3691

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Dose-response Relationship of Subcutaneous Long-Acting HIV Capsid Inhibitor GS-6207

Eric Daar,[1] Cheryl McDonald,[2] Gordon Crofoot,[3] Peter Ruane,[4] Gary Sinclair,[5] Rebecca Begley,[6] Edwin DeJesus,[7] Mezgebe Berhe,[8] Moti N. Ramgopal,[9] Ya-Pei Liu,[6] Diana M. Brainard,[6] Robert H. Hyland,[6] Heena Patel,[6] Martin Rhee[6]

[1]The Lundquist Institute, Harbor-UCLA Medical Center, Torrance, CA; [2]Texas Centers for Infectious Disease Associates, Fort Worth, TX; [3]The Crofoot Research Center, Houston, TX; [4]Ruane Clinical Research Group, Inc., Los Angeles, CA; [5]Prism Health North Texas, Dallas, TX; [6]Gilead Sciences, Inc., Foster City, CA; [7]Orlando Immunology Center, Orlando, FL; [8]North Texas Infectious Diseases Consultants, Dallas; [9]Midway Immunology and Research Center, Fort Pierce, FL

Introduction

- GS-6207 is a novel, first-in-class inhibitor of HIV-1 capsid protein (CA) suited for long-acting regimens
- GS-6207 can meet significant unmet medical needs:
 - A new mechanism of action for heavily treatment-experienced people living with multidrug-resistant HIV
 - Reduction of daily pill burden through less frequent dosing
- Highly desirable in vitro profile of GS-6207 for heavily treatment-experienced people
 - Similar antiviral activity across all major HIV-1 subtypes[1]
 - Unique resistance profile, with full activity against NRTI-, NNRTI-, INSTI-, and PI-resistant mutants[1,2]
 - High potency against naturally occurring polymorphisms in group-specific antigen (Gag; including CA, p24)[2,3]
- In healthy volunteers without HIV infection, single SC doses of GS-6207 at ≥100 mg supported dosing intervals ≥12 weeks[4,5]
 - GS-6207 was well tolerated
 - The most common adverse events (AEs) were self-limiting Grade 1 injection-site reactions[4]
- We now report the antiviral activity and safety of SC GS-6207 in people living with HIV (PLWH); safety data are currently blinded

