Clinical Significance of gp120 Polymorphisms, TMR IC_{50} FC, and HIV-1 Subtype in BRIGHTE

M. Gartland[1], P. Ackerman[2], F. Mannino[3], L. Garside[4], A. Clark[5], A. Pierce[1], M. Krystal[2], C. Llamoso[2], M. Lataillade[2]

[1]ViiV Healthcare, Research Triangle Park, United States; [2]ViiV Healthcare, Branford, United States; [3]GlaxoSmithKline, Collegeville, United States; [4]GlaxoSmithKline, London, United Kingdom; [5]ViiV Healthcare, London, United Kingdom

Poster 0503

Introduction

- Fostemsavir (FTR) is a prodrug of temsavir (TMR), a first-in-class, investigational attachment inhibitor being developed for heavily treatment-experienced (HTE) adults living with multi-drug–resistant HIV-1 who are unable to form a viable combination antiretroviral (ARV) regimen out of remaining fully active agents.[1,2]
- TMR binds to HIV-1 gp120, preventing viral attachment to, and entry into, host CD4+ T cells and other immune cells (**Figure 1**).[1,2]
- Among clinical isolates of HIV-1, a broad (>6-log) range of *in vitro* susceptibility to TMR has been observed, likely due to the substantial diversity in HIV-1 gp120.[3,4]
- Previous studies have identified amino acid substitutions at 4 gp120 positions that may influence HIV-1 susceptibility to TMR: S375H/I/M/N/T, M426L/P, M434I/K, and M475I (**Figure 2**).[4–6]
- A reliable clinical cut-off for *in vitro* FTR susceptibility tests has not yet been determined.

Figure 1. Mechanism of Action of TMR[1,2]

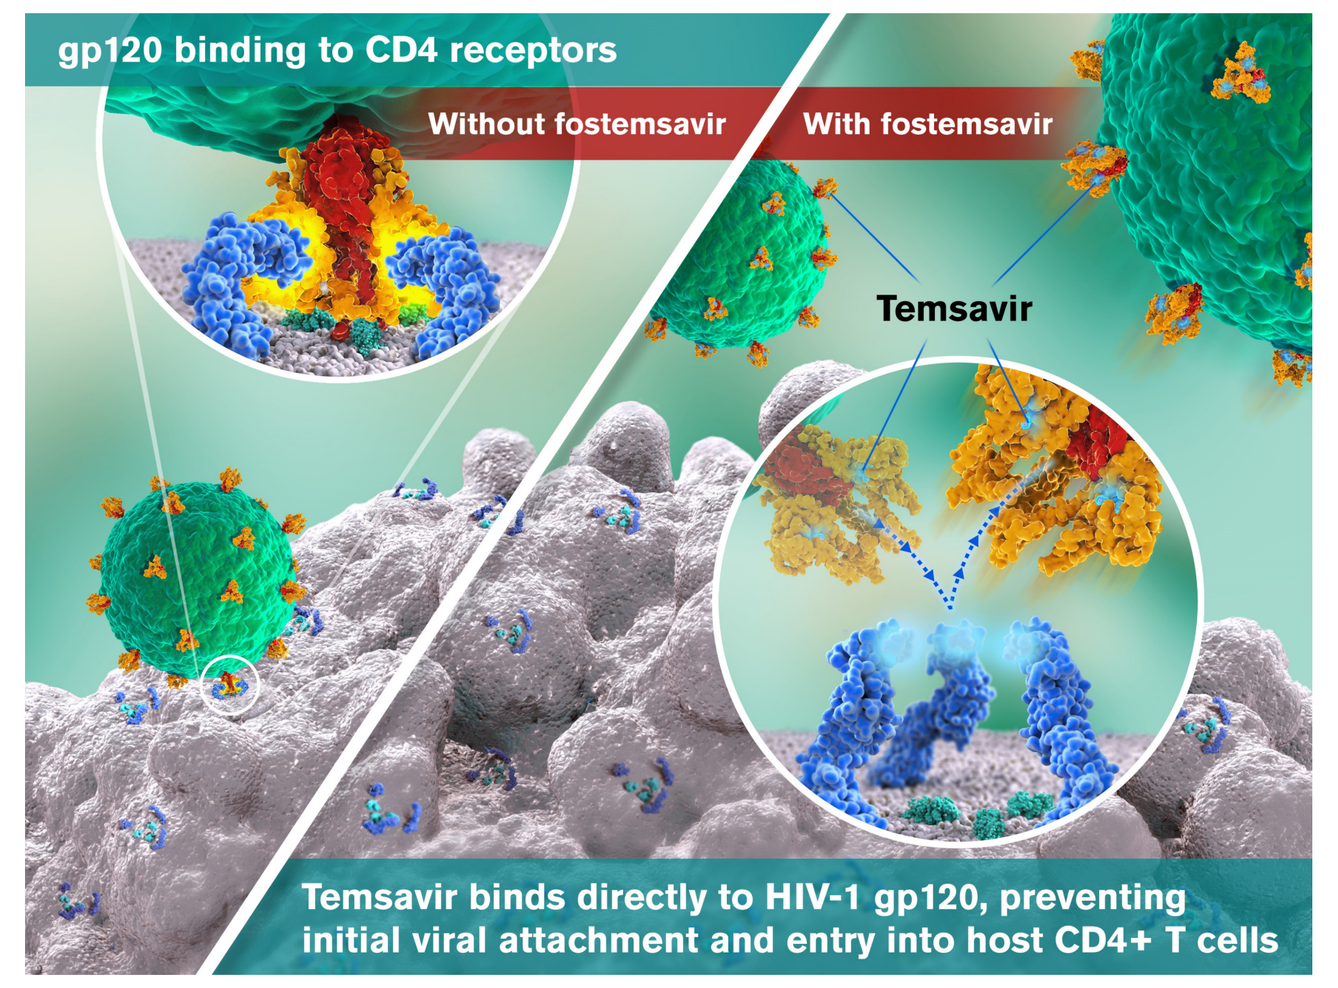
gp120 binding to CD4 receptors
Without fostemsavir
With fostemsavir
Temsavir
Temsavir binds directly to HIV-1 gp120, preventing initial viral attachment and entry into host CD4+ T cells

