Safety, PK, and Efficacy of Low-Dose B/F/TAF in Children ≥2 Years Old Living With HIV

3929

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Carina A. Rodriguez,[1] Kulkanya Chokephaibulkit,[2] Afaaf Liberty,[3] Renate Strehlau,[4] Riana van Zyl,[4] Pope Kosalaraksa,[5] Coleen K. Cunningham,[4]
Eric J. McGrath,[6] Natella Rakhmanina,[7] Heather Maxwell,[8] Danielle Porter,[8] Sophia R. Majeed,[8] Shaolan S. Xiang,[8] Diana M. Brainard,[8] Cheryl Pikora[8]

[1]University of South Florida, Tampa, FL; [2]Mahidol University, Nakhon Pathom, Thailand; [3]Chris Hani Baragwanath Hospital, Johannesburg, South Africa; [4]Rahima Moosa Mother and Child Hospital, University of the Witwatersrand Faculty of Health Sciences, Johannesburg; [5]Khon Kaen University, Khon Kaen, Thailand; [6]Children's Hospital of Michigan, Detroit; [7]The Children's Research Institute, Children's National Hospital, Washington, DC; [8]Gilead Sciences, Inc., Foster City, CA

Introduction

- Few antiretroviral (ARV) options exist for very young children living with HIV and no single-tablet regimen (STR) is used or approved for this population
- Bictegravir (BIC; B) is a novel, unboosted integrase strand transfer inhibitor (INSTI), with a high resistance barrier and low potential for drug-drug interactions
- BIC has been coformulated with emtricitabine (FTC; F) and tenofovir alafenamide (TAF) into a once-daily STR (B/F/TAF)
- B/F/TAF is approved for use and is a guideline-recommended regimen in children weighing ≥25 kg living with HIV[1-4]
- B/F/TAF has been formulated as a low-dose STR for children aged ≥2 y and weighing 14–<25 kg
 - Low-dose STR strength is B/F/TAF 30/120/15 mg (60% of full-strength STR)
 - Can be taken without regard to food
- This is the 1st study to report the pharmacokinetics (PK), safety, and efficacy of B/F/TAF in young children aged ≥2 y living with HIV

Objectives

- Primary: to determine the plasma PK of BIC, and evaluate the safety and tolerability of B/F/TAF through 24 wk of treatment in virologically suppressed adolescents (Cohort 1) and children (Cohorts 2 and 3) living with HIV
- Secondary: to evaluate the safety and tolerability of B/F/TAF for 48 wk, and its antiviral activity at 24 and 48 wk, in virologically suppressed adolescents (Cohort 1) and children (Cohorts 2 and 3) living with HIV
- **Cohort 3 is the focus of the present analyses**

Methods

Study Design

Eligibility Criteria
- HIV-1 RNA <50 copies/mL for ≥6 mo
- CD4 count ≥200 cells/μL
- eGFR ≥90 mL/min/1.73 m² (Schwartz)

- Phase 2/3, open-label, multicenter, multicohort, single-arm study (NCT02881320)
 - Part A: iPK was assessed to confirm dose of B/F/TAF
 - Part B: following dose confirmation and IDMC review* of short-term safety from Part A, additional participants were enrolled to complete the cohort and initiate enrollment into the next younger cohort

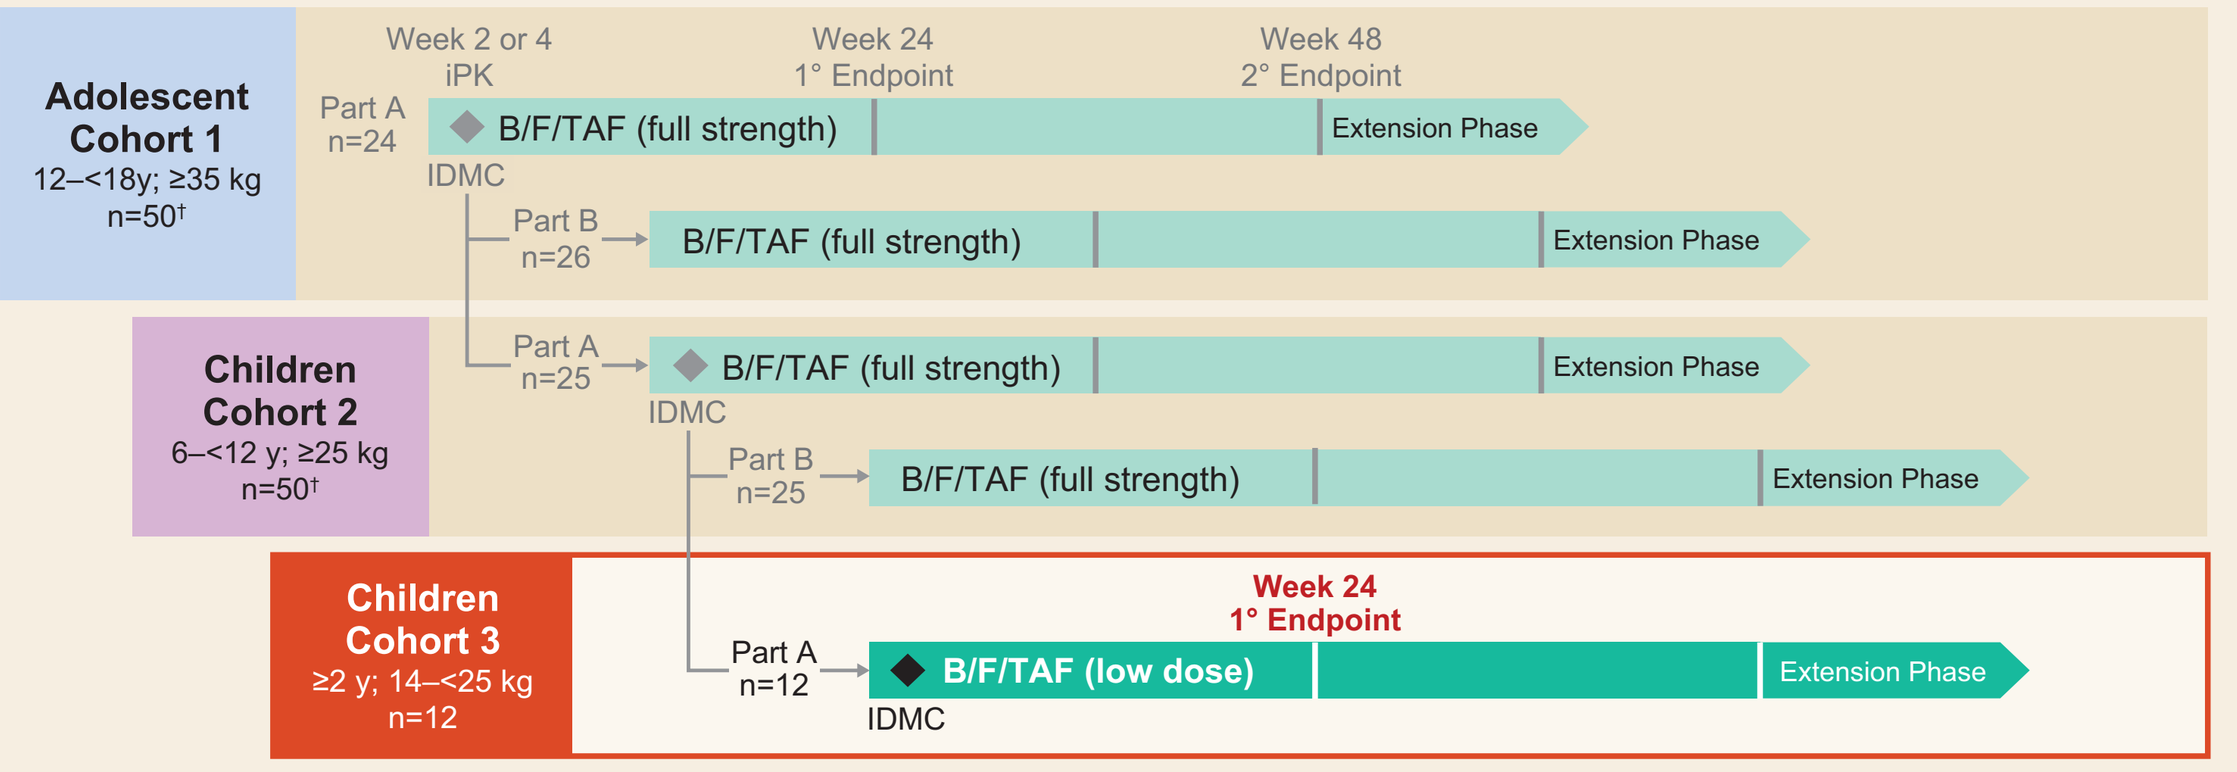
Week 2 or 4
iPK
Week 24
1° Endpoint
Week 48
2° Endpoint
Adolescent Cohort 1
12–<18y; ≥35 kg
n=50†
Part A
n=24
B/F/TAF (full strength)
Extension Phase
IDMC
Part B
n=26
B/F/TAF (full strength)
Extension Phase
Children Cohort 2
6–<12 y; ≥25 kg
n=50†
Part A
n=25
B/F/TAF (full strength)
Extension Phase
IDMC
Part B
n=25
B/F/TAF (full strength)
Extension Phase
Children Cohort 3
≥2 y; 14–<25 kg
n=12
Week 24
1° Endpoint
Part A
n=12
B/F/TAF (low dose)
Extension Phase
IDMC