GS-6207: First-in-Class HIV Capsid Inhibitor

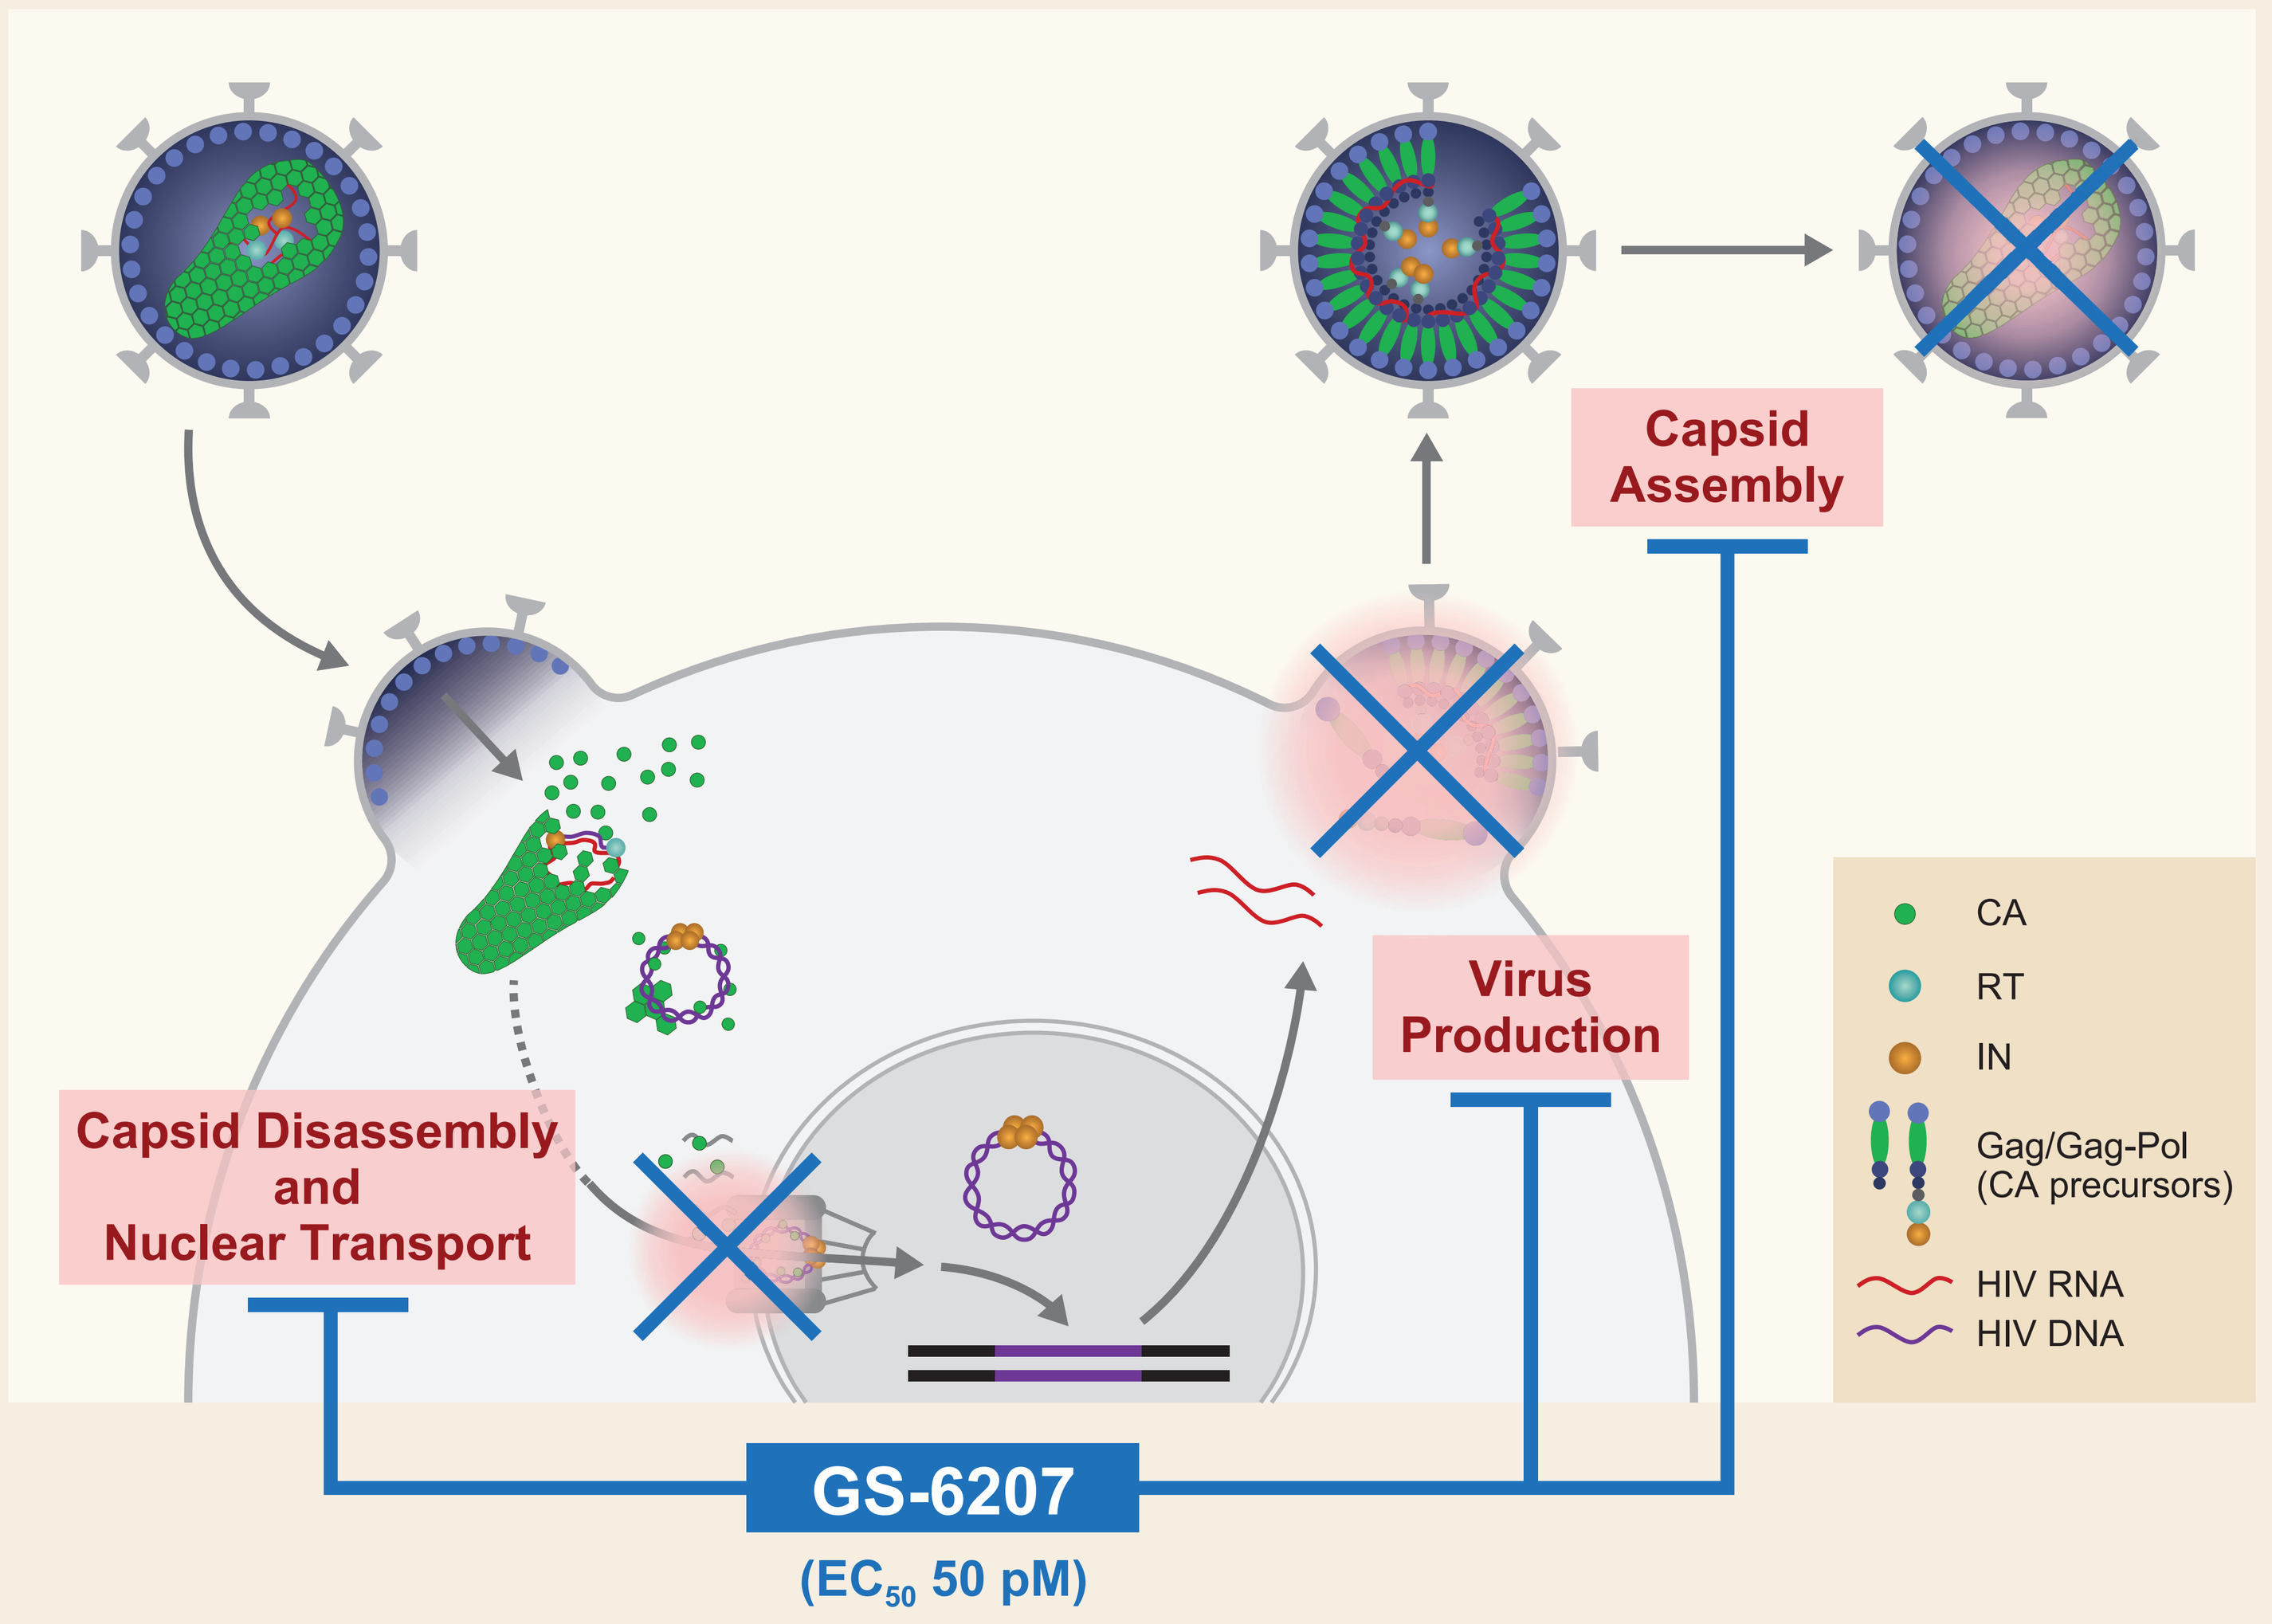
Capsid Assembly
Virus Production
Capsid Disassembly and Nuclear Transport
CA
RT
IN
Gag/Gag-Pol (CA precursors)
HIV RNA
HIV DNA
GS-6207
(EC50 50 pM)