Figure 2. 3D Ribbon Structure of gp120 Bound to TMR[7]

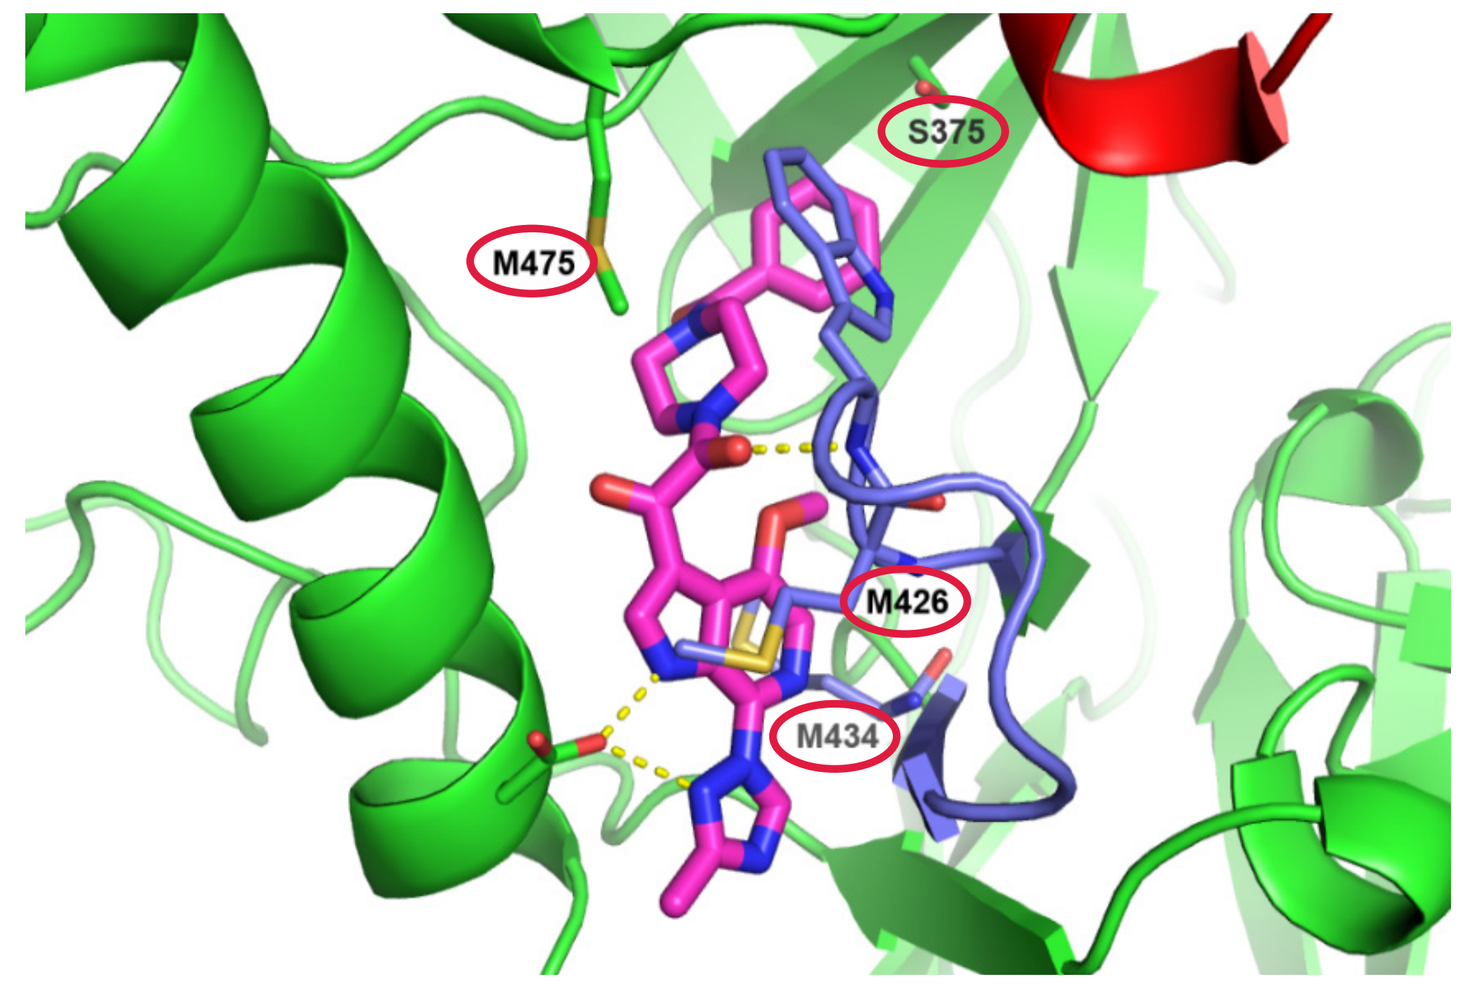
S375
M475
M426
M434

- BRIGHTE (NCT02362503; **Figure 3**) is an ongoing Phase 3 study investigating the efficacy and safety of FTR plus optimized background therapy (OBT) in HTE individuals who were failing their current regimen (confirmed HIV-1 RNA ≥400 c/mL).[1,2]
- Here we present the impact of key baseline (BL) factors including gp120 polymorphisms, TMR IC_{50} fold-change (FC), and HIV-1 subtype on short-term virologic outcomes and durability of response to FTR in the Randomized Cohort (RC).

