ST VINCENT'S HOSPITAL SYDNEY

A FACILITY OF ST VINCENT'S HEALTH AUSTRALIA

ST VINCENT'S CENTRE FOR APPLIED MEDICAL RESEARCH

Dolutegravir with tenofovir disoproxil fumarate-emtricitabine as HIV post-exposure prophylaxis in gay and bisexual men

J. McAllister,[1] J. M. Towns,[2] A. McNulty,[3] A. B. Pierce,[4] R. Foster,[5] R. Richardson,[1] A. Carr[1]

[1]HIV, Immunology & Infectious Diseases Unit and Centre for Applied Medical Research, St Vincent's Hospital, Sydney, NSW, Australia, [2]Melbourne Sexual Health Centre, Melbourne, VIC, Australia, [3]Sydney Sexual Health Centre, Sydney, NSW, Australia, [4]Department of Infectious Diseases, The Alfred Hospital, Melbourne, VIC, Australia, [5]Clinic 16, Royal North Shore Hospital, Sydney, NSW, Australia

Introduction

- Antiretroviral drugs as HIV nonoccupational postexposure prophylaxis (NPEP) is recommended by the World Health Organisation[1]
- Up to one-third of gay or bisexual men (GBM) do not complete 28 days of NPEP[2]
- Adverse events (AEs) are the likely primary cause of NPEP non-completion
- Co-formulated TDF/FTC (Truvada; TVD) is the preferred NRTI backbone for NPEP[3,4]
- Choices for a 3rd drug include PIs, integrase inhibitors, NNRTIs and entry inhibitors[5,6,7,8]
- 3-drug NPEP discontinuation
 - higher with LPVr, DRVr or MVC NPEP
 - lower with raltegravir (RAL), rilpivirine (RPV)[5,6,7,8]
- Limitations of current NPEP regimens
 - PIs: GI side-effects, drug-drug interactions and act after HIV integration[5,6]
 - RAL can cause acute muscle toxicity and twice-daily dosing is required[6]
 - RPV must be taken with food[7]
- Dolutegravir (DTG) is an attractive 3rd drug for NPEP:
 - dosed once-daily
 - potent
 - safe and well tolerated
 - relatively few drug/drug interactions
 - T_{max} 2-4 hours post-dose; and
 - mode of action pre-HIV integration[9,10]
- DTG not previously evaluated for NPEP
- We investigated the completion rate, safety and adherence to TVD + DTG as 3-drug NPEP

Methods

- Open-label, single-arm study at 3 sexual health clinics and 2 emergency departments in Australia
- One hundred HIV-uninfected GBM requiring 3-drug PEP received DTG plus TVD for 28 days
- The primary endpoint was PEP failure (premature cessation or primary HIV infection through Week 12)
- Additional endpoints were: adherence by self-report (n=98); pill count (n=55); plasma tenofovir levels (n=82); plasma DTG levels (n=80);and safety (clinical and laboratory adverse events [AEs])
- Adherence and adverse events (laboratory & clinical) assessed at Week 1,2 and 4

.

Acknowledgements

- Study funded by ViiV Healthcare

Results

Baseline characteristics

- Recruitment 1st August 2014 – 30th October 2015
- Prior NPEP 41%
 - Mean doses 1 (SD 2)
- Risk behaviour
 - receptive anal sex 82%
 - substance use 61%
 - condomless sex 70%
 - HIV+ source 40%
 - HIV RNA detectable 10%
 - HIV RNA not detectable 8%
- Time from exposure (hours)
 - to assessment 25 (IQR 14-39)
 - to 1st NPEP dose 27.5 (IQR 17-40)

Completion

468 GBM eligible for 3-drug PEP
(mean age 33, SD 9, range 18-62 years)

↓

104 GBM consented
(mean age 33, SD 10, range 17-62 years)

↓

4 GBM excluded

source tested HIV negative 2
unable to commit to study visits 1
eGFR < 60 mL/min/1.73m2 1

↓

100 GBM enrolled
(mean age 33, SD 9 range 17-62 years)

↓

Completed 28 days of NPEP
n=90

Did not complete NPEP
n=10
(median 9, IQR 2-16 days)
lost to follow-up 9
adverse event 1

Adherence

Self-report % (n=98)		Pill count % (n=55)		Day-28 plasma drug level ≥ inhibitory quotient (TDF ≥40ng/mL; n=82) (DTG ≥64ng/mL; n=80)	
TVD	98%	TVD	98%	TNV	85%
DTG	98%	DTG	98%	DTG	99%

measured a mean 15 (SD 8) hrs after last dose of DTG or TVD

Safety / adverse events

Clinical AEs (n=98)