EC_{50}, half-maximal effective concentration determined in peripheral blood mononuclear cells using wild-type HIV-1 patient isolates with various HIV-1 subtypes; IN, integrase; Pol, polymerase; RT, reverse transcriptase.

- Inhibition of multiple CA-dependent functions essential for viral replication

Objectives

- **Primary:** to assess the antiviral activity of GS-6207 in reducing plasma HIV-1 RNA over 10 days after a single SC dose
- **Secondary:** to assess the safety and tolerability of GS-6207

Methods

Study Design

Key inclusion criteria:
- HIV-1 RNA 5000–400,000 copies/mL
- CD4+ cell count >200 cells/mm[3]
- Naïve to CA and IN inhibitors
- Experienced off ARV medications ≥12 months

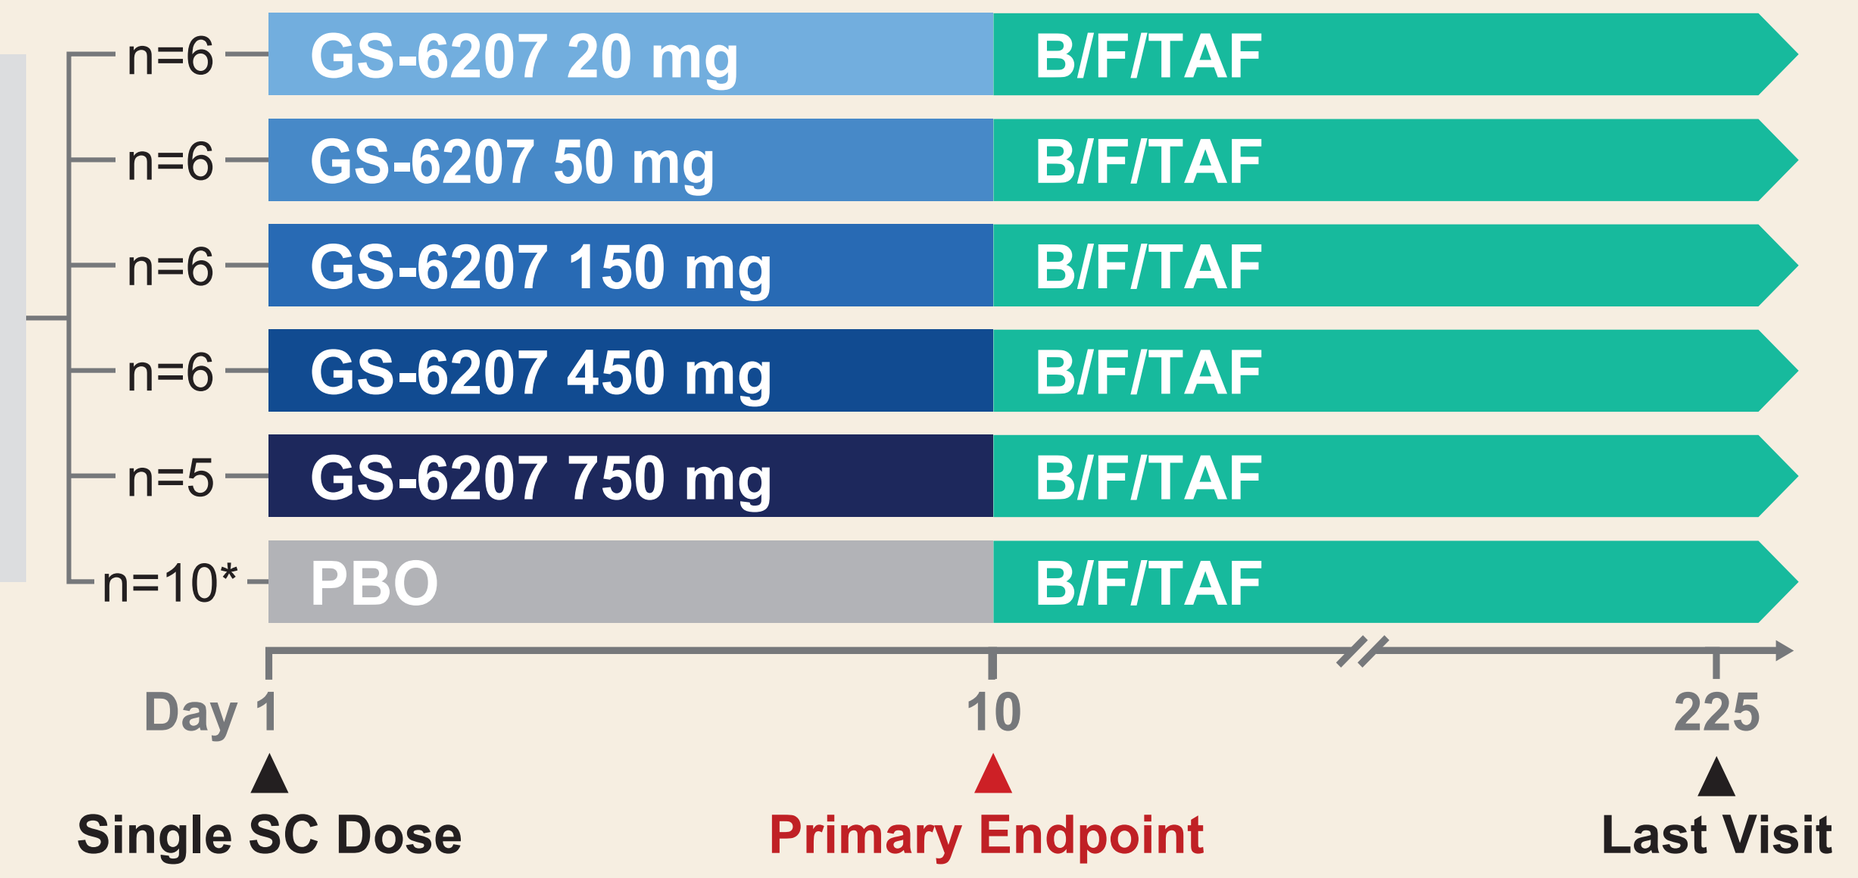
n=6 GS-6207 20 mg B/F/TAF
n=6 GS-6207 50 mg B/F/TAF
n=6 GS-6207 150 mg B/F/TAF
n=6 GS-6207 450 mg B/F/TAF
n=5 GS-6207 750 mg B/F/TAF
n=10* PBO B/F/TAF
Day 1
10
225
Single SC Dose
Primary Endpoint
Last Visit