Figure 3. Study Design

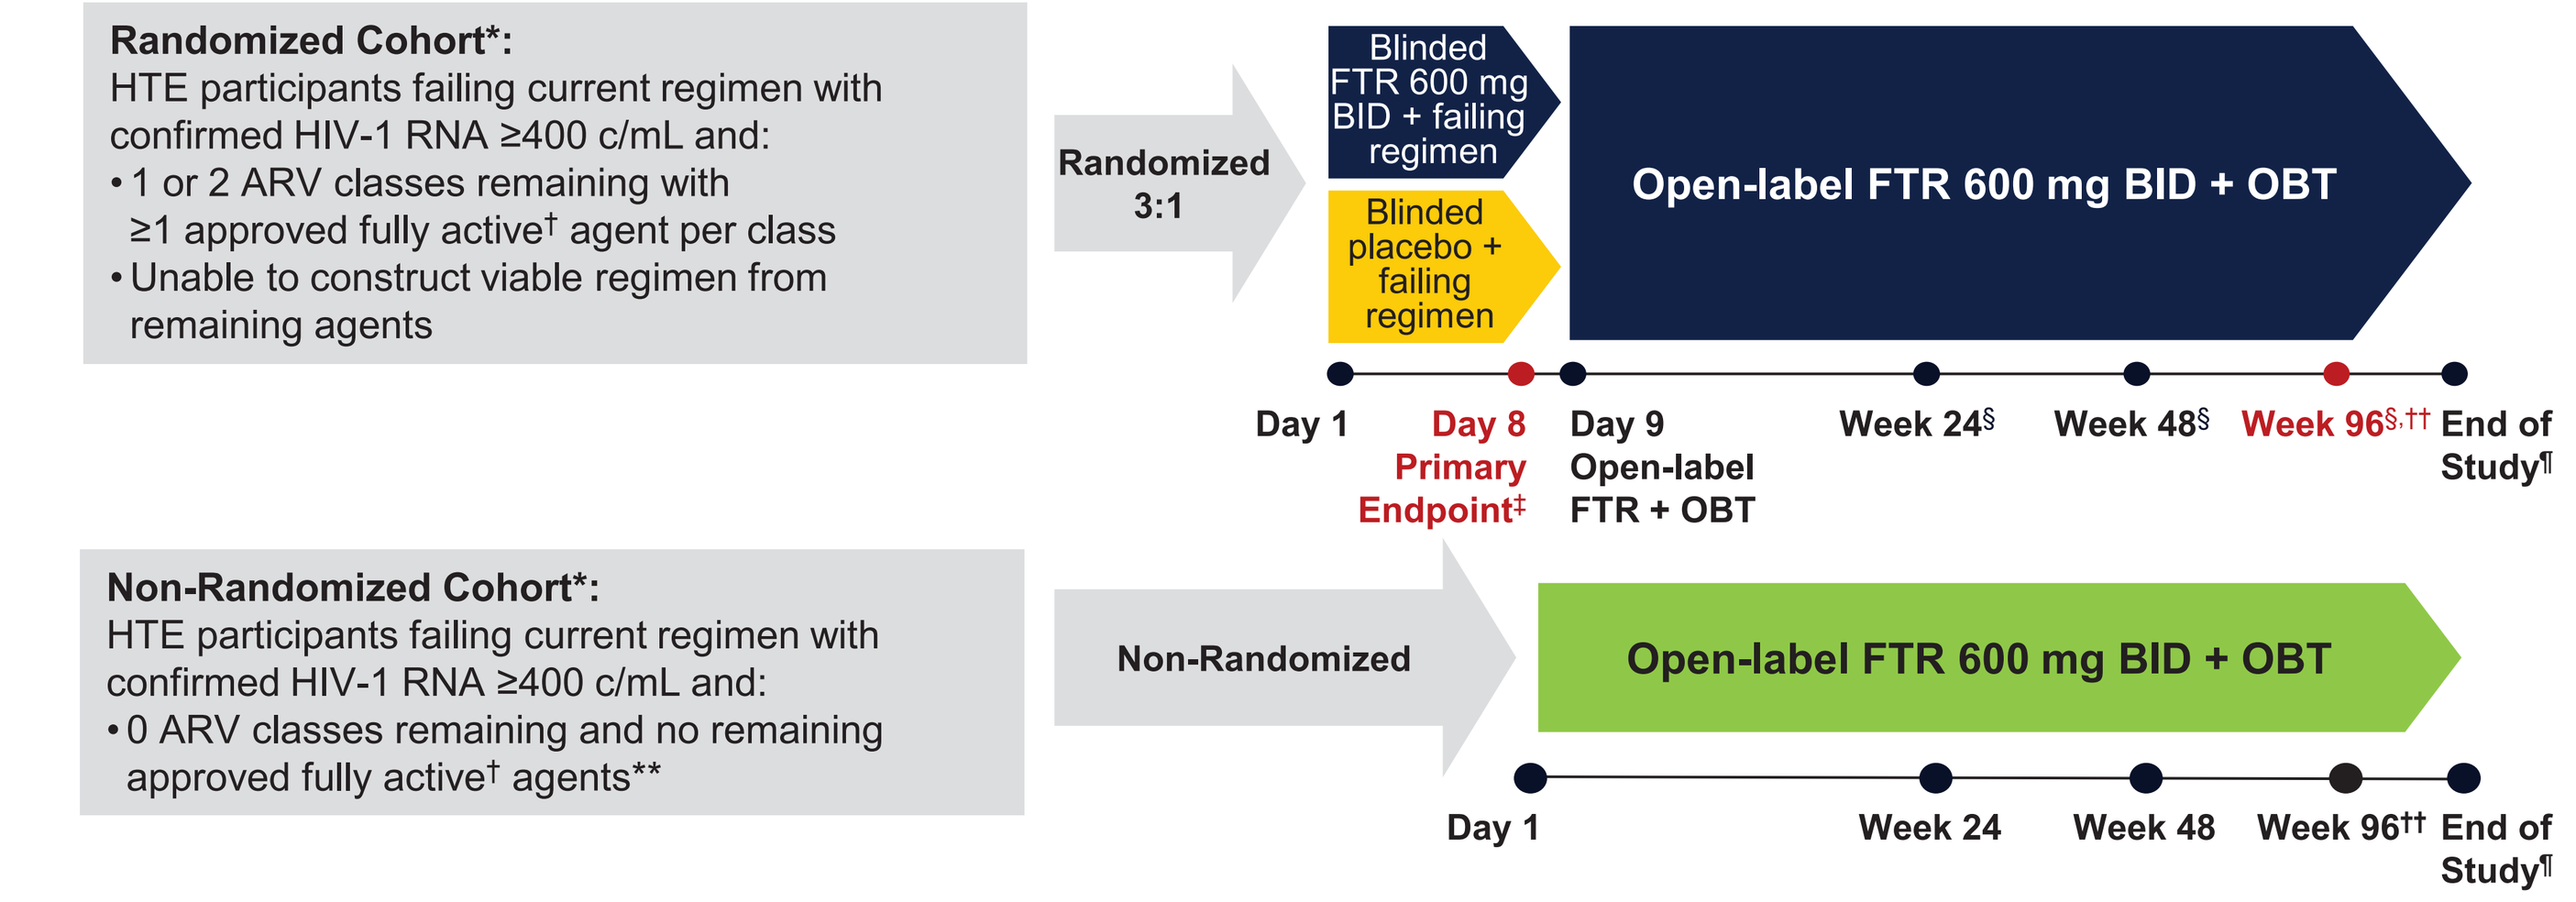
Randomized Cohort*:
HTE participants failing current regimen with confirmed HIV-1 RNA ≥400 c/mL and:
• 1 or 2 ARV classes remaining with ≥1 approved fully active† agent per class
• Unable to construct viable regimen from remaining agents
Randomized 3:1
Blinded FTR 600 mg BID + failing regimen
Blinded placebo + failing regimen
Open-label FTR 600 mg BID + OBT
Day 1
Day 8 Primary Endpoint‡
Day 9 Open-label FTR + OBT
Week 24§
Week 48§
Week 96§,††
End of Study¶
Non-Randomized Cohort*:
HTE participants failing current regimen with confirmed HIV-1 RNA ≥400 c/mL and:
• 0 ARV classes remaining and no remaining approved fully active† agents**
Non-Randomized
Open-label FTR 600 mg BID + OBT
Day 1
Week 24
Week 48
Week 96††
End of Study¶

