Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

3815

Drug Levels, Adherence, and Risks for Low Adherence in the DISCOVER PrEP Study

Susanne Doblecki-Lewis,[1] Olamide Dosekun,[2] Moti Ramgopal,[3] Jay Gladstein,[4] Mezgebe Berhe,[4] Kevin Nguyen,[5] Jason Hindman,[5] Yongwu Shao,[5] Ramin Ebrahimi,[5] Diana M. Brainard,[5] Moupali Das,[5] Scott McCallister,[5] Adriano Lazzarin,[6] Gerold F. Lang,[4] Peter L. Anderson[7]

[1]University of Miami, FL; [2]Imperial College Healthcare NHS Trust, London, UK; [3]Midway Research Center, Fort Pierce, FL; [4]APLA Health, Los Angeles, CA; [5]Gilead Sciences, Inc., Foster City, CA; [6]Ospedale San Raffaele, Milano, Italy; [7]University of Colorado Anschutz Medical Campus, Aurora

Introduction

- Medication adherence is a key correlate of HIV pre-exposure prophylaxis (PrEP) effectiveness[1]
- DISCOVER is an ongoing, randomized, controlled trial comparing emtricitabine/tenofovir alafenamide (F/TAF) with emtricitabine/tenofovir disoproxil fumarate (F/TDF) for PrEP
- During DISCOVER, study drug adherence was monitored by self-report, pill counts, and drug level measurement in red blood cells by dried blood spot (DBS) analysis
- At the primary endpoint (100% of participants completed 48 wk and 50% completed 96 wk):
 - DISCOVER demonstrated that F/TAF had noninferior efficacy vs F/TDF for HIV PrEP, as well as superior biomarker findings with respect to renal function and bone mineral density[2]
 - Adherence was high by self-report, pill count, and DBS[3]

Objectives

- To assess adherence with F/TAF vs F/TDF through 96-wk treatment and factors associated with low adherence in DISCOVER

Methods

Study Design

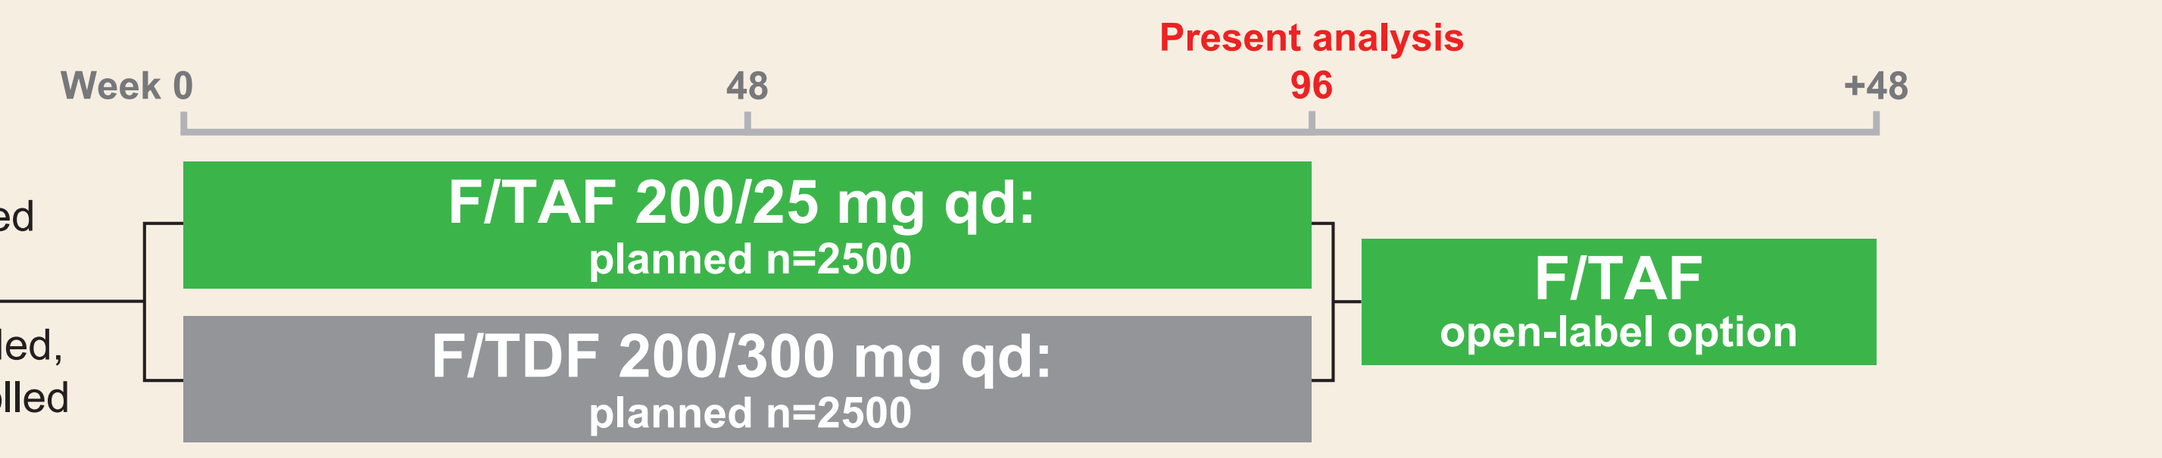
Week 0
48
Present analysis
96
+48
MSM or TGW aged ≥18 y
Randomized 1:1
Double blinded, active controlled
F/TAF 200/25 mg qd: planned n=2500
F/TDF 200/300 mg qd: planned n=2500
F/TAF open-label option

MSM, men who have sex with men; TGW, transgender women.

- Eligibility: high sexual risk of HIV
 - 2+ unique condomless anal sex partners in past 12 wk, or rectal gonorrhea/chlamydia or syphilis in past 24 wk
 - HIV and hepatitis B virus negative, and estimated glomerular filtration rate (Cockcroft-Gault) ≥60 mL/min
 - Prior use of PrEP allowed
- Conducted in Europe and North America in cities/sites with high HIV incidence
- Assessments: safety, adherence, HIV lab testing, and HIV risk behavior

Adherence Support Offered at Study Visits

- Adherence counseling
- Personal communications from site staff, as needed
- Optional daily text messaging
- Periodic email updates

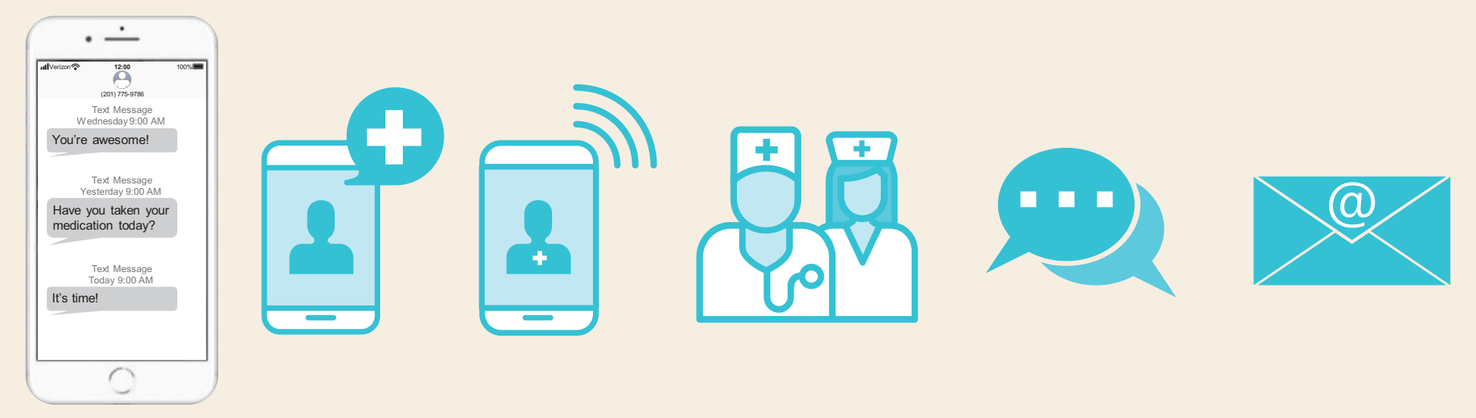

DBS Substudy

- To evaluate adherence by tenofovir-diphosphate (TFV-DP) concentrations in DBS, a 10% representative subset of DISCOVER was preselected

Adherence Cutoffs by TFV-DP DBS Concentrations

Adherence Cutoffs[4-6]	TFV-DP DBS Concentration, fmol/Punch(es)* F/TAF	F/TDF
High adherence (≥4 tablets/wk)	≥900	≥700
Medium adherence (2–3 tablets/wk)	≥450–<900	≥350–<700
Low adherence (<2 tablets/wk)	<450	<350

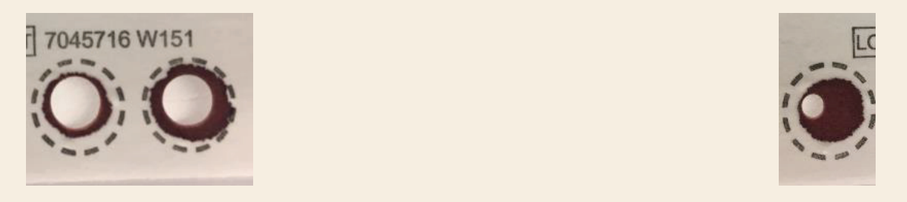
7045716 W151

2 x 7 mm 3 mm

*Reported concentrations based on one 3-mm (F/TDF) or two 7-mm (F/TAF) punch disks.