*Independent data monitoring committee (IDMC) review occurred when 50% of participants reached Week 12 and all completed intensive PK (iPK) visit; †Previously reported.[5,6] CD4, cluster of differentiation-4; eGFR, estimated glomerular filtration rate.

Study Assessments

- **PK:** intensive and sparse PK samples collected to examine steady-state exposure of BIC, FTC, and TAF
- **Safety:** adverse events (AEs) and clinical laboratory abnormalities
- **Efficacy:** HIV-1 RNA and CD4 cell count
- **Palatability and acceptability:** questionnaires and facial scale
- **Adherence:** assessed by pill count at each visit

Results

Part A Disposition: PK Substudy

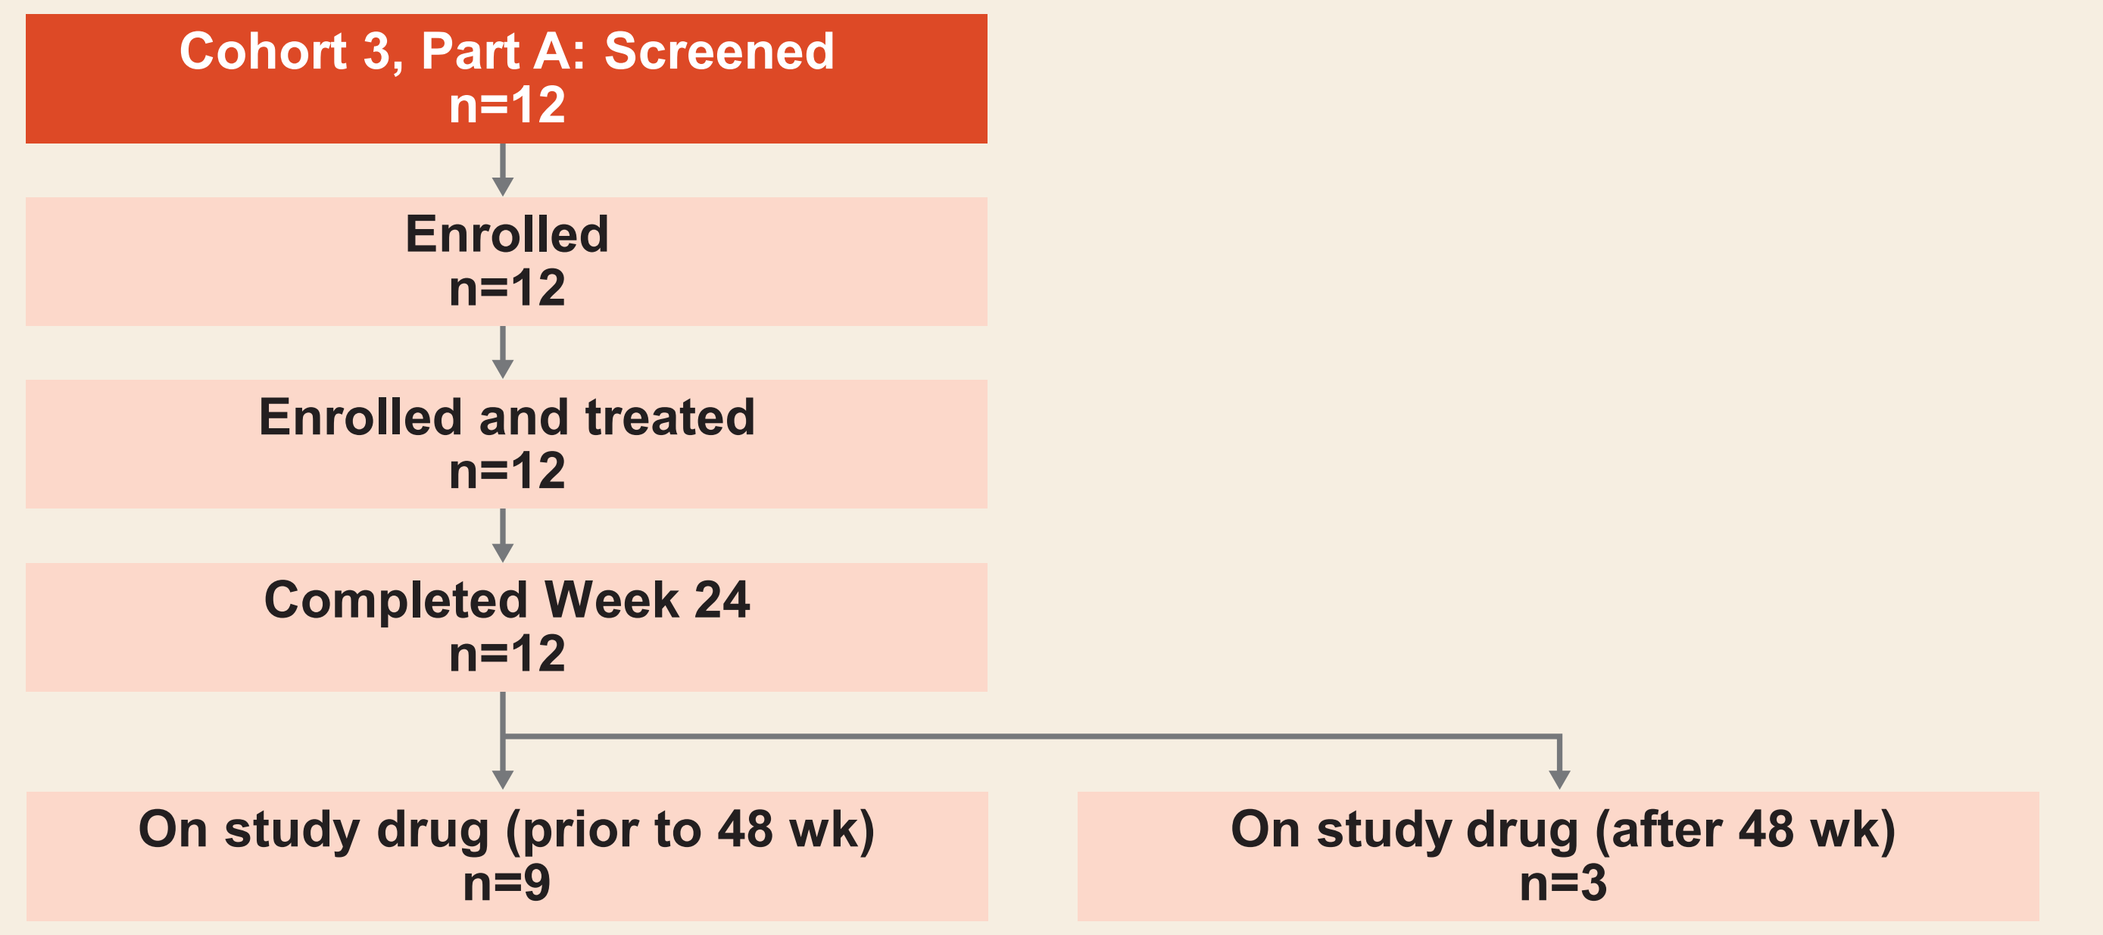
Cohort 3, Part A: Screened
n=12
Enrolled
n=12
Enrolled and treated
n=12
Completed Week 24
n=12
On study drug (prior to 48 wk)
n=9
On study drug (after 48 wk)
n=3