*The 10 participants receiving placebo (PBO) consisted of 2 from each dose cohort; participants were randomized to receive either active treatment (n=6, except n=5 for 750-mg cohort) or PBO (n=2). ARV, antiretroviral; B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; CD4, cluster of differentiation-4.

- Phase 1b, double-blind, randomized, PBO-controlled, dose-ranging study (NCT03739866)
- Primary endpoint: maximal reduction of plasma HIV-1 RNA through Day 10
- Secondary endpoint: safety and tolerability of GS-6207
- All participants were required to start B/F/TAF on Day 10
- Antiviral activity data were unblinded; safety data remain blinded given that GS-6207 is expected to be detectable for >6 months[2]

Results

Demographics and Baseline Characteristics*

	GS-6207 20 mg or PBO n=8	GS-6207 50 mg or PBO n=8	GS-6207 150 mg or PBO n=8	GS-6207 450 mg or PBO n=8	GS-6207 750 mg or PBO n=7	Total N=39
Age, median year (Min, Max)	35 (23, 50)	28 (19, 56)	36 (24, 56)	29 (20, 59)	26 (20, 65)	33 (19, 65)
Female, n (%)	1 (13)	0	1 (13)	0	2 (29)	4 (10)
Race, n (%)						
White	4 (50)	5 (63)	4 (50)	5 (63)	3 (43)	21 (54)
Black	2 (25)	2 (25)	3 (38)	3 (38)	2 (29)	12 (31)
Asian	1 (13)	1 (13)	0	0	1 (14)	3 (8)
Other	1 (13)	0	1 (13)	0	1 (14)	3 (8)
BMI, median kg/m² (Min, Max)	25 (21, 38)	25 (21, 28)	26 (20, 34)	25 (23, 29)	25 (19, 34)	25 (19, 38)
HIV-1 RNA, median log_{10} copies/mL (Q1, Q3)	4.5 (4.1, 4.9)	4.3 (4.2, 4.7)	4.6 (4.3, 4.6)	4.5 (4.4, 4.6)	4.6 (4.3, 4.9)	4.5 (4.3, 4.7)
CD4 cells/μL, median (Q1, Q3)	472 (395, 542)	594 (459, 662)	388 (309, 581)	430 (260, 611)	491 (321, 707)	463 (359, 614)
ARV treatment naïve, n (%)	8 (100)	6 (75)	4 (50)	7 (88)	7 (100)	32 (82)

*Data were pooled from 6 active (5 in 750-mg cohort) and 2 PBO participants in each cohort as data are currently blinded. BMI, body mass index; Max, maximum; Min, minimum; Q, quartile.

Duration of Follow-up*

Duration, day	GS-6207 20 mg or PBO n=8	GS-6207 50 mg or PBO n=8	GS-6207 150 mg or PBO n=8	GS-6207 450 mg or PBO n=8	GS-6207 750 mg or PBO n=7	Total N=39
Mean (SD)	160 (20)	232 (9)	226 (0.9)	221 (21)	84 (45)	187 (60)
Median	156	227	225	226	86	225
Q1, Q3	146, 174	226, 240	225, 227	222, 233	30, 127	156, 227
Min, Max	135, 191	225, 247	225, 227	169, 233	16, 129	16, 247

*Data were pooled from 6 active (5 in 750-mg cohort) and 2 PBO participants in each cohort as data are currently blinded. SD, standard deviation.