Results

Participant Disposition Through Week 96

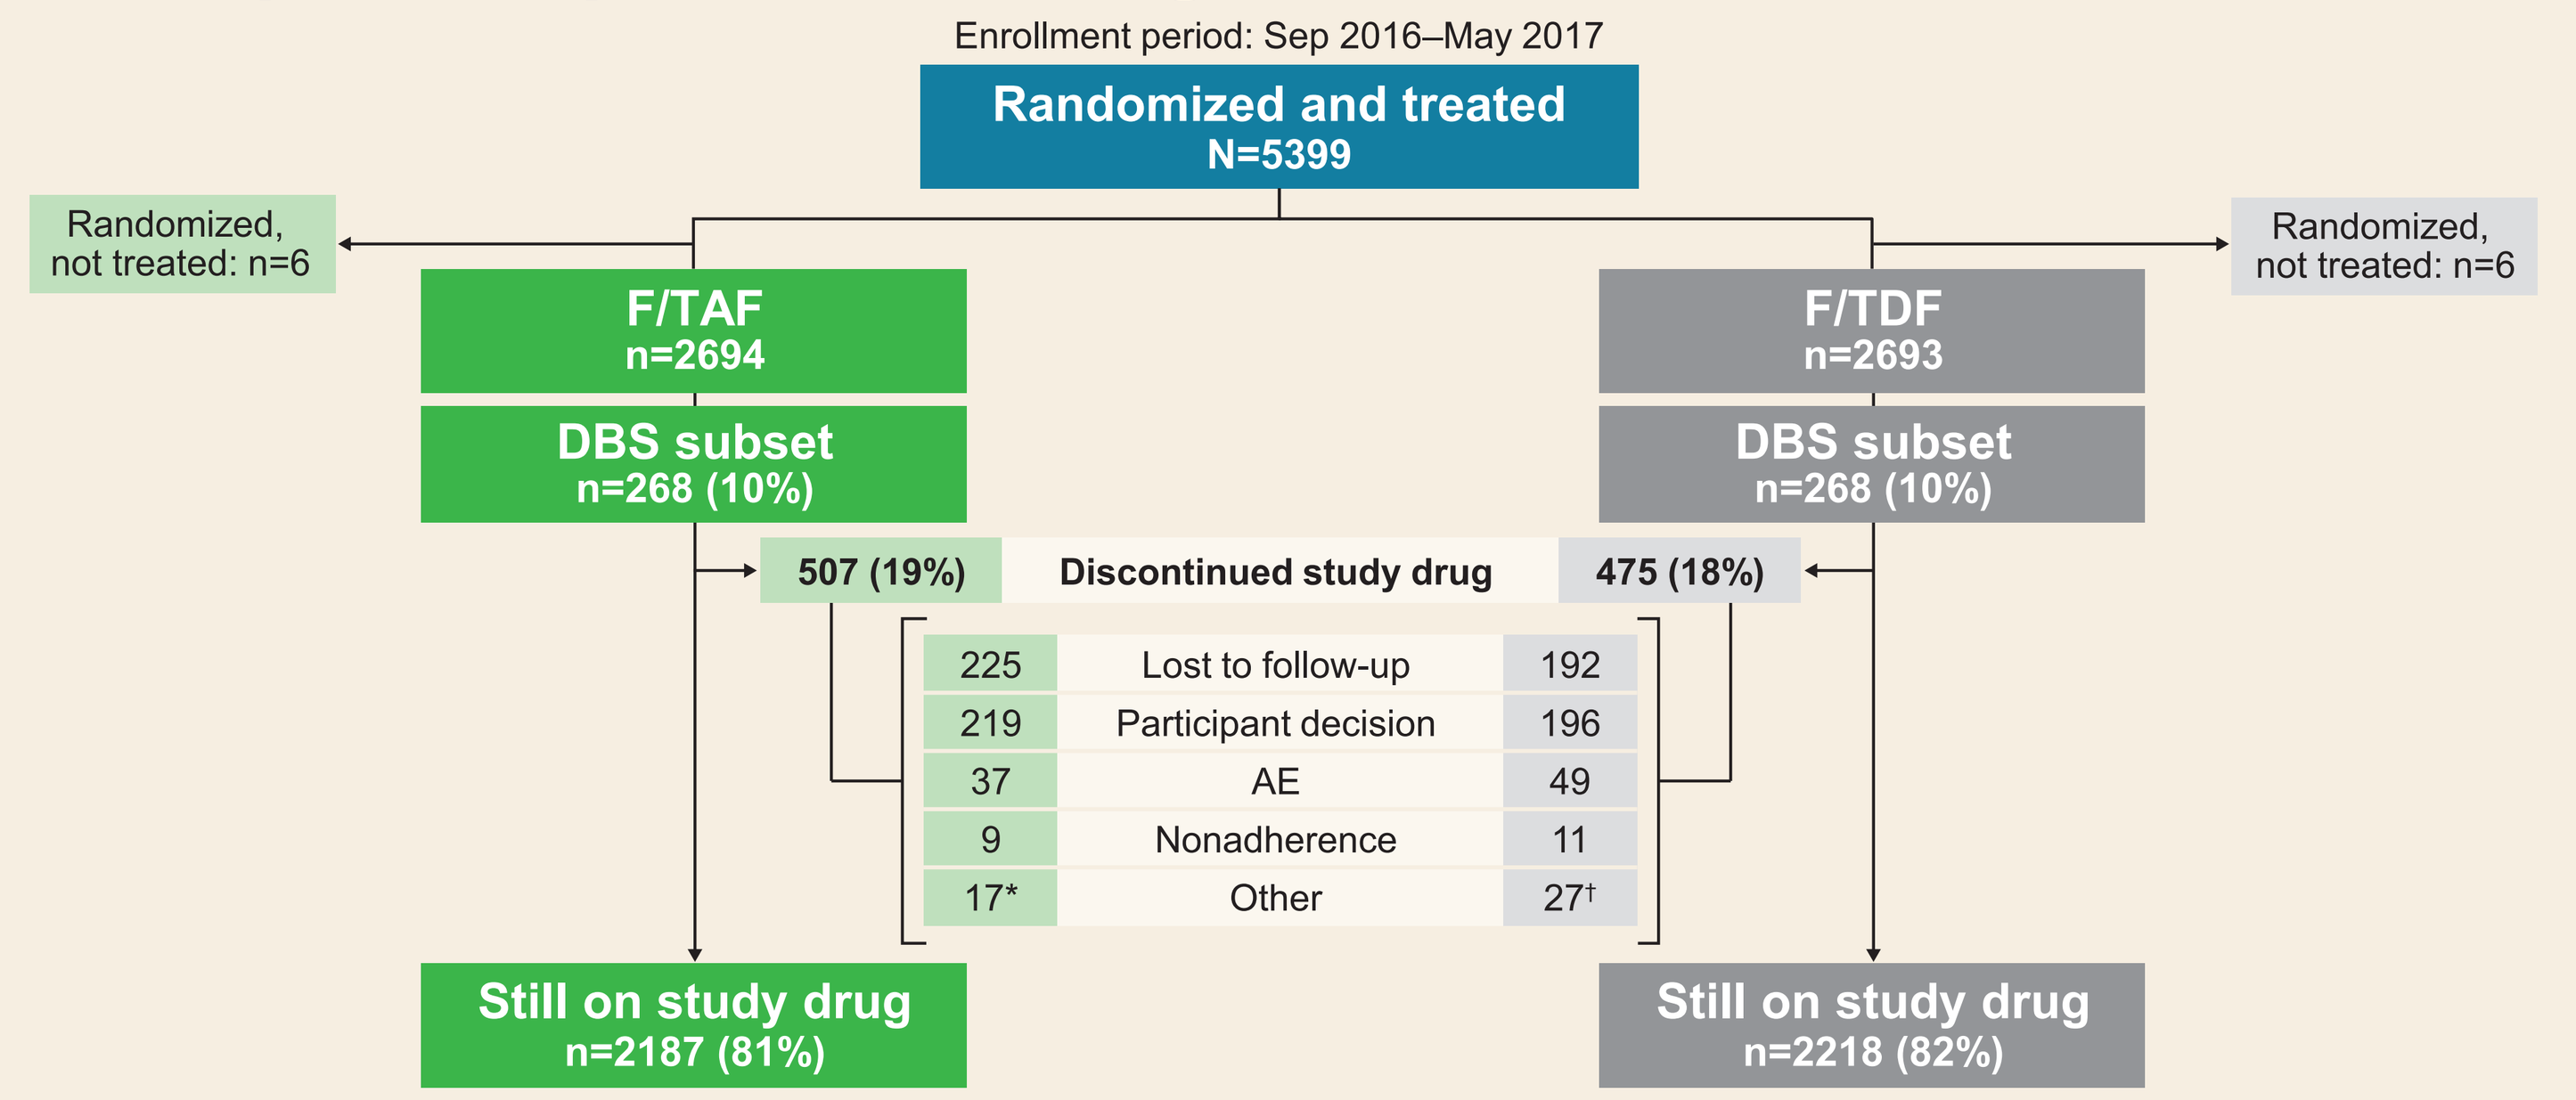
Enrollment period: Sep 2016–May 2017
Randomized and treated N=5399
Randomized, not treated: n=6
F/TAF n=2694
DBS subset n=268 (10%)
F/TDF n=2693
DBS subset n=268 (10%)
Randomized, not treated: n=6
507 (19%) Discontinued study drug 475 (18%)
225 Lost to follow-up 192
219 Participant decision 196
37 AE 49
9 Nonadherence 11
17* Other 27†
Still on study drug n=2187 (81%)
Still on study drug n=2218 (82%)

*Includes investigator's discretion (n=7), HIV-1 infection (n=5), protocol violation (n=4), and death (n=1); †Includes investigator's discretion (n=11), HIV-1 infection (n=10), protocol violation (n=3), and death (n=3). AE, adverse event.