- Median (quartile [Q] 1, Q3) duration of exposure: 42.3 wk (40.1, 49.3)

Baseline Characteristics

	Children ≥2 y; 14–<25 kg n=12
Median age, y (range)	6 (3–9)
Median weight, kg (range)	20.1 (14.6–24.1)
Female, n (%)	7 (58)
Race, n (%)	
Asian	5 (42)
Black	7 (58)
Country, n (%)	
South Africa	3 (25)
Thailand	5 (42)
USA	4 (33)
HIV-1 RNA <50 copies/mL, n (%)	12 (100)
Median CD4 cell count/μL (Q1, Q3)	841 (703, 1238)
Median eGFR, mL/min/1.73 m² (Q1, Q3)	151.0 (141.5, 167.0)
Vertical transmission, n (%)	12 (100)
Previous ARVs, n (%)	
NRTIs (AZT or ABC, 3TC)	12 (100)
INSTI (RAL)	1 (8)
NNRTI (NVP or EFV)	6 (50)
PI (LPV/r)	5 (42)

3TC, lamivudine; ABC, abacavir; AZT, zidovudine; EFV, efavirenz; LPV/r, ritonavir-boosted lopinavir; (N)NRTIs, (non)nucleoside reverse-transcriptase inhibitors; NVP, nevirapine; PI, protease inhibitor; RAL, raltegravir.

Overall Safety

Participants, n (%)	Children ≥2 y; 14–<25 kg n=12
Any grade AE	10 (83)
Grade 3 or 4 AE	0
AE related to study drug	3 (25)
Serious AE	0
AE leading to study drug discontinuation	0
Death	0

- Most common AEs were upper respiratory tract infection (n=3 [25%]), and abdominal pain, constipation, diarrhea, vomiting, viral upper respiratory tract infection, enuresis, cough, and rhinorrhea (each n=2 [17%])
 - No other AE occurred in >1 participant and all AEs were mild–moderate in severity
- 3 participants had AEs considered related to study drug: neutropenia, abdominal pain, irritability, and social avoidant behavior
- Grade 3 or 4 laboratory abnormalities: decreased neutrophils (n=2 [17%]) and increased creatinine (n=1 [8%])

Intensive BIC PK: B/F/TAF Low-Dose Tablet*

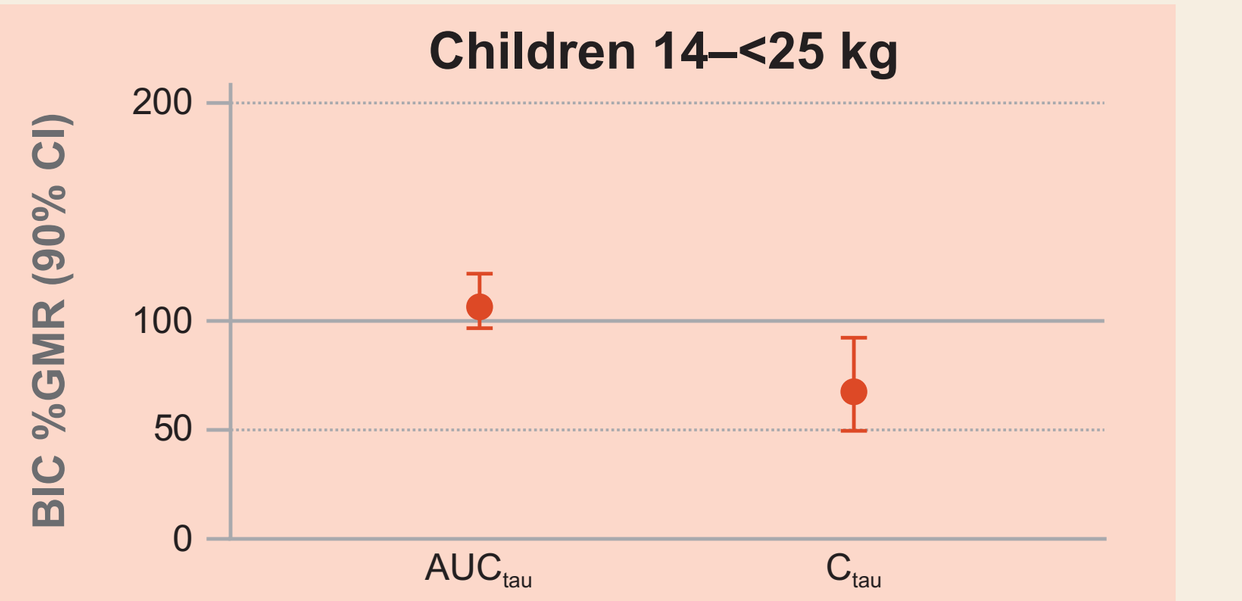
Children 14–<25 kg
BIC %GMR (90% CI)
200
100
50
0
AUCtau
Ctau