Subcutaneous GS-6207: Antiviral Activity

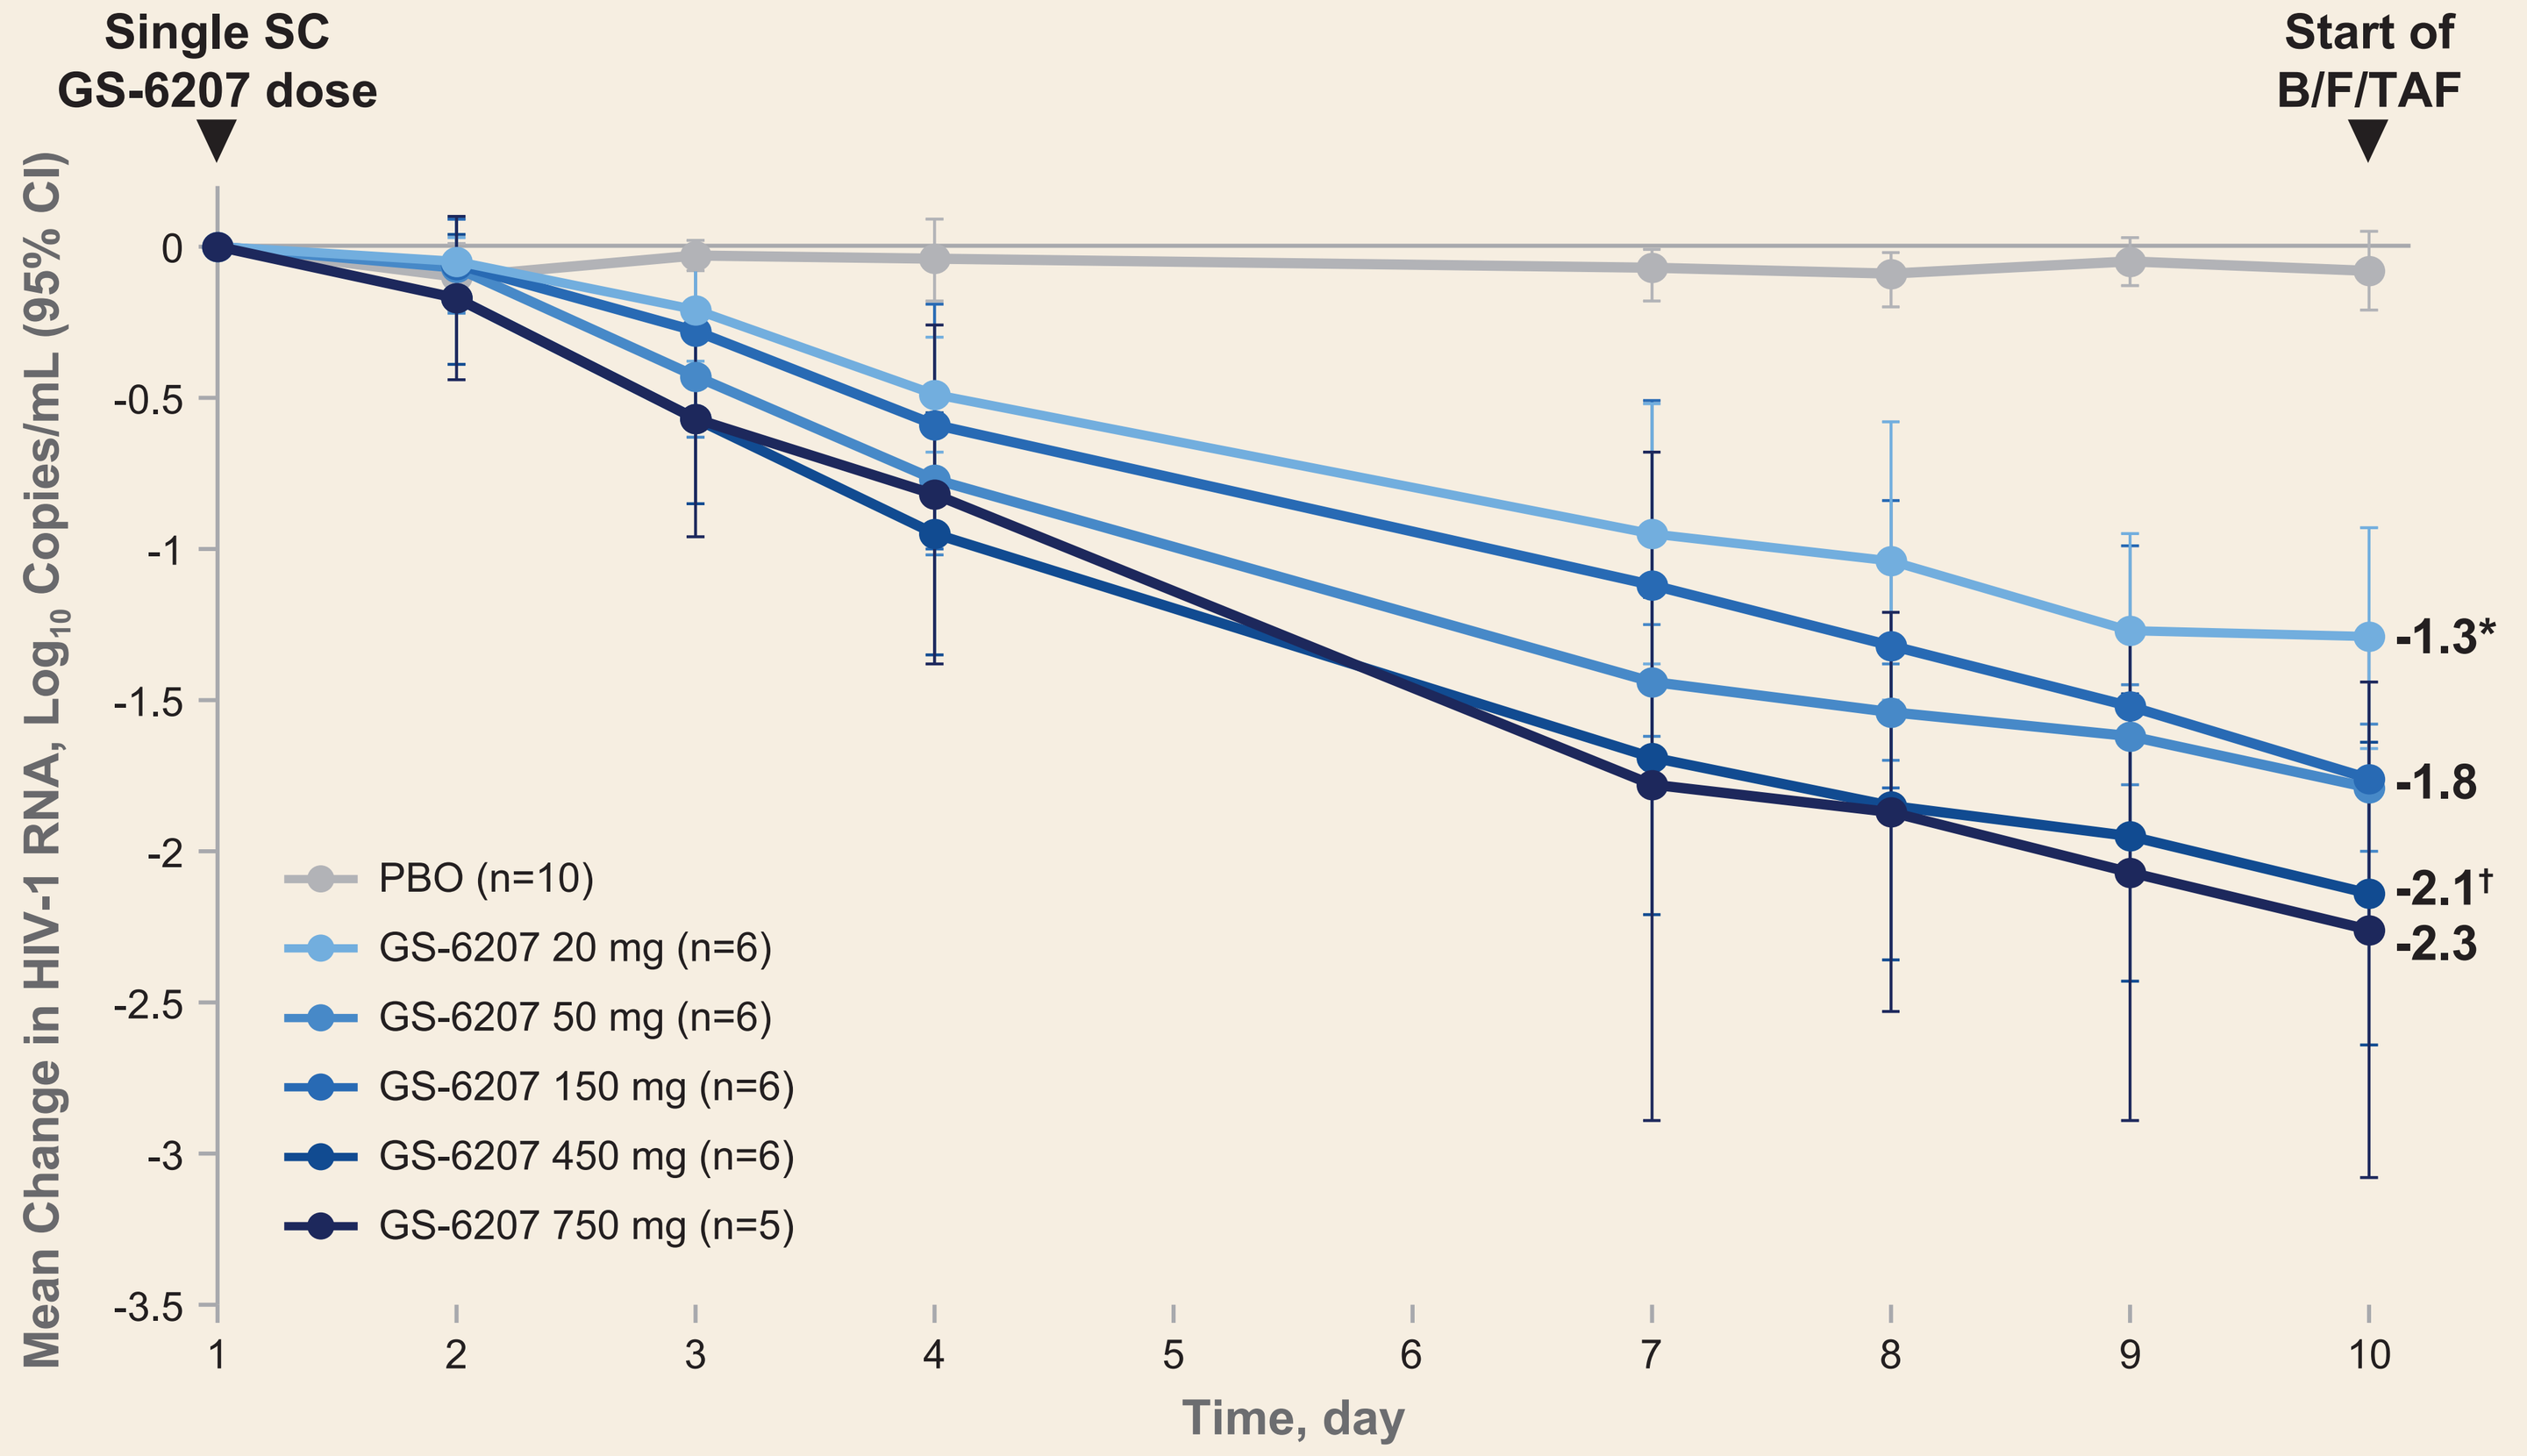
Single SC GS-6207 dose
Start of B/F/TAF
Mean Change in HIV-1 RNA, Log10 Copies/mL (95% CI)
0
-0.5
-1
-1.5
-2
-2.5
-3
-3.5
PBO (n=10)
GS-6207 20 mg (n=6)
GS-6207 50 mg (n=6)
GS-6207 150 mg (n=6)
GS-6207 450 mg (n=6)
GS-6207 750 mg (n=5)
-1.3*
-1.8
-2.1†
-2.3
1
2
3
4
5
6
7
8
9
10
Time, day