Demographics and Baseline Characteristics

		Safety Analysis Set F/TAF n=2694	Safety Analysis Set F/TDF n=2693	DBS Subset F/TAF n=268	DBS Subset F/TDF n=268
Demographics	Median age, y (range)	34 (18–76)	34 (18–72)	36 (20–73)	33 (18–64)
	MSM, n (%)	2649 (98)	2664 (99)	265 (99)	266 (99)
	TGW, n (%)	45 (2)	29 (1)	3 (1)	2 (<1)
	Race, n (%)				
	White	2264 (84)	2247 (84)	221 (83)	233 (87)
	Black*	240 (9)	234 (9)	29 (11)	21 (8)
	Asian	113 (4)	120 (4)	13 (5)	8 (3)
	Other	74 (3)	87 (3)	4 (1)	6 (2)
	Hispanic/Latinx ethnicity, n (%)	635 (24)	683 (25)	57 (21)	72 (27)
Other Characteristics	Baseline F/TDF PrEP, n (%)	465 (17)	440 (16)	50 (19)	40 (15)
	≥4-y college education, n (%)	1543 (58)	1535 (57)	149 (56)	151 (57)
	Circumcision, n (%)	1485 (55)	1513 (57)	136 (51)	155 (58)
	Recreational drug use, n (%)†	1785 (67)	1786 (67)	174 (65)	185 (70)
	Binge drinking, n (%)‡	1476 (56)	1506 (56)	141 (54)	165 (62)
Prior STIs and Sexual Behavior	History of syphilis, n (%)§	230 (9)	263 (10)	26 (10)	27 (10)
	History of rectal gonorrhea, n (%)§	274 (10)	262 (10)	25 (9)	30 (11)
	History of chlamydia, n (%)§	342 (13)	333 (12)	37 (14)	34 (13)
	Condomless receptive anal sex, mean no. of partners (SD)†	4 (6)	3 (6)	3 (6)	3 (4)

*Includes black/mixed race; †In 3 mo prior to screening; ‡≥6 drinks on 1 occasion; §In 24 wk prior to screening. SD, standard deviation; STIs, sexually transmitted infections.

Primary Efficacy Endpoint: Noninferiority Maintained at Week 96

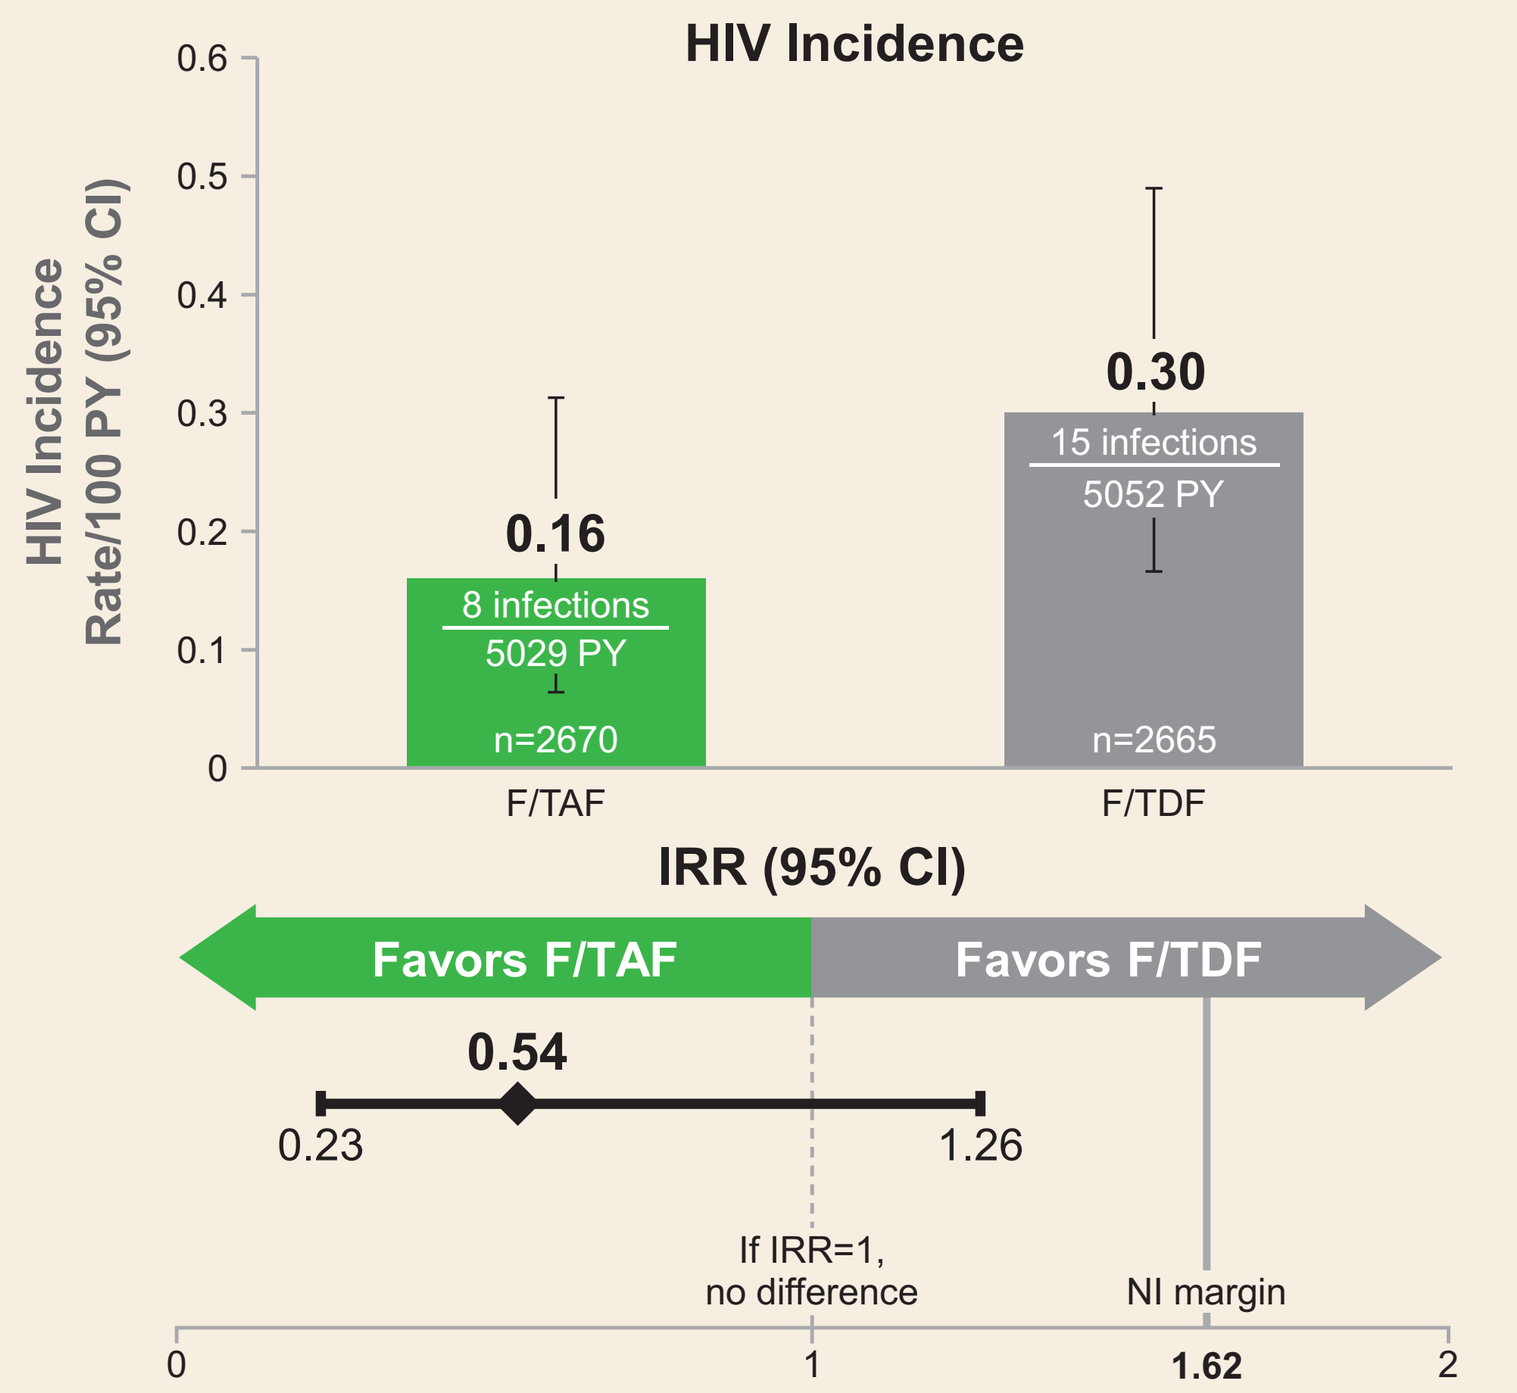
HIV Incidence
HIV Incidence Rate/100 PY (95% CI)
0.16
8 infections / 5029 PY
n=2670
F/TAF
0.30
15 infections / 5052 PY
n=2665
F/TDF
IRR (95% CI)
Favors F/TAF
Favors F/TDF
0.54
0.23
1.26
If IRR=1, no difference
NI margin
0
1
1.62
2