*There were no screening temsavir IC_{50} criteria. †Fully active is based on susceptibility (current or historical resistance measures) and availability (the participant is tolerant of, eligible for, and willing to take [in the case of enfuvirtide] the ARV). ‡FTR demonstrated superior efficacy compared with placebo after 8 days of functional monotherapy. §Measured from the start of open-label FTR 600 mg BID + OBT. The last participant initiated OBT in August 2016. ¶The study is expected to be conducted until an additional option, rollover study, or marketing approval is in place. **Use of investigational agents as part of OBT was permitted. ††Week 96 database lock August 14, 2018. ClinicalTrials.gov Identifier: NCT02362503; EudraCT Number: 2014-002111-41.

Methods

- RC participants, with fully active ARVs available in 1–2 classes, were randomized (3:1) to blinded FTR 600 mg (n=203) or placebo (n=69) twice daily (BID) plus failing regimen for 8 days of functional monotherapy, followed by open-label FTR 600 mg BID plus OBT (N=272).
- The impacts of BL factors (gp120 polymorphisms, TMR IC_{50} FC relative to reference virus, and HIV-1 subtype) on changes in HIV-1 RNA from Day 1 to 8, proportion of participants with a clinically relevant (>0.5 $\log_{10}$) decrease in HIV-1 RNA at Day 8, and virologic response (HIV-1 RNA <40 c/mL) by FDA Snapshot analysis[8] at Week 96 were evaluated.

Figure 4. BL Characteristics of Evaluable Participants (A) gp120 Polymorphisms of Interest (B) TMR IC_{50} FC (C) HIV-1 Subtype

