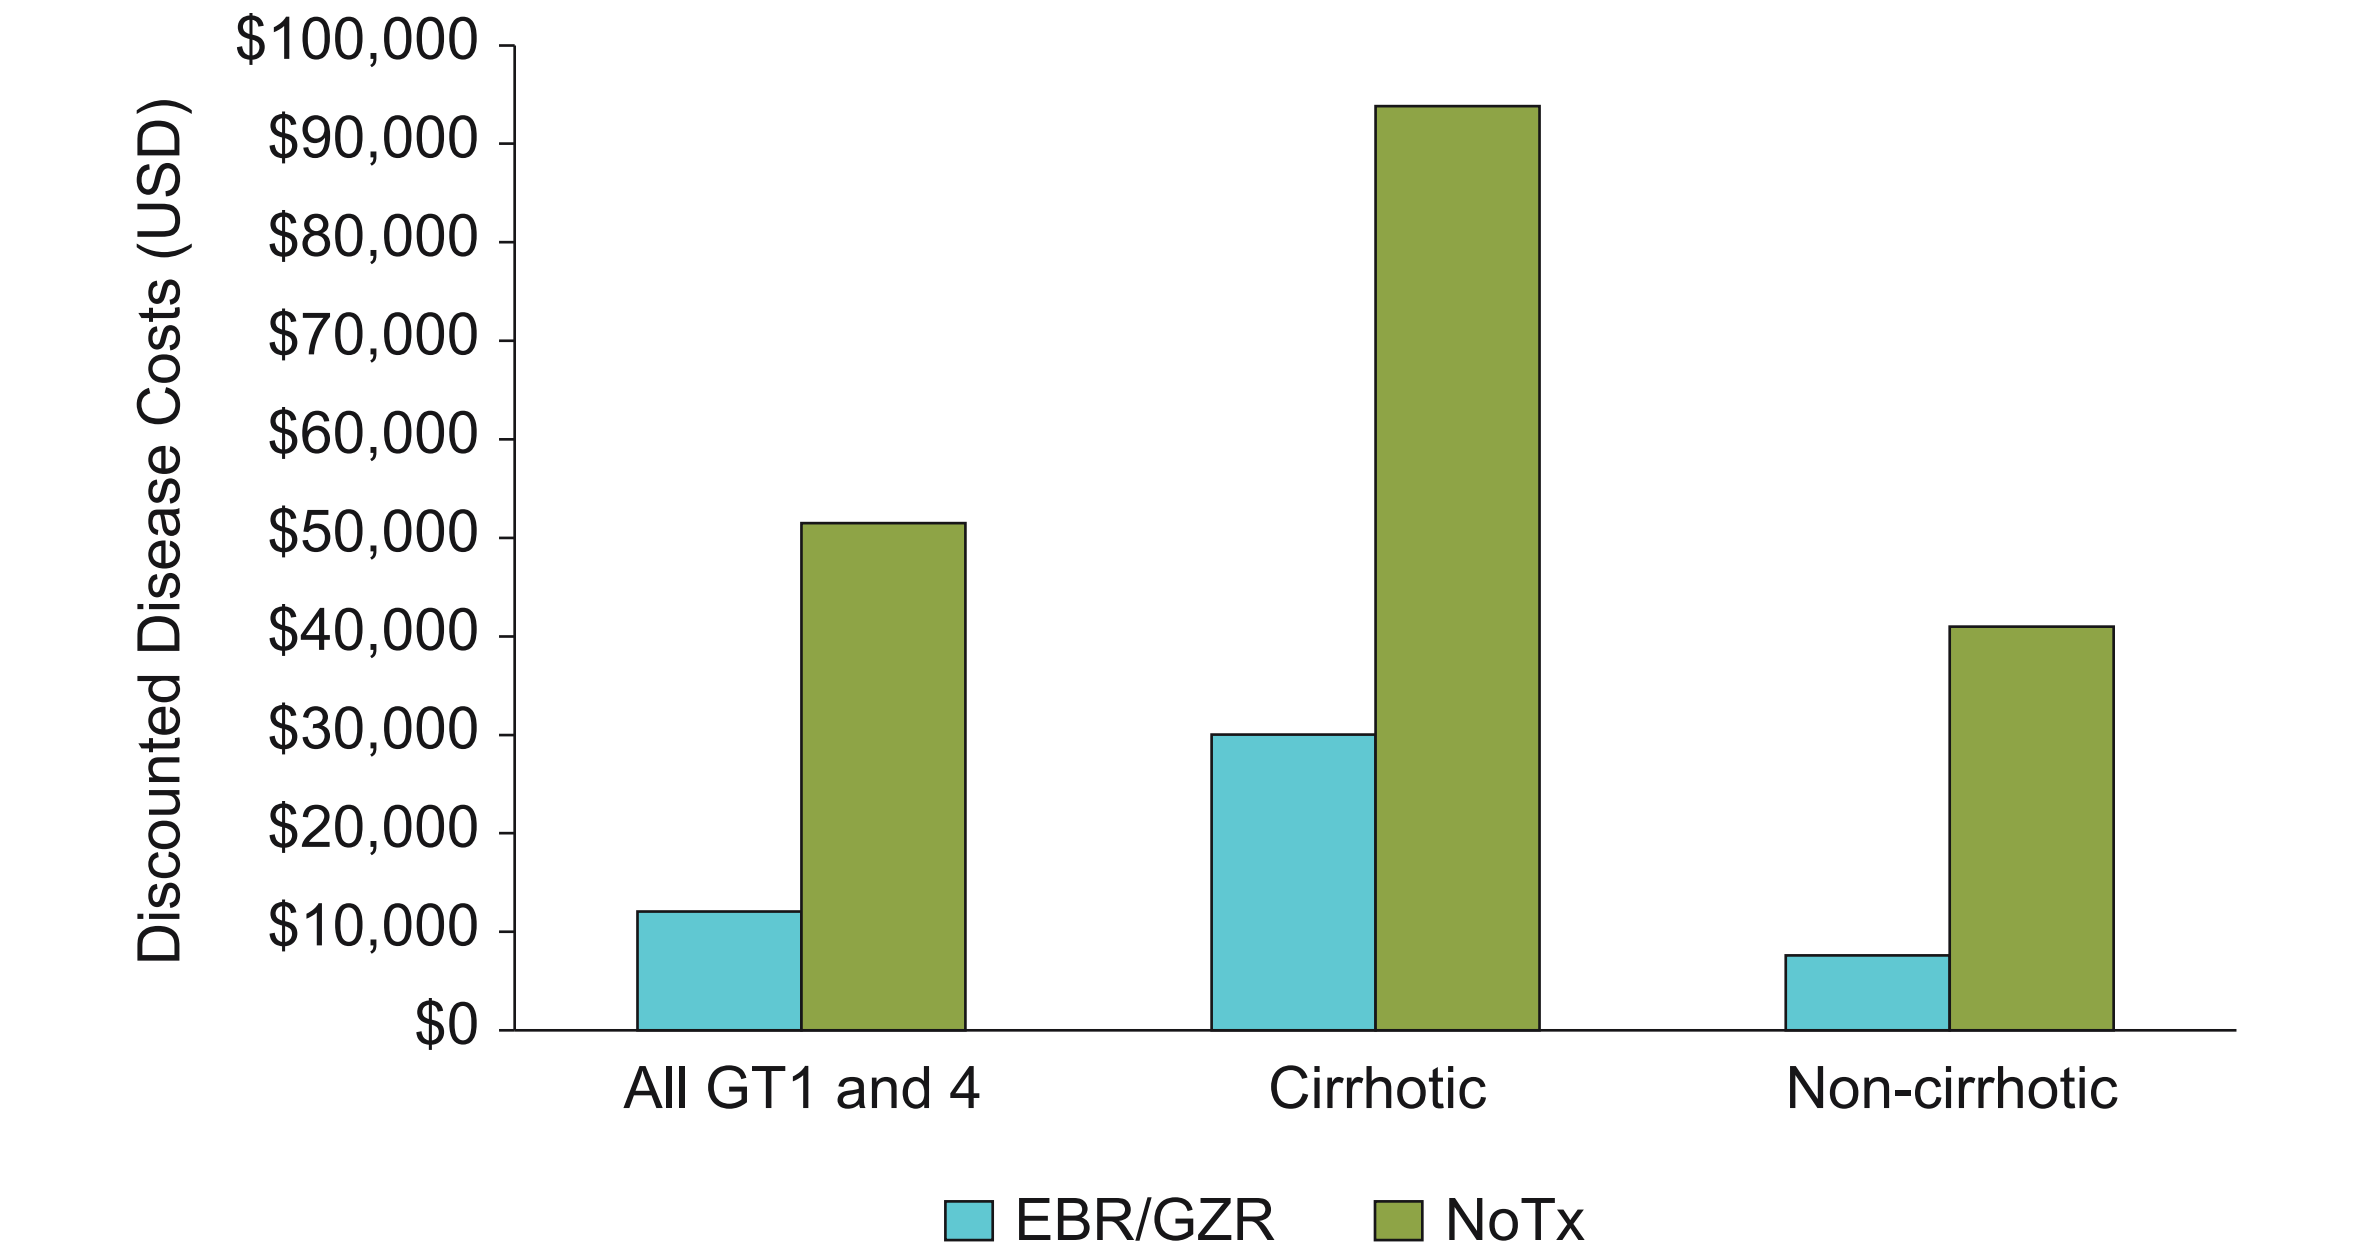
Discounted Disease Costs (USD)
$100,000
$90,000
$80,000
$70,000
$60,000
$50,000
$40,000
$30,000
$20,000
$10,000
$0
All GT1 and 4
Cirrhotic
Non-cirrhotic
EBR/GZR
NoTx