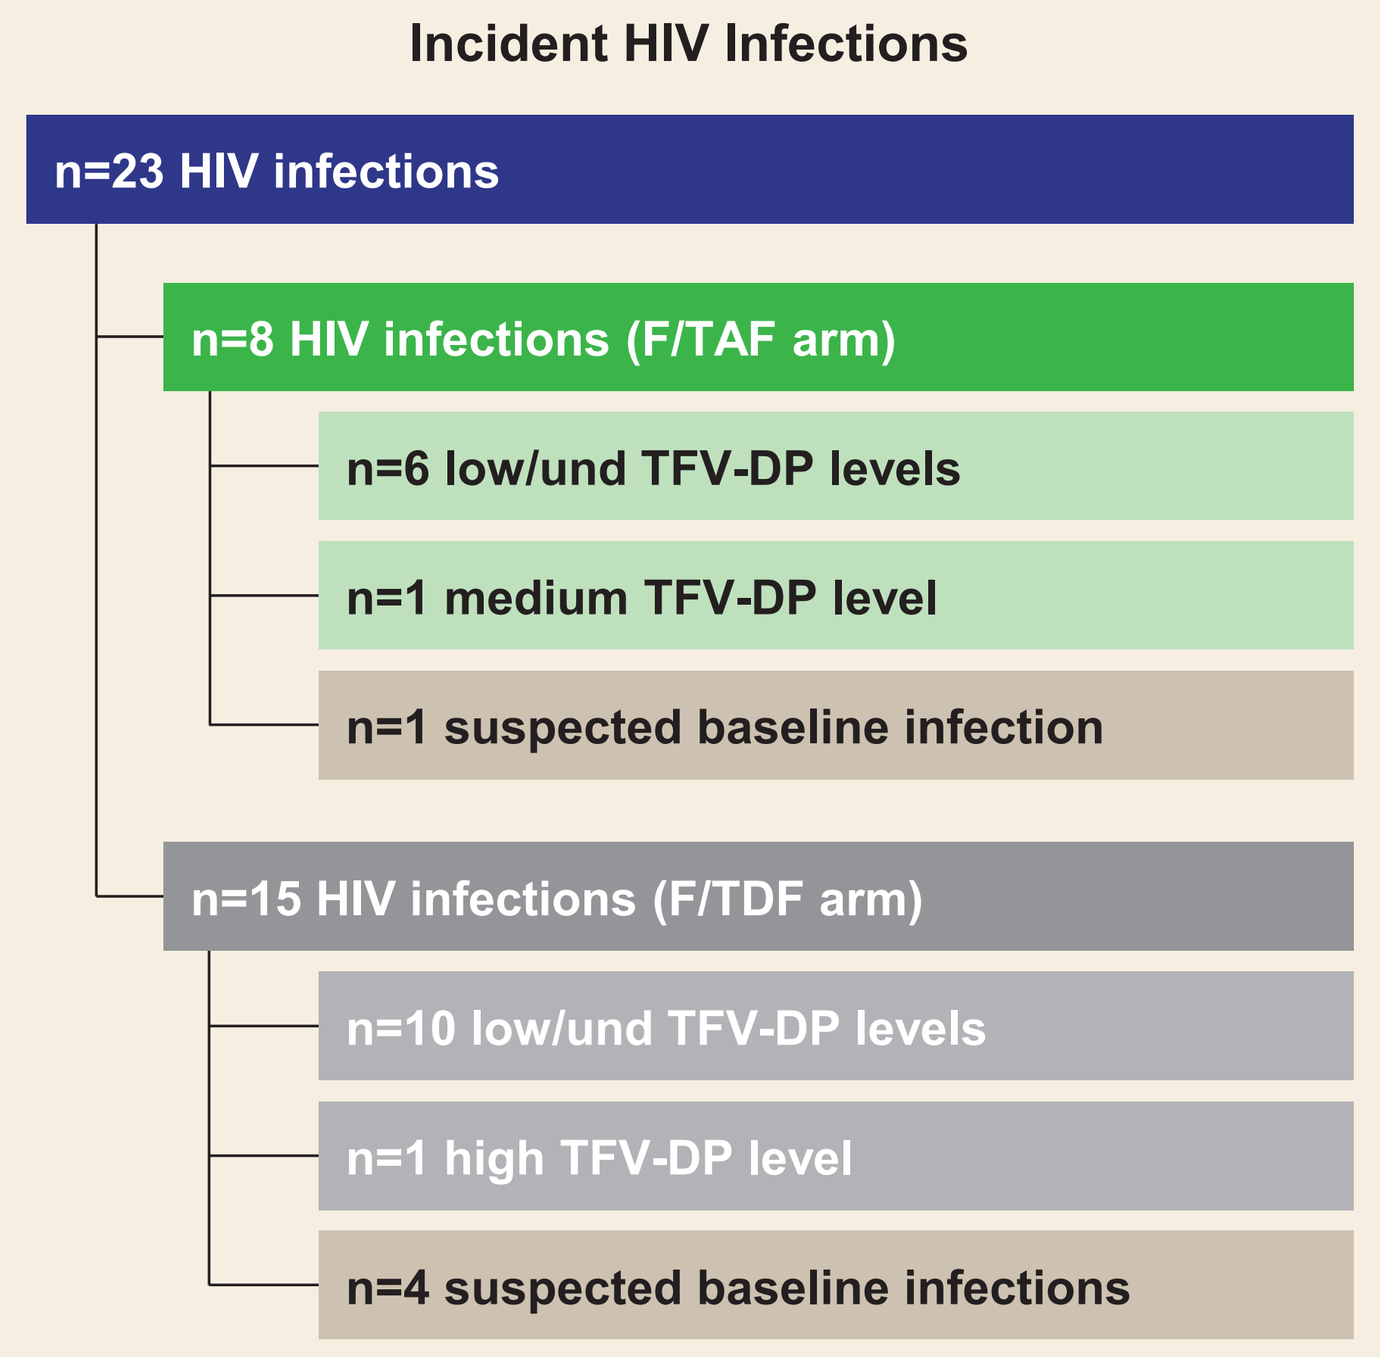
Incident HIV Infections
n=23 HIV infections
n=8 HIV infections (F/TAF arm)
n=6 low/und TFV-DP levels
n=1 medium TFV-DP level
n=1 suspected baseline infection
n=15 HIV infections (F/TDF arm)
n=10 low/und TFV-DP levels
n=1 high TFV-DP level
n=4 suspected baseline infections

CI, confidence interval; IRR, incidence rate ratio; NI, noninferiority; PY, person-year; und, undetectable.