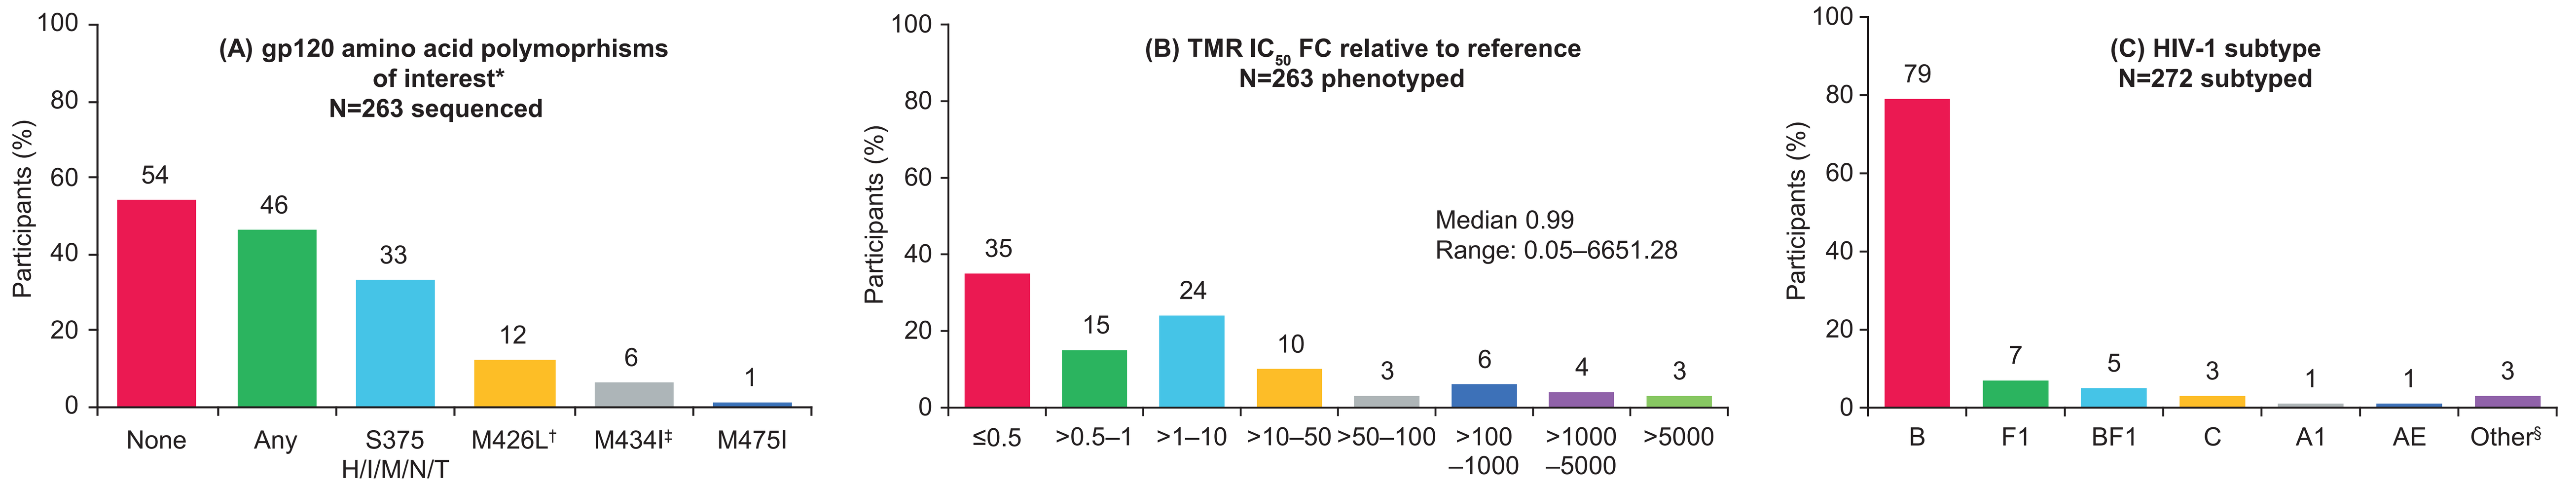
(A) gp120 amino acid polymoprhisms of interest* N=263 sequenced
Participants (%)
100 80 60 40 20 0
54 46 33 12 6 1
None Any S375 H/I/M/N/T M426L† M434I‡ M475I
(B) TMR IC50 FC relative to reference N=263 phenotyped
Median 0.99
Range: 0.05–6651.28
35 15 24 10 3 6 4 3
≤0.5 >0.5–1 >1–10 >10–50 >50–100 >100 –1000 >1000 –5000 >5000
(C) HIV-1 subtype N=272 subtyped
79 7 5 3 1 1 3
B F1 BF1 C A1 AE Other§

Figures 4 and 7 are for the total RC, N=272
Figures 5 and 6 are for the FTR treatment group of the RC, N=203

Footnotes for Figures 4 to 7: *S375H/I/M/N/T, M426L/P, M434I/K, and M475I. Numbers include mixtures. †No M426P was detected. ‡No M434K was detected. §Other includes: non-analyzable/not reported, G, AG or recombinant virus/mixtures.

Figure 5. Change in HIV-1 RNA from Day 1 at Day 8 FTR Group by (A) BL gp120 Polymorphisms of Interest (B) BL TMR IC_{50} FC (C) HIV-1 Subtype

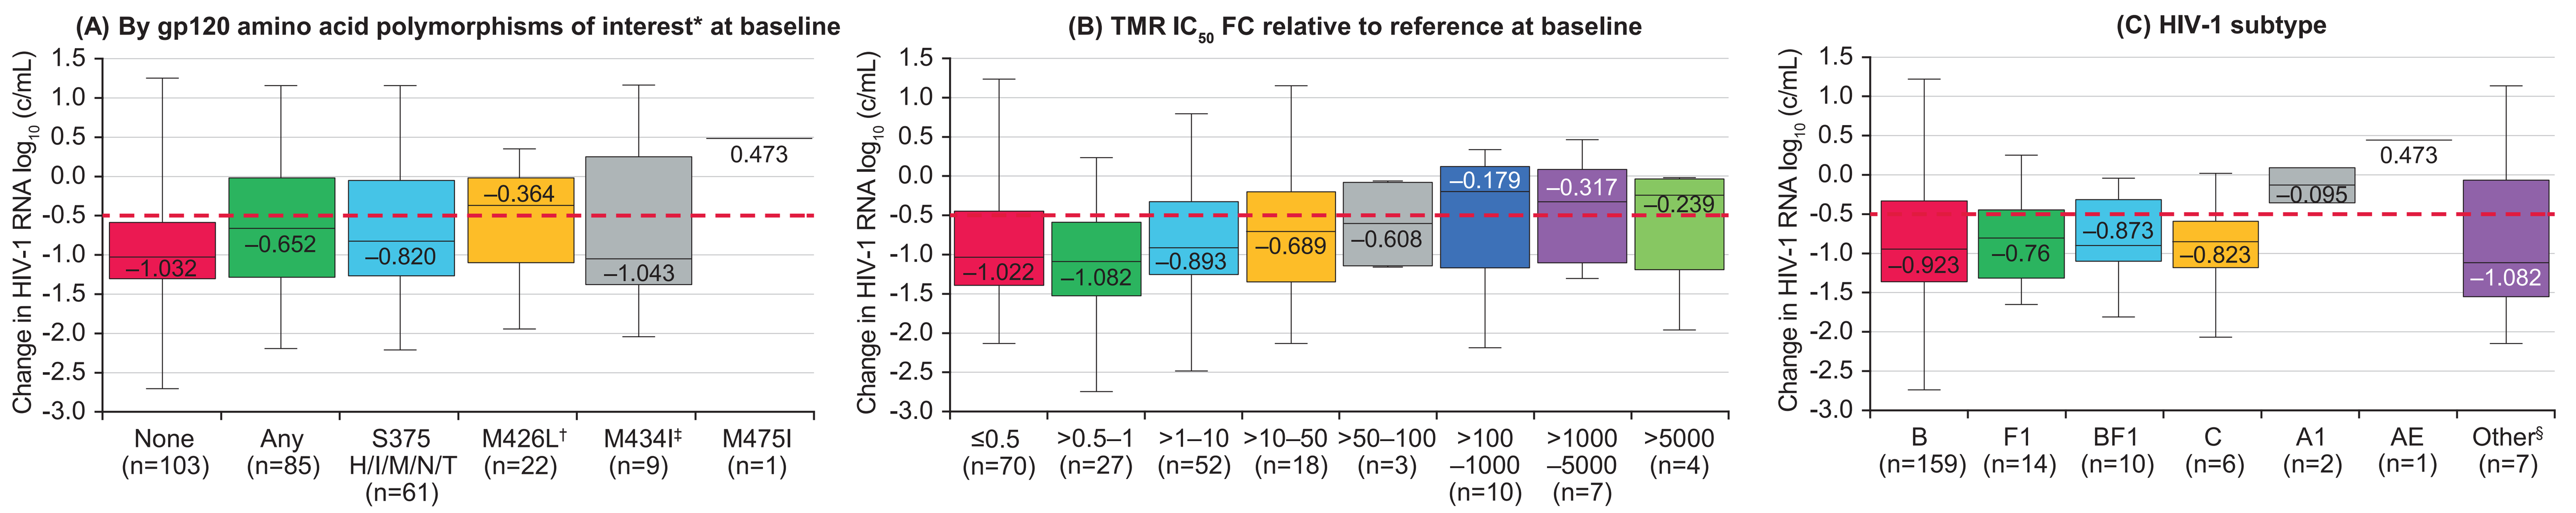
(A) By gp120 amino acid polymorphisms of interest* at baseline
Change in HIV-1 RNA log10 (c/mL)
1.5 1.0 0.5 0.0 -0.5 -1.0 -1.5 -2.0 -2.5 -3.0
–1.032 –0.652 –0.820 –0.364 –1.043 0.473
None (n=103) Any (n=85) S375 H/I/M/N/T (n=61) M426L† (n=22) M434I‡ (n=9) M475I (n=1)
(B) TMR IC50 FC relative to reference at baseline
–1.022 –1.082 –0.893 –0.689 –0.608 –0.179 –0.317 –0.239
≤0.5 (n=70) >0.5–1 (n=27) >1–10 (n=52) >10–50 (n=18) >50–100 (n=3) >100 –1000 (n=10) >1000 –5000 (n=7) >5000 (n=4)
(C) HIV-1 subtype
–0.923 –0.76 –0.873 –0.823 –0.095 0.473 –1.082
B (n=159) F1 (n=14) BF1 (n=10) C (n=6) A1 (n=2) AE (n=1) Other§ (n=7)

