529

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Absence of GS-6207 Phenotypic Resistance in HIV Gag Cleavage Site Mutants and Isolates With Resistance to Existing Drug Classes

Nicolas A. Margot, Renee R. Ram, Martin Rhee, Christian Callebaut — Gilead Sciences, Inc., Foster City, CA

Introduction

- HIV capsid protein (CA; p24) is a product of HIV group-specific antigen (gag) processing by HIV protease, which plays a key role in the HIV life cycle (Fig 1)
- GS-6207 is a first-in-class HIV CA inhibitor (CAI; Fig 2), with a multistage mode of action and picomolar potency (Fig 3)[1]
- GS-6207 binds to the interface between 2 CA monomers, and prevents CA-mediated nuclear entry of viral DNA, HIV assembly, and proper capsid formation (Fig 3)[1]
- GS-6207 physicochemical properties make GS-6207 suitable as a long-acting injectable agent: picomolar antiviral potency, low predicted clearance, and low aqueous solubility[1]
- In clinical studies, a single SC dose of GS-6207 sustained measurable concentrations in HIV-negative participants for ≥12 wk and demonstrated potent antiviral activity in people living with HIV (PLWH) over 10 d (up to 2.2-$\log_{10}$ decline in HIV-1 RNA; Fig 4)[2,3]
- In vitro dose-escalation studies identified variants in the CA portion of gag (L56I, M66I, Q67H, K70N, N74D/S, and T107N) associated with reduced susceptibility to GS-6207 coupled with reduced viral fitness[4]
- Treatment of PLWH with protease inhibitors (PIs) has led to the emergence of gag cleavage site mutations (GCSMs) associated with PI resistance,[5-7] which could alter GS-6207 antiviral activity
- In addition, the presence of naturally occurring HIV gag polymorphisms could affect the potency of GS-6207, as is the case for maturation inhibitors (MIs), such as bevirimat (BVM), which targets the final gag cleavage step prior to CA release[8]
- Here, we studied the antiviral activity of GS-6207 in HIV-1 mutants harboring GCSMs and/or naturally occurring gag polymorphisms, as well as in mutants with resistance to existing drug classes

Fig 1: HIV-1 Capsid Core Formation

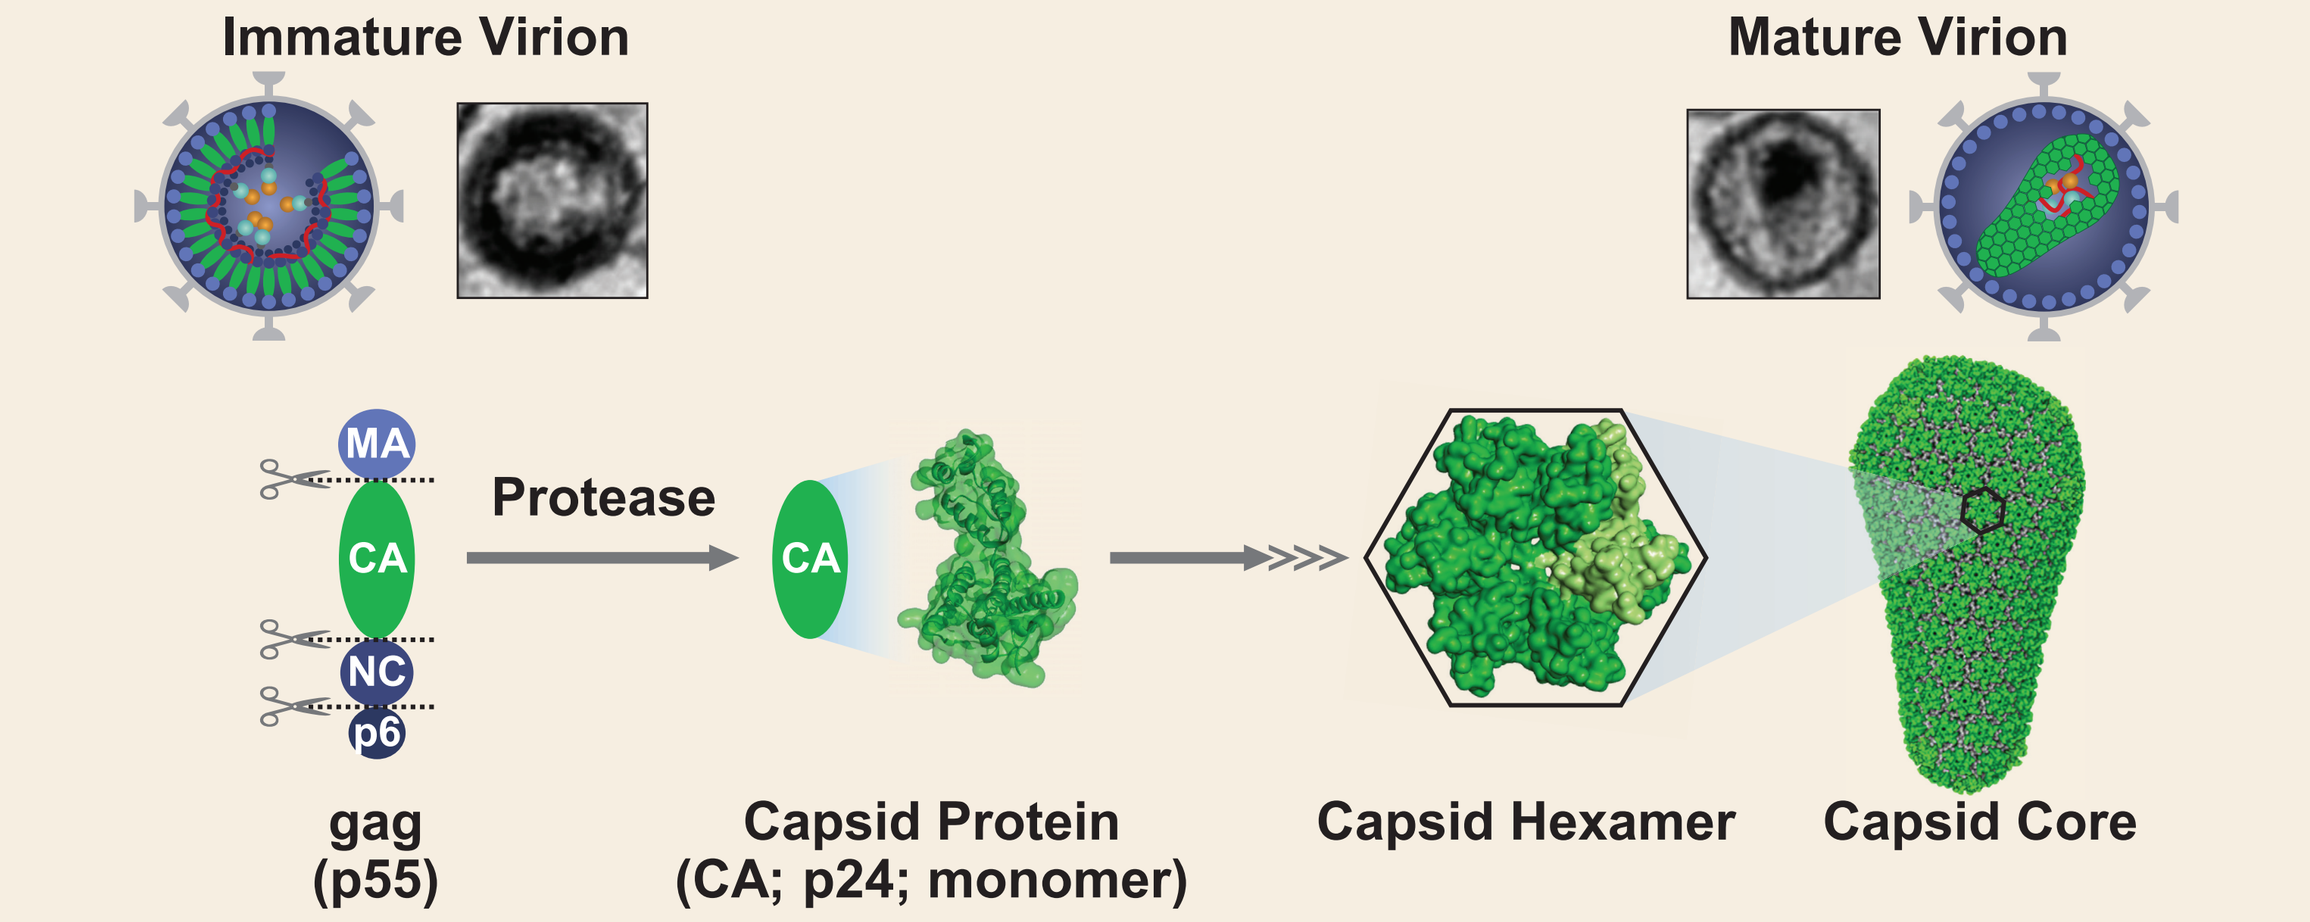
Immature Virion
Mature Virion
MA
CA
NC
p6
Protease
CA
gag
(p55)
Capsid Protein
(CA; p24; monomer)
Capsid Hexamer
Capsid Core

MA, matrix; NC, nucleocapsid.