*Change (mean) on Day 10 in 20-mg cohort was -1.3 log_{10} copies/mL while maximal change (mean) through Day 10 was -1.4 log_{10} copies/mL; †Change (mean) on Day 10 in 450-mg cohort was -2.1 log_{10} copies/mL while maximal change (mean) through Day 10 was -2.2 log_{10} copies/mL. CI, confidence interval.

Antiviral Activity Through Day 10

Maximal Change in HIV-1 RNA From Baseline, Log_{10} Copies/mL	GS-6207 20 mg n=6	GS-6207 50 mg n=6	GS-6207 150 mg n=6	GS-6207 450 mg n=6	GS-6207 750 mg n=5	PBO n=10
Mean	-1.4	-1.8	-1.8	-2.2	-2.3	-0.2
95% CI	-1.7, -1.0	-2.3, -1.3	-2.0, -1.6	-2.7, -1.7	-3.1, -1.4	-0.3, -0.1
Median	-1.4	-1.7	-1.8	-2.2	-2	-0.2
Q1, Q3	-1.6, -1.2	-2.3, -1.6	-1.9, -1.6	-2.5, -1.8	-2.9, -2.0	-0.2, -0.1
Min, Max	-1.7, -0.8	-2.4, -1.2	-2.1, -1.5	-2.9, -1.6	-3.0, -1.5	-0.4, 0.0

- All participants who received GS-6207 ≥50 mg SC had a mean ≥1.8 log_{10} copies/mL reduction in HIV-1 RNA through Day 10
- At doses of 20 to 750 mg, mean GS-6207 concentrations on Day 10 were 0.7- to 20.5-fold higher than the protein-adjusted 95% effective concentration for wild-type HIV-1

Dose-response Relationship Between GS-6207 and Antiviral Activity: E_{max} Model*

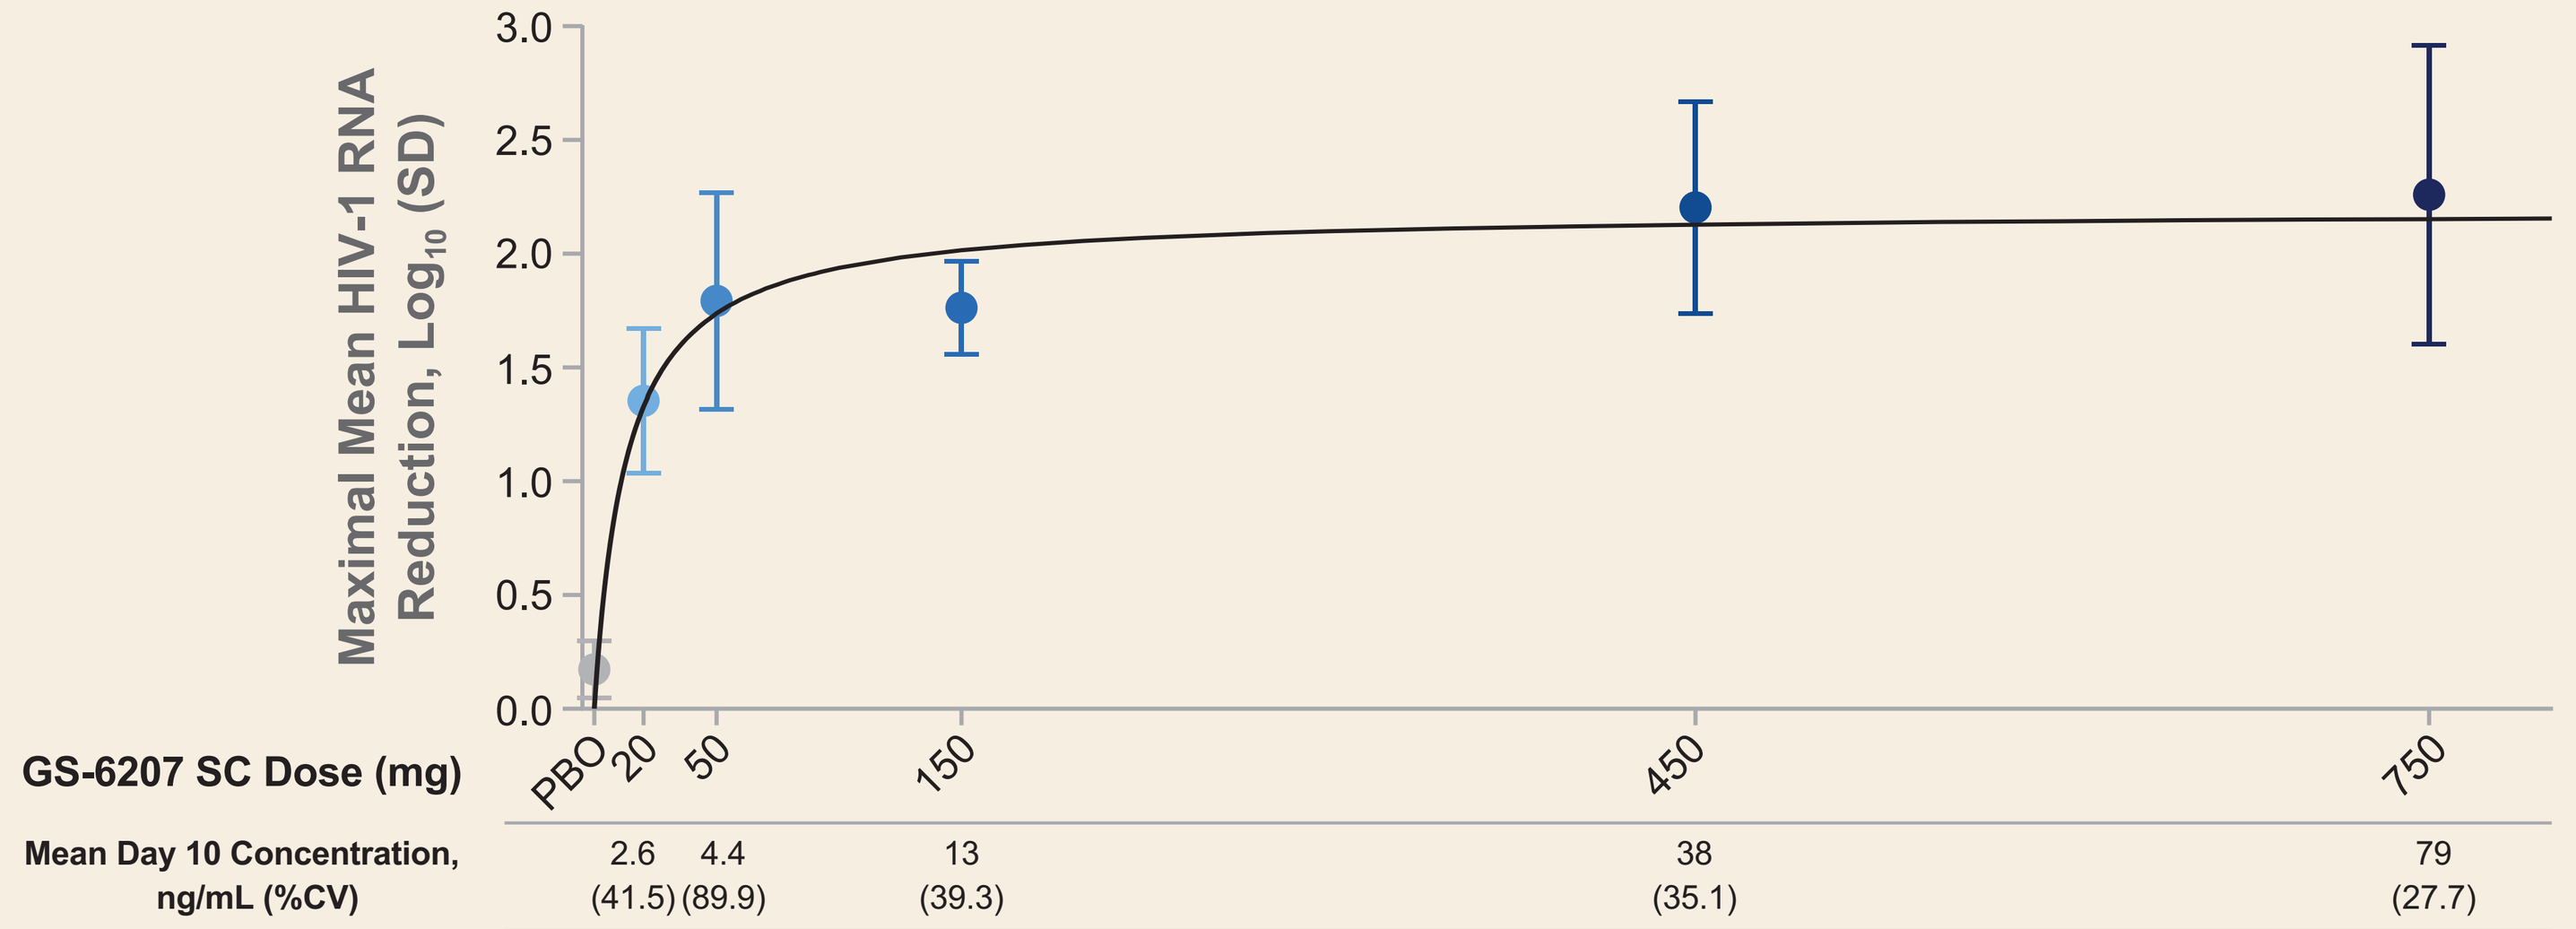
Maximal Mean HIV-1 RNA Reduction, Log10 (SD)
3.0
2.5
2.0
1.5
1.0
0.5
0.0
GS-6207 SC Dose (mg)
PBO
20
50
150
450
750
Mean Day 10 Concentration, ng/mL (%CV)
2.6 (41.5)
4.4 (89.9)
13 (39.3)
38 (35.1)
79 (27.7)