- EBR/GZR was associated with more quality-adjusted life years (QALYs) than no treatment in all genotypes studied, and ICURs were less than $6000/QALY for all genotypes over 30 years (**Table 4**), and $1500/QALY over lifetime

Table 4: Base case results over 30-year time horizon

	No Treatment	EBR/GZR		
Result	GT1 and 4	GT1a	GT1b	GT4
Discounted QALYs	10.8146	13.4935	13.4761	13.5131
Discounted costs ($)	$51,513	$66,567	$66,814	$66,291
ICUR vs no treatment ($/QALYs)	--	$5,620	$5,749	$5,477

Conclusions

- Use of EBR/GZR for the treatment of CHC in patients receiving OAT in the United States was projected to prevent a considerable number of cases of cirrhosis, decompensated cirrhosis, HCC, liver transplants, and liver-related death over 30 years compared to no treatment
- Thus EBR/GZR was projected to be a cost-effective therapy for CHC GT1- and 4-infected patients on OAT in the United States

References

1. World Health Organization. Hepatitis C Fact sheet. Available at: http://www.who.int/mediacentre/factsheets/fs164/en/.
2. Gower E, Estes C, Blach S, et al. *J Hepatol.* 2014;61(1 Suppl):S45-57.
3. Rafiq SM, Banik GR, Khan S, et al. *Infect Disord Drug Targets.* 2014;14:93-100.
4. World Health Organization. Guidelines for the Screening, Care and Treatment of Persons with Hepatitis C Infection, 2016. Available at: http://www.who.int/hiv/pub/hepatitis/hepatitis-c-guidelines/en/.
5. Dore GJ, Altice F, Litwin AH, et al. *Ann Intern Med.* 2016;165:625-634.
6. Dore GJ, Grebely J, Altice F, et al. Presented at: The Liver Meeting 2016®; November 11-15, 2016; Boston, MA, USA.
7. Martin NK, Vickerman P, Miners A, et al. *Hepatology.* 2012;55:49-57.
8. Chhatwal J, Kanwal F, Roberts MS, et al. *Ann Intern Med.* 2015;162(6):397-406.

Sponsorship

Merck & Co., Inc.

Copyright © 2017 Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc. Kenilworth, NJ, USA. All rights reserved.