Adherence by Self Report and Pill Count: Overall Study Population

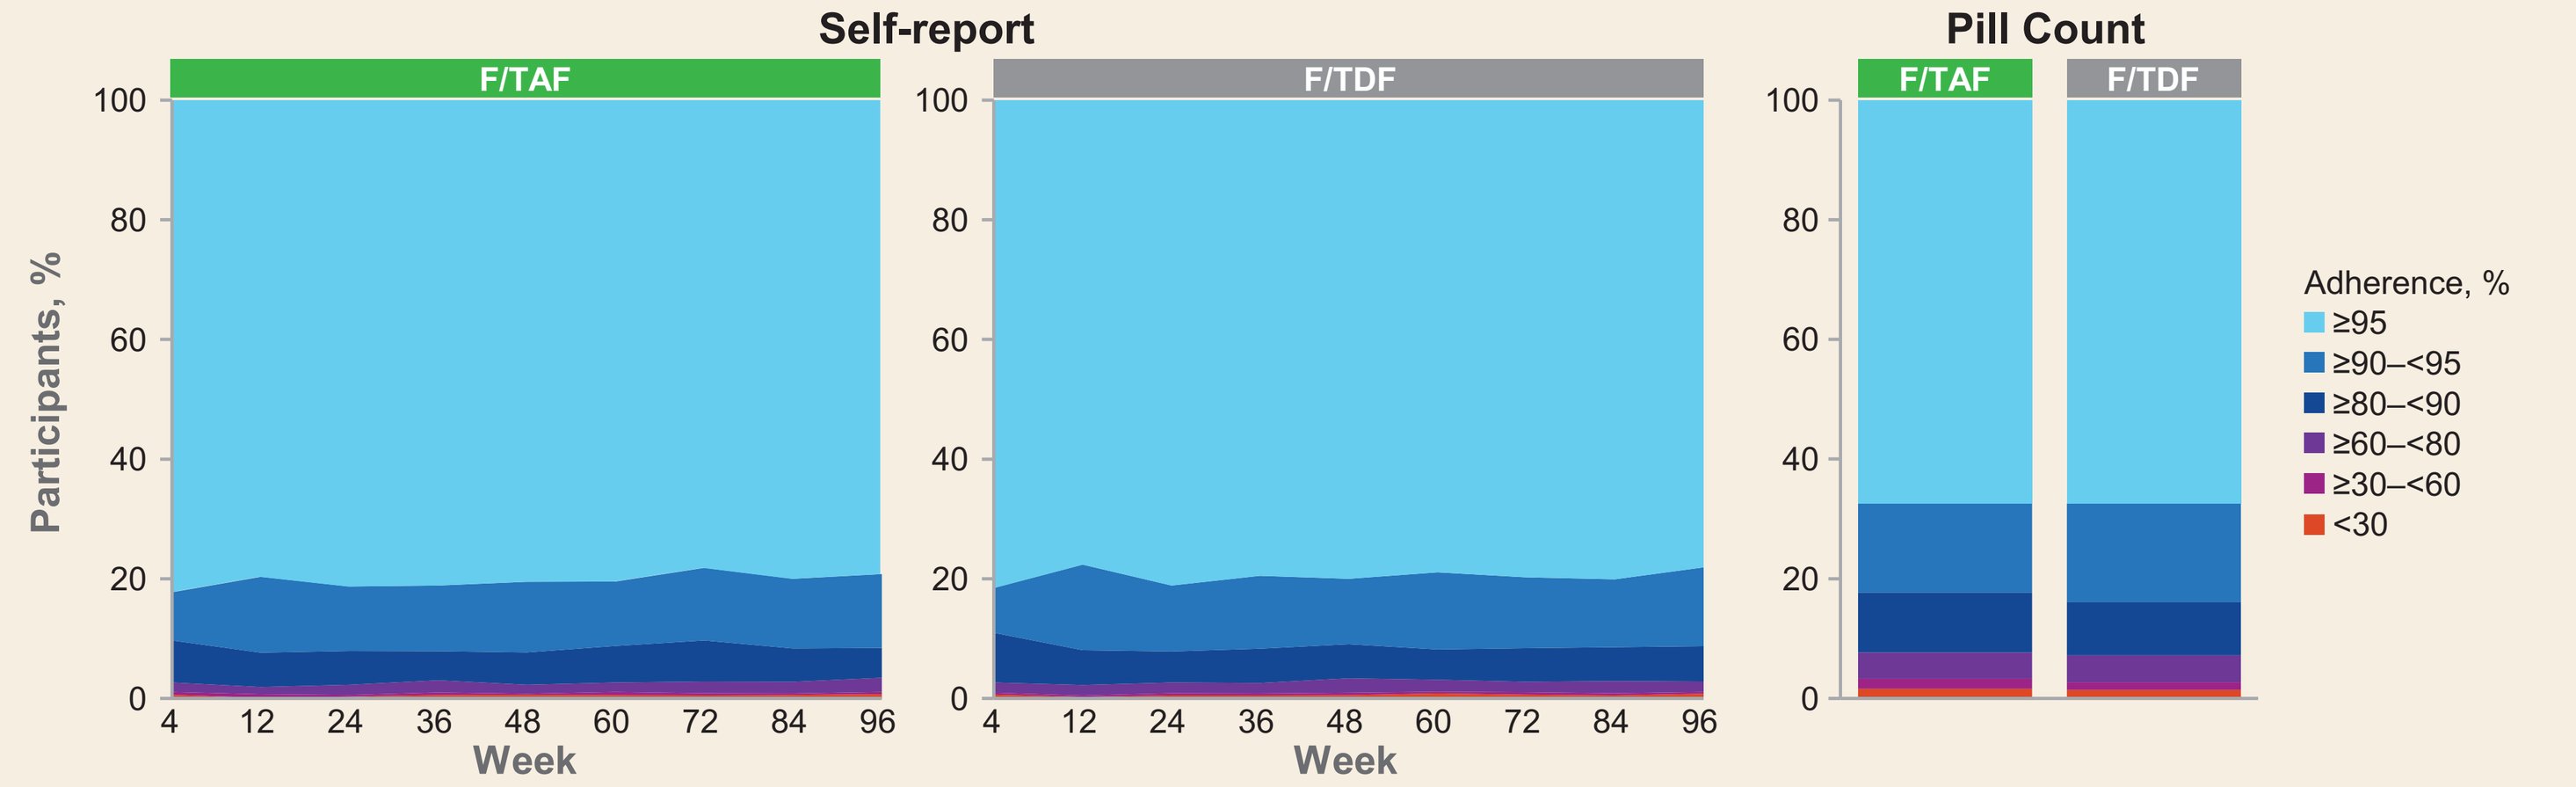
Self-report
Pill Count
F/TAF
F/TDF
Participants, %
Week
Adherence, %
≥95
≥90–<95
≥80–<90
≥60–<80
≥30–<60
<30

- 78–82% of participants in both treatment groups self-reported ≥95% adherence over 96 wk
- Median adherence by pill count over 96 wk: 98% in both F/TAF and F/TDF groups
 - ≥95% adherence: 68% in F/TAF and 67% in F/TDF groups

Adherence by TFV-DP DBS Concentration

DBS Subset (n=536)

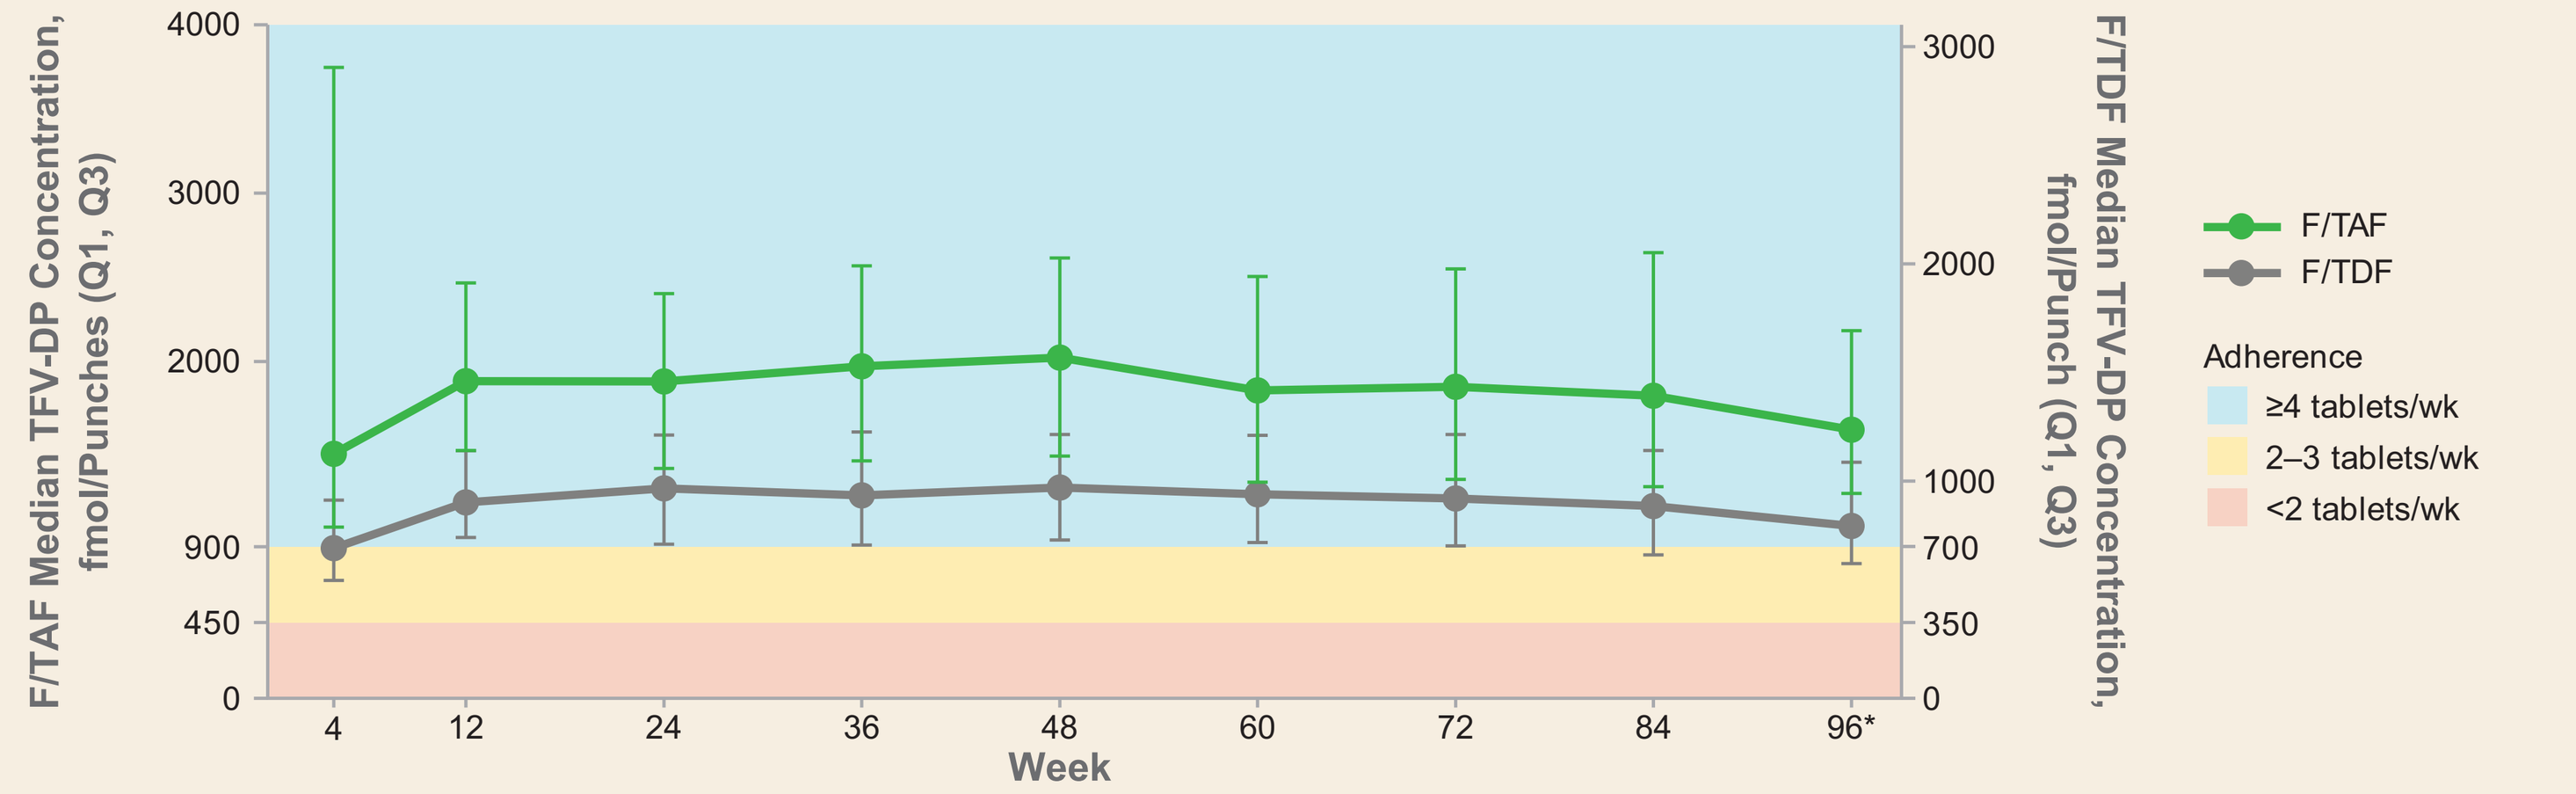
F/TAF Median TFV-DP Concentration, fmol/Punches (Q1, Q3)
F/TDF Median TFV-DP Concentration, fmol/Punch (Q1, Q3)
Week
F/TAF
F/TDF
Adherence
≥4 tablets/wk
2–3 tablets/wk
<2 tablets/wk