*iPK sampling was conducted in children to obtain exposure estimates; exposure in children (test) was compared with that in adults (reference); geometric mean ratios (GMRs) and 90% confidence intervals (CIs) were constructed around PK parameters of interest; taking into account higher variability in PK in younger children, exposure was considered equivalent to adults if 90% CI fell within prespecified lack of alteration boundary of 50–200%; equivalency boundary was established based on totality of safety and efficacy data available for B/F/TAF. AUC_{tau}, area under curve over dosing interval; C_{tau}, trough concentration.

- BIC AUC_{tau} was similar in children weighing 14–<25 kg relative to adults
- BIC C_{tau} mean estimate was lower in children weighing 14–<25 kg vs adults, but remained ~12-fold above the protein-adjusted 95% effective concentration (162 ng/mL) for wild-type virus

Population BIC PK: B/F/TAF Full-Strength Tablet* Previously Reported[5,6]

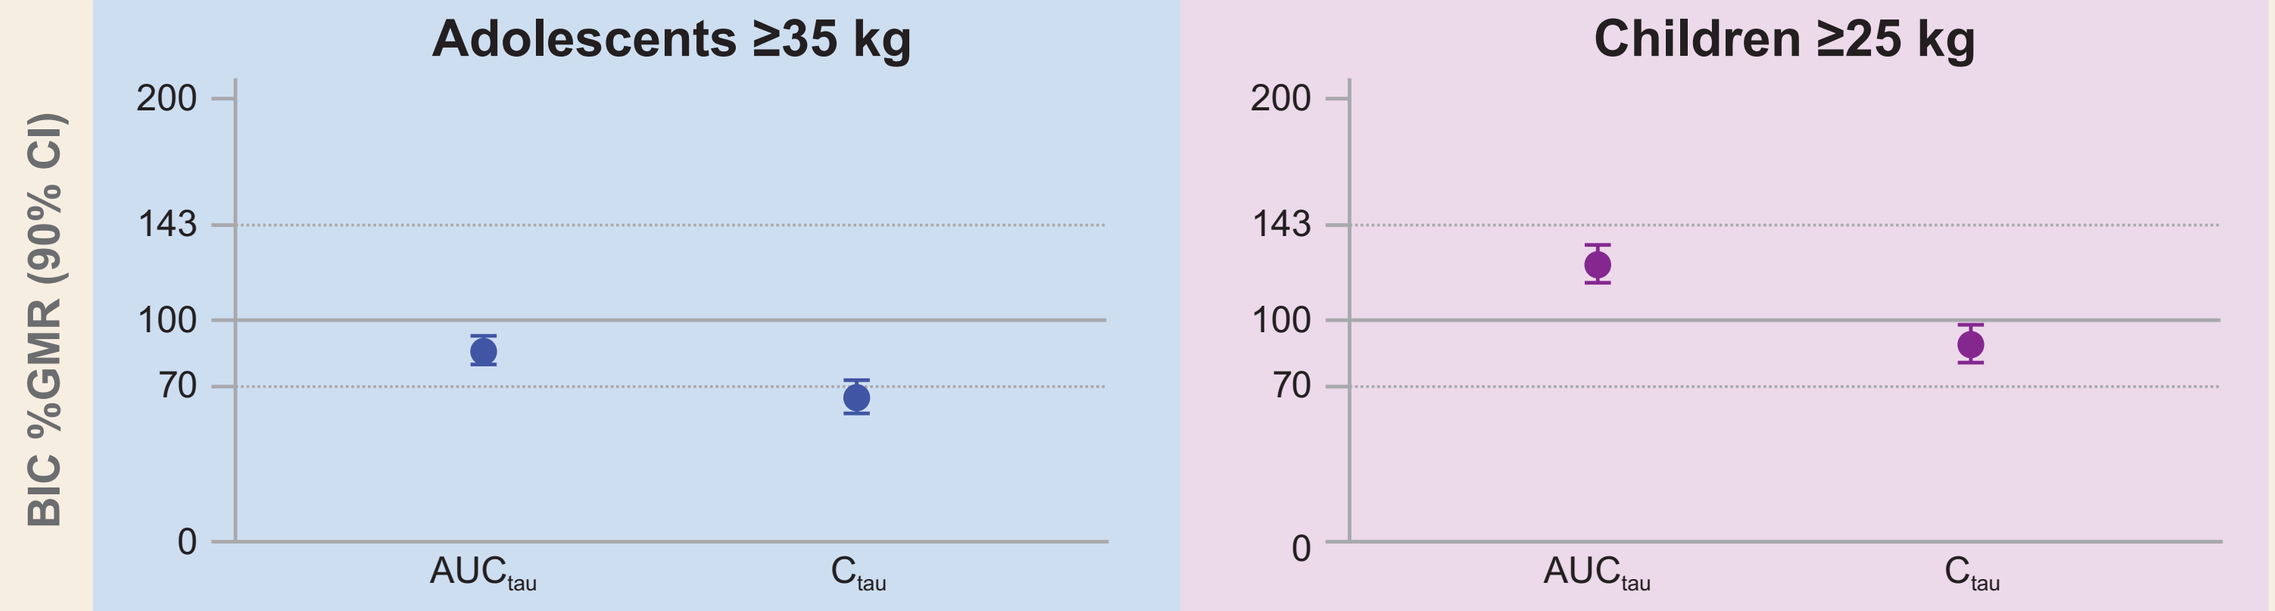
Adolescents ≥35 kg
Children ≥25 kg
BIC %GMR (90% CI)
200
143
100
70
0
AUCtau
Ctau

*Population PK analyses using intensive and sparse samples from adolescents and children were conducted; exposure in adolescents or children (test) was compared with that in adults (reference); GMRs and 90% CIs were constructed around PK parameters of interest; exposure in children was considered equivalent to adults if 90% CI fell within prespecified lack of alteration boundary of 70–143%.