*Each dot represents mean HIV-1 RNA reduction in each dosed group. CV, coefficient of variation; E_{max}, maximal effect.

- Following SC administration of GS-6207 in PLWH, E_{max} on Day 10 postdose was predicted to be a 2.2 log_{10} decline in HIV-1 RNA
- Mean GS-6207 concentrations ≥4.4 ng/mL are predicted to provide near maximal antiviral activity

Safety Summary: Blinded Data

Participants, n (%)	GS-6207 20 mg or PBO n=8	GS-6207 50 mg or PBO n=8	GS-6207 150 mg or PBO n=8	GS-6207 450 mg or PBO n=8	GS-6207 750 mg or PBO n=7	Total N=39
AEs						
Any AE	7 (88)	6 (75)	7 (88)	7 (88)	6 (86)	33 (85)
Injection-site reaction	1 (13)	5 (63)	5 (63)	5 (63)	6 (86)	22 (56)
Grade 3 or 4	0	0	0	1 (13)	0	1 (3)
SAE*	1 (13)	0	0	1 (13)	0	2 (5)
AE leading to discontinuation	0	0	0	0	0	0
Death	0	0	0	0	0	0
Laboratory abnormalities						
Grade 3 or 4	2 (25)	0	3 (38)	2 (25)	1 (14)	8 (21)

*1 participant with preexisting cardiovascular risk factors had Grade 3 serious AE (SAE) of atrial fibrillation on Day 113 postdose following methamphetamine use; same participant experienced Grade 4 SAEs of coronary artery disease and acute myocardial infarction, Grade 3 AE of unstable angina and angina pectoris (all on Day 191), and Grade 3 SAE of noncardiac chest pain (Day 195); all were considered not related to GS-6207; another participant experienced Grade 2 SAE of small intestinal obstruction on Day 57 not related to GS-6207.

- The most common AEs were mild to moderate reactions at injection site (56%; n=22), including pain (49%; n=19) and erythema (28%; n=11), which were self-limiting and resolved in a few days after dosing
- Grade 3 or 4 laboratory abnormalities in ≥2 participants were transient creatine kinase elevations (n=3; attributed to recent exertion), asymptomatic amylase elevations (n=2), and urine blood attributed to menses (n=2), none of which were considered clinically relevant

Conclusions

- Single SC doses of GS-6207 resulted in potent antiviral activity
 - HIV-1 RNA declined over 10 days: mean 1.4 to 2.3 log_{10} copies/mL
- Mean GS-6207 concentrations ≥4.4 ng/mL are predicted to provide near maximal antiviral activity
- In a blinded safety review, GS-6207 and PBO were generally safe and well tolerated
 - The most common AEs were self-limiting, mild to moderate injection-site reactions
- These results support further evaluation of GS-6207 as a long-acting ARV agent in 2 ongoing clinical trials in PLWH, with a 6-month dosing interval:
 - In treatment-naïve PLWH (NCT04143594)
 - In heavily treatment-experienced PLWH (NCT04150068)

References: 1. Yant SR, et al. CROI 2019, poster 480; 2. Margot N, et al. CROI 2020, poster 529; 3. Margot N, et al. EACS 2019, poster PE13/22; 4. Begley R, et al. EACS 2019, oral PS13/7; 5. Sager JE, et al. CROI 2019, oral O-13.

Acknowledgments: We extend our thanks to the participants, their families, and all participating study investigators and staff: D. Asmuth, P. Benson, M. Berhe, G. Crofoot, E. Daar, C. McDonald, A. Mills, O. Osiyemi, M.N. Ramgopal, E. DeJesus, P.J. Ruane, and G.I. Sinclair. This study was funded by Gilead Sciences, Inc.

Presented at Conference on Retroviruses and Opportunistic Infections, March 8–11, 2020, Boston, MA

©2020 Gilead Sciences, Inc. All rights reserved.