- 67 (68%) reported 144 subjective AEs possibly attributable to study drug
- 98% of AEs were grade 1-2
- There were no unexpected AEs and no serious AEs
- 1 pt ceased NPEP because of grade-3 headache

Subjective AEs possibly related to NPEP occurring in ≥5%

Subjective AEs	Percent
Fatigue	26%
Nausea	25%
Diarrhoea	21%
Headache	10%
Abdominal pain/cramps	9%
Flatus	9%
Vivid dreams	7%

Laboratory AEs

Laboratory test	Grade 1 – Grade 2	Grade 3 – Grade 4
Chemistries [n (%)]		
↑Creatinine	10/93 (11)	
Hyperglycaemia	1/91 (1)	
Hypophosphataemia	5/91 (5)	
↑Bilirubin	4/93 (4)	1/89 (1)
↑Alanine aminotransferase	22/89 (25)	1/93 (1)
↑Creatine kinase	8/92 (9)	1/92 (1)
↑Lipase	7/92 (8)	1/84 (1)
↑Amylase	7/92 (8)	1/85 (1)
Hyperlactataemia	8/91 (9)	1/89 (1)
Urinalysis [n (%)]		
Proteinuria	10/88 (11)	
Haematuria	1/88 (1)	
Glycosuria	2/88 (2)	

- No pt ceased NPEP for any lab AE
- Most common laboratory AE was raised alanine aminotransferase (25%)
 - only 1 pt had a Grade 3 or 4 ALT
 - no clinical hepatitis
 - no pt with serum bilirubin ≥2xULN had abnormal level of conjugated or unconjugated bilirubin
- Mean eGFR decrease at Day 28 was 14 mL/minBSAc (SD 17, p=0.001)
 - eGFR <60 mL occurred in 3 pts

HIV infection

- No HIV infection through Week 12

Conclusions

- **DTG + TVD were well tolerated as NPEP with high levels of completion (90%) and adherence**
 - **Rates similar to those using single-tablet NPEP with TDF-FTC-RPV**
- **Adherence 98% by self report and pill count, but only 85% had plasma tenofovir ≥40ng/mL at Day 28**

References

1. World Health 1. World Health Organization. Consolidated guidelines on the use of antiretroviral drugs for treating and preventing HIV infection 2016. Available at: http://www.who.int/hiv/pub/arv/arv-2016/en/ Last accessed 2nd July 2016
2. Ford N, Irvinea C, Shubberb A et al. Adherence to HIV postexposure prophylaxis: a systematic review and meta-analysis. AIDS 2014, 28:2721–2727
3. Thomas R, Galanakis C, Vezina S et al. Adherence to post-exposure prophylaxis (PEP) and incidence of HIV seroconversion in a major North American cohort. PLOS ONE | DOI:10.1371/journal.pone.0142534 November 11, 2015
4. Ford N, Shubber Z, Calmy A et al. Choice of Antiretroviral Drugs for Postexposure prophylaxis for adults and adolescents: a systematic review. Clin Infect Dis 2015:60 (Suppl 3) • S171 – S176
5. Fatkenheuer G, Jessen H, Jung N et al. PEPDar: a randomized prospective noninferiority study of ritonavir-boosted darunavir for HIV post-exposure prophylaxis. HIV Med. 2016;17:453-59.
6. McAllister J, Read P, McNulty A et al. Raltegravir-emtricitabine-tenofovir as HIV nonoccupational post-exposure prophylaxis in men who have sex with men: safety, tolerability and adherence. men: safety, tolerability and adherence. HIV Med. 2014;1:13-22.
7.Foster R, McAllister J Read TR et al. Single-tablet emtricitabine-rilpivirine-tenofovir as HIV post-exposure prophylaxis in men who have sex with men. Clin Infect Dis 2015;61(8):1336-41
8. Leal L, Leon A, Torres B et al. Tenofovir/Emtricitabine Plus LPV/r vs MVC or Raltegravir for PEP: 2 Randomized Trials. In: Program and abstracts of the Conference on Retroviruses and Opportunistic Infections (Seattle) Washington: International Antiviral Society-USA, 2015:959
9.Raffi F, Rachlis A, Stellbrink H-J et al. Once-daily dolutegravir versus raltegravir in antiretroviral-naïve adults with HIV-1 infection: 48 week results from the randomised, double blind, non-inferiority SPRING-2 study. Lancet 2013; 381: 735-37.9. 10. ViiV Healthcare & The Glaxo-SmithKline Group: Clinical Investigators Brochure for GSK1349572 Version 06 14-Feb 2013.
Organization. Consolidated
10. ViiV Healthcare & The Glaxo-SmithKline Group: Clinical Investigators Brochure for GSK1349572 Version 06 14-Feb 2013.