Cases (n=23) and Controls (n=114)†

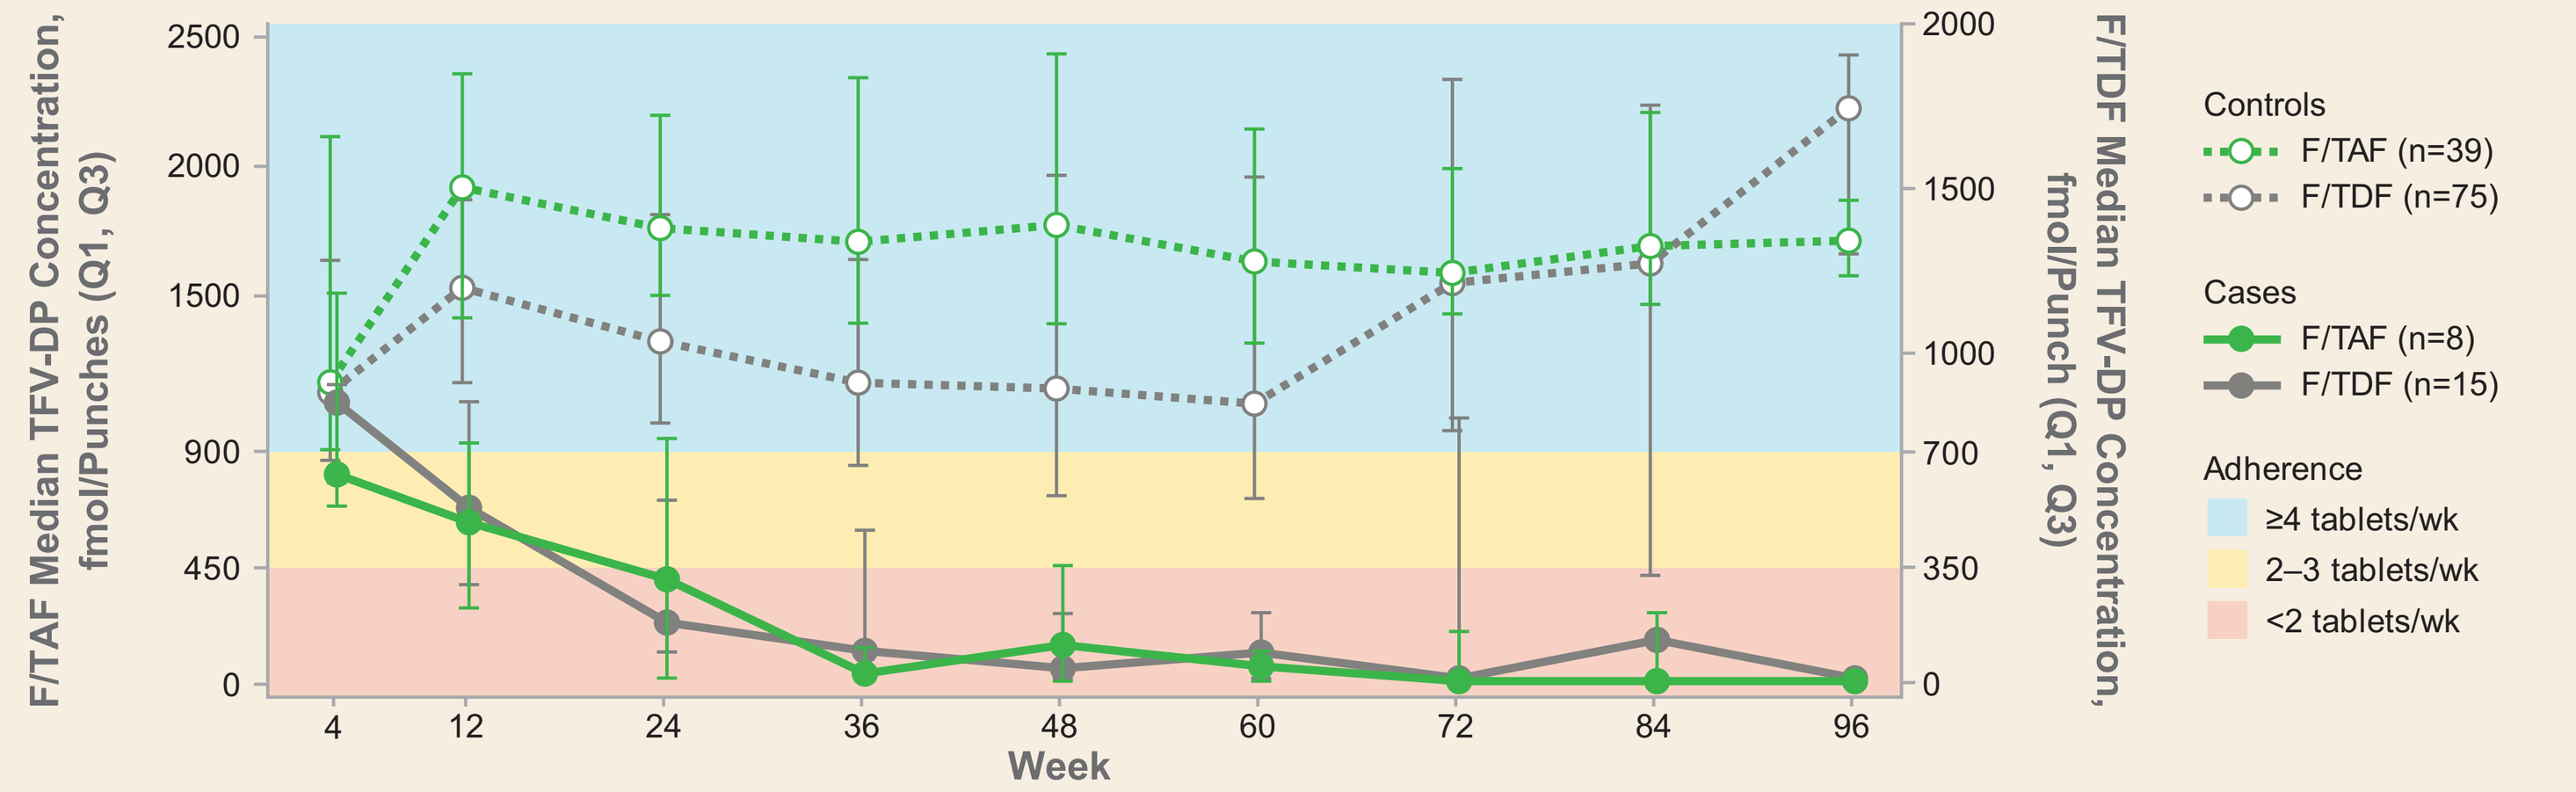
F/TAF Median TFV-DP Concentration, fmol/Punches (Q1, Q3)
F/TDF Median TFV-DP Concentration, fmol/Punch (Q1, Q3)
Week
Controls
F/TAF (n=39)
F/TDF (n=75)
Cases
F/TAF (n=8)
F/TDF (n=15)
Adherence
≥4 tablets/wk
2–3 tablets/wk
<2 tablets/wk

*Data from primary analysis cut when 50% of study population reached Week 96; †Not all cases and controls had data at every study visit. Q, quartile.