Box and whisker plots
Vertical lines = min, max
Box = Q1 to Q3
Horizontal line = median

Figure 6. Virologic Response Category at Day 8 FTR Group by (A) BL gp120 Polymorphisms of Interest (B) BL TMR IC_{50} FC (C) HIV-1 Subtype

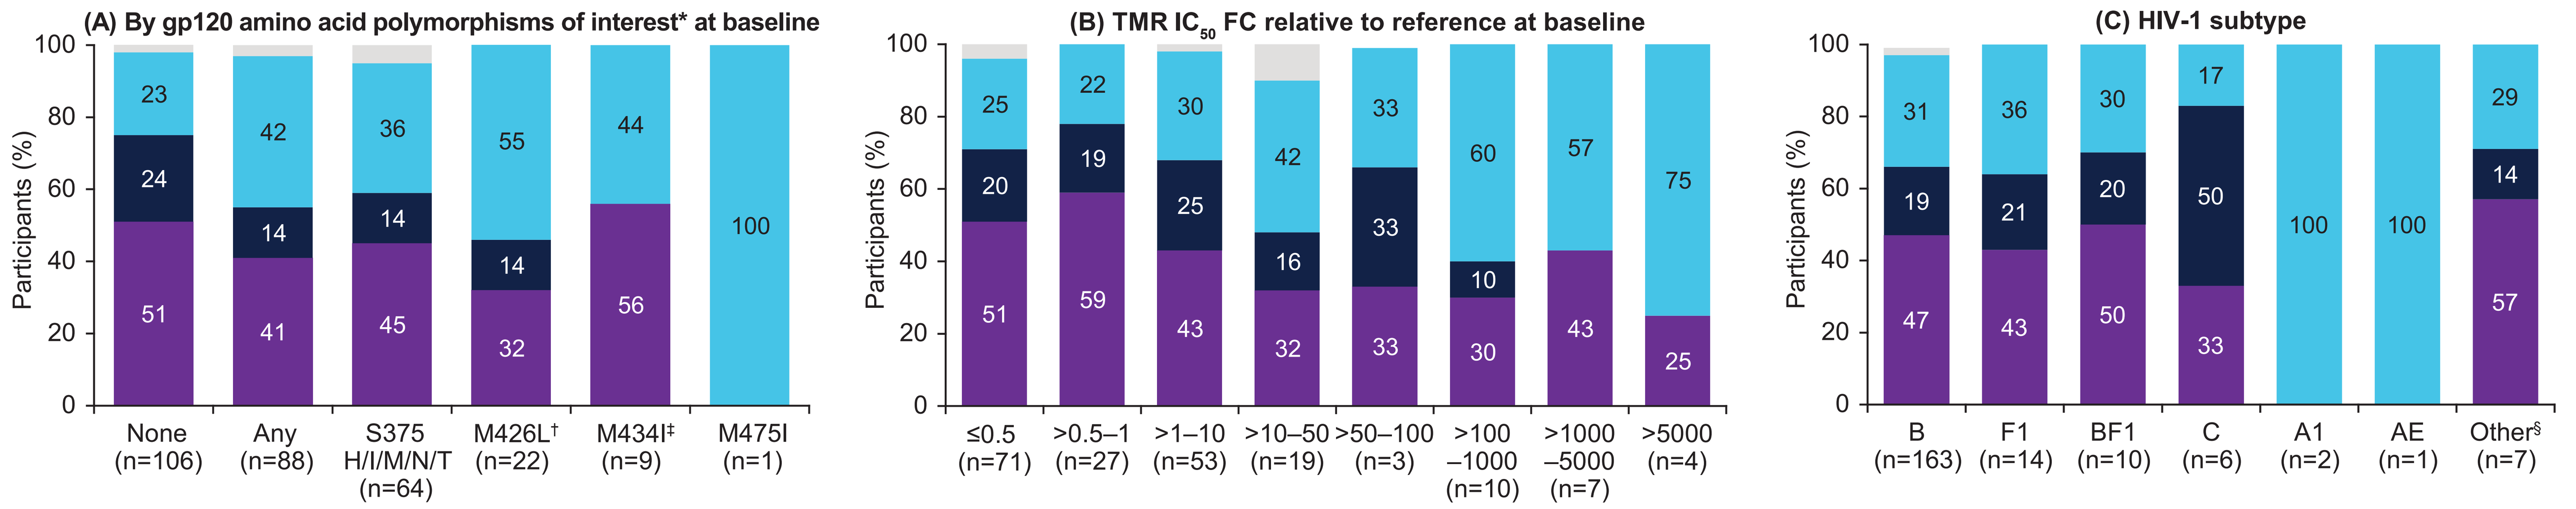
(A) By gp120 amino acid polymorphisms of interest* at baseline
Participants (%)
100 80 60 40 20 0
23 24 51
42 14 41
36 14 45
55 14 32
44 56
100
None (n=106) Any (n=88) S375 H/I/M/N/T (n=64) M426L† (n=22) M434I‡ (n=9) M475I (n=1)
(B) TMR IC50 FC relative to reference at baseline
25 20 51
22 19 59
30 25 43
42 16 32
33 33 33
60 10 30
57 43
75 25
≤0.5 (n=71) >0.5–1 (n=27) >1–10 (n=53) >10–50 (n=19) >50–100 (n=3) >100 –1000 (n=10) >1000 –5000 (n=7) >5000 (n=4)
(C) HIV-1 subtype
31 19 47
36 21 43
30 20 50
17 50 33
100
100
29 14 57
B (n=163) F1 (n=14) BF1 (n=10) C (n=6) A1 (n=2) AE (n=1) Other§ (n=7)