Fig 2: GS-6207 Structure

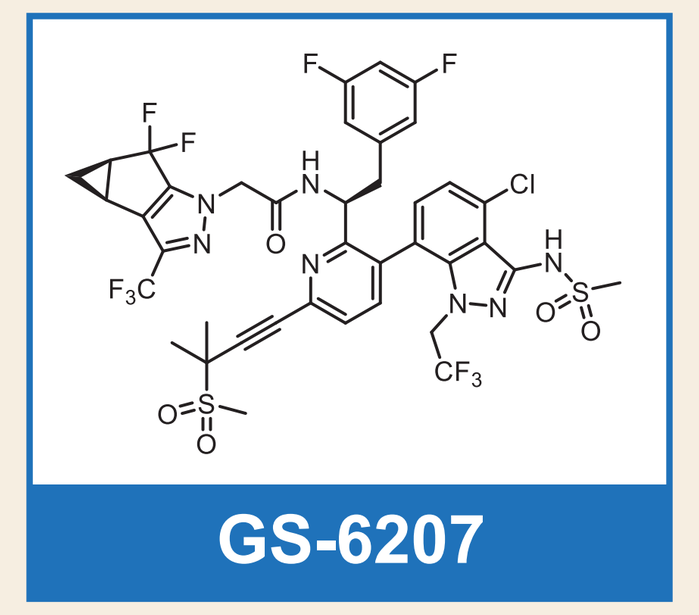
GS-6207

Fig 3: Multistage Mode of Action of GS-6207

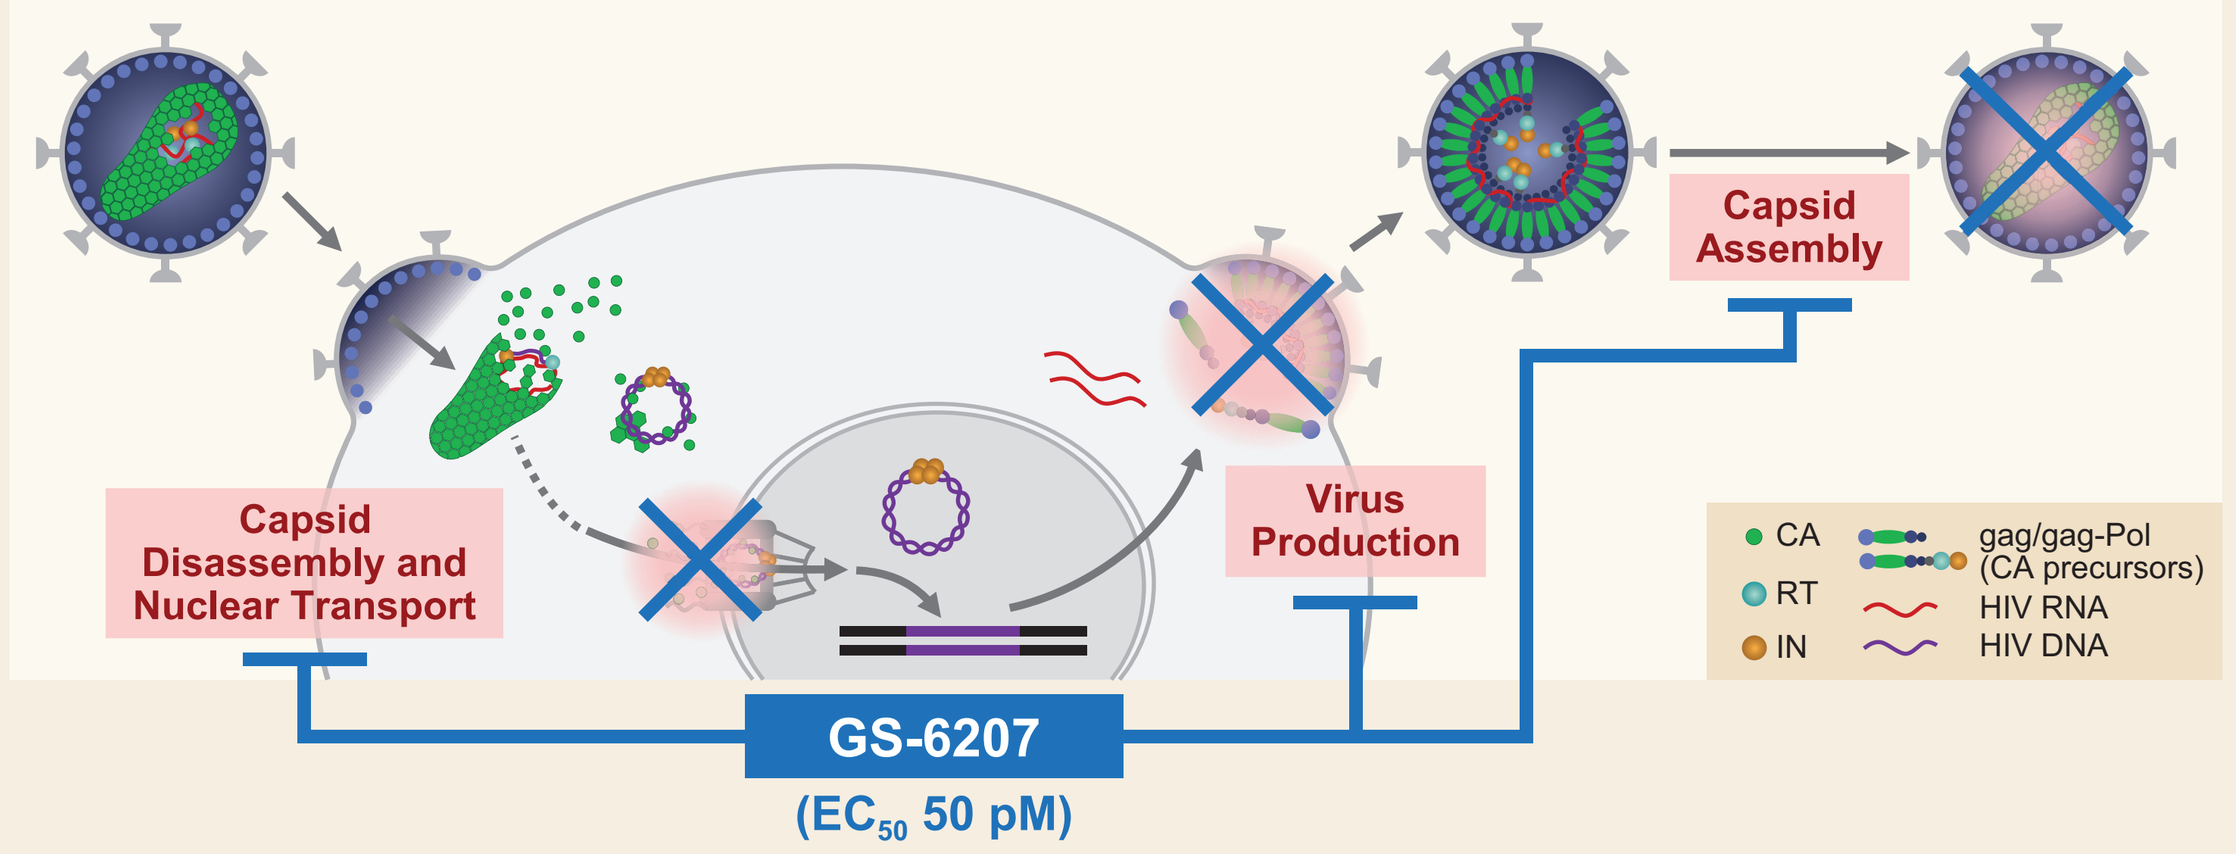
Capsid Assembly
Capsid Disassembly and Nuclear Transport
Virus Production
GS-6207
(EC50 50 pM)
CA
RT
IN
gag/gag-Pol (CA precursors)
HIV RNA
HIV DNA

EC_{50}, half-maximal effective concentration determined in peripheral blood mononuclear cells using wild-type (WT) HIV-1 patient isolates with various HIV-1 subtypes. IN, integrase; Pol, polymerase; RT, reverse transcriptase.

Fig 4: Phase 1b Viral Response

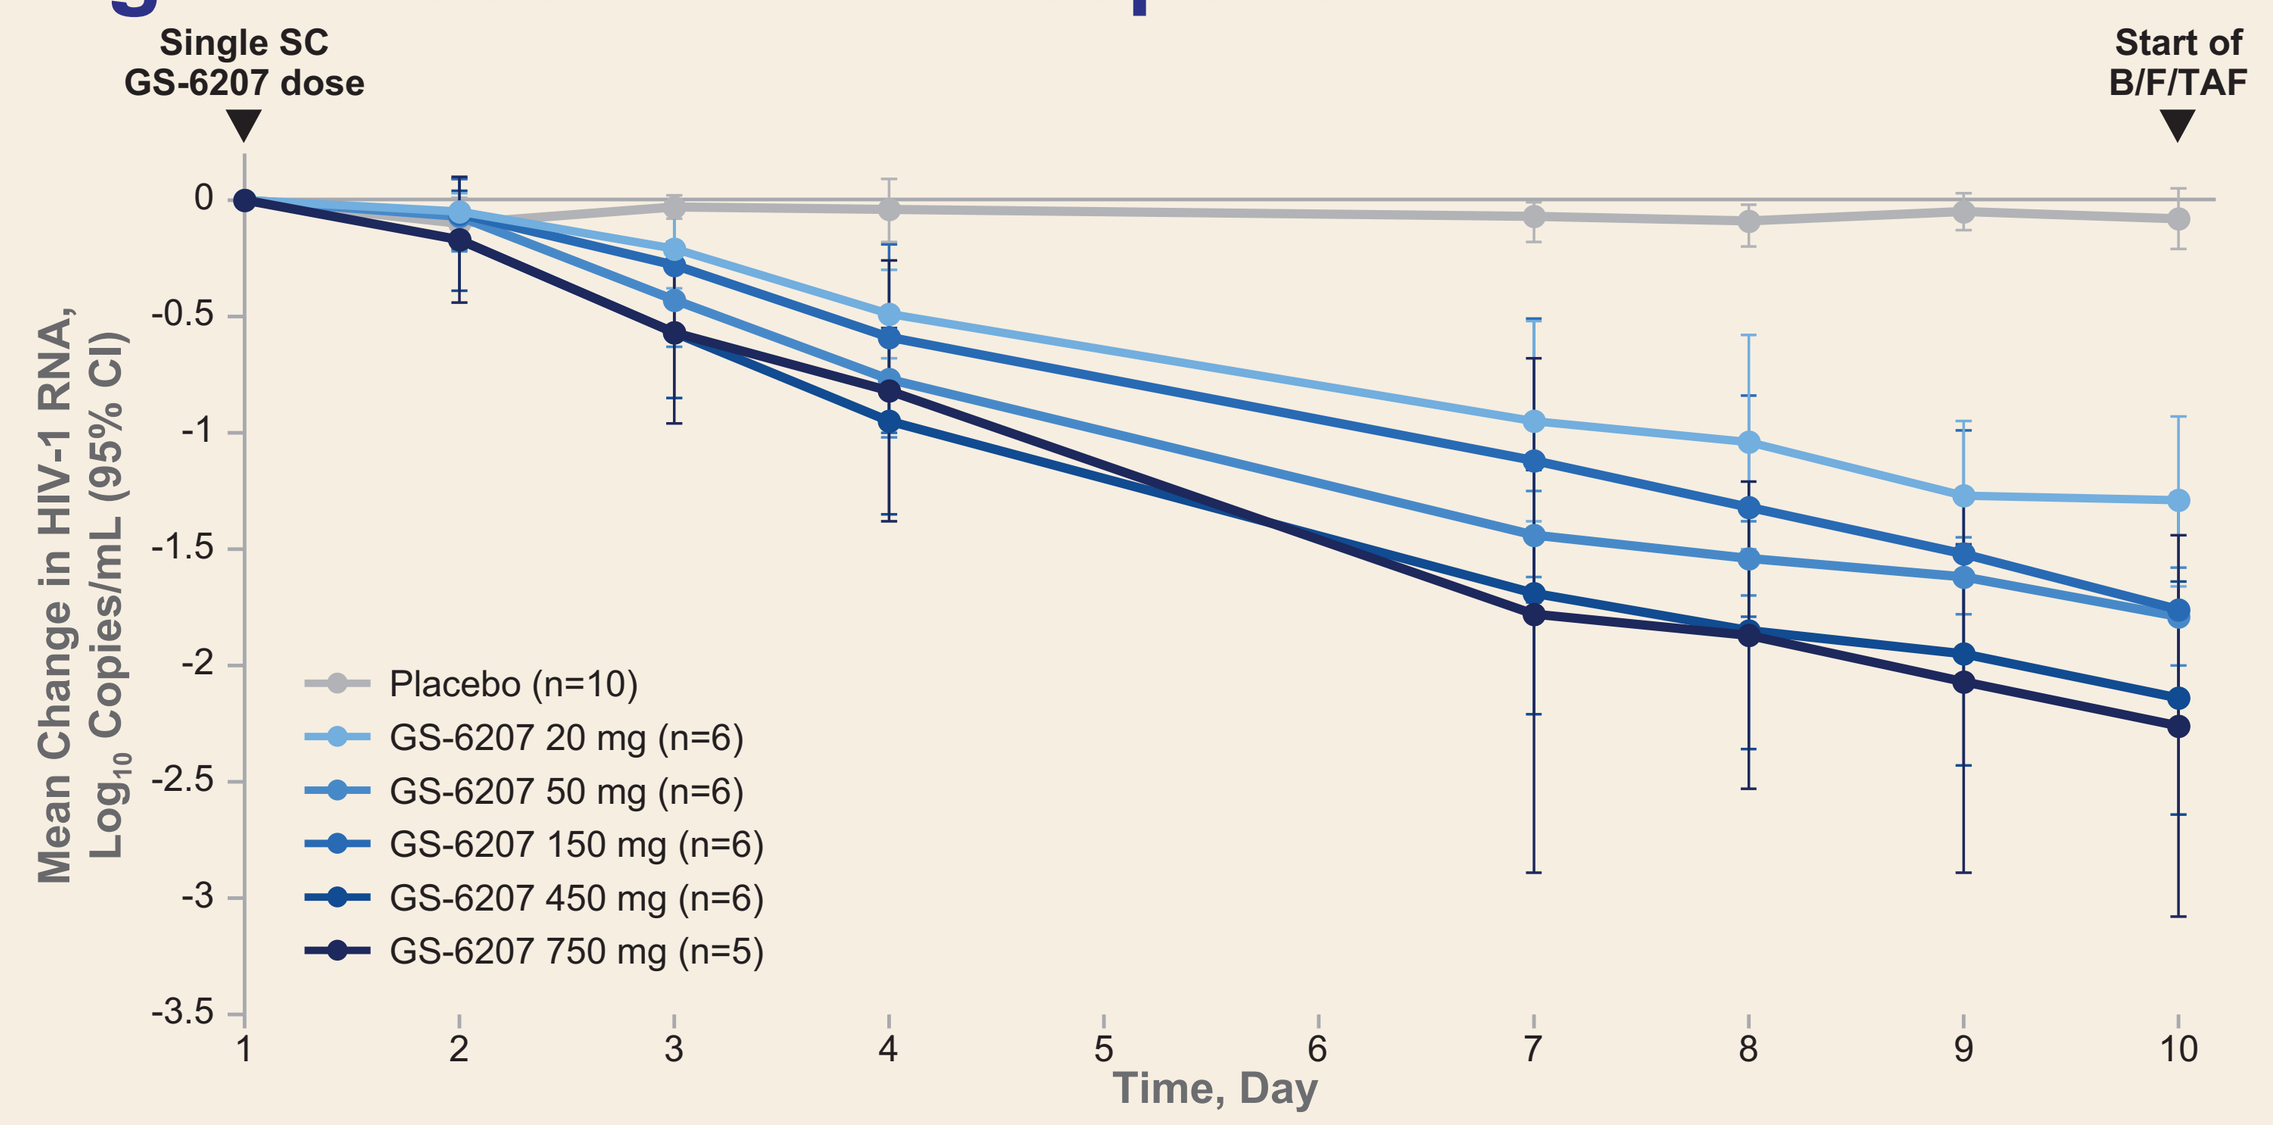
Single SC GS-6207 dose
Start of B/F/TAF
Mean Change in HIV-1 RNA, Log10 Copies/mL (95% CI)
Time, Day
Placebo (n=10)
GS-6207 20 mg (n=6)
GS-6207 50 mg (n=6)
GS-6207 150 mg (n=6)
GS-6207 450 mg (n=6)
GS-6207 750 mg (n=5)

B/F/TAF, bictegravir/emtricitabine/tenofovir alafenamide; CI, confidence interval.

Methods

- Single and double mutants (N=19) containing GCSMs with or without PI resistance mutations V82A or I84V (K436E, I437T, I437V ± V82A, K436E + I437T, A431V ± V82A, A431V ± I84V, L449H, L449V, L449F ± I84V, Q430R ± I84V, P453L ± I84V, L363F, L363M, or A364V) were constructed by site-directed mutagenesis (GENEWIZ, South Plainfield, NJ) of the proviral infectious clone pXXLAI[8]
- Patient-derived HIV isolates were generated from plasma samples from treatment-experienced (TE) and -naïve (TN) PLWH from past Gilead clinical studies
- The HIV-1 gag-protease fragment from the plasma samples (N=51; 36 TE and 15 TN) were amplified by polymerase chain reaction (PCR), and the unique SfoI and XmaI sites were used to clone the PCR products into the HIV-1 molecular clone pXXLAI using In-Fusion® cloning (Takara Bio USA, Inc., Mountain View, CA)
- HIV-1 constructs were transfected into 293T cells and viral isolates were harvested after 24 h
- Susceptibility (EC_{50}) of HIV-1 isolates to GS-6207 and control drugs was measured in a 5-d multicycle antiviral assay in MT-2 cells, and compared with WT
- The panel of mutants with resistance to existing drug classes (PI, nucleoside RT inhibitor [NRTI], non-NRTI [NNRTI], and IN strand transfer inhibitor [INSTI]; n=40) was analyzed using the PhenoSense® GT or PhenoSense Integrase assays (Monogram Biosciences, South San Francisco, CA)

Results

Table 1: Gag Cleavage Site Mutations in HIV-1 Isolates

	No. of Isolates With GCSMs							
	CA/SP1 GCS		NC/SP2 GCS				SP2/p6 GCS	
Isolate Type	L363F/M	A364V	Q430R	A431V	K436E/S	I437T/V	L449H/V/F	P453L
SDM (n=19)	2	1	2	3	2	4	4	2
TE (n=36)*	—	—	—	13	—	5	5	11
TN (n=15)	—	—	—	—	—	—	—	—

*The 36 TE isolates comprised 24 isolates with GCSMs and 12 without GCSMs. —, none; GCS, gag cleavage site; SDM, site-directed mutant.