TFV-DP DBS Concentrations: Cases, Controls, and at Time of HIV Diagnosis

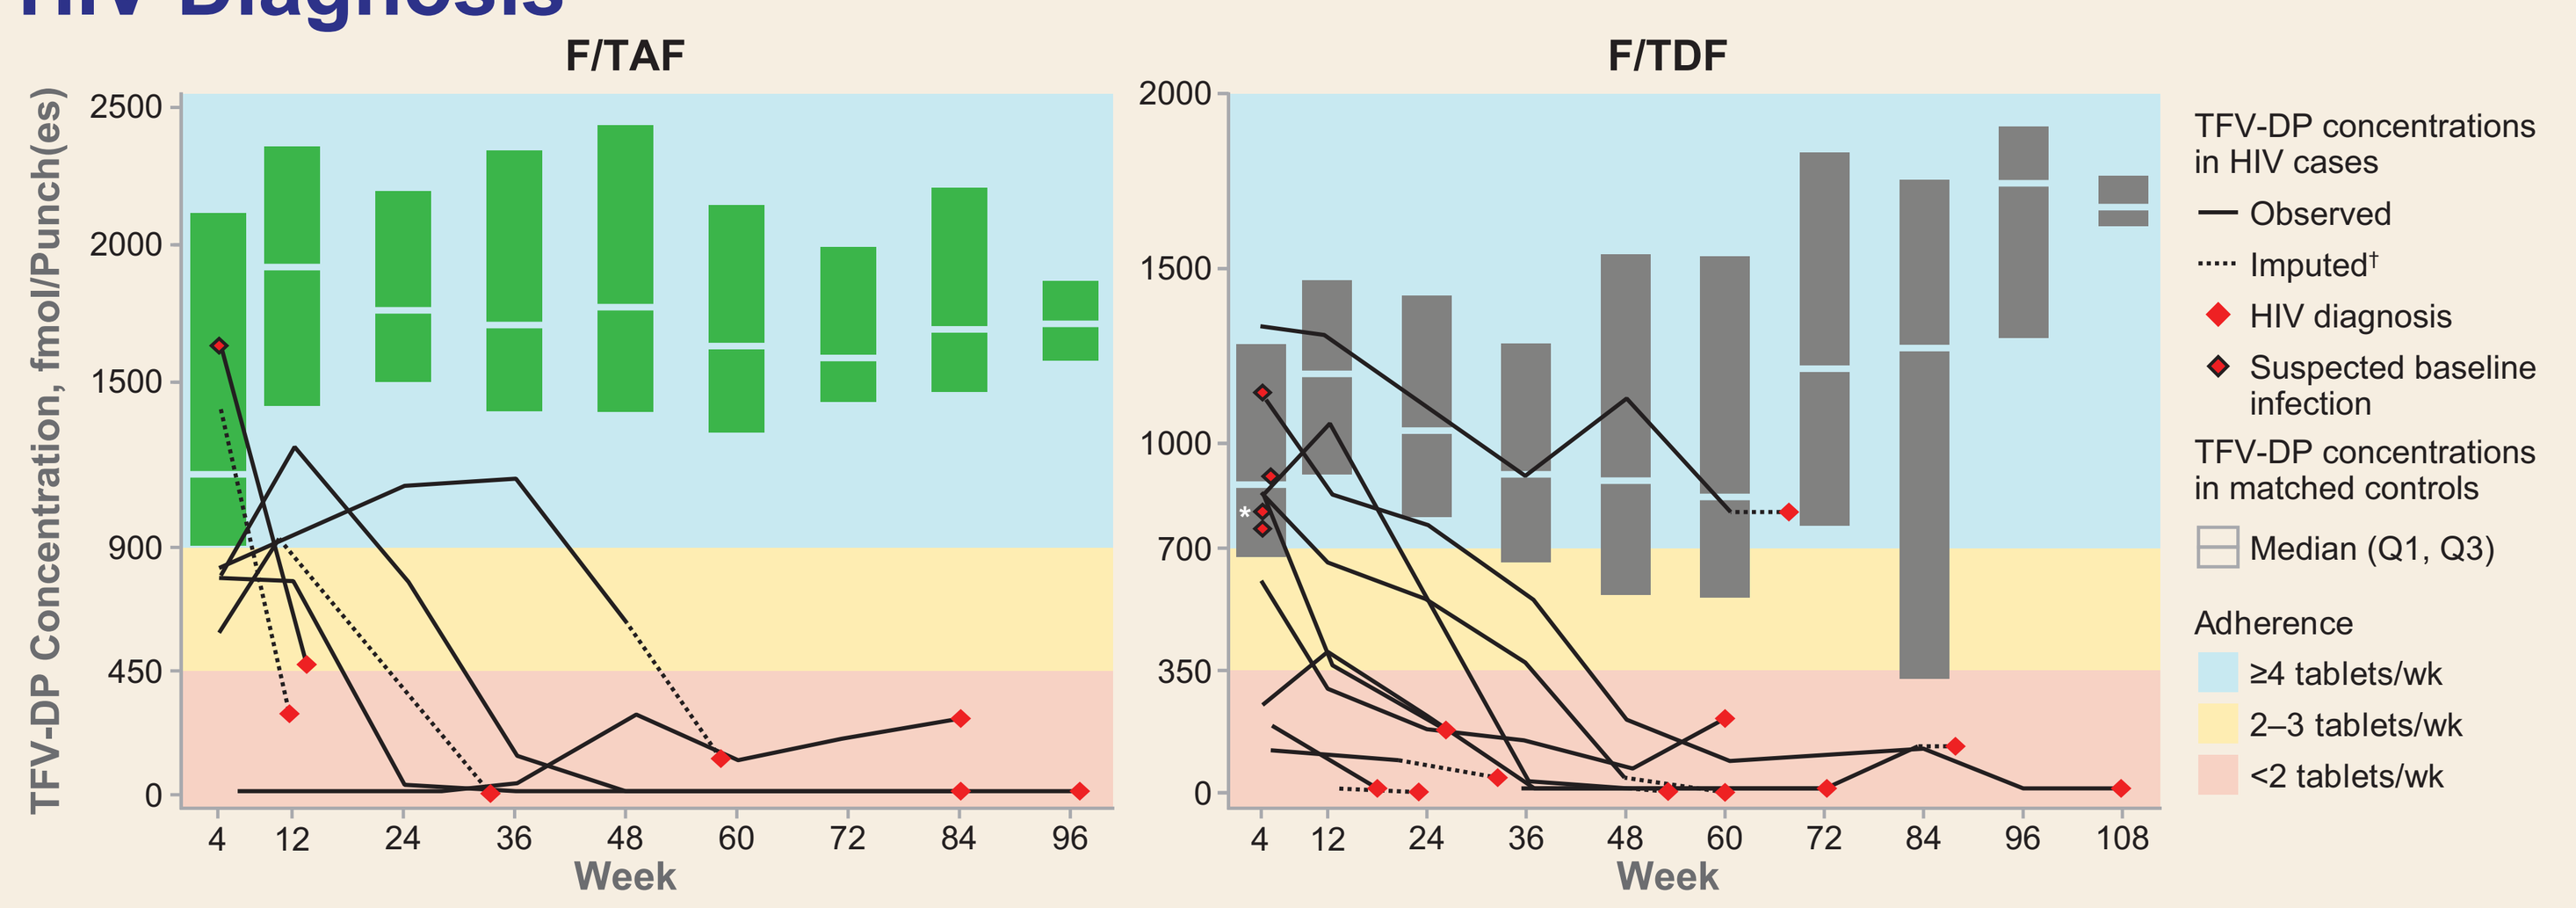
F/TAF
F/TDF
TFV-DP Concentration, fmol/Punch(es)
Week
TFV-DP concentrations in HIV cases
Observed
Imputed†
HIV diagnosis
Suspected baseline infection
TFV-DP concentrations in matched controls
Median (Q1, Q3)
Adherence
≥4 tablets/wk
2–3 tablets/wk
<2 tablets/wk

*Participant originally diagnosed with HIV infection at Week 12 visit, but retrospective viral load testing of Week 4 sample confirmed participant had acquired HIV infection prior to Week 4; †If at time of HIV diagnosis TFV-DP levels from DBS were missing, TFV-DP from previous visit was carried forward up to last dose date and then decayed by number of half-lives (20 d for F/TAF and 17 d for F/TDF) since last dose date to day of diagnosis.