Response category HIV-1 RNA reduction:
- >1.0 $\log_{10}$
- >0.5 to ≤1.0 $\log_{10}$
- ≤0.5 $\log_{10}$
- Missing data

Figure 7. Virologic Response Category at Week 96 (Snapshot Analysis) by (A) BL gp120 Polymorphisms of Interest (B) BL TMR IC_{50} FC (C) HIV-1 Subtype

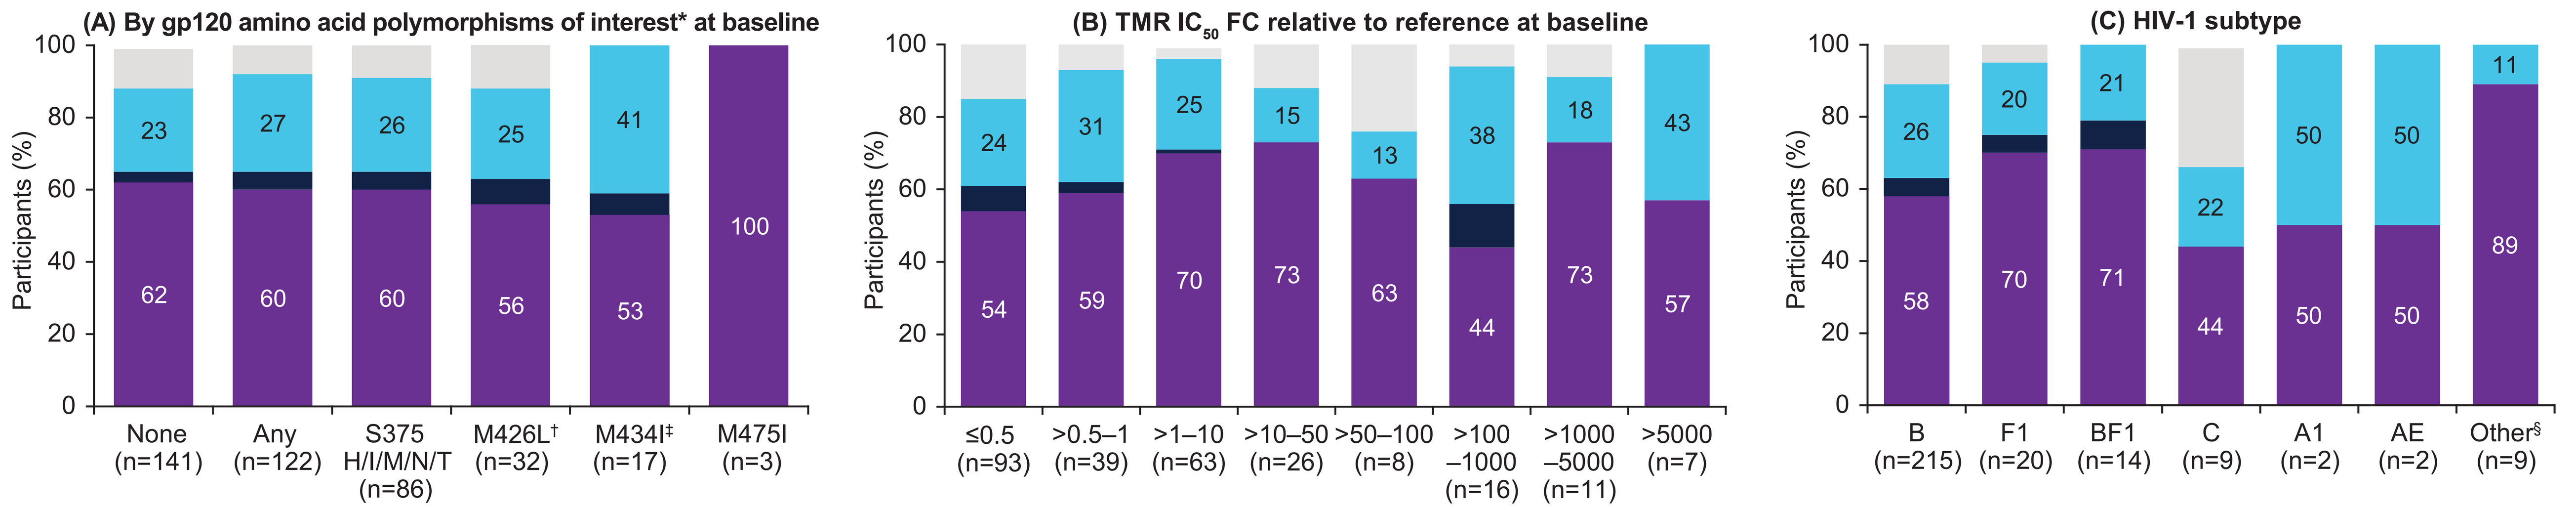
(A) By gp120 amino acid polymorphisms of interest* at baseline
Participants (%)
100 80 60 40 20 0
23 62
27 60
26 60
25 56
41 53
100
None (n=141) Any (n=122) S375 H/I/M/N/T (n=86) M426L† (n=32) M434I‡ (n=17) M475I (n=3)
(B) TMR IC50 FC relative to reference at baseline
24 54
31 59
25 70
15 73
13 63
38 44
18 73
43 57
≤0.5 (n=93) >0.5–1 (n=39) >1–10 (n=63) >10–50 (n=26) >50–100 (n=8) >100 –1000 (n=16) >1000 –5000 (n=11) >5000 (n=7)
(C) HIV-1 subtype
26 58
20 70
21 71
22 44
50 50
50 50
11 89
B (n=215) F1 (n=20) BF1 (n=14) C (n=9) A1 (n=2) AE (n=2) Other§ (n=9)