- BIC AUC_{tau} was similar in adolescents and children weighing ≥25 kg compared with adults
- BIC C_{tau} was similar in children and adults
- BIC C_{tau} was lower in adolescents vs adults, but >11-fold above the protein-adjusted 95% effective concentration for wild-type virus

Intensive PK Data

	PK Parameter*	Children ≥2 y; 14–<25 kg n=12†	Adults n=74–77‡	Child/Adult GMR% (90% CI)
FTC	AUC_{tau}, h·ng/mL	14,576	11,790	124 (110, 139)
	C_{max}, ng/mL	3473	2004	173 (144, 209)
	C_{tau}, ng/mL	80§	90	89 (49, 161)
TAF	AUC_{tau}, h·ng/mL	282	195	145 (115, 182)
	C_{max}, ng/mL	393	227	173 (140, 214)

*Geometric mean; †1 participant was excluded from FTC summary due to noncompliance with study drug; ‡Pooled iPK data from four Phase 3 studies in adults with HIV. §n=10; C_{max}, maximal concentration.

- Exposures of FTC and TAF were within the safe and efficacious ranges of historical data in adults and adolescents following administration of approved FTC/TAF-containing products[7,8]

Estimated Glomerular Filtration Rate (Schwartz)

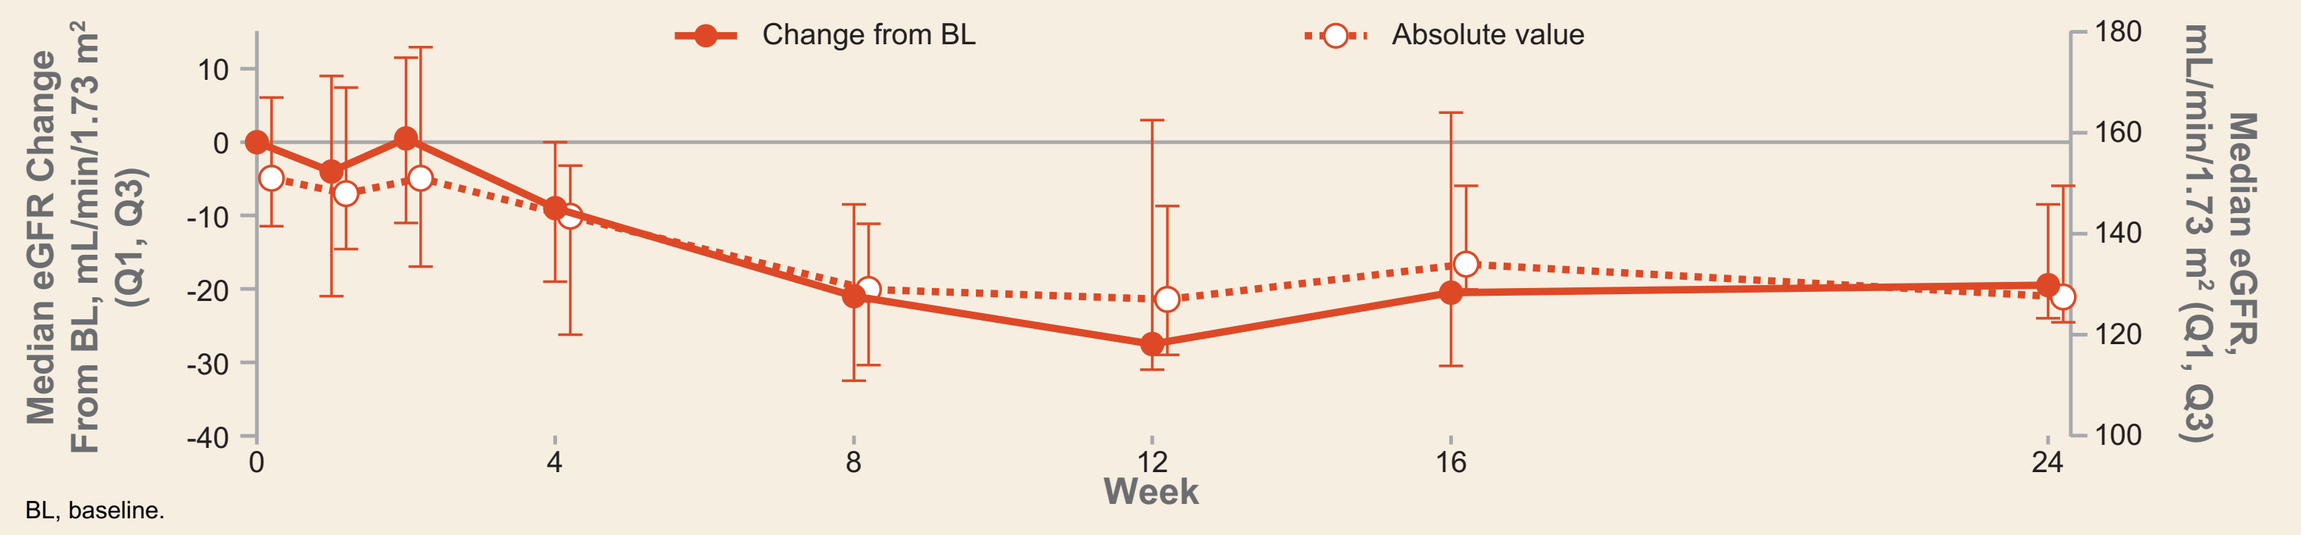
Change from BL
Absolute value
Median eGFR Change From BL, mL/min/1.73 m² (Q1, Q3)
10
0
-10
-20
-30
-40
Median eGFR, mL/min/1.73 m² (Q1, Q3)
180
160
140
120
100
0
4
8
12
16
24
Week

BL, baseline.

- Median changes in eGFR ranged from 0.5 to -27.5 mL/min/1.73 m² between Weeks 1 and 24
- Changes in eGFR in children weighing 14–<25 kg were consistent with the known renal creatinine transporter effect of BIC[9,10] and not considered clinically significant