- TFV-DP concentrations at HIV diagnosis visit were significantly lower in participants diagnosed with HIV (cases) vs uninfected matched controls (p <0.001)
 - F/TAF: cases, median 204 fmol/punches (Q1, Q3 13, 385); controls, 1725 (1414, 2216)
 - F/TDF: cases, median 133 fmol/punch (Q1, Q3 13, 755); controls, 1075 (712, 1533)

Baseline Characteristics Associated With Low (<2 tablets/wk) Adherence (OR >1) as Assessed by DBS

Univariate Logistic Regression*	Comparison	OR Estimate (95% CI)
Using F/TDF for PrEP at baseline	No vs yes	2.91 (1.14, 7.44)
Race	Black vs nonblack	2.37 (1.17, 4.79)
Region	US vs ex-US	2.27 (1.28, 4.04)
Age	< vs ≥25 y	2.22 (1.07, 4.60)
Highest level of education	< vs ≥4-y college	2.17 (1.29, 3.65)

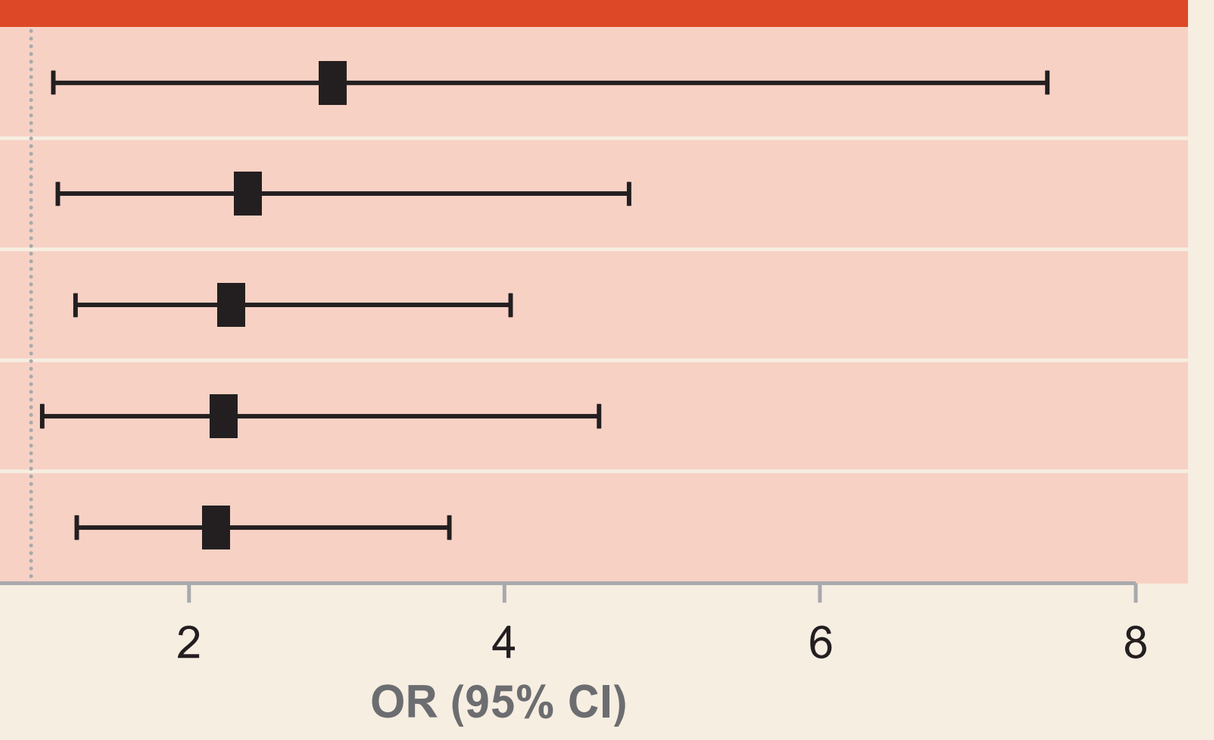
0
2
4
6
8
OR (95% CI)

*Baseline characteristics that were not statistically significant in univariate model were: recreational drug use, ethnicity, diagnosis of rectal gonorrhea, rectal chlamydia, or syphilis in 6 mo prior to screening, binge alcohol use, no. of partners for condomless receptive anal sex in 3 mo prior to screening, and circumcision status.

Multivariate Logistic Regression†	Comparison	OR Estimate (95% CI)
Region	US vs ex-US	1.98 (1.10, 3.56)
Highest level of education	<4 y vs ≥4 y college	2.21 (1.30, 3.75)

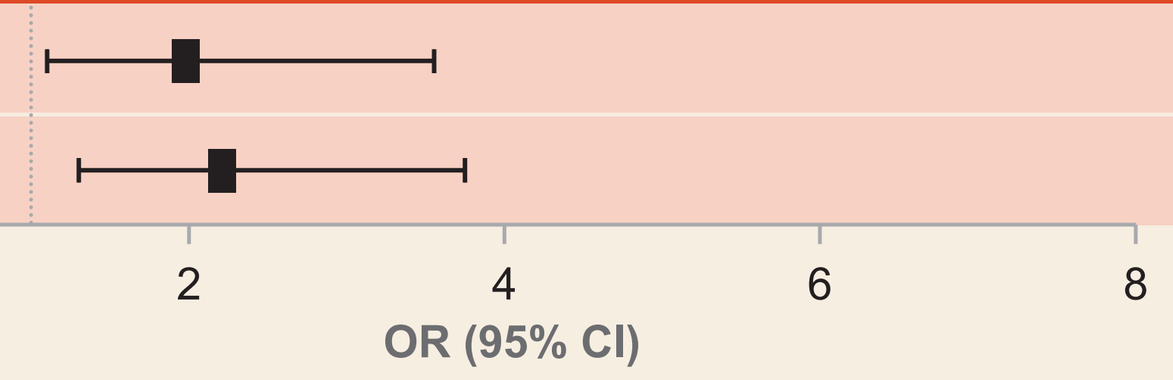
0
2
4
6
8
OR (95% CI)

†Stepwise logistic regression was used to screen for significant predictors in multivariate model, which included: age group (< or ≥25 y), race (black or nonblack), ethnicity, region, education level, alcohol use, recreational drug use, no. of partners for condomless receptive anal sex (≤ or >3), STIs, and baseline F/TDF for PrEP. OR, odds ratio.

Conclusions

- Overall, DISCOVER participants were highly adherent to study medications by self-report and pill count and, correspondingly, low rates of HIV acquisition were reported in both treatment arms
- Most people who acquired HIV-1 had low adherence with dosing of <2 tablets/wk (16/18 [89%])
- In an exploratory univariate logistic regression, 5 baseline characteristics were associated with low adherence (<2 tablets/wk): not using F/TDF at baseline, black race, US residence, age <25 y, and <4-y college education
- In a multivariate analysis, only US residence and <4-y college education remained strongly associated with low adherence, as selected by stepwise logistic regression
- Notably, in this population with high levels of self-reported condomless anal sex partners, lab-identified rectal chlamydia/gonorrhea, recreational drug use (67%), and binge drinking (56%), these factors were not associated with low adherence

References: 1. Buchbinder SP, et al. Top Antivir Med 2018;25:138-42; 2. Hare CB, et al. CROI 2019, abstr 104; 3. Spinner C, et al. IAS 2019, abstr TUAC0403LB; 4. Anderson PL, et al. Antimicrob Agents Chemother 2017;62:e01710-7; 5. Yager J, et al. CROI 2019, abstr 463; 6. Zheng JH, et al. J Pharm Biomed Anal 2016;122:16-20.

Acknowledgments: We extend our thanks to the participants, their families, and all participating study investigators and staff: *Austria:* B Haas, A Rieger; *Canada:* J Brunetta, JJ de Wet, J Szabo, C Tremblay, B Trottier; *Denmark:* J Gerstoft, G Kronborg, C Larsen, D Larsen; *France:* E Cua, J-M Molina, P Philibert, G Pialoux; *Germany:* H Jessen, G Knecht, I Krznaric, C Spinner; *Ireland:* C Bergin, P Mallon; *Italy:* A Antinori, A Lazzarin; *Netherlands:* M Prins; *Spain:* J Coll, M Crespo, J del Romero Gerrero, D Podzamczer; *UK:* V Apea, A Clarke, O Dosekun, R Gilson, S Kegg, C Leen, N Nwokolo, F Post, I Reeves, G Schembri, S Taylor; *USA:* D Asmuth, A Avery, P Benson, M Berhe, I Brar, C Brinson, JH Burack, T Campbell, M Cespedes, M Coleman, CM Creticos, GE Crofoot, FA Cruickshank, E Daar, E DeJesus, W Dinges, S Doblecki-Lewis, T Donovan, J Flamm, JE Gallant, J Gladstein, RM Grant, R Grossberg, J Halperin, WD Hardy, CB Hare, S Hassler, R Hengel, K Henry, T Hodge, S Hosek, M Iandorio, A LaMarca, C Lucasti, S Mannheimer, CT Martorell, M Markowitz, K Mayer, A Mills, S Morris, K Mounzer, O Ogbuagu, O Osiyemi, A Petroll, J Phoenix, MN Ramgopal, B Rashbaum, GJ Richmond, PJ Ruane, L Salazar, AJ Scarsella, M Scott, P Shalit, JL Stephens, MA Thompson, G Voskuhl, BH Wade, DA Wohl, K Workowski, B Young. This study was funded by Gilead Sciences, Inc.

Presented at Conference on Retroviruses and Opportunistic Infections, March 8–11, 2020, Boston, MA

©2020 Gilead Sciences, Inc. All rights reserved.