Response category (HIV-1 RNA c/mL):
- <40
- >40 to <400
- ≥400
- Missing data

Results

- Among evaluable participants in the RC at BL (**Figure 4**):
 - 46% (122/263) had a relevant gp120 polymorphism present.
 - There was a broad range of TMR IC_{50} FC (0.05 to >5000; median 0.99-fold).
 - 74% (195/263) and 87% (229/263) had TMR IC_{50} FC <10 and <100, respectively.
 - The majority (79%, 216/272) had HIV-1 subtype B virus.
- At Day 8 of functional monotherapy (**Figures 5 and 6**):
 - Median change in HIV-1 RNA was smaller among participants with vs. without BL gp120 polymorphisms of interest (–0.65 $\log_{10}$ vs. –1.03 $\log_{10}$); however, 55% (48/88) of participants with BL gp120 polymorphisms achieved a viral load reduction >0.5 $\log_{10}$.
 - Participants with BL TMR IC_{50} FC >100 had a median change in HIV-1 RNA of <0.5 $\log_{10}$ at Day 8; however, 38% (8/21) of participants with BL TMR IC_{50} FC >100 achieved >0.5 $\log_{10}$ decline.
 - Similar proportions of participants with subtype B (66%, 108/163,) vs. non-B (65%, 26/40) HIV-1 achieved >0.5 $\log_{10}$ decline in HIV-1 RNA, although the number of participants with non-B subtypes, including AE, was small (n=40 and n=1, respectively).
 - Consistent with previous observations,[3] the gp120 of the subtype AE virus carried both S375H and M475I and this participant had no reduction in HIV-1 RNA at Day 8.
- At Week 96 of open-label FTR + OBT (**Figure 7**):
 - The proportions of participants with HIV-1 RNA <40 c/mL were comparable across all BL factors evaluated.

Conclusions

- In the Randomized Cohort of heavily-treatment-experienced participants in the BRIGHTE trial:
 - There was a broad range of TMR IC_{50} FC at baseline but most (87%) were <100-fold.
 - Increased baseline TMR IC_{50} FC, or the presence of predefined gp120 polymorphisms at positions of interest, did not preclude participants from achieving a reduction in HIV-1 RNA of >1 $\log_{10}$ c/mL at Day 8 and did not impact durability of response (HIV-1 RNA <40 c/mL) to FTR + OBT through Week 96 of therapy.
 - Virologic response at Day 8 of FTR functional monotherapy and at subsequent timepoints on FTR + OBT (Week 96) was not reliably predicted by baseline factors (genotypic, phenotypic, or HIV-1 subtype), but remained highly context dependent.

Acknowledgments

We would like to thank all of the BRIGHTE clinical trial participants and their families.

ViiV Healthcare and GSK personnel: Marcia Wang and Jill Slater.

Monogram Biosciences: Carmeliza Santos.

Professional medical writing and editorial assistance was provided by Esther Race at SciMentum, funded by ViiV Healthcare.

References

1. Kozal M, et al. 16th European AIDS Conference; October 25–27, 2017; Milan, Italy: Abstract PS8/5. (NEJM, in press).
2. Lataillade M, et al. IAS Congress; July 21–24, 2019; Mexico City, Mexico: Oral presentation MOAB0102.
3. Nowicka-Sans B, et al. Antimicrob Agents Chemother 2012;56:3498–3507.
4. Zhou N, et al. J Antimicrob Chemother. 2014;69(3):573–581.
5. Ray N, et al. J Acquir Immune Defic Syndr. 2013;64(1):7–15.
6. Lataillade M, et al. J Acquir Immune Defic Syndr. 2018;77(3):299–307.
7. Pancera M, et al. Nat Chem Biol. 2017;13(10):1115–1122.
8. U.S. Food and Drug Administration, Center for Drug Evaluation and Research. Guidance for Industry 2015. Available from: https://www.fda.gov/regulatory-information/search-fda-guidance-documents/human-immunodeficiency-virus-1-infection-developing-antiretroviral-drugs-treatment. Accessed January 27, 2020.

Conference on Retroviruses and Opportunistic Infections; March 8–11, 2020; Boston, MA