Palatability and Acceptability

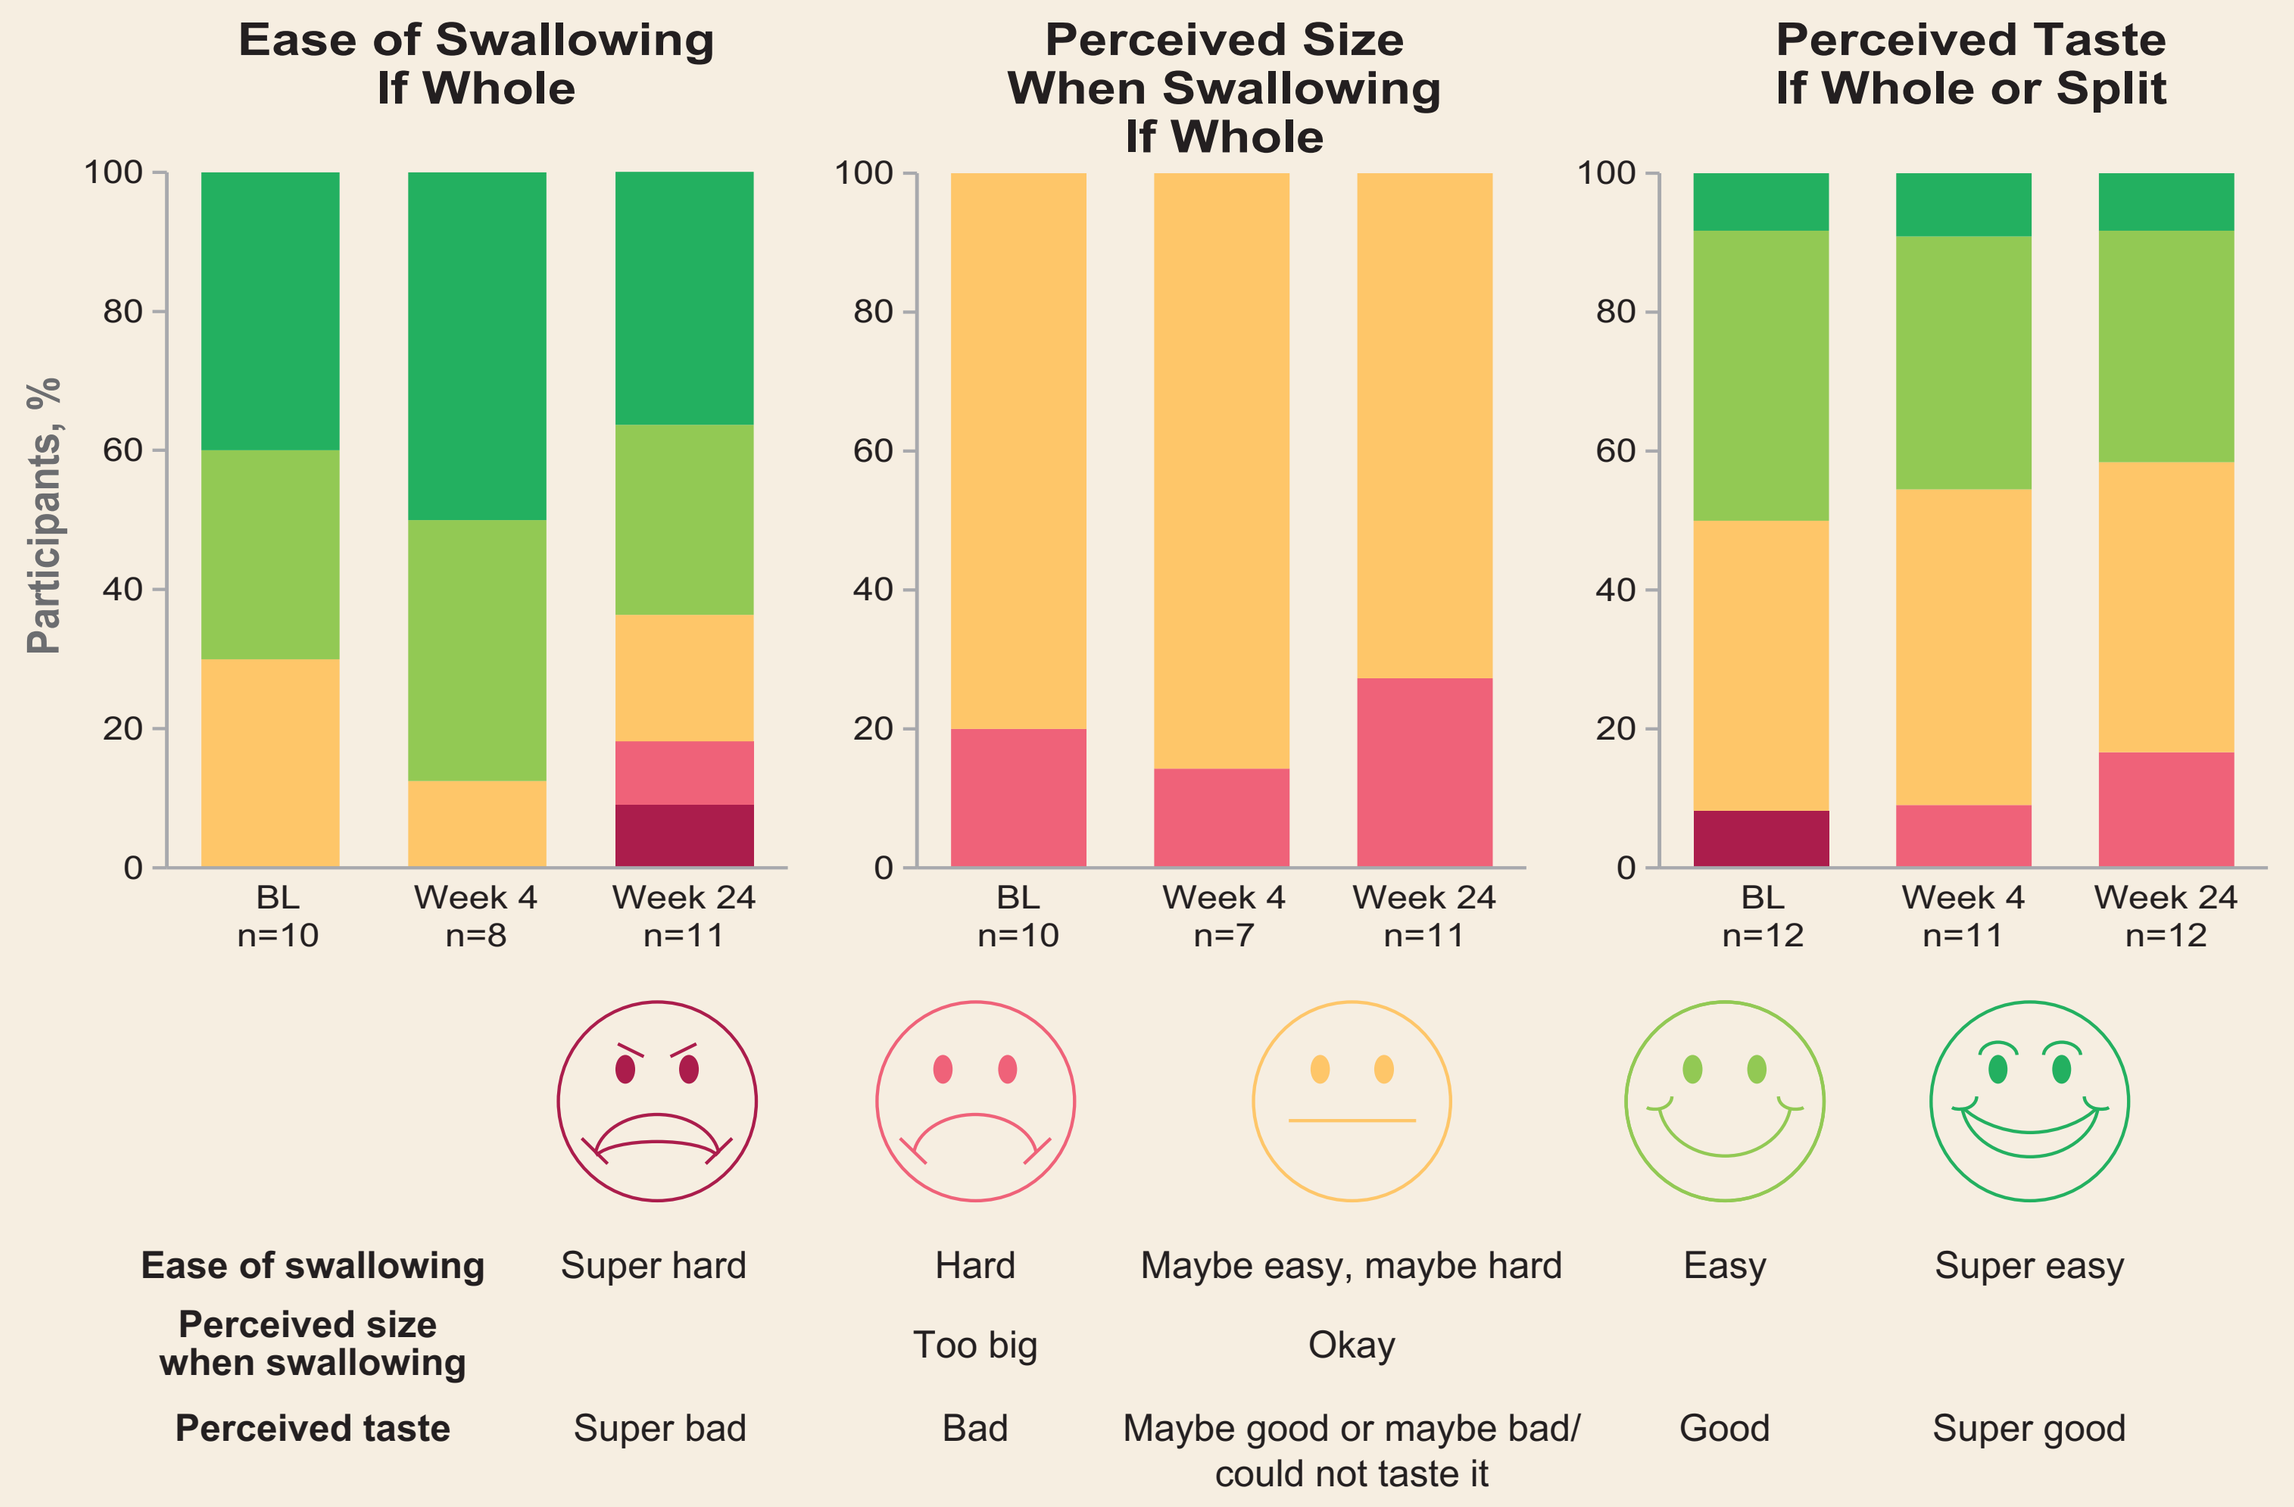
Ease of Swallowing If Whole
Perceived Size When Swallowing If Whole
Perceived Taste If Whole or Split
Participants, %
100
80
60
40
20
0
BL n=10
Week 4 n=8
Week 24 n=11
BL n=10
Week 4 n=7
Week 24 n=11
BL n=12
Week 4 n=11
Week 24 n=12
Ease of swallowing
Super hard
Hard
Maybe easy, maybe hard
Easy
Super easy
Perceived size when swallowing
Too big
Okay
Perceived taste
Super bad
Bad
Maybe good or maybe bad/ could not taste it
Good
Super good