Table 2: Drug Susceptibility Summary

	Mean Drug Susceptibilities: EC_{50} Fold-Change From WT (range)					
Isolate Type	GS-6207 (CA)	DRV (PI)	ATV (PI)	BVM (MI)	GSK-795 (MI)	TAF (NRTI)
SDM (n=19)	1.1 (0.7–1.9)	1.2 (0.5–2.3)	1.8 (0.4–3.6)	12 (0.3–>64)	31 (0.7–267)	0.9 (0.5–1.4)
TE (n=36)	0.9 (0.4–1.8)	21 (0.5–>112)	35 (0.5–>66)	40 (0.5–>64)	94 (0.5–>333)	0.8 (0.3–1.4)
With GCSM (n=24)	1.0 (0.4–1.8)	22 (0.5–>112)	36 (0.7–>66)	40 (0.5–>64)	85 (0.5–>333)	0.8 (0.3–1.4)
Without GCSM (n=12)	0.8 (0.5–1.4)	20 (0.5–>112)	31 (0.5–>66)	39 (0.6–>64)	112 (0.6–>333)	0.7 (0.3–1.4)
TN (n=15)	0.9 (0.6–1.6)	1.0 (0.5–1.7)	1.0 (0.5–1.9)	42 (1.7–>64)	72 (1.1–>333)	0.9 (0.5–1.5)

ATV, atazanavir; DRV, darunavir; GSK-795, GSK-3532795/BMS-955176; TAF, tenofovir alafenamide.

Fig 5: Drug Susceptibilities in SDM HIV-1 Isolates With GCSMs (n=19)*

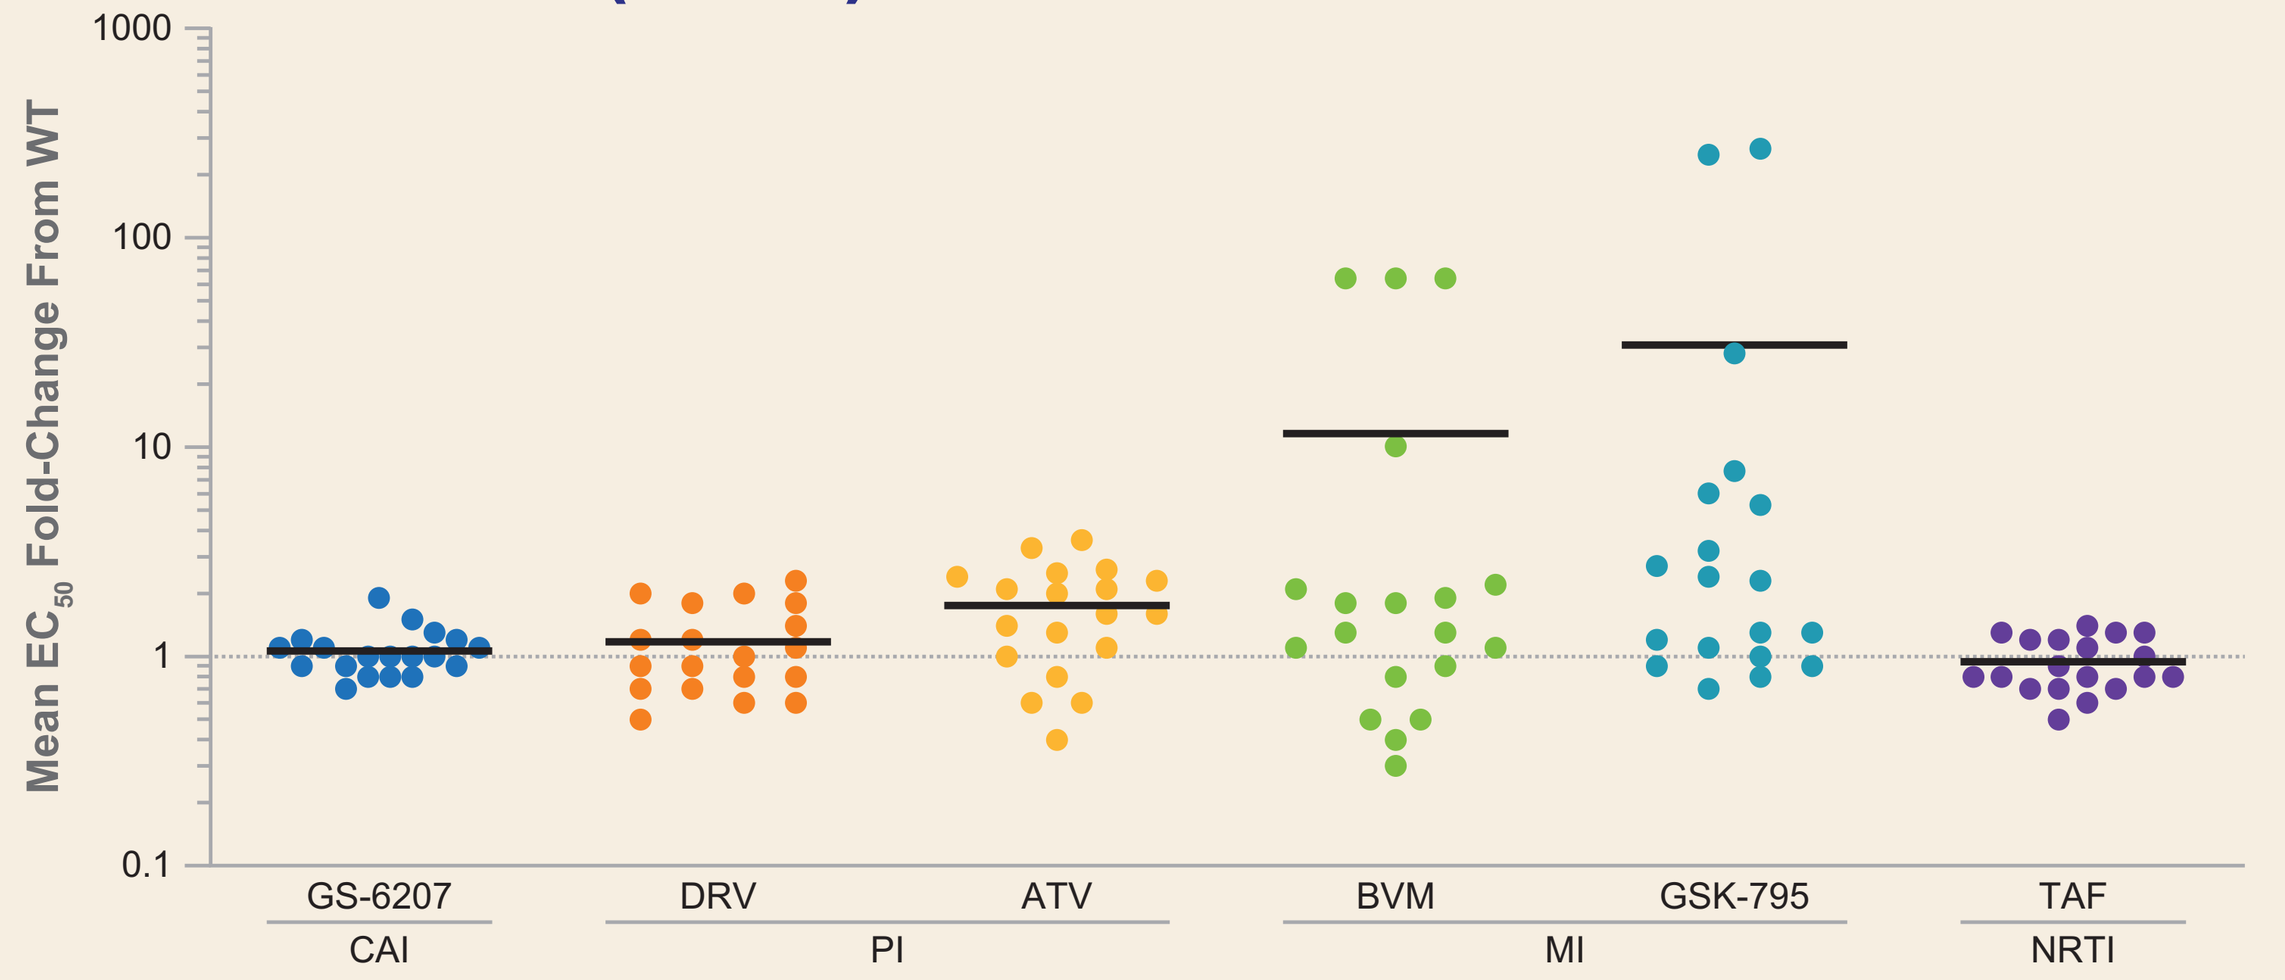
Mean EC50 Fold-Change From WT
GS-6207
DRV
ATV
BVM
GSK-795
TAF
CAI
PI
MI
NRTI

**Each dot* represents a single isolate; *black lines* indicate mean EC_{50} fold-change for each drug.

- In SDM viruses containing GCSMs, GS-6207 showed near WT potency (average EC_{50} 102 vs 95 pM for WT), with limited variability across all 19 mutants
- Low-level reduced susceptibility to DRV and ATV was noted in some SDM viruses with GCSMs, the highest observed with the K436E + I437T mutant at 2.3- and 3.6-fold above WT, respectively

Fig 6: Drug Susceptibilities in Treatment-Experienced HIV-1 Isolates

With GCSMs (n=24)*

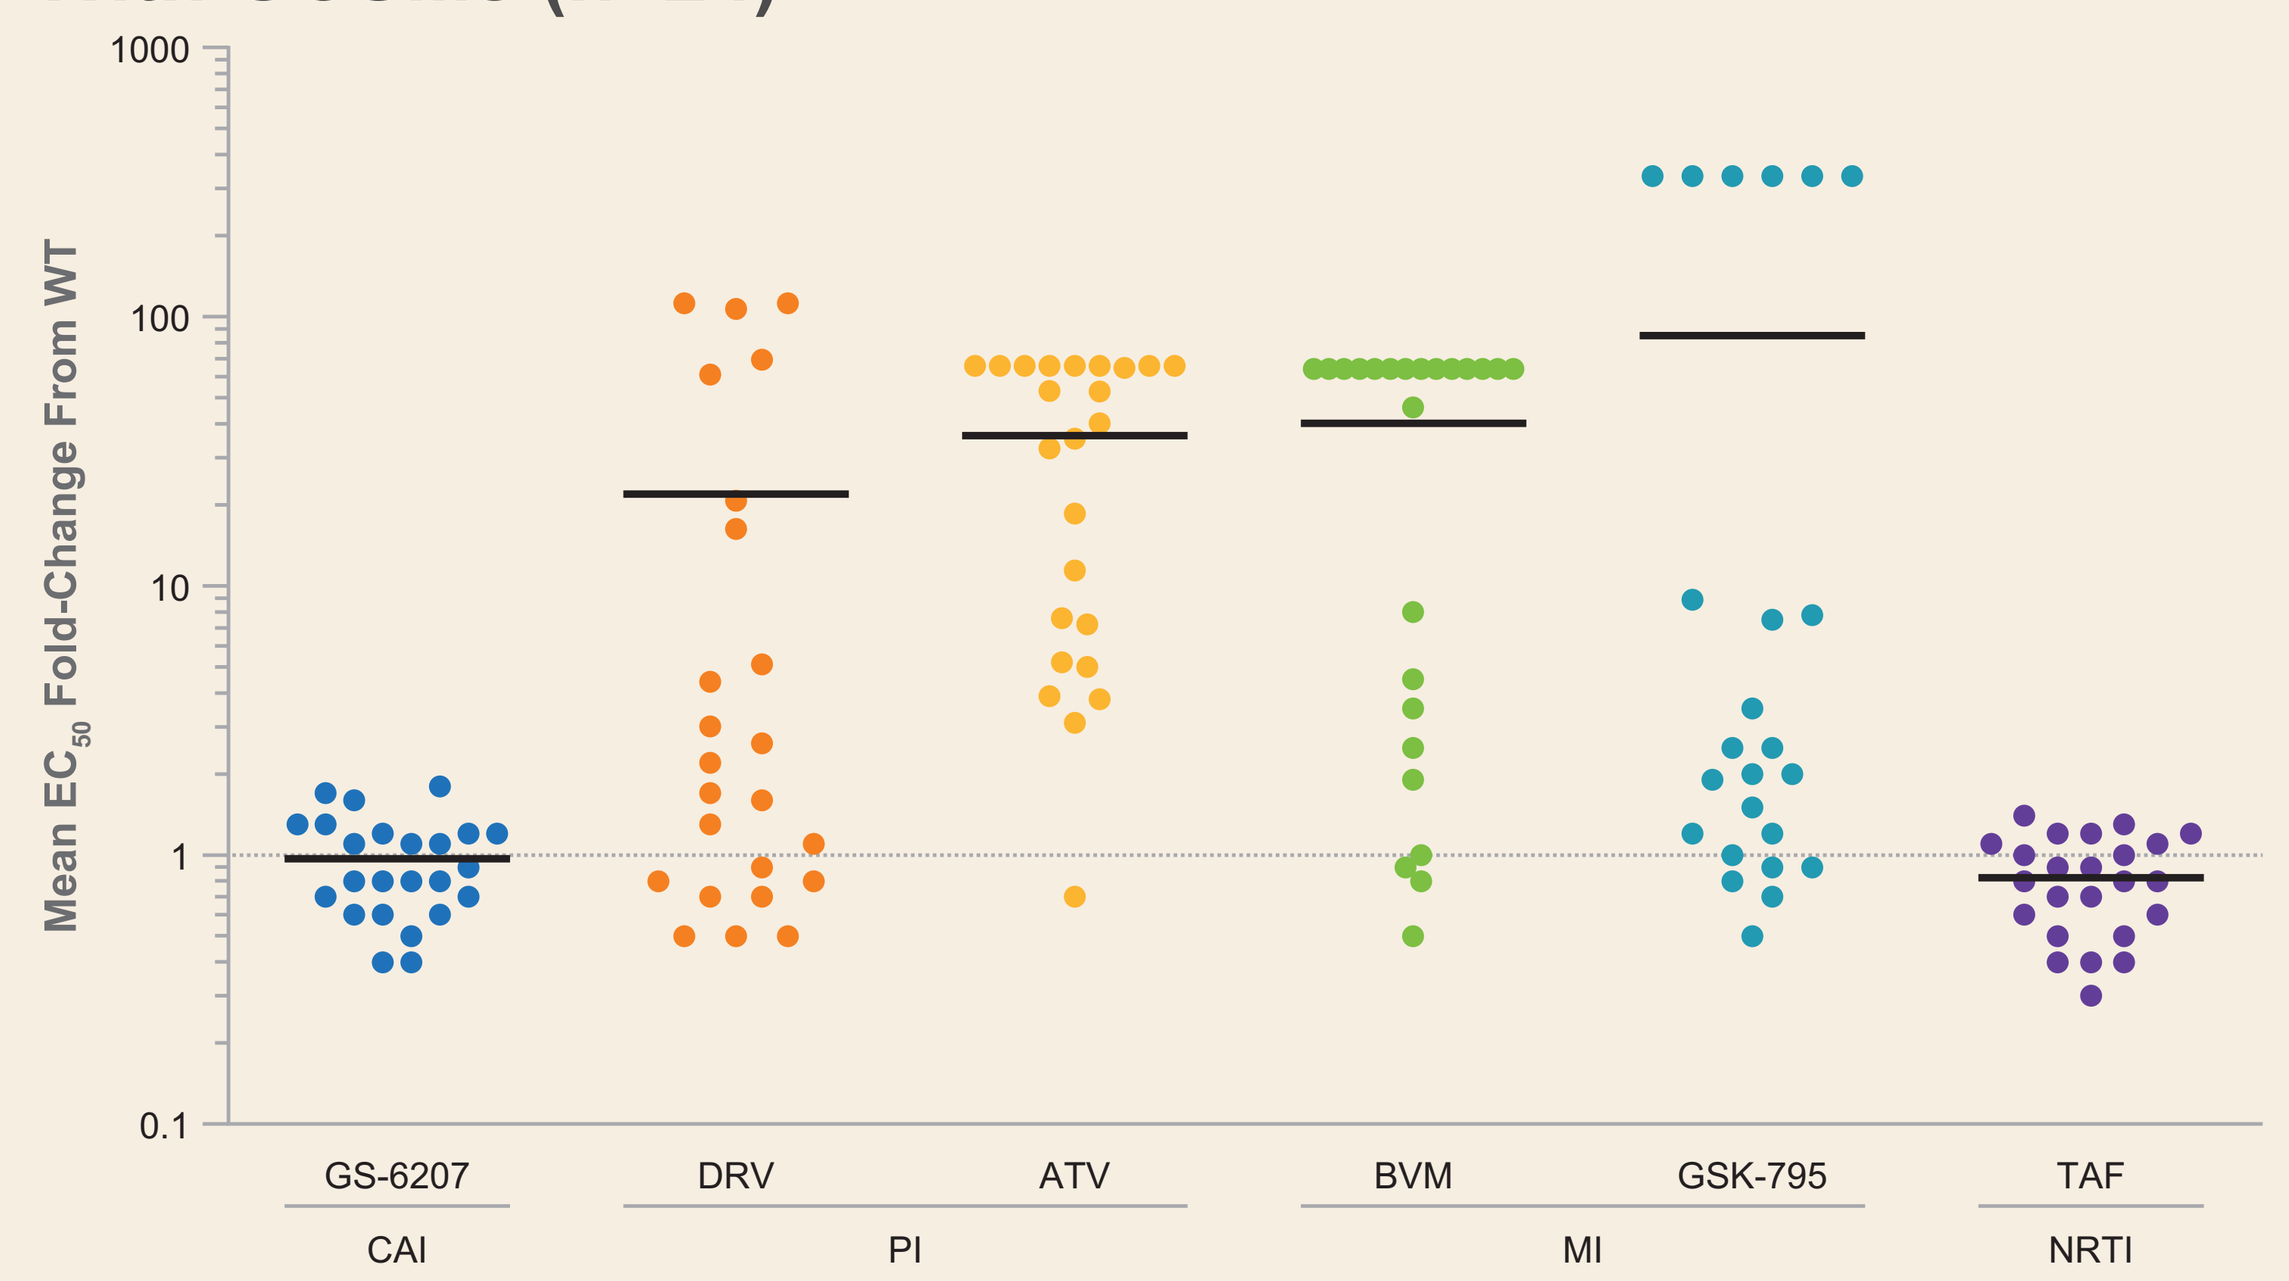
Mean EC50 Fold-Change From WT
GS-6207
DRV
ATV
BVM
GSK-795
TAF
CAI
PI
MI
NRTI

Without GCSMs (n=12)

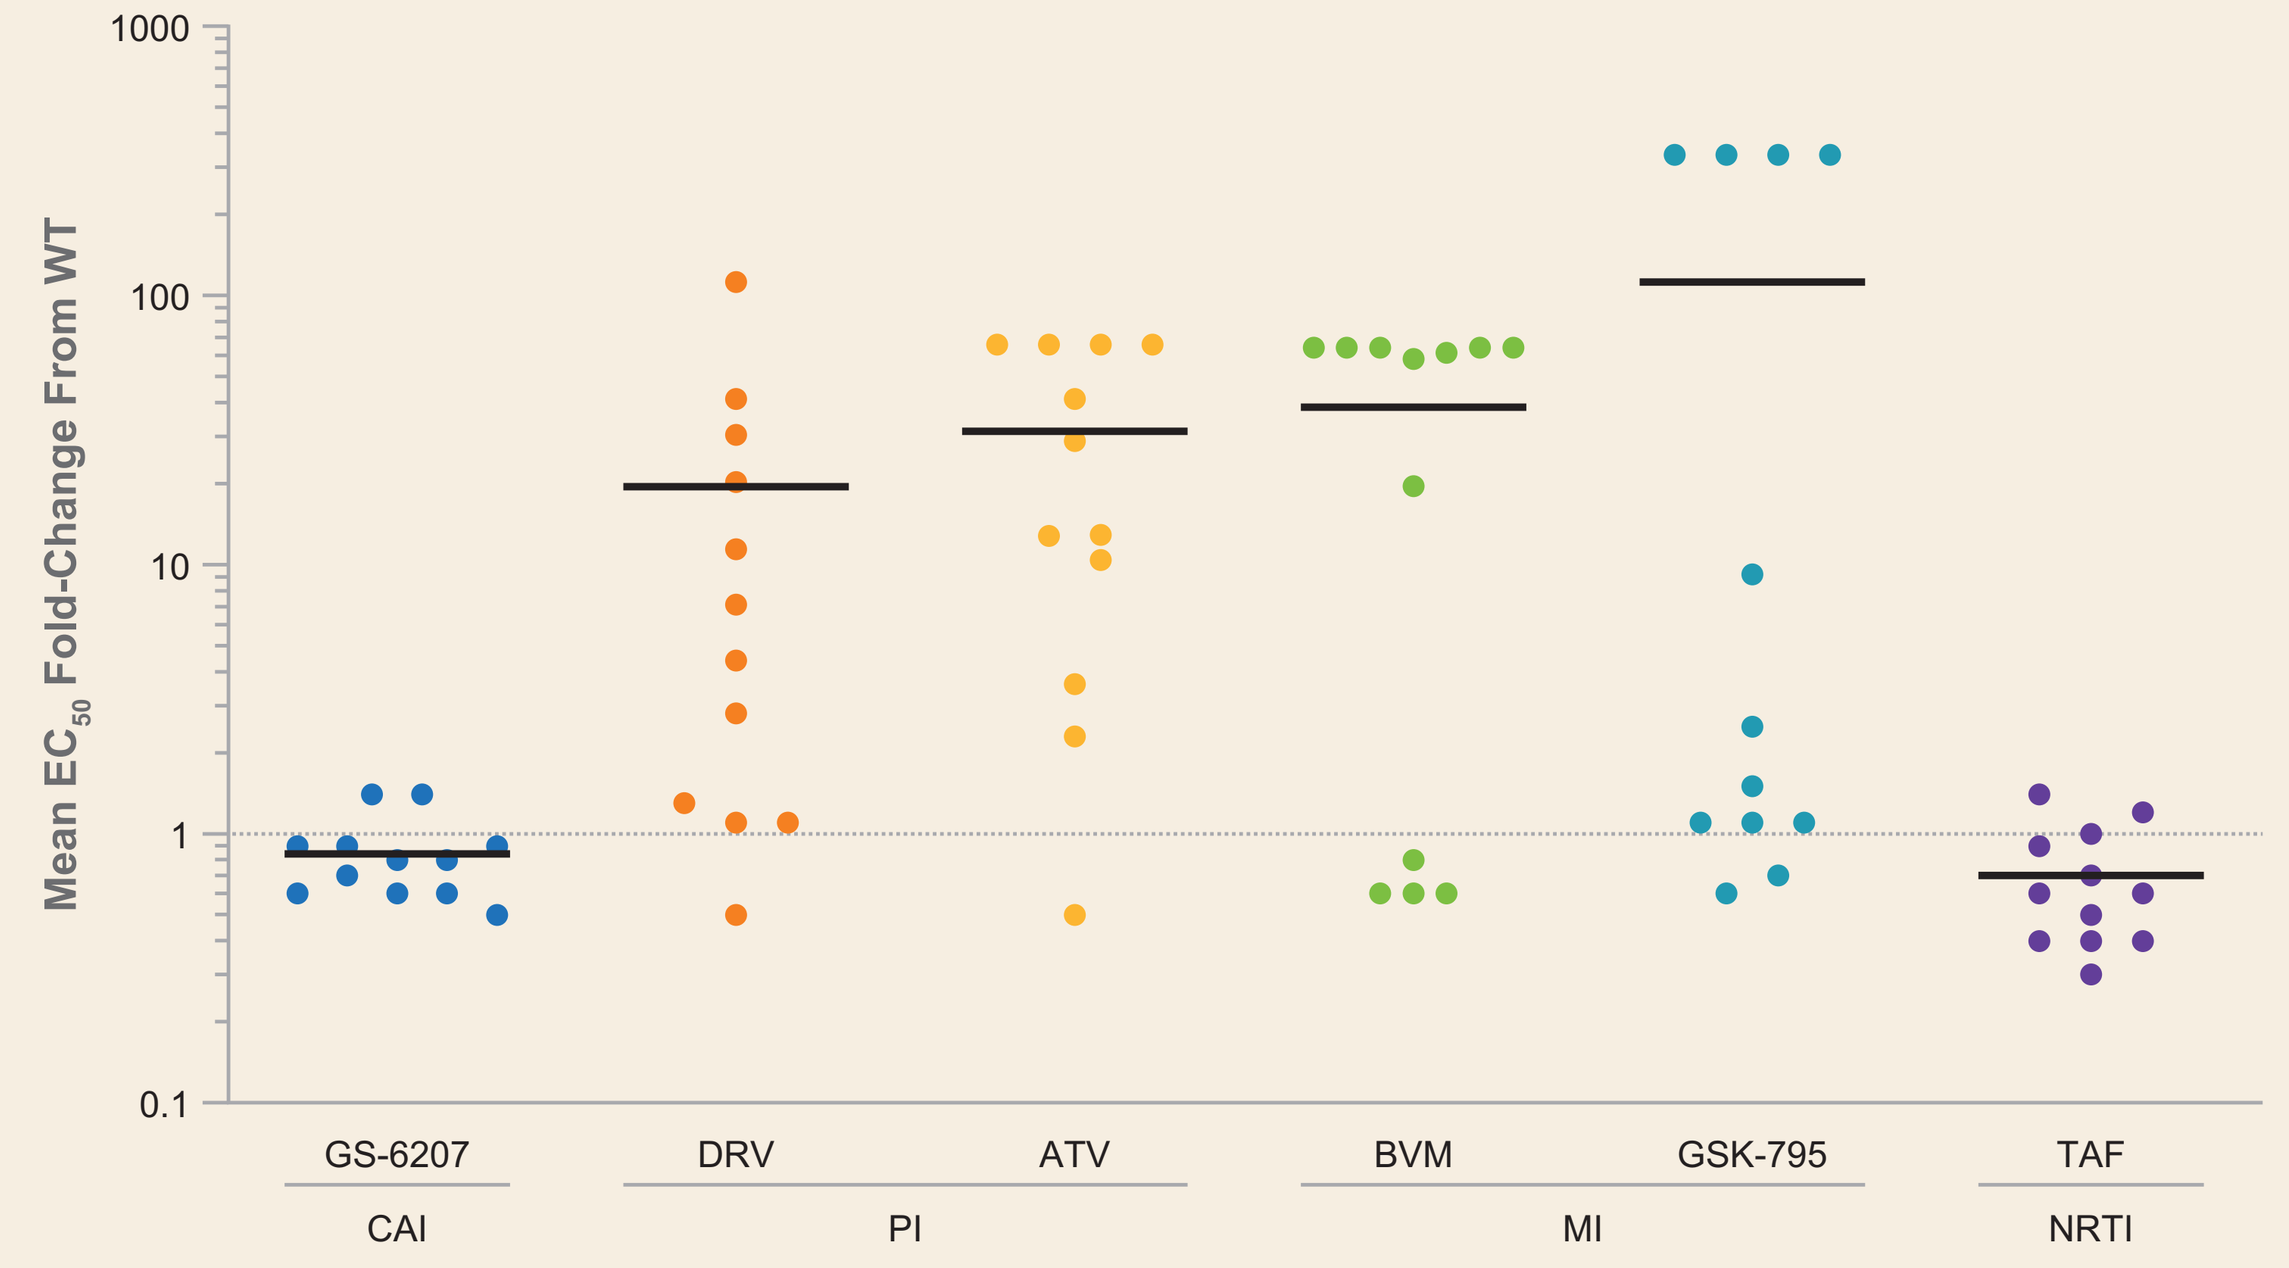
Mean EC50 Fold-Change From WT
GS-6207
DRV
ATV
BVM
GSK-795
TAF
CAI
PI
MI
NRTI

**Each dot* represents a single isolate; *black lines* indicate mean EC_{50} fold-change for each drug.

- In isolates from TE PLWH, GS-6207 also displayed near WT potency (average EC_{50} 89 pM), with minimal variability across all 36 isolates, regardless of presence of GCSMs
- TE isolates with PI resistance mutations demonstrated 21- and 34-fold reduced susceptibility to DRV and ATV, respectively

Fig 7: Drug Susceptibilities in Treatment-Naïve HIV-1 Isolates (n=15)*

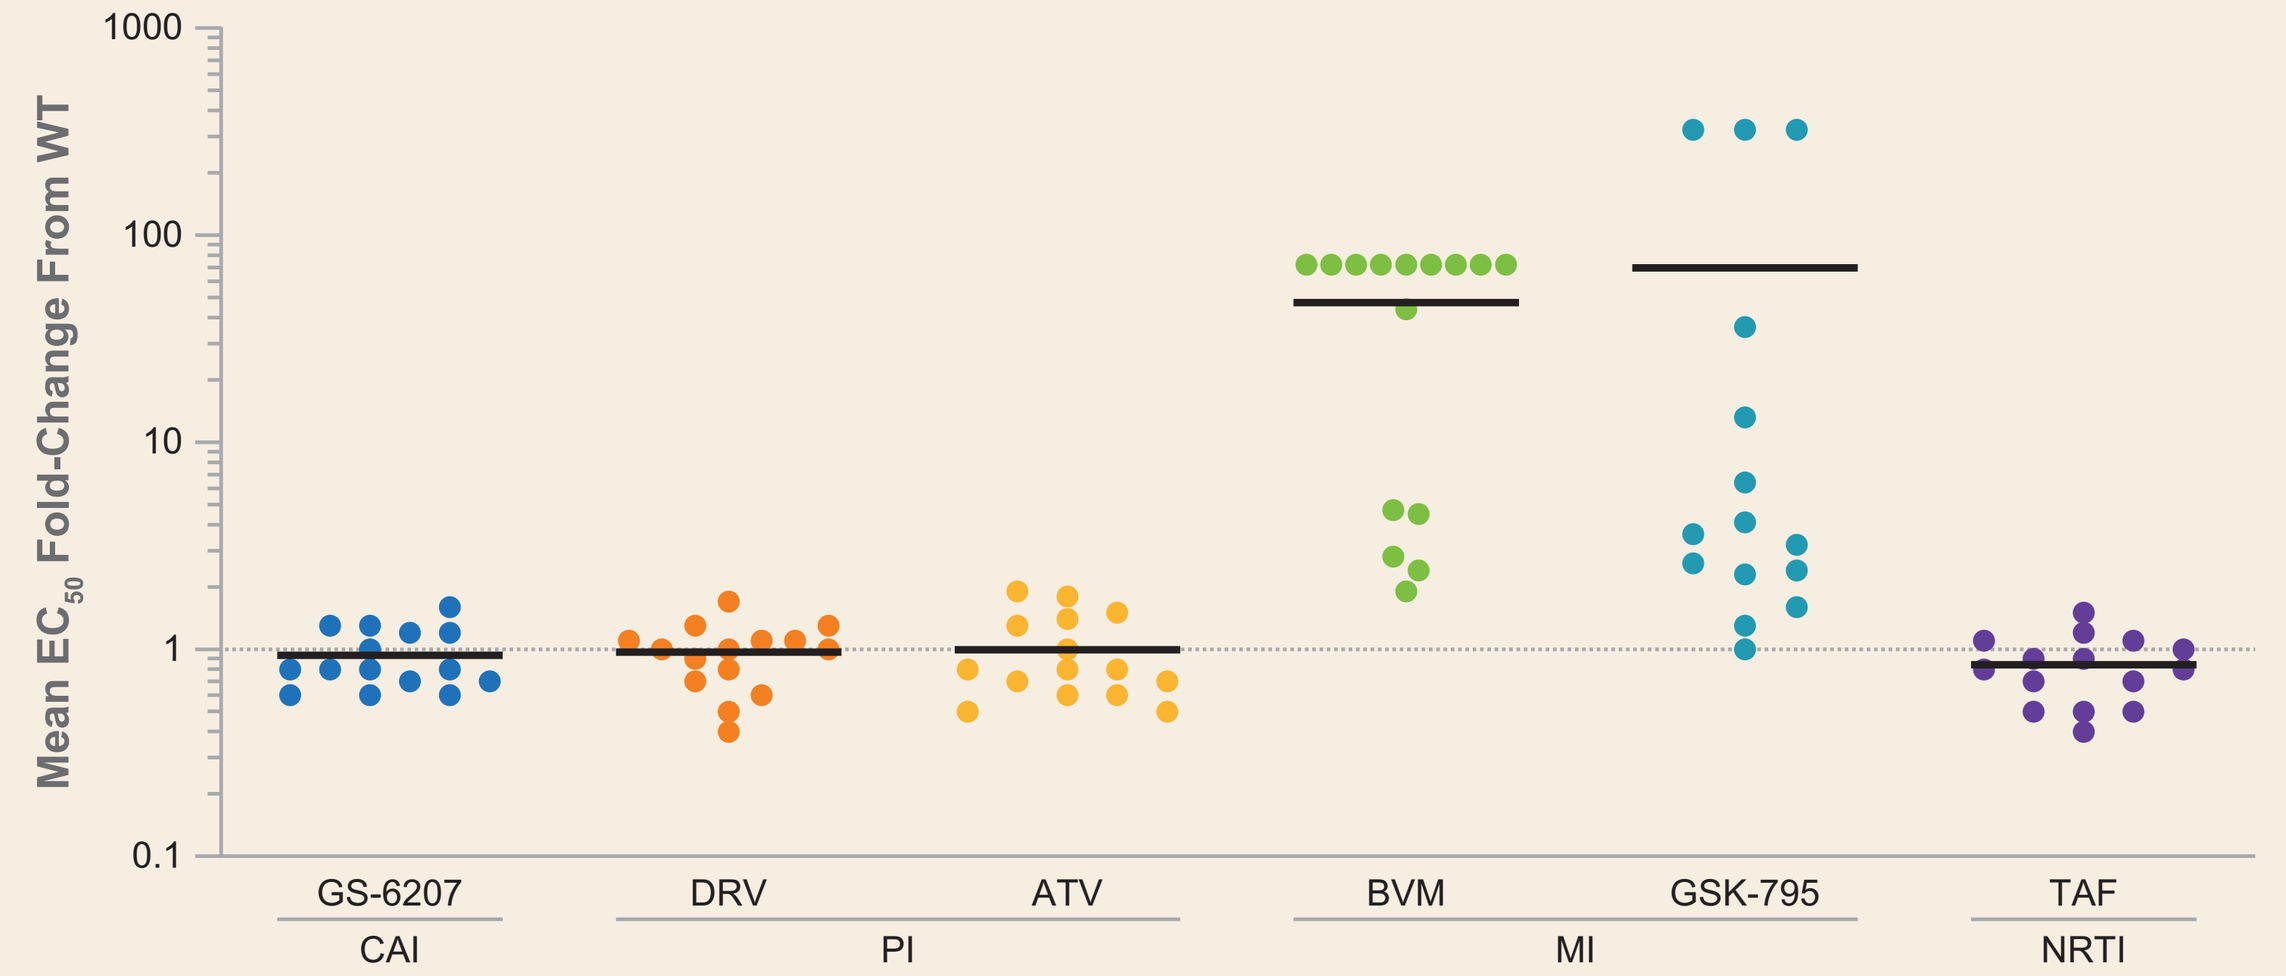
Mean EC50 Fold-Change From WT
GS-6207
DRV
ATV
BVM
GSK-795
TAF
CAI
PI
MI
NRTI

**Each dot* represents a single isolate; *black lines* indicate mean EC_{50} fold-change for each drug.

- In isolates from TN PLWH, GS-6207 exhibited similar near WT potency as previous isolates (average EC_{50} 88 pM), with minimal variability across all 15 isolates
- MIs such as BVM and GSK-795 showed significantly reduced potency in both TN and TE isolates

Fig 8: Drug Susceptibilities in HIV-1 Isolates With Resistance Mutations to the 4 Main Drug Classes (n=40)*

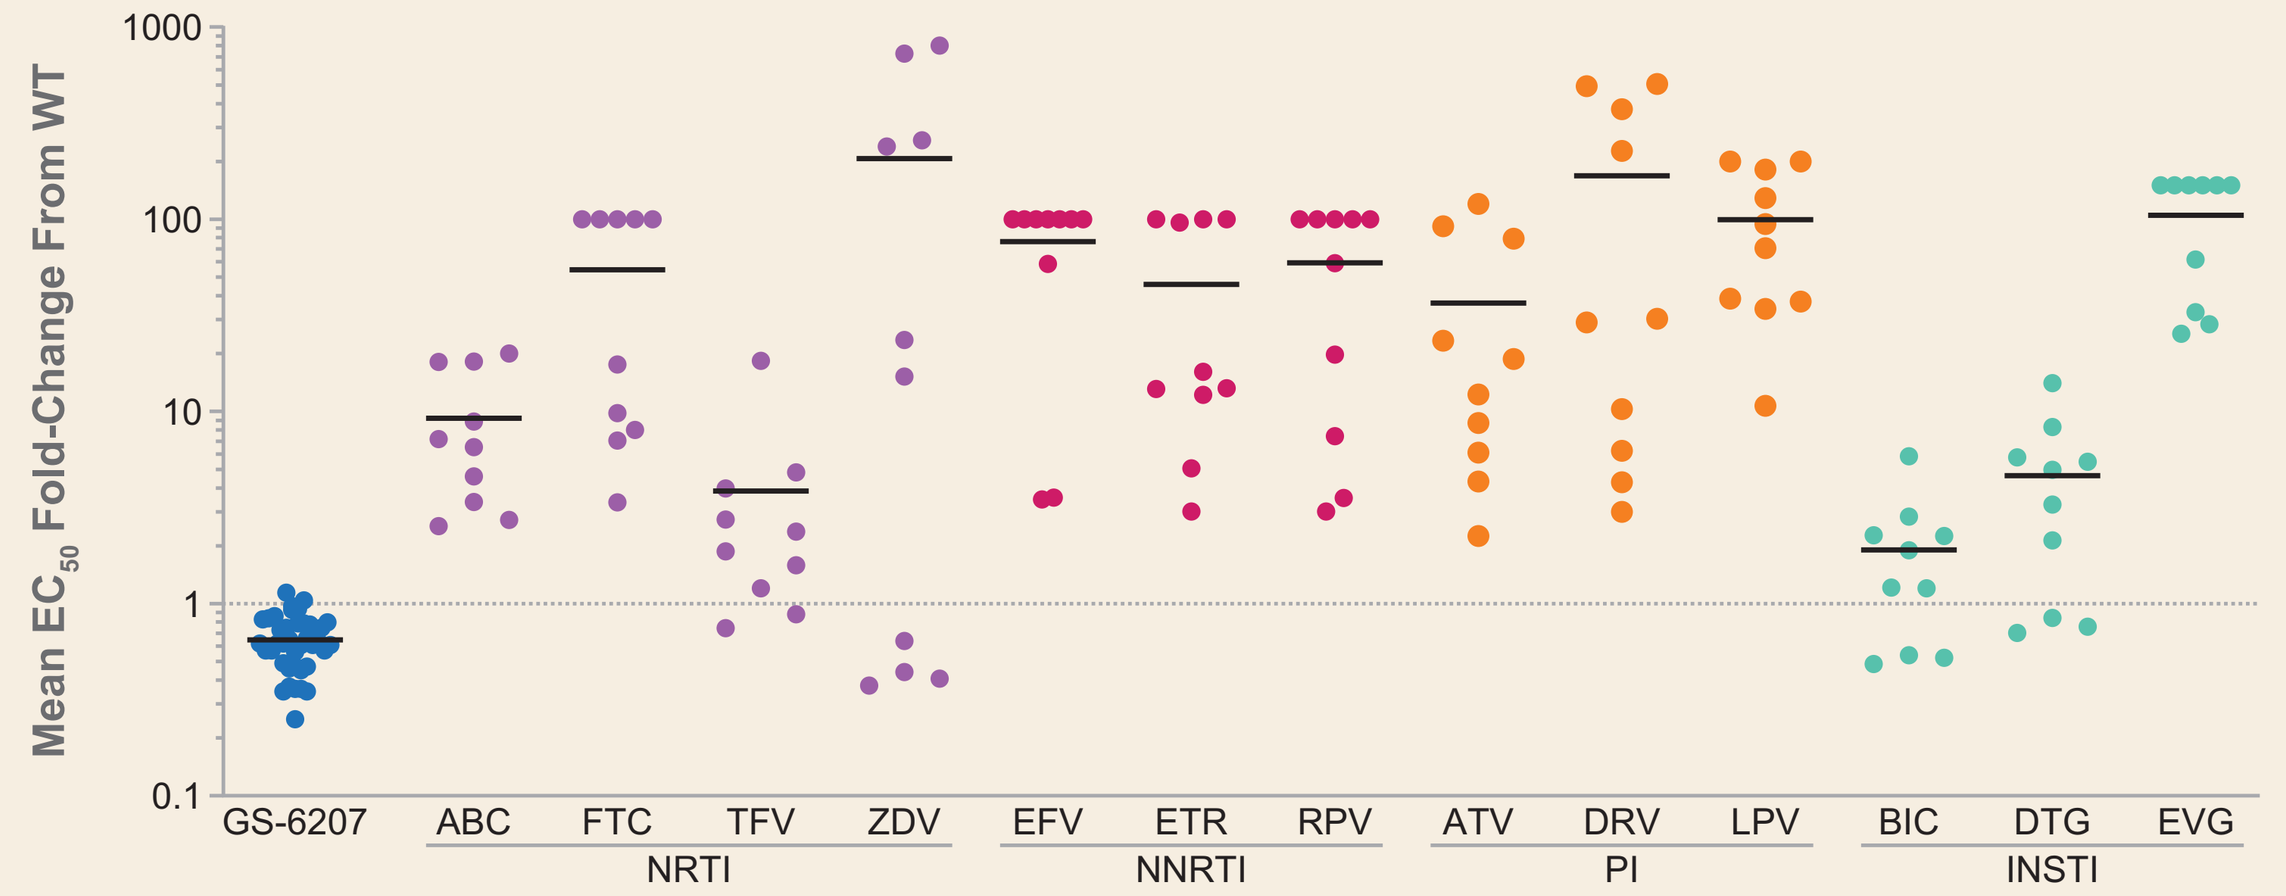
Mean EC50 Fold-Change From WT
GS-6207
ABC
FTC
TFV
ZDV
EFV
ETR
RPV
ATV
DRV
LPV
BIC
DTG
EVG
NRTI
NNRTI
PI
INSTI

**Each dot* represents a single isolate; *black lines* indicate mean EC_{50} fold-change for each drug; 10 resistant isolates were tested per drug class; data for GS-6207 include all 40 mutants tested; NRTI-resistant isolates each contained a combination of RT mutations M41L, A62V, K65R, D67N, T69ins, K70R, L74V, V75I, F77L, F116Y, Q151M, M184V, L210W, T215Y/F, and/or K219Q; NNRTI-resistant isolates each contained a combination of RT mutations A98G, L100I, K101E/H/P, K103N/S, V106A/I/M, E138A/G/K/Q/R, V179D/F/L/T, Y181C/I/V, Y188C/H/L, G190A/E/Q/S, P225H, and/or F227C; PI-resistant isolates each contained a combination of protease mutations D30N, V32I, L33F, M46I/L, I47V, G48V, I50L/V, I54L/V, V82A, I84V, N88D, and/or L90M; and INSTI-resistant isolates each contained a combination of IN mutations E92Q, T97A, Y143C/R, Q148H/R/K, and/or N155H. ABC, abacavir; BIC, bictegravir; DTG, dolutegravir; EFV, efavirenz; ETR, etravirine; EVG, elvitegravir; FTC, emtricitabine; LPV, lopinavir; RPV, rilpivirine; ZDV, zidovudine.

- No cross-resistance to GS-6207 was observed in the panel of mutants with resistance to NRTIs, NNRTIs, PIs, and INSTIs

Conclusions

- The picomolar potency of GS-6207 was unchanged (fold-change <2) in the presence of common GCSMs in both SDMs and patient-derived isolates
- The presence of naturally occurring polymorphisms in gag (including CA) and/or protease mutations in patient-derived isolates did not affect the high potency of GS-6207
- These observations underscore the absence of naturally occurring gag polymorphisms conferring resistance against GS-6207, in contrast to MIs
 - This confirms that the mode of action of GS-6207 is distinct from that of MIs
- Viruses with resistance mutations to the 4 main antiretroviral classes were not cross-resistant to GS-6207
- Viral isolates from TN and a wide variety of TE PLWH were equally susceptible to GS-6207, underlining its potential for treatment in all PLWH regardless of their antiretroviral therapy history

References: **1.** Yant S, et al. CROI 2019, poster 1504; **2.** Daar E, et al. EACS 2019, poster PE3/17; **3.** Sager J, et al. CROI 2019, oral 13; **4.** Yant S, et al. IAS 2019, poster TUPEA075; **5.** Clavel F, Mammano F. Viruses 2010;2;1411-26; **6.** Dam E, et al. PLoS Pathog 2009;5:e1000345; **7.** Nijhuis M, et al. PLoS Med 2007;4:e36; **8.** Margot N, et al. Antimicrob Agents Chemother 2009;54:2345-53.

©2020 Gilead Sciences, Inc. All rights reserved.

Presented at Conference on Retroviruses and Opportunistic Infections, March 8–11, 2020, Boston, MA