- Palatability and acceptability assessments:
 - Questionnaire to ask the participant/parent:
 - Whether tablet was broken in half to allow it to be swallowed
 - If broken in half, whether both halves were taken within 10 min
 - Facial scale and age-appropriate labels to rate:
 - Ease or difficulty in swallowing tablet (if tablet was taken whole only)
 - Acceptability of tablet shape
 - Acceptability of tablet size
 - Assessment of tablet taste
- Mean (standard deviation [SD]) adherence to B/F/TAF was 96.5% (6.1%)
- Nos. of children who split tablet: n=2 (BL), n=3 (Week 4), and n=1 (Week 24)
- For children who had tablet split, all were able to take both halves one right after the other (within 10 min)
- At planned assessments, all children aged 3 y (n=3) swallowed tablet whole: 1 at all time points, 1 at Week 24 only, and 1 at BL (Day 1) only

B/F/TAF STR Size Comparisons*

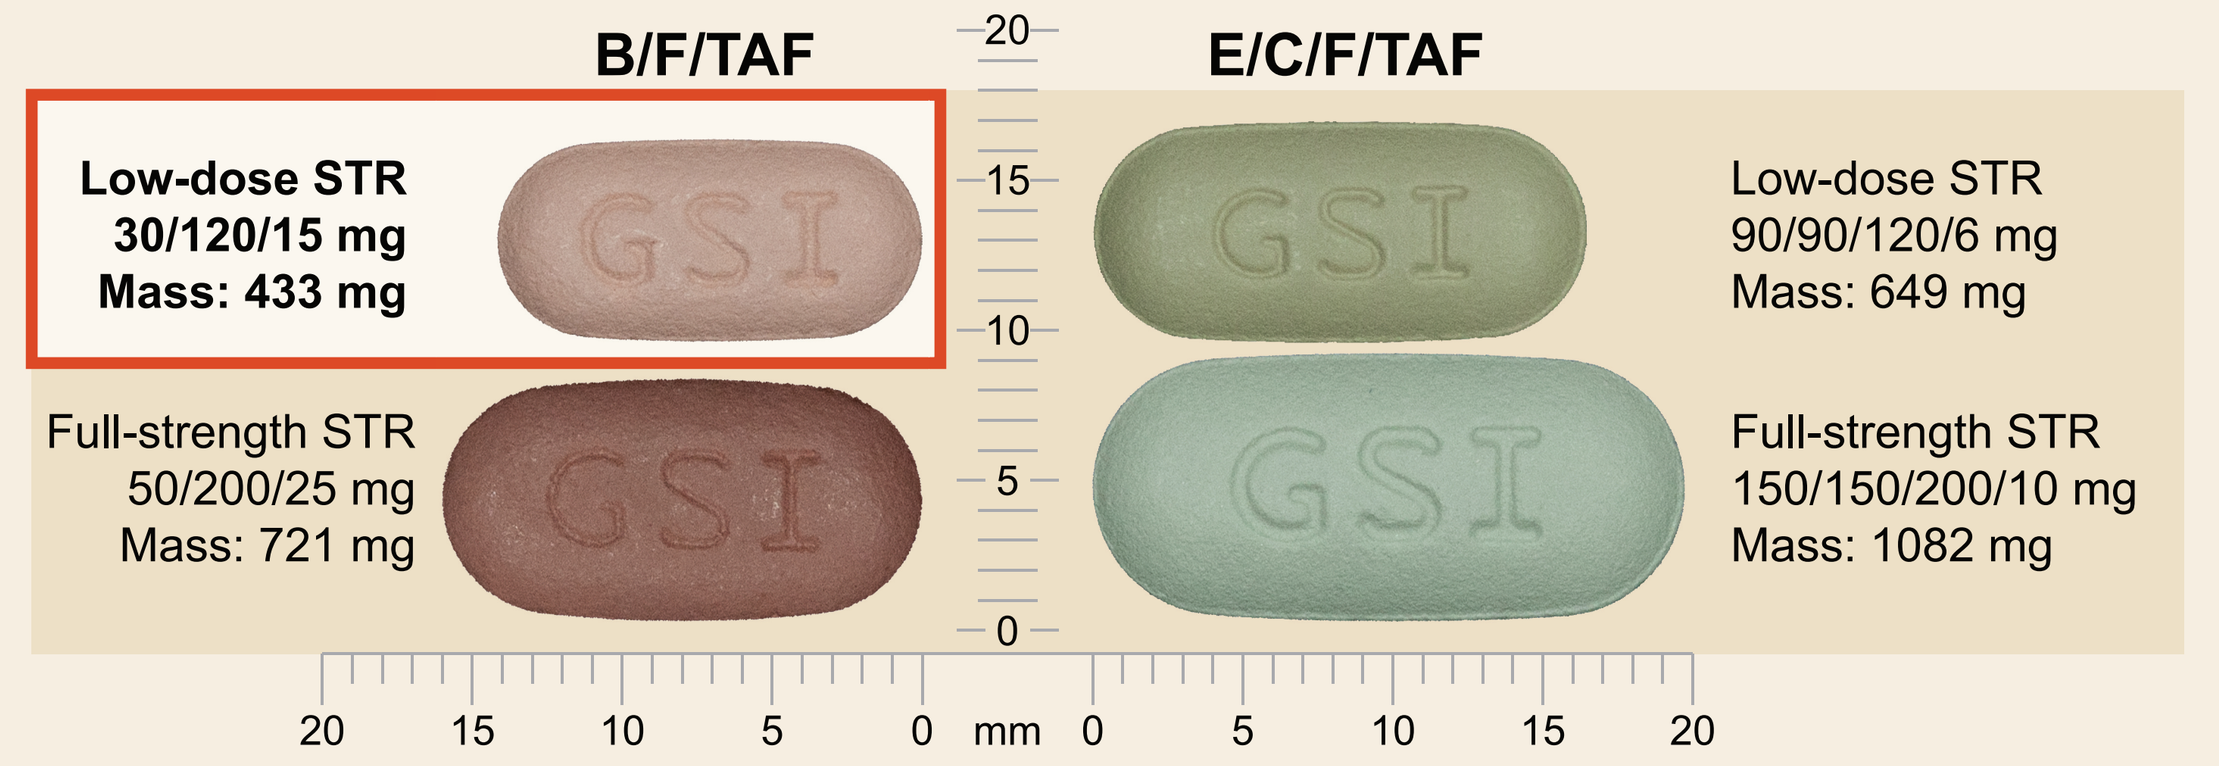
B/F/TAF
E/C/F/TAF
Low-dose STR
30/120/15 mg
Mass: 433 mg
Full-strength STR
50/200/25 mg
Mass: 721 mg
Low-dose STR
90/90/120/6 mg
Mass: 649 mg
Full-strength STR
150/150/200/10 mg
Mass: 1082 mg
GSI
20
15
10
5
0
mm

*Approved in USA for use in adults, adolescents, and children weighing ≥25 kg; final commercial trade dress may change (note: tablet size is not intended to compare clinical efficacy and safety, indications, dosing regimens, or treatment adherence).

Efficacy: Virologic Outcome

- Using a missing = excluded analysis, virologic suppression (HIV-1 RNA <50 copies/mL) was maintained in all 12 participants (100%) at Week 24
- At Week 24, mean change (SD) in CD4 cell count was -66 cells/μL (180.7) and mean change in CD4% was -0.9 (4.64)
- No participants met the criteria for resistance analysis

Conclusions

- In virologically suppressed children (aged ≥2 y; weight 14–<25 kg):
 - The B/F/TAF low-dose STR was well tolerated
 - All AEs were mild–moderate and there were no serious AEs, deaths, or AEs that led to discontinuation
 - B/F/TAF demonstrated high rates of adherence and maintained virologic suppression
 - Ability to swallow the low-dose STR was high, even down to age 3 y
 - Exposures of BIC, FTC, and TAF were consistent with the ranges of exposures observed in adults in Phase 3 trials of B/F/TAF
- Efficacy and safety were consistent with results from Phase 3 trials of B/F/TAF in adults, which showed high proportions with viral suppression, no resistance, and good tolerability
- These data support further pediatric studies of B/F/TAF, which may be an important unboosted INSTI option for HIV-infected young children aged ≥2 y and able to swallow a tablet
 - An additional 10 children have been enrolled in Cohort 3, Part B (current total n=22)
- The evaluation of other formulations of B/F/TAF in younger children who are unable to swallow tablets is planned

For information on relative bioavailability of the B/F/TAF low-dose STR, see Majeed S, et al. CROI 2020, poster 3194

References: 1. AIDSinfo. https://aidsinfo.nih.gov/guidelines/html/2/pediatric-arv/45/whats-new-in-the-guidelines: 12/19; 2. Biktarvy [package insert]. Foster City, CA: Gilead Sciences, Inc: 6/19; 3. Gallant JE, et al. J Acquir Immune Defic Syndr 2017;75:61-6; 4. Tsiang M, et al. Antimicrob Agents Chemother 2016.60:7086-97; 5. Cotton M, et al. International Workshop on HIV Pediatrics 2018, oral 4; 6. Gaur A, et al. CROI 2017, poster 424; 7. Descovy [package insert]. Foster City, CA: Gilead Sciences, Inc.; 4/16; 8. Genvoya [package insert]. Foster City, CA: Gilead Sciences, Inc.; 4/16; 9. Zhang H, et al. International Workshop on Clinical Pharmacology of Antiviral Therapy 2017, poster 50; 10. Zhang H, et al. British HIV Association 2017; oral O2.

Acknowledgments: We extend our thanks to the participants, their families, and all participating study investigators and staff: *South Africa:* A Liberty, R Strehlau, R Van Zyl; *Thailand:* K Chokephaibulkit, P Kosalaraksa; *USA:* C Rodriguez, C Cunningham, E McGrath, N Rakhmanina. This study was funded by Gilead Sciences, Inc.

Presented at Conference on Retroviruses and Opportunistic Infections, March 8–11, 2020, Boston, MA

©2020 Gilead Sciences, Inc. All